506

PRTX007, an Oral Prodrug of a Novel Interferon-centric Toll-like Receptor 7 (TLR7) Agonist, Proposed for Curative Therapy of Chronic Hepatitis D and Functional Cure of Chronic Hepatitis B Infection

J. Appleman, S. Webber, S. Zook, C. Scribner

Primmune Therapeutic, Inc.

The objective of our program is to supplant pegylated interferon by the oral TLR7 agonist prodrug PRTX007 as a more effective, well-tolerated immunotherapeutic in the treatment of of chronic Hepatitis D and B. PRTX007 directly engages plasmacytoid dendritic cells, which subsequently induces an interferon-like response in target cells and engages other immune cells, notably CD8+ T-cells and NK cells.

Circulating IFNs too poorly tolerated, most systemic TLR7 agonists too inflammatory

- Exogenously administered PEG-IFN must be at high, poorly tolerated circulating IFNα levels to induce meaningful ISGs in target cells
- This causes safety/tolerability issues and is dose limiting
- Like PEG-IFN, TLR7 agonists elicit antiviral response via IFN-mediated stimulation of ISGs
- Prior TLR7 agonists were suboptimal and produced undesirable cytokines due to concurrent stimulation of NF-κB and/or other TLR pathways

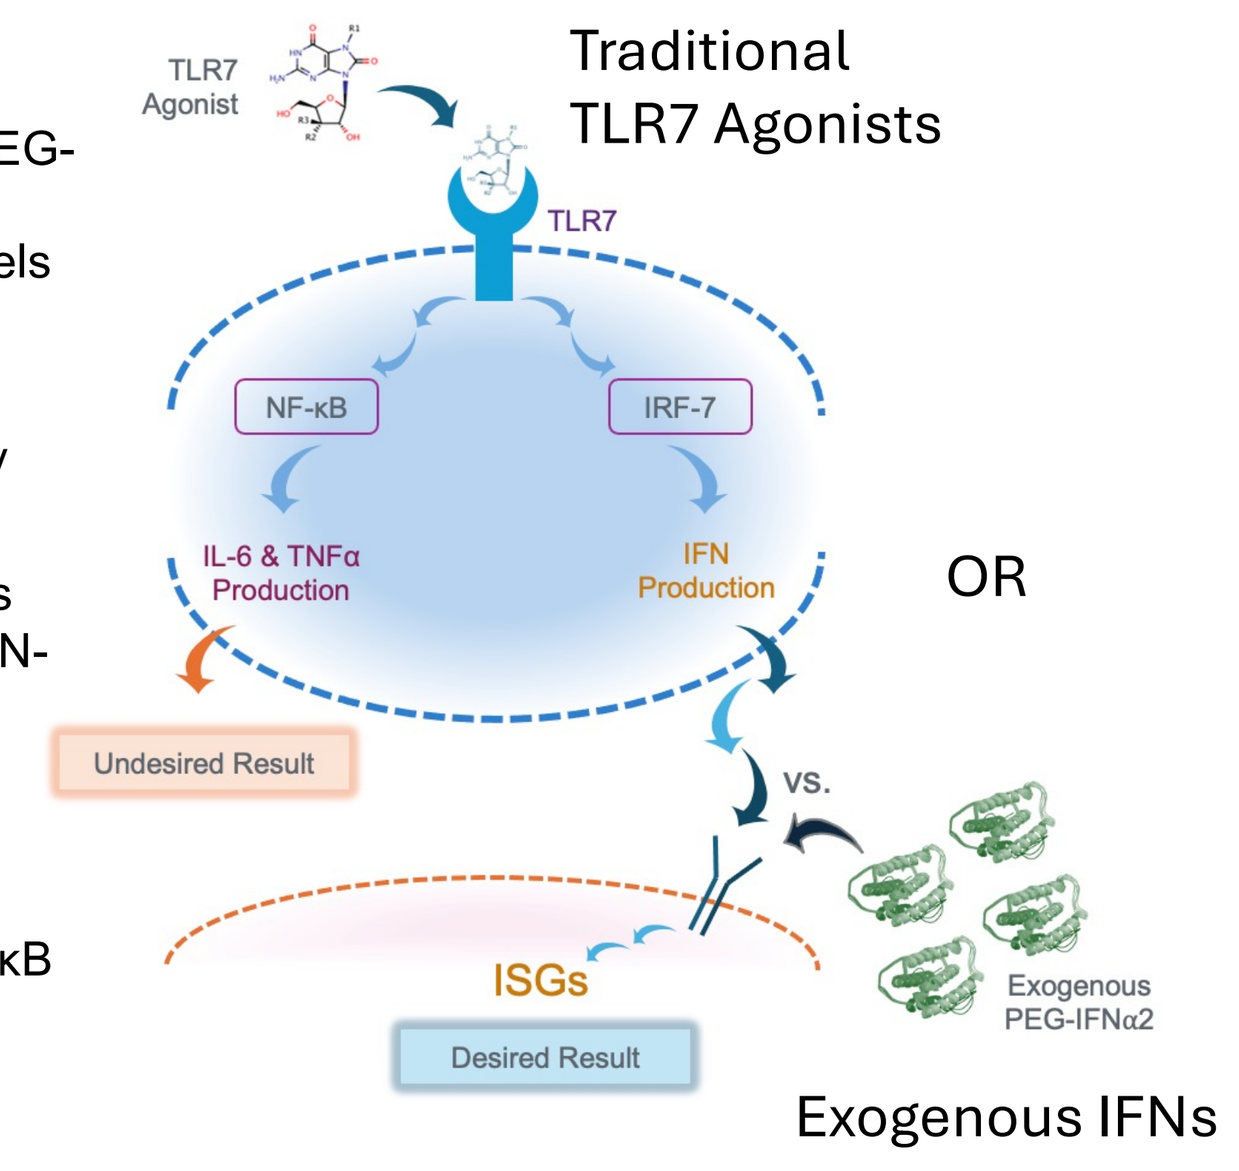
TLR7 Agonist
Traditional TLR7 Agonists
TLR7
NF-κB
IRF-7
IL-6 & TNFα Production
IFN Production
Undesired Result
OR
vs.
ISGs
Desired Result
Exogenous PEG-IFNα2
Exogenous IFNs

Primmune's novel approach induces ISGs in target cells without drawbacks of previous generations of TLR7 agonists or IFNs

- We sought to identify TLR7 agonists that selectively **activate downstream ISGs/interferon** while **avoiding undesirable activation** of the NF-κB pathway or other TLR pathways
- We have successfully…
 - Designed TLR7 agonists that preferentially stimulate IRF-7 with minimal effects on NF-κB or other TLR pathways
 - Demonstrated desired effects *in vitro*
 - Demonstrated desired effects *in vivo* in non-human primates (NHPs)
 - Confirmed this desirable profile is recapitulated in healthy volunteers

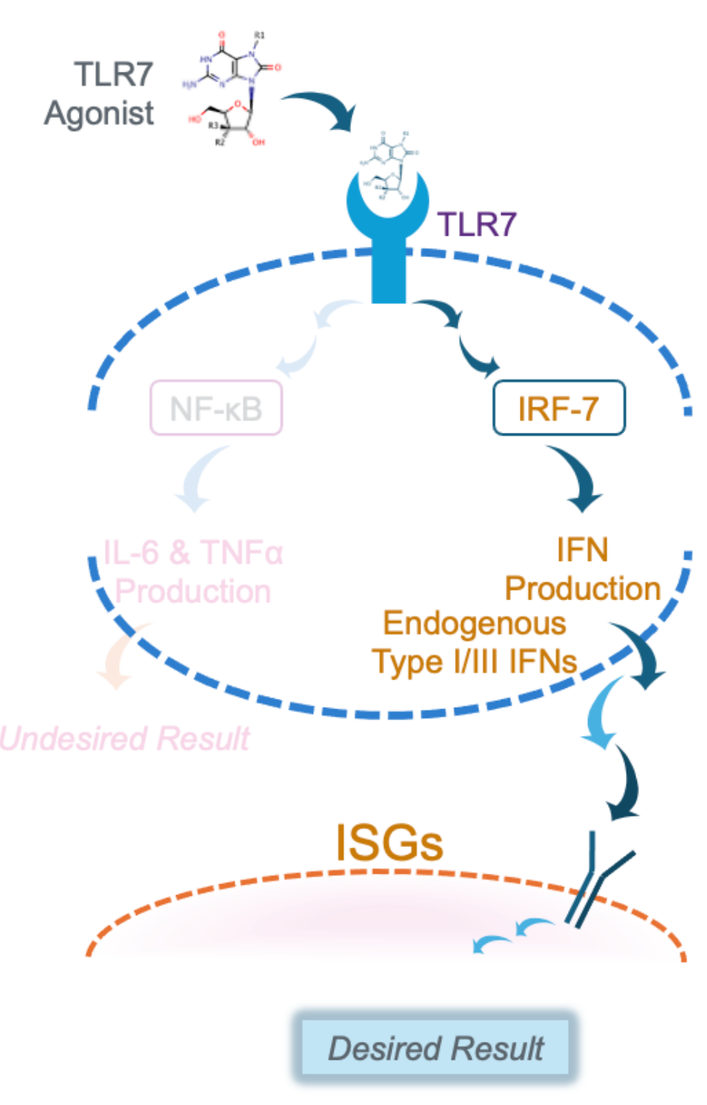
TLR7 Agonist
TLR7
NF-κB
IRF-7
IL-6 & TNFα Production
IFN Production
Endogenous Type I/III IFNs
Undesired Result
ISGs
Desired Result

PRTX007 Primes the Immune System to Respond to the Presence of Viral Infection or Transformed Cells (Precancerous Lesions, Solid Tumors)

- **PRTX007 engages a variety of immune responses to mitigate threats to the health of the host**
- **Key are those initiated by direct activation of plasmacytoid dendritic cells (pDCs).**

INNATE — **pDC activation mediates:**
- therapeutic engagement of NK cells and other elements of the antiviral and antitumor innate immune response
- production of soluble factors including all Type I/III IFNs

ADAPTIVE — **Activated pDCs drive the adaptive immune response primarily through:**
- reversal of intratumoral CD8+ T-cell anergy
- differentiation and activation of naïve T-cells to CD8+ T cells

NEW! — **TLR7-mediated engagement of trained immunity via other cell types including monocytes:**
- We and collaborators have recently discovered additional mechanisms of disease control engaged by Primmune's proprietary TLR7-specific agonists

PRTX007 CLINICAL PROFILE

- **Orally** administered prodrug
- **Systemically** distributed agonist
- **Safe** and **well-tolerated**
- **Every other day administration** enables long-term immune pressure without counter-regulation
- **IFN centric immune activation** with little to no activation of systemic pro-inflammatory cytokines
- **Readily combinable** with other treatment and prevention modalities

Clinical study: relevant pharmacologic activities have been demonstrated in healthy volunteers

HEALTHY VOLUNTEER STUDY DESIGN

SAD
N=72
8/arm; (6 active:2 placebo)

MAD
N=40
10/arm; (8 active:2 placebo)
PO QOD x 7 doses (13 days)

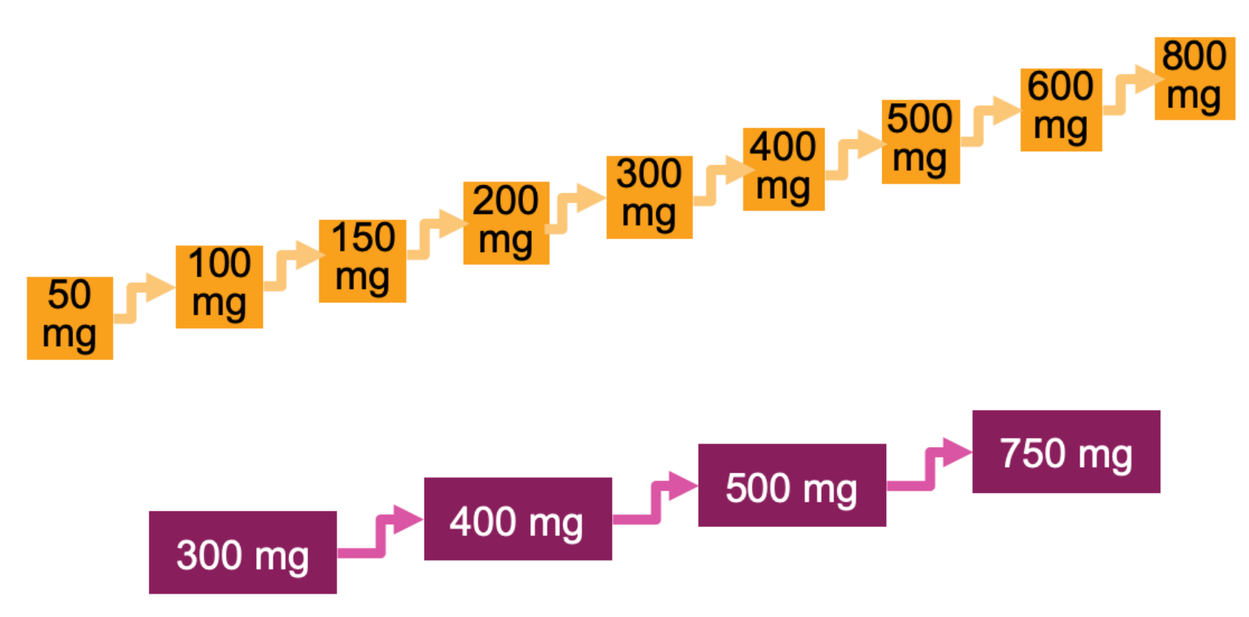
50 mg
100 mg
150 mg
200 mg
300 mg
400 mg
500 mg
600 mg
800 mg
300 mg
400 mg
500 mg
750 mg

PHARMACOKINETICS

- Short duration of pulsatile systemic exposure to PRX034
- Rapid OAT3-mediated kidney clearance (by design)

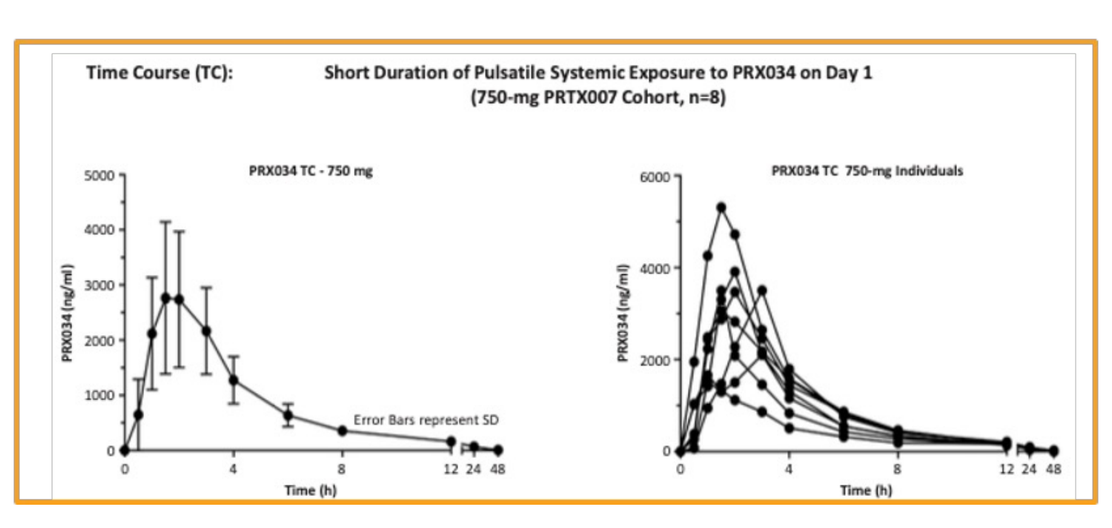
Time Course (TC):
Short Duration of Pulsatile Systemic Exposure to PRX034 on Day 1
(750-mg PRTX007 Cohort, n=8)
PRX034 TC - 750 mg
PRX034 TC 750-mg Individuals
Error Bars represent SD
Time (h)

- Dose proportional exposure increase
- PRTX007 ≈ 2/3 PRX034 exposure (all doses)
- No accumulation

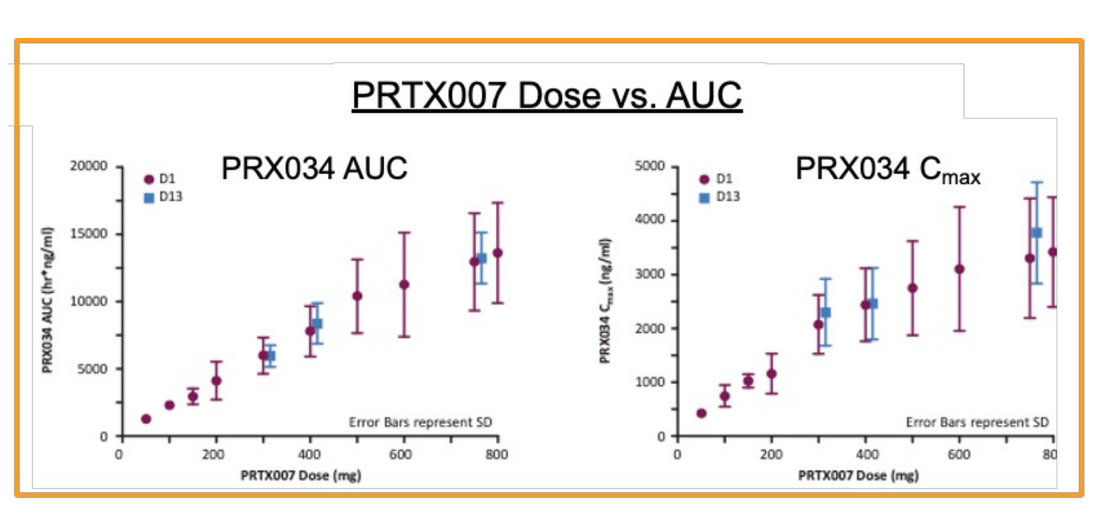
PRTX007 Dose vs. AUC
PRX034 AUC
PRX034 C_{max}
D1
D13
Error Bars represent SD
PRTX007 Dose (mg)

SAFETY AND TOLERABILITY

- 0% Serious adverse events (SAE)
- 0% Discontinuations due to a TEAE
- 0% Severe (Grade ≥3) TEAEs

AEs (X from N=130)	Placebo (n=26)	PRTX007 (n=104)	PRTX007 Percent (X/104=%) Grade Response: Grade 1	Grade 2	Grade 3
Nervous System Disorders					
Headache (40)	4	17[a]	12%	5%	0%
Hepatobilliary Disorders					
Elevated ALT (5)	0	5	5%	0%	0%
General Disorders					
Fever (transient) (4)	1	3	3%	0%	0%

[a] Nineteen additional headaches in the PRTX007-treated group were coded as unrelated to drug. There was no dose dependence

Headache
Generally mild; no dose-response

Fever
Seen after dose #1 or #2 in 2 highest MAD dose groups. Resolved within 24-36 hours and did not recur upon subsequent dosing

ALT elevation
All Grade 1/mild – all <2x ULN, except 1 at ~2.8x ULN (400 mg). No symptoms or associated changes in AST, alkaline phosphatase or bilirubin. No dose response: 300 mg n=2; 400 mg n=2; 500 mg n=0; 750 mg n=1

PHARMACODYNAMIC RESPONSE MARKERS

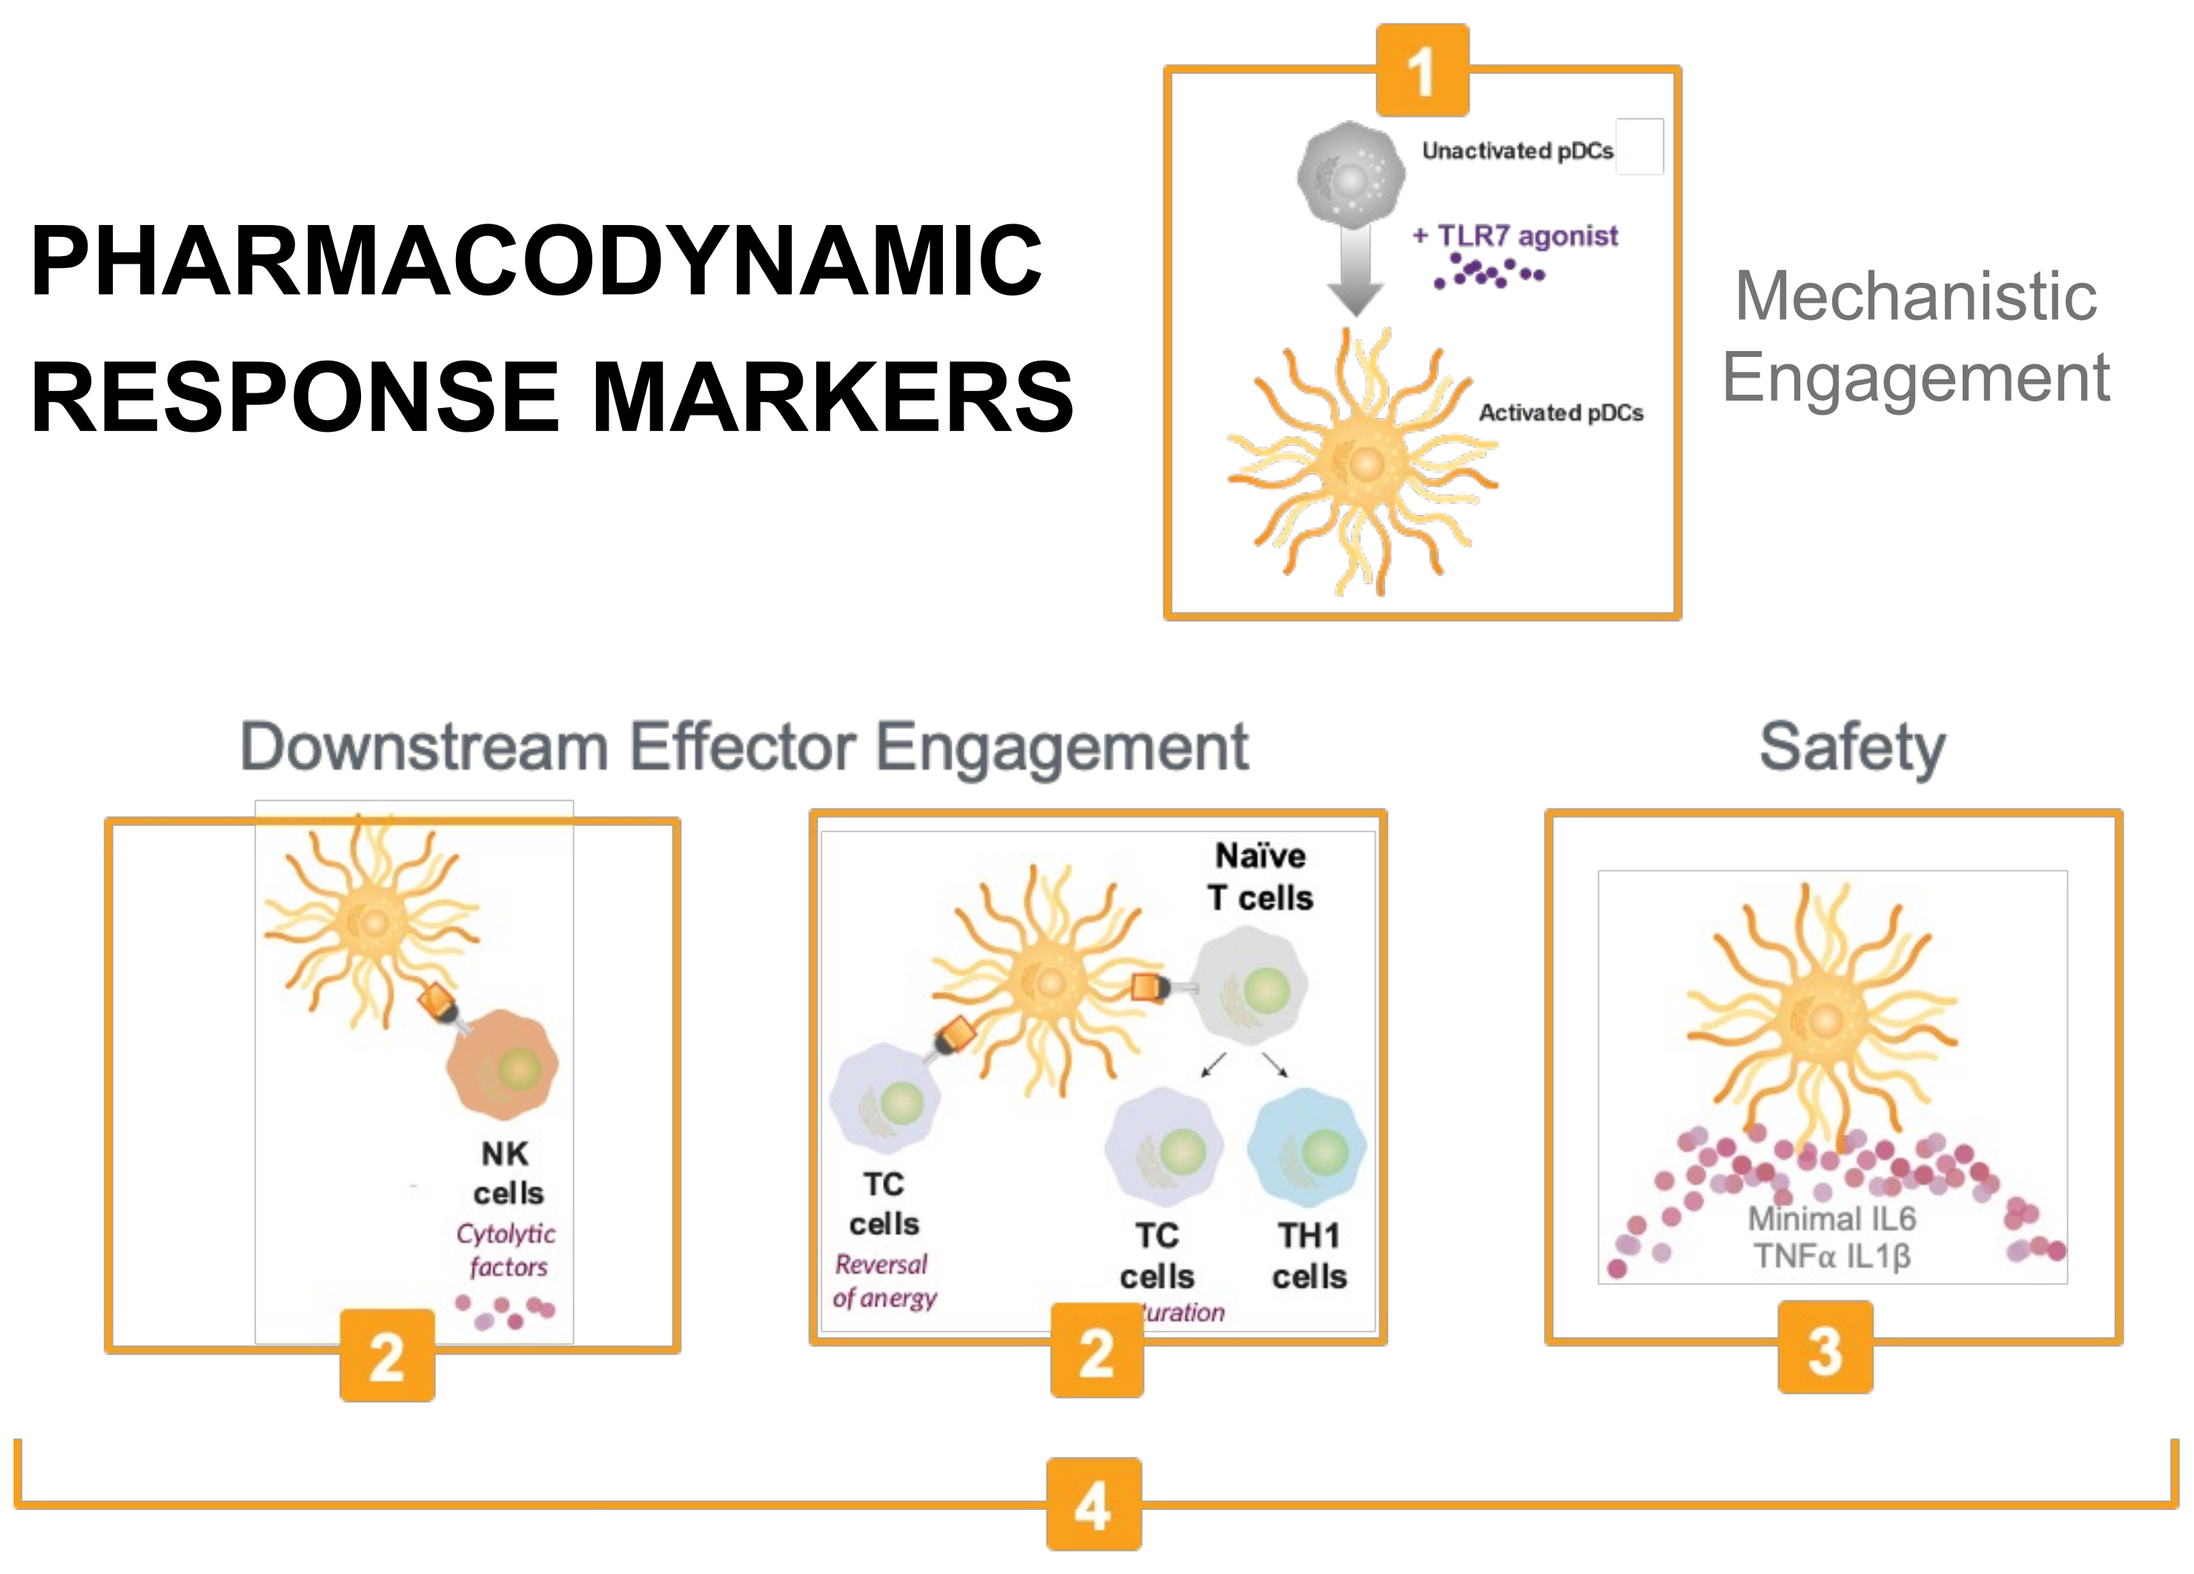
1
Unactivated pDCs
+ TLR7 agonist
Activated pDCs
Mechanistic Engagement
Downstream Effector Engagement
Safety
NK cells
Cytolytic factors
Naïve T cells
TC cells
Reversal of anergy
TC cells
TH1 cells
Minimal IL6
TNFα IL1β
2
2
3
4

1. Selected biomarker fingerprint demonstrates engagement of MoA (interferon-mediated gene signatures in circulating cells downstream of pDC engagement)
2. Cellular markers indicate engagement of additional effector cells (CD8+ T cells and NK cells)
3. Circulating markers support safety (lack of circulating proinflammatory factors)
4. Detailed time course measurements in MAD cohorts demonstrate induction of desired responses without counter-regulation (QOD dosing)
 - Dose response
 - Build in interferon response
 - Activation of NK and CD8+ T-cells

Key pharmacodynamic markers

IFN-like Immune Response in Circulating Immune Cells

Single Dose (SAD) or First Dose (MAD): Dose Responsive Increase in ISGs at Doses ≥200 mg

Heat Map of Peak Response (R_{max}) Following 1st Dose

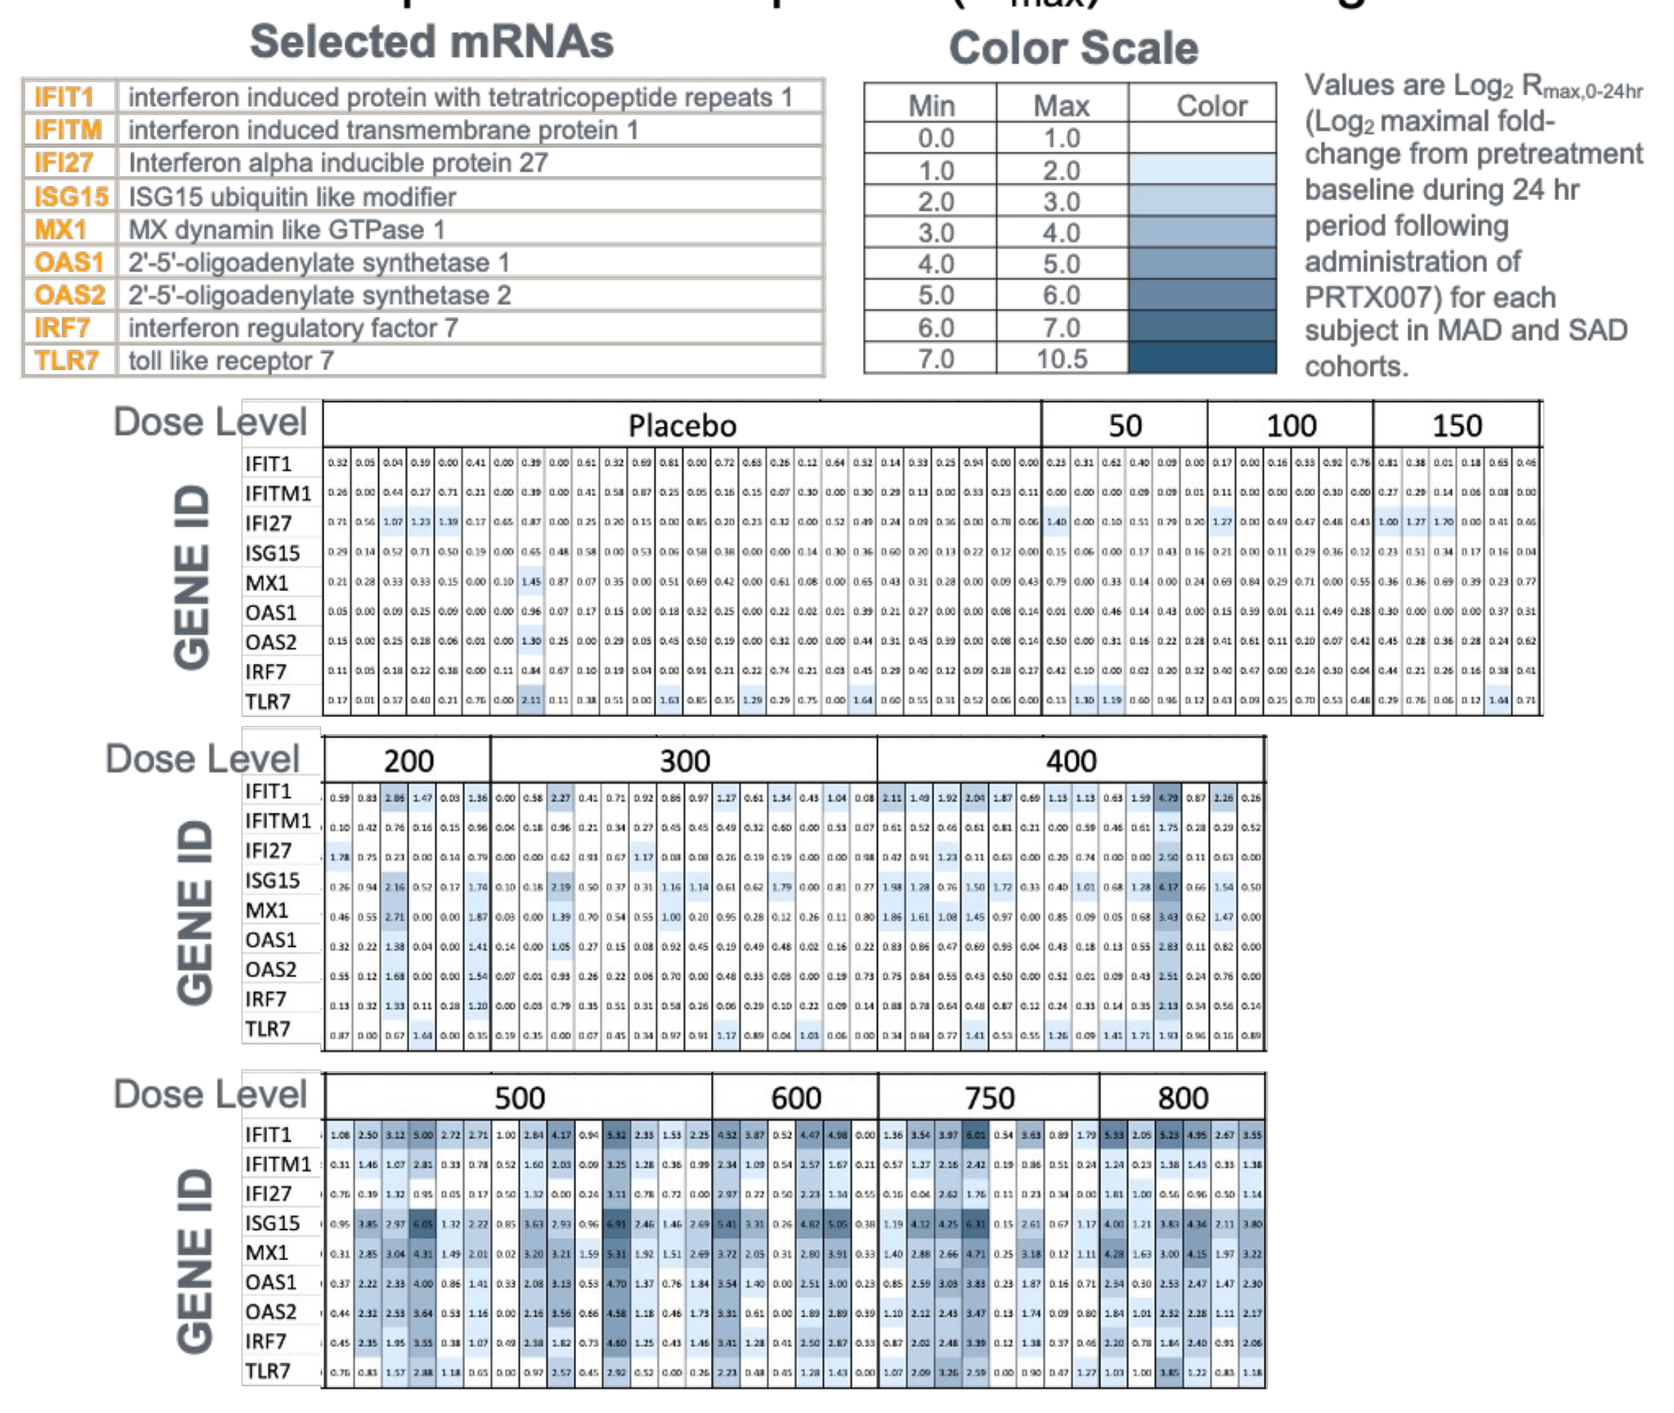
Selected mRNAs
IFIT1 interferon induced protein with tetratricopeptide repeats 1
IFITM interferon induced transmembrane protein 1
IFI27 Interferon alpha inducible protein 27
ISG15 ISG15 ubiquitin like modifier
MX1 MX dynamin like GTPase 1
OAS1 2'-5'-oligoadenylate synthetase 1
OAS2 2'-5'-oligoadenylate synthetase 2
IRF7 interferon regulatory factor 7
TLR7 toll like receptor 7
Color Scale
Values are $\text{Log}_2\ R_{max,0\text{-}24hr}$ (Log_2 maximal fold-change from pretreatment baseline during 24 hr period following administration of PRTX007) for each subject in MAD and SAD cohorts.
Dose Level
Placebo
50
100
150
200
300
400
500
600
750
800
GENE ID

Individual Day 1 (1st Dose) R_{max}

(examples of response for individual mRNAs)

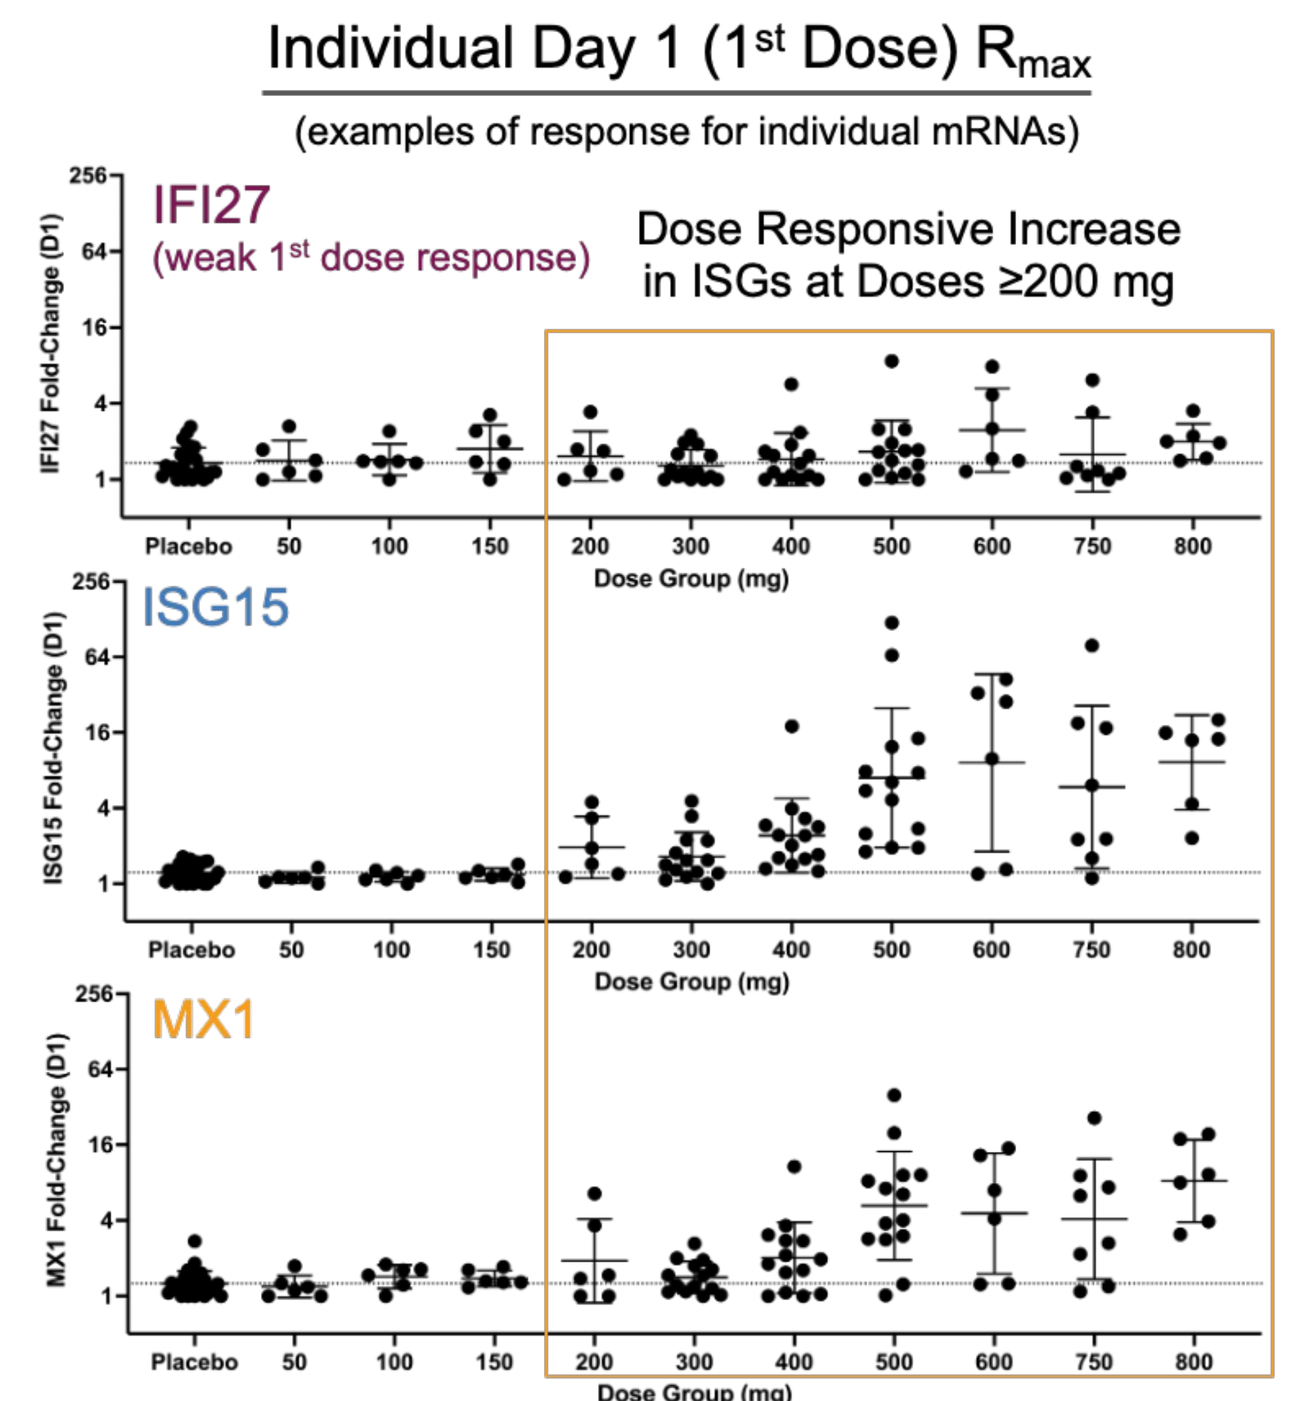
IFI27
(weak 1st dose response)
Dose Responsive Increase in ISGs at Doses ≥200 mg
ISG15
MX1
Placebo
Dose Group (mg)

Proportion of responders and intensity of D1 response increases with dose

Multi-Dose Response - Upregulation of ISGs without Non-Responders (500 and 750 mg))

Population Response to Repeated Doses
(750 mg PRTX007 Cohort, n=8 receiving drug)

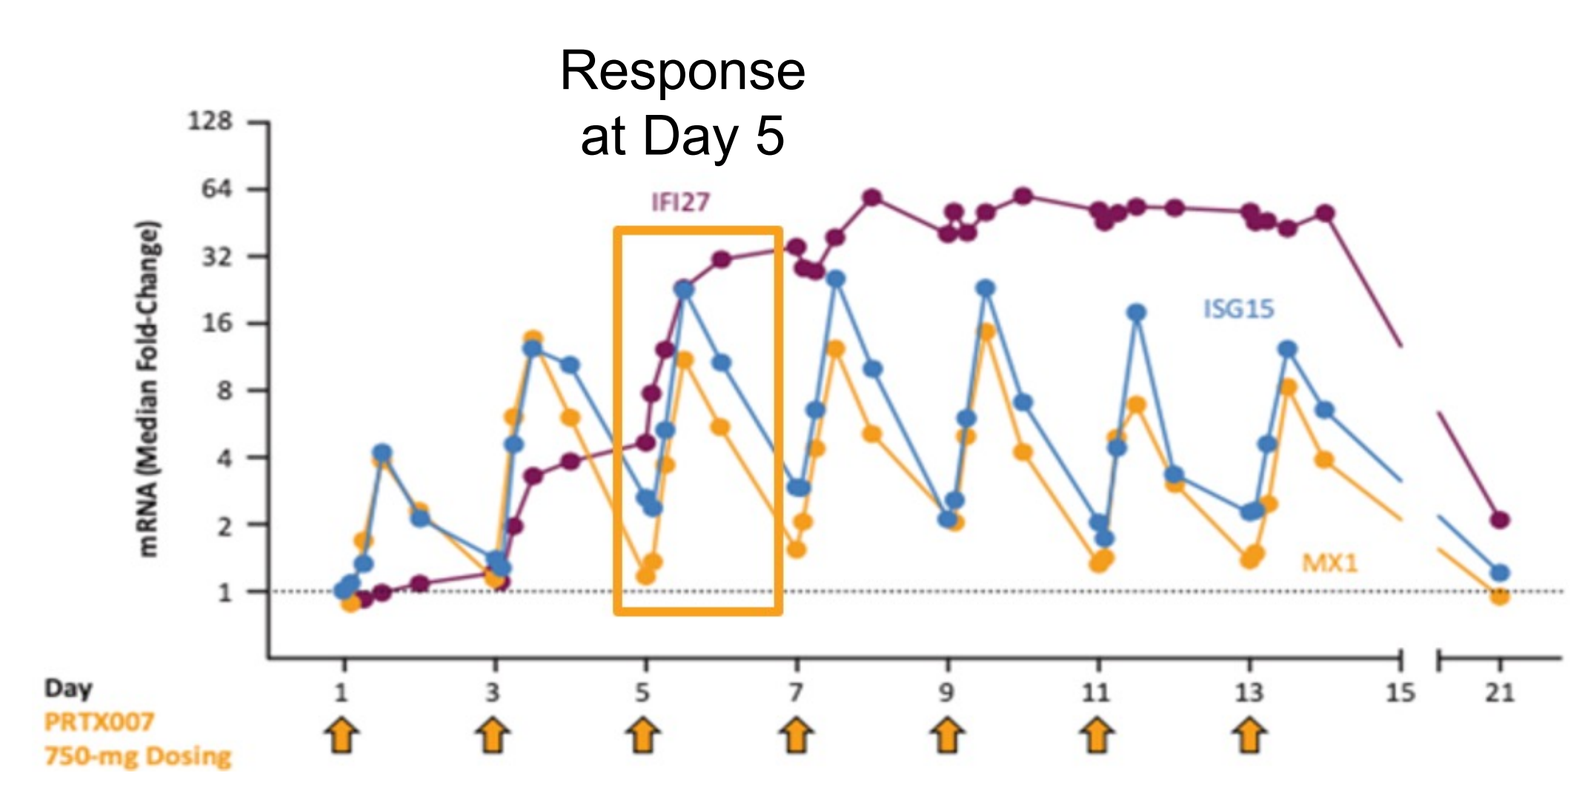
Response at Day 5
IFI27
ISG15
MX1
mRNA (Median Fold-Change)
Day
PRTX007 750-mg Dosing

Achieved steady-state ISG mRNA response in a few doses, resulting in sustained immune pressure

Day 5 (steady-state) dose response:
- Nearly everyone responds robustly to 500-750 mg (including subjects unresponsive to 1st dose)
- Significant response by some, but not all, subjects at 300 and 400 mg

Individual Day 5 (3rd Dose) R_{max}

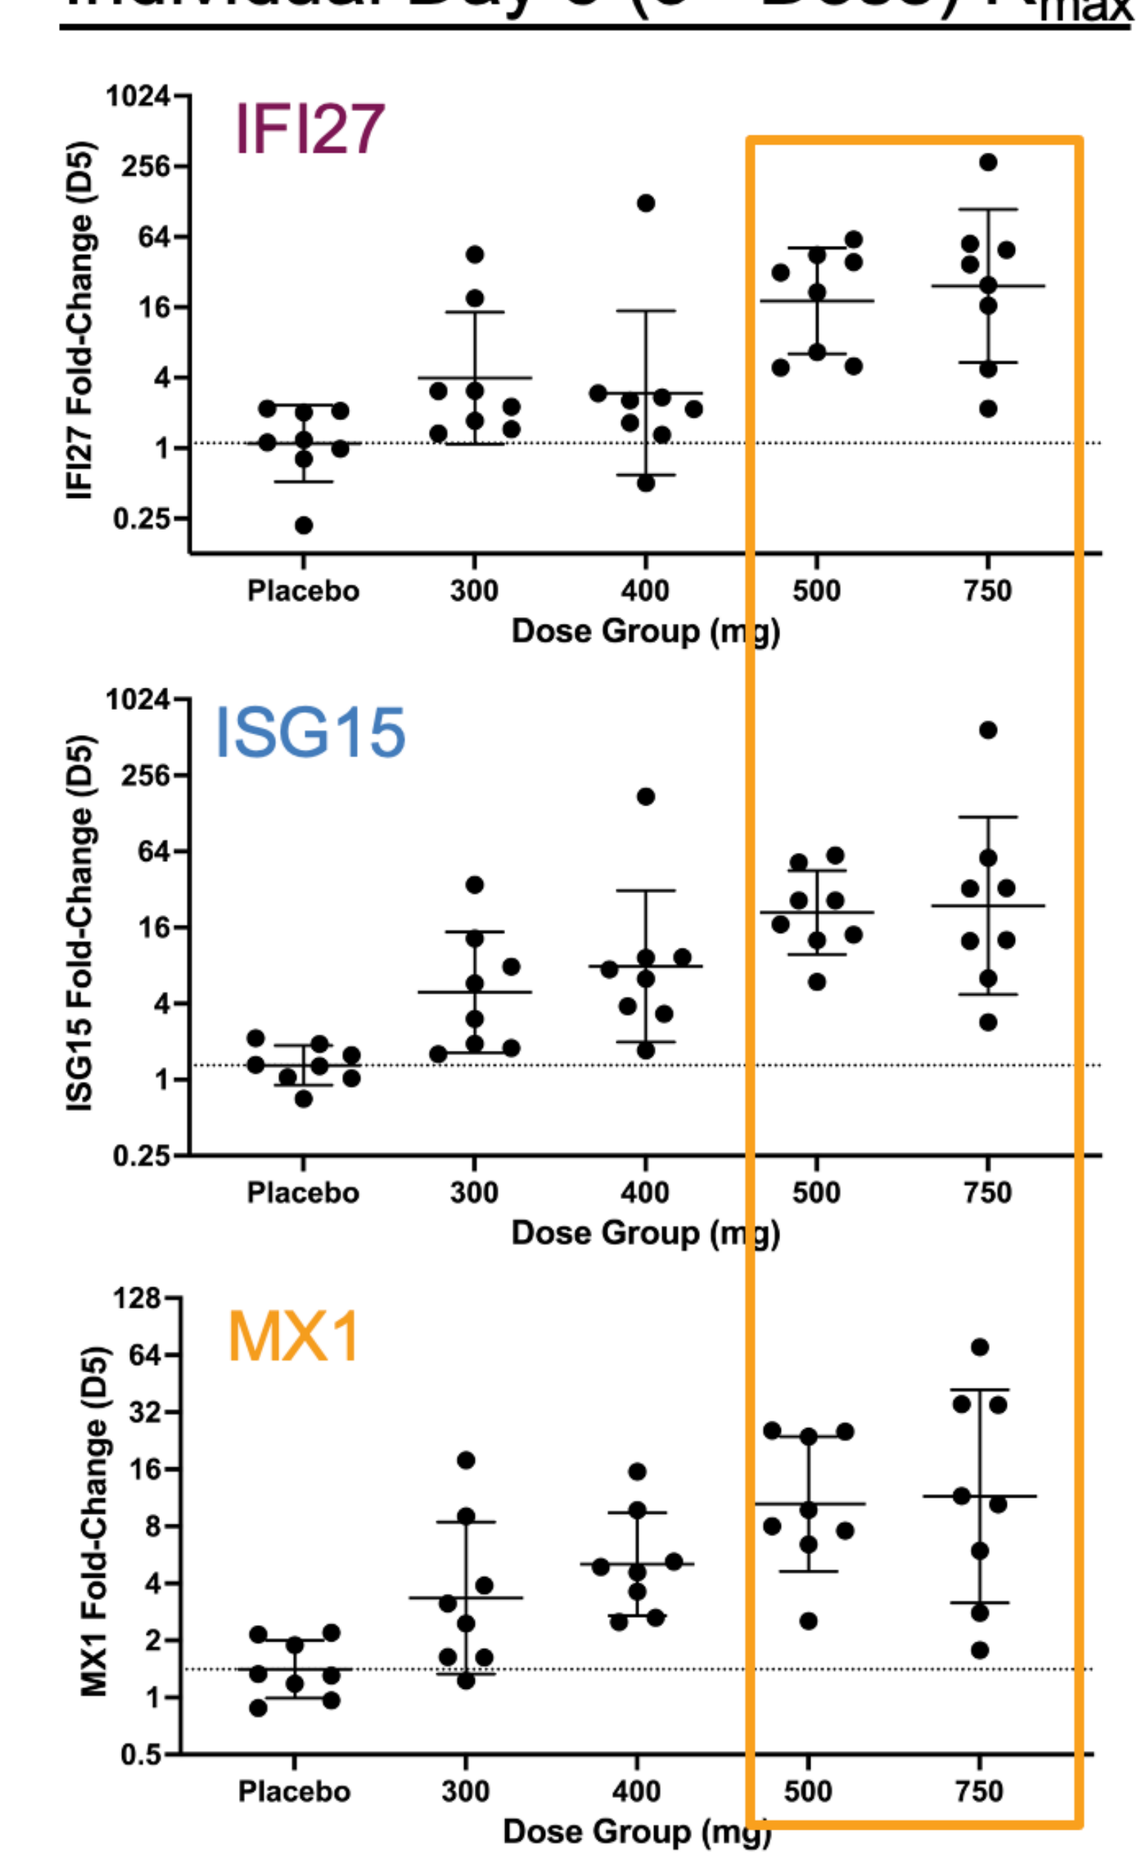
IFI27
ISG15
MX1
IFI27 Fold-Change (D5)
ISG15 Fold-Change (D5)
MX1 Fold-Change (D5)
Placebo
300
400
500
750
Dose Group (mg)

Activation of CD8+ T Cells and NK Cells

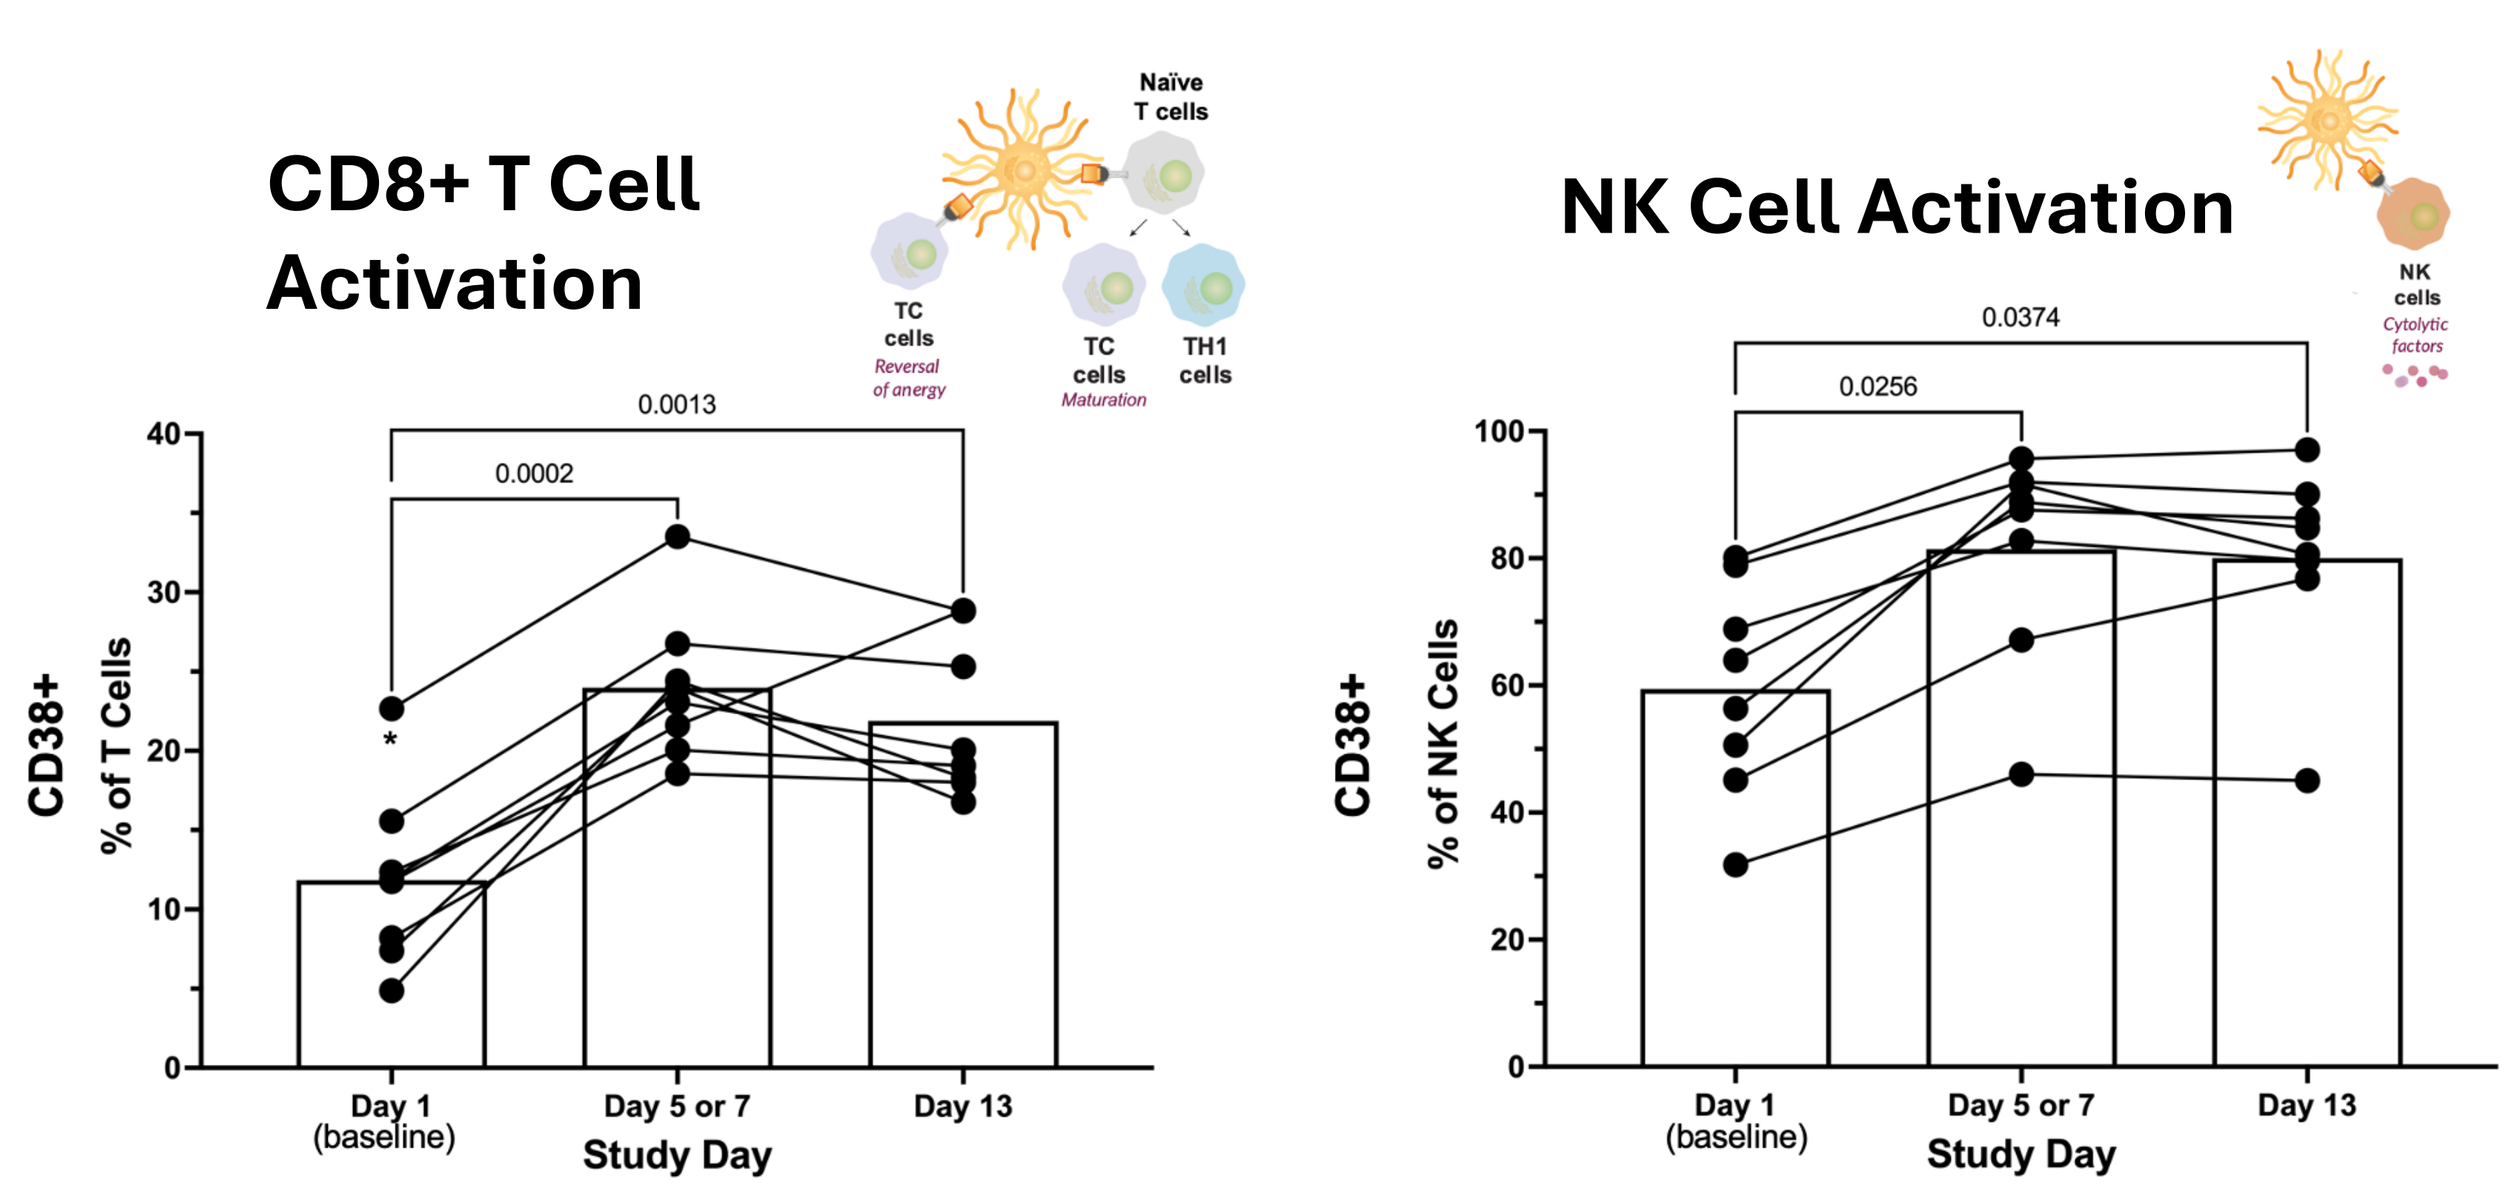
CD8+ T Cell Activation
Naïve T cells
TC cells
Reversal of anergy
TC cells
Maturation
TH1 cells
0.0013
0.0002
CD38+ % of T Cells
Day 1 (baseline)
Day 5 or 7
Day 13
Study Day
NK Cell Activation
NK cells
Cytolytic factors
0.0374
0.0256
CD38+ % of NK Cells

Data from 750 mg MAD Cohort. No evidence of activation of CD4+ T cells of B cells

Avoids Systemic Inflammation

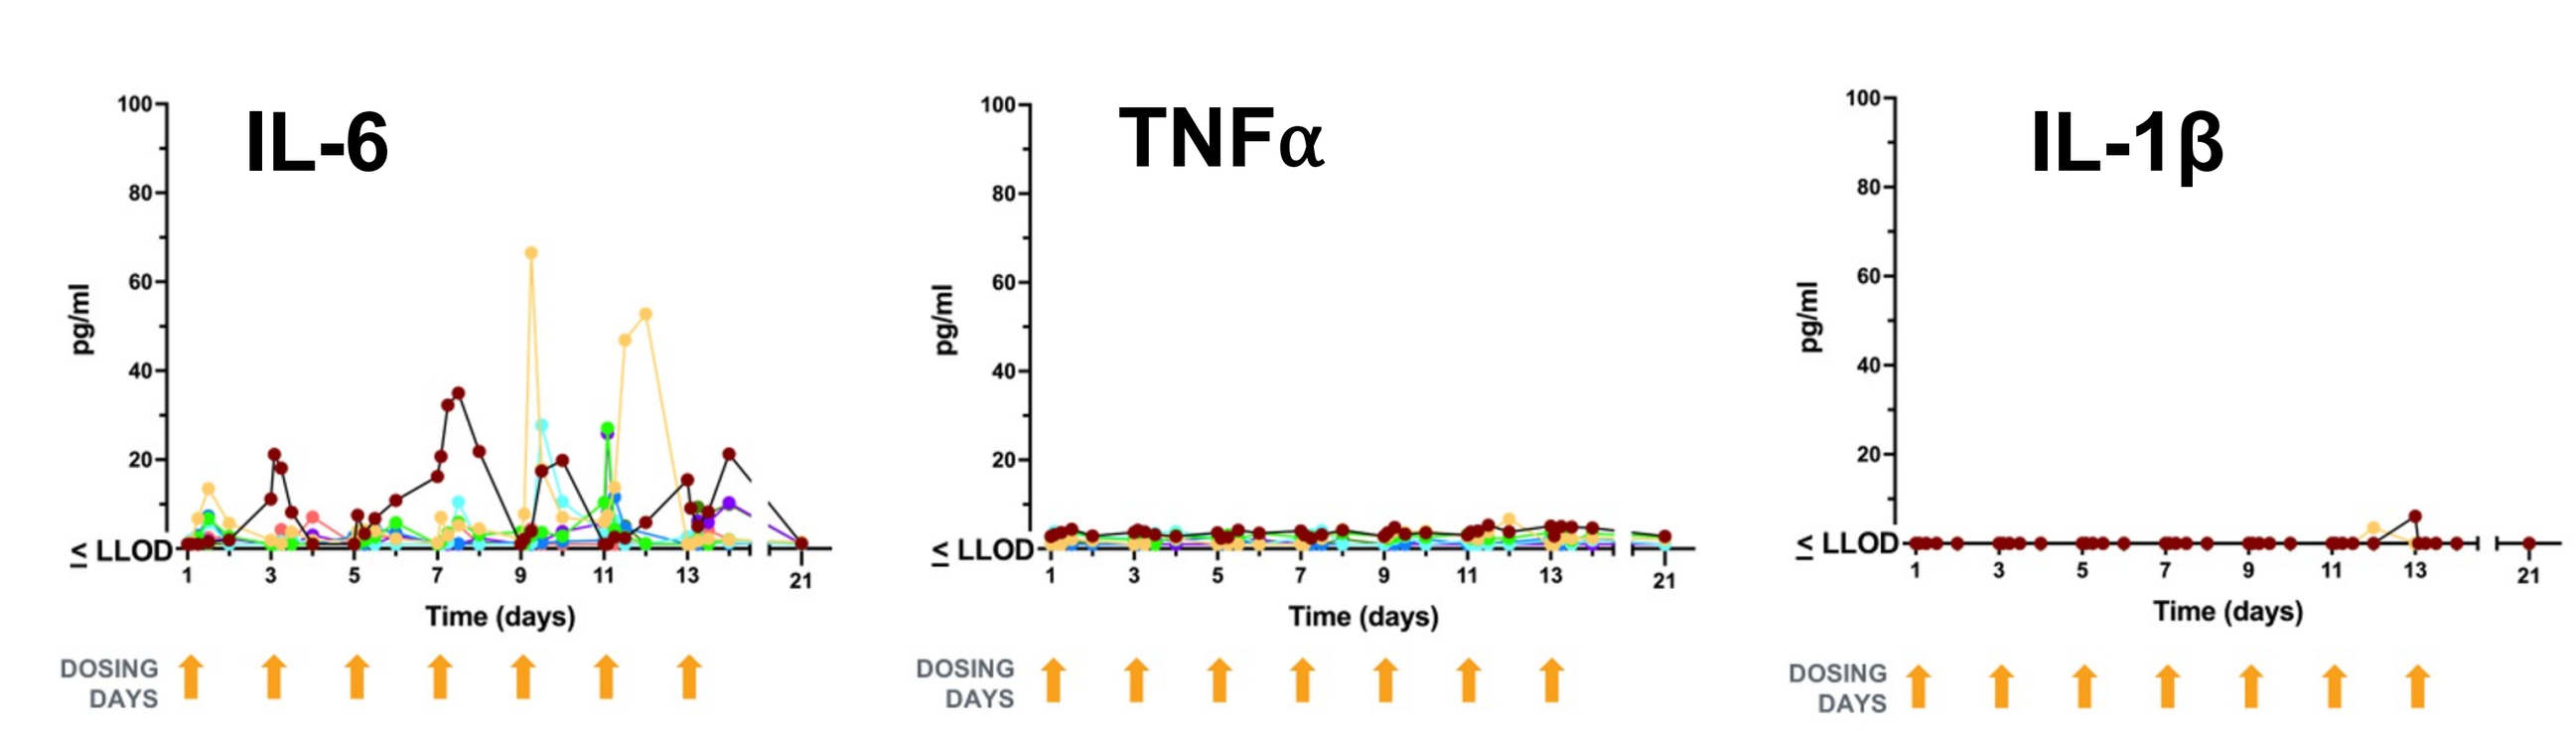
IL-6
TNFα
IL-1β
pg/ml
≤ LLOD
Time (days)
DOSING DAYS

Complete ime courses of proinflammatory factors in plasma for all eight subjects receiving 750 mg PRTX007

In conclusion:

- ➢ pDC-mediated interferon-like responses at target cells and engagement of innate and adaptive immunity (e.g. CD8+ T cells and NK cells) without undesirable pro-inflammatory cytokine responses allows us to maintain antiviral immune pressure without the limitations of PEG-IFN and other TLR agonists.
- ➢ The magnitude of ISG induction exceeds that achieved with PEG-IFN (figure at right) which should be predictive of therapeutic benefit.
- ➢ All of these propoperties are encouraging for utility in the treatment of chronic viral diseases like HBV and HDV, acute viral infections and in the treatment of solid tumors.

ISG Induction by PRTX007 Substantially Exceeds That by PEG-IFN

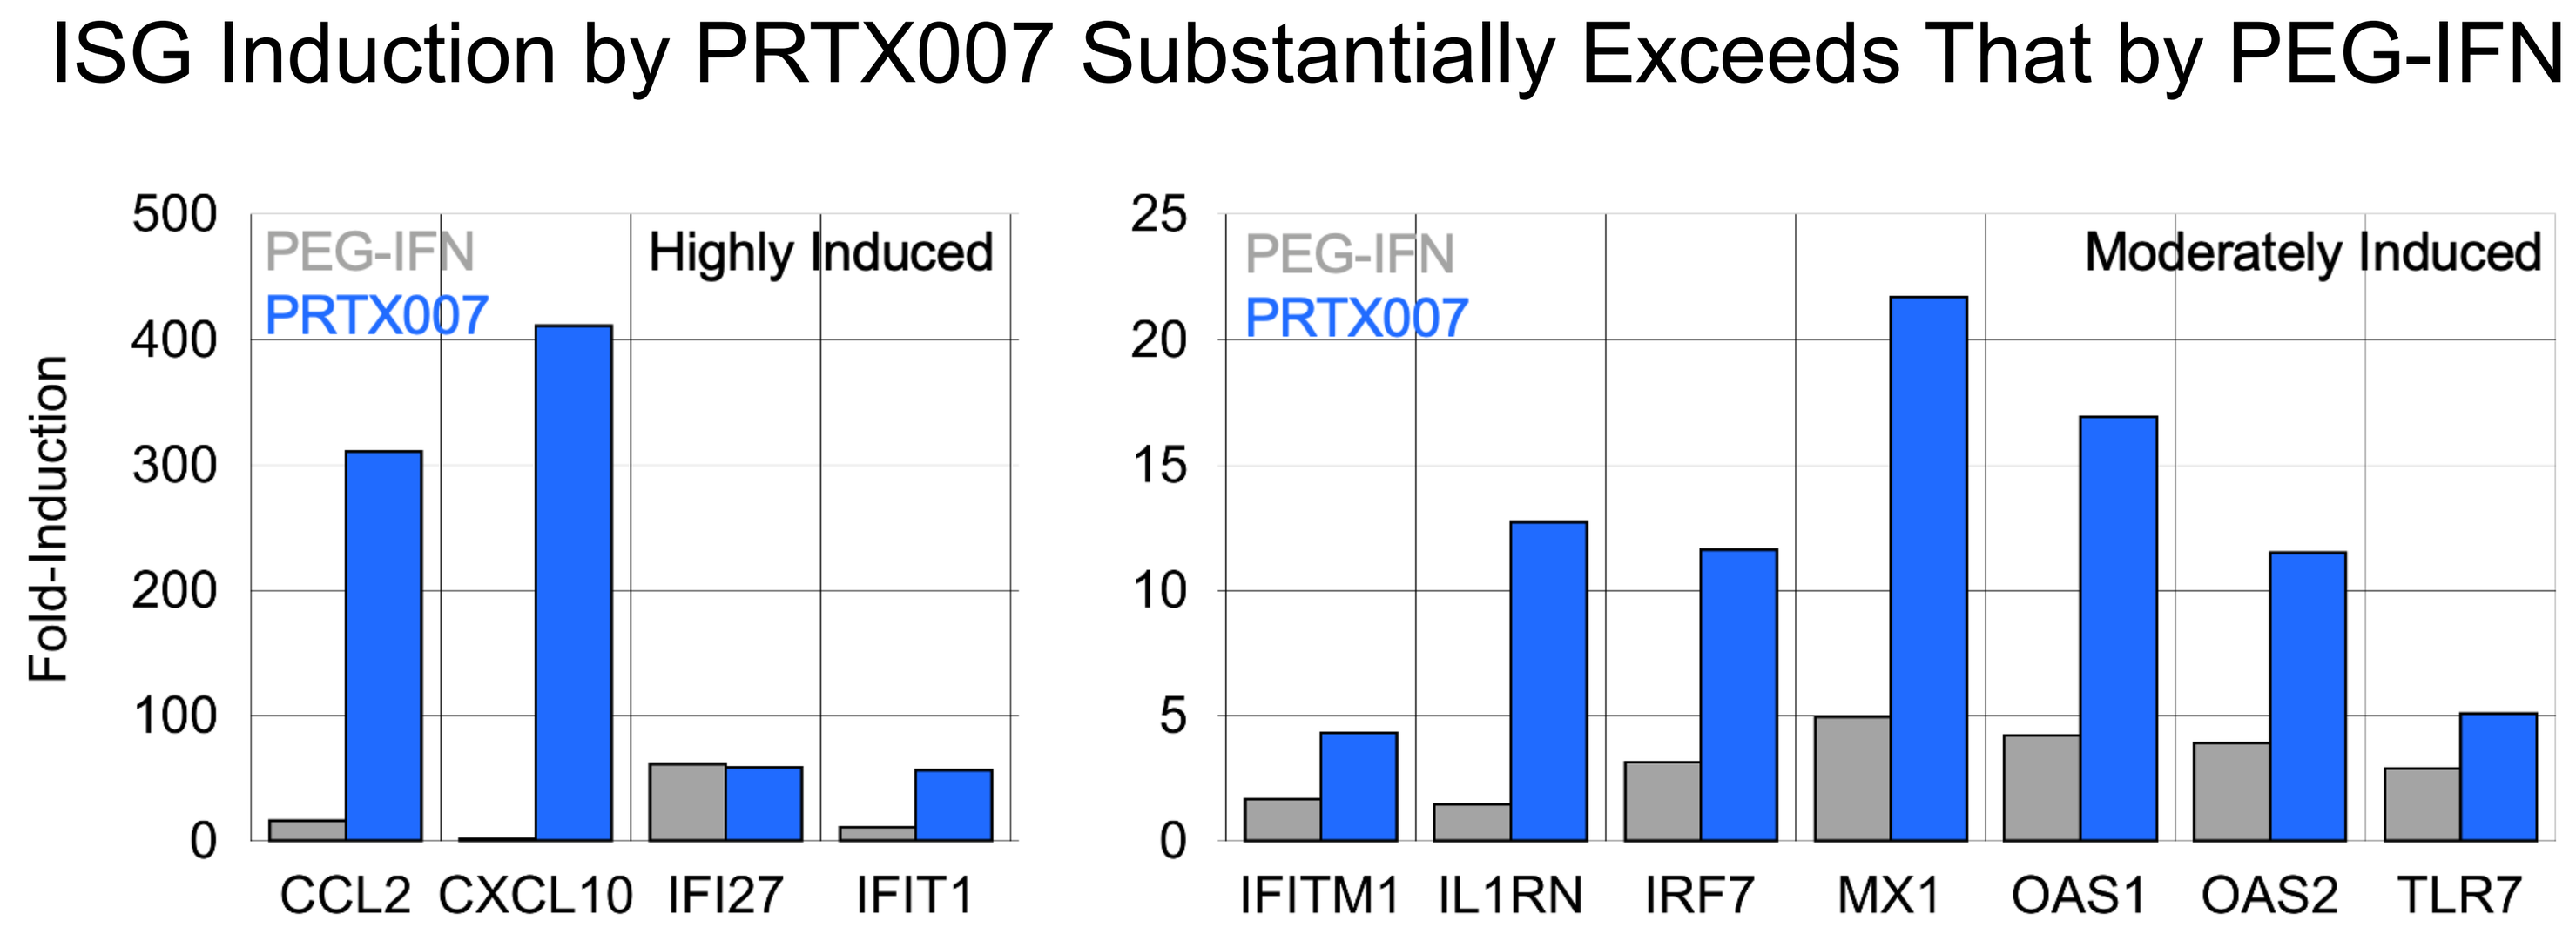
PEG-IFN
PRTX007
Highly Induced
Moderately Induced
Fold-Induction
CCL2
CXCL10
IFI27
IFIT1
IFITM1
IL1RN
IRF7
MX1
OAS1
OAS2
TLR7

Data for PEG-IFN for treatment D7 for robust antiviral responders; Taylor et al. Changes in Gene Expression during Pegylated Interferon and Ribavirin Therapy of Chronic Hepatitis C Virus Distinguish Responders from Nonresponders to Antiviral Therapy (2007) 81(7):3391

Data for PRTX007 from D5 response in 750 mg multidose cohort in healthy volunteer study