Activation of innate and adaptive immune response with a clinical stage TLR7 agonist prodrug PRTX007

CT180

Charlotte R. Lemech[1], Christopher Argent[1], Curtis L. Scribner[2], Andrew Sharabi[3], Richard Daniels[4], James R. Appleman[4]

[1]Scientia Clinical Research, LTD, Randwick, NSW, Australia; [2]Curtis L. Scribner, Oakland, CA, USA; [3]Department of Radiation Medicine, University of California at San Diego, San Diego, CA, USA; [4]Primmune Therapeutics Inc., Carlsbad, CA, USA

Background

- Use of the toll-like receptor 7 (TLR7) pathway to stimulate innate and adaptive immunity for treatment of cancer and multiple viral diseases has been well characterized
- Historically, systemically administered TLR7 agonists present a variety of challenges in providing therapeutic benefit that have been successfully addressed with PRTX007
- PRX034, an orally administered TLR7 agonist comprised of the prodrug PRTX007, elicits a robust, well-tolerated immune response involving activation of plasmacytoid dendritic cells (pDCs), CD8+ T cells, and natural killer (NK) cells throughout the body with no evidence of nuclear factor κB (NF-κB)–mediated inflammation[1,2]; it is expected to also activate these cells in cancer patient tumor microenvironments
- We performed a first-in-human, phase 1, prospective, randomized, double-blind, placebo-controlled, single-ascending dose (SAD) and multiple-ascending dose (MAD) study of PRTX007, and reported interim results on the following[2]:
 - PRTX007 demonstrated a favorable safety profile with no serious adverse events (SAEs) and a lack of the adverse events (AEs) historically associated with TLR7 agonists
 - Robust systemic immune induction was achieved, without counter-regulation or evidence of systemic inflammation, in healthy volunteers (HVs) receiving PRTX007 in the 300- to 500-mg MAD cohorts
- Here we report clinical pooled safety for the phase 1 study and new data on the immune correlate endpoints for the 750-mg cohort
- Data from preclinical studies (including primates) and this phase 1 clinical study suggest a novel mechanism of action (MOA) for PRX034. Like other classical TLR7 agonists, PRX034 stimulates systemic immune induction via activation of pDCs; however, unlike the classical TLR7 agonists, downstream activation is restricted to NK cells (innate immunity) and naïve and cytotoxic T cells (adaptive immunity), with minimal activation of B cells (**Figure 1**). The activation and downstream cascade elicit effective antitumor and antiviral responses

Mechanism of Action (MOA)

Activation of pDCs by TLR7 Agonist PRX034 Elicits Effective Antitumor and Antiviral Responses

Figure 1. MOA for PRX034 is focused on pDC activation

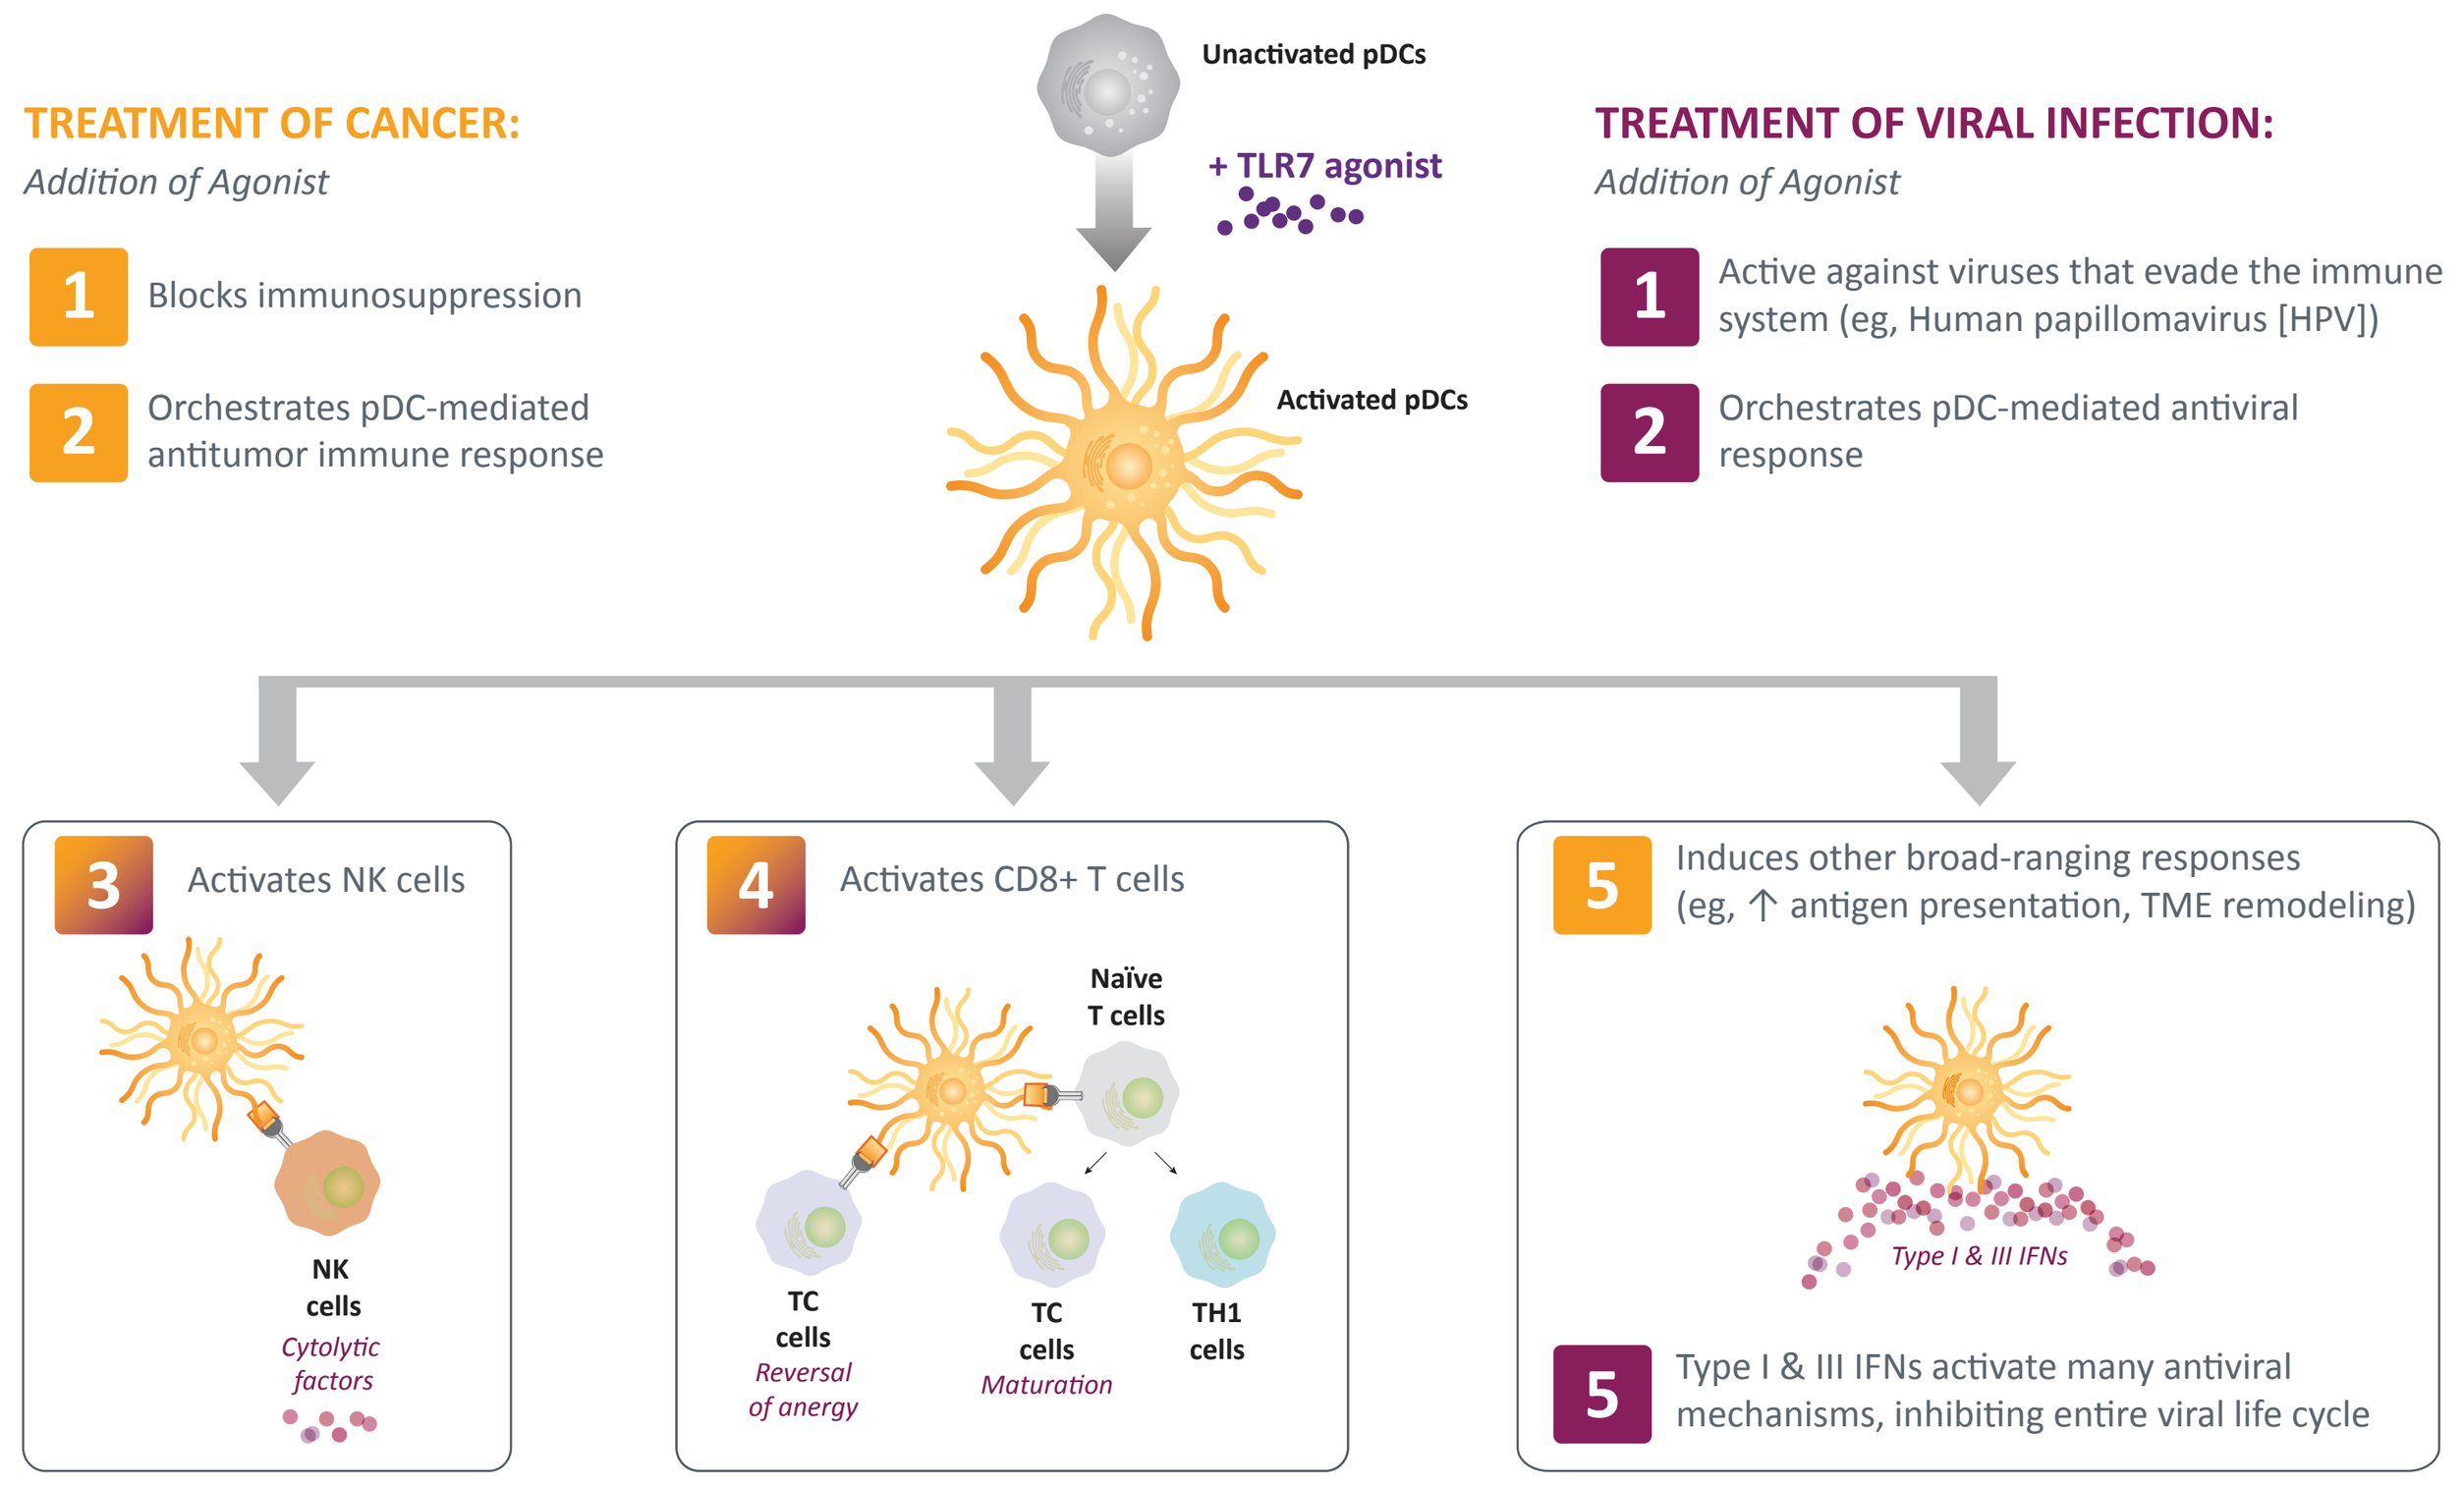
Unactivated pDCs
+ TLR7 agonist
Activated pDCs
TREATMENT OF CANCER:
Addition of Agonist
1 Blocks immunosuppression
2 Orchestrates pDC-mediated antitumor immune response
TREATMENT OF VIRAL INFECTION:
Addition of Agonist
1 Active against viruses that evade the immune system (eg, Human papillomavirus [HPV])
2 Orchestrates pDC-mediated antiviral response
3 Activates NK cells
NK cells
Cytolytic factors
4 Activates CD8+ T cells
Naïve T cells
TC cells
Reversal of anergy
TC cells
Maturation
TH1 cells
5 Induces other broad-ranging responses (eg, ↑ antigen presentation, TME remodeling)
Type I & III IFNs
5 Type I & III IFNs activate many antiviral mechanisms, inhibiting entire viral life cycle

IFN=interferon; NK=natural killer; TC=cytotoxic T cell; TH1= T helper type 1; TME=tumor and its surrounding microenvironment; TLR7=toll-like receptor 7.

- **PRTX007** is an oral prodrug that delivers the TLR7 agonist **PRX034** throughout the body
- PRX034 pharmacologic activities are more restricted than most TLR7 agonists
 - Avoids induction of NFκB-mediated biosynthesis of proinflammatory factors
 - Minimizes activation of B cells
- **PRX034** pharmacologic activity engages CD8+ T cells and NK cells, the two most important immune cells for killing cancer cells
- **Avoids systemic inflammation while delivering therapeutic benefit**

PRX118 and the Mouse Model

Experimental Design

Figure 2. Experimental design for assessing mono and combination therapy of PRX118 in a mouse tumor model

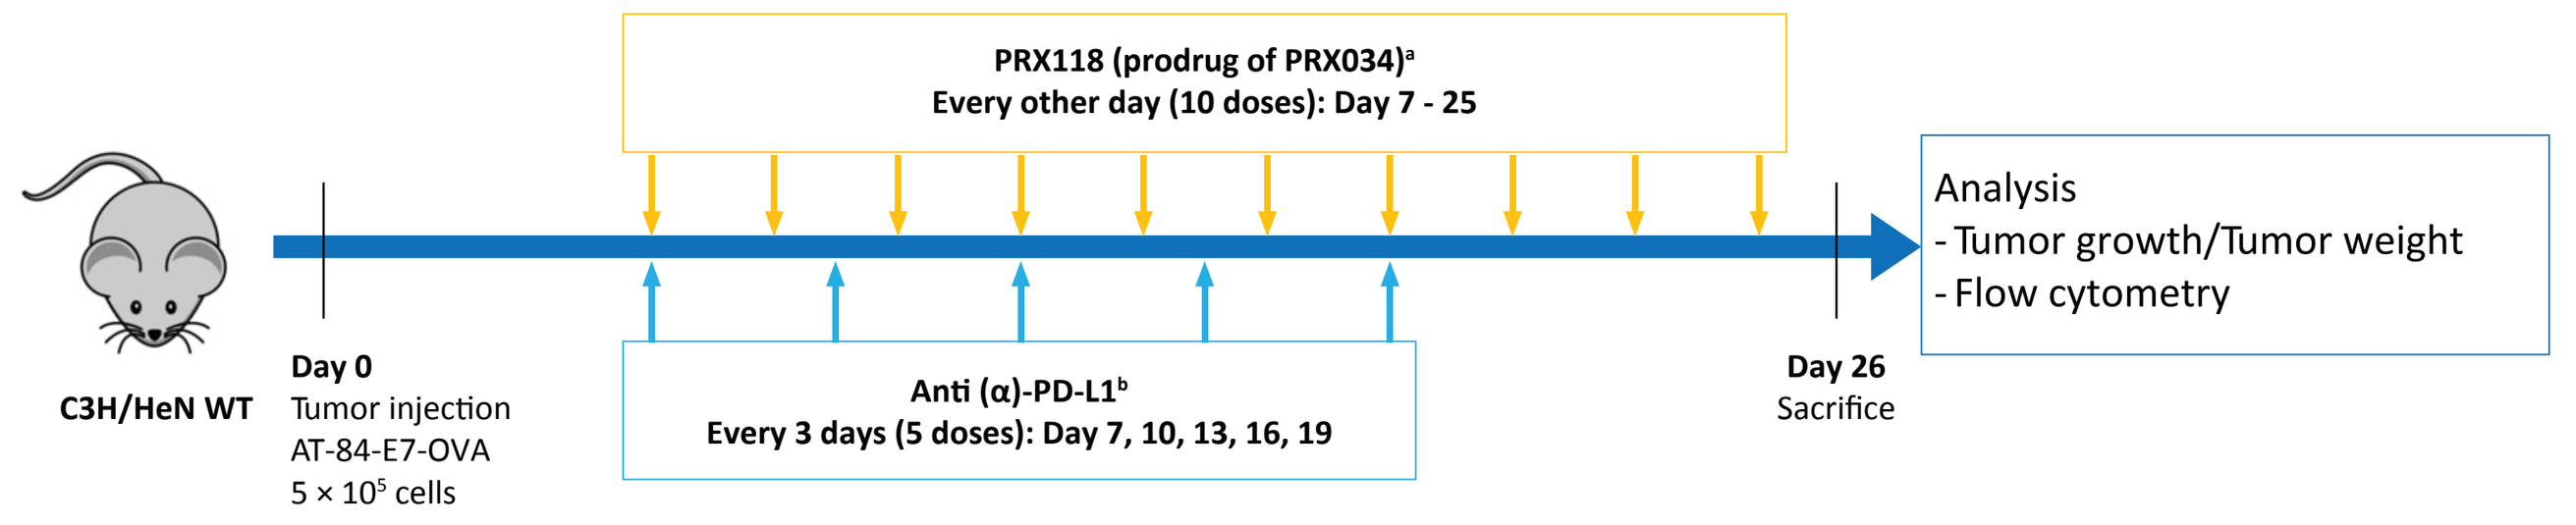
PRX118 (prodrug of PRX034)[a]
Every other day (10 doses): Day 7 - 25
C3H/HeN WT
Day 0
Tumor injection
AT-84-E7-OVA
5 × 10[5] cells
Anti (α)-PD-L1[b]
Every 3 days (5 doses): Day 7, 10, 13, 16, 19
Day 26
Sacrifice
Analysis
- Tumor growth/Tumor weight
- Flow cytometry

Tumor cell
AT-84-E7-OVA
5 × 10[5] cells in right flank

Mouse
C3H/HeN, female, 12 weeks old

Analysis
- Tumor growth
- Flow cytometry

Groups (n=7 or 8 per group)

1. Control (PBS)	(n=8)
2. α-PD-L1	(n=8)
3. PRX118	(n=7)
4. PRX118 + α-PD-L1	(n=7)

PRX118: 100 mg/kg, every 2 days, total 10 doses, i.p.
α-PD-L1: 200 µg/mouse, every 3 days, total 5 doses, i.p.

i.p.=intraperitoneal; PBS=phosphate buffered saline.
[a]PRX118, a mouse-compatible prodrug surrogate for PRTX007 that converts to PRX034.
[b]Anti (α)-PD-L1, an antibody directed against programmed cell death ligand 1 (PD-L1) and used an immune checkpoint inhibitor.

- Preclinical testing of the MOA was carried out in a mouse tumor model (**Figure 2**)
- Mono and combination immunotherapy of PRX118, the mouse-compatible prodrug surrogate for PRTX007, was assessed in a murine tumor model expressing the E7 oncogene from the human papillomavirus (HPV)[3,4]

PRX118 Mono and Combination Immunotherapy in Murine Tumor Model Expressing the E7 Oncogene From the HPV Demonstrate Importance of TLR7 Agonist

Figure 3. Tumor volume reduction and enhanced infiltration of tumor-associated CD8+ T-cells observed after mono and combination therapy

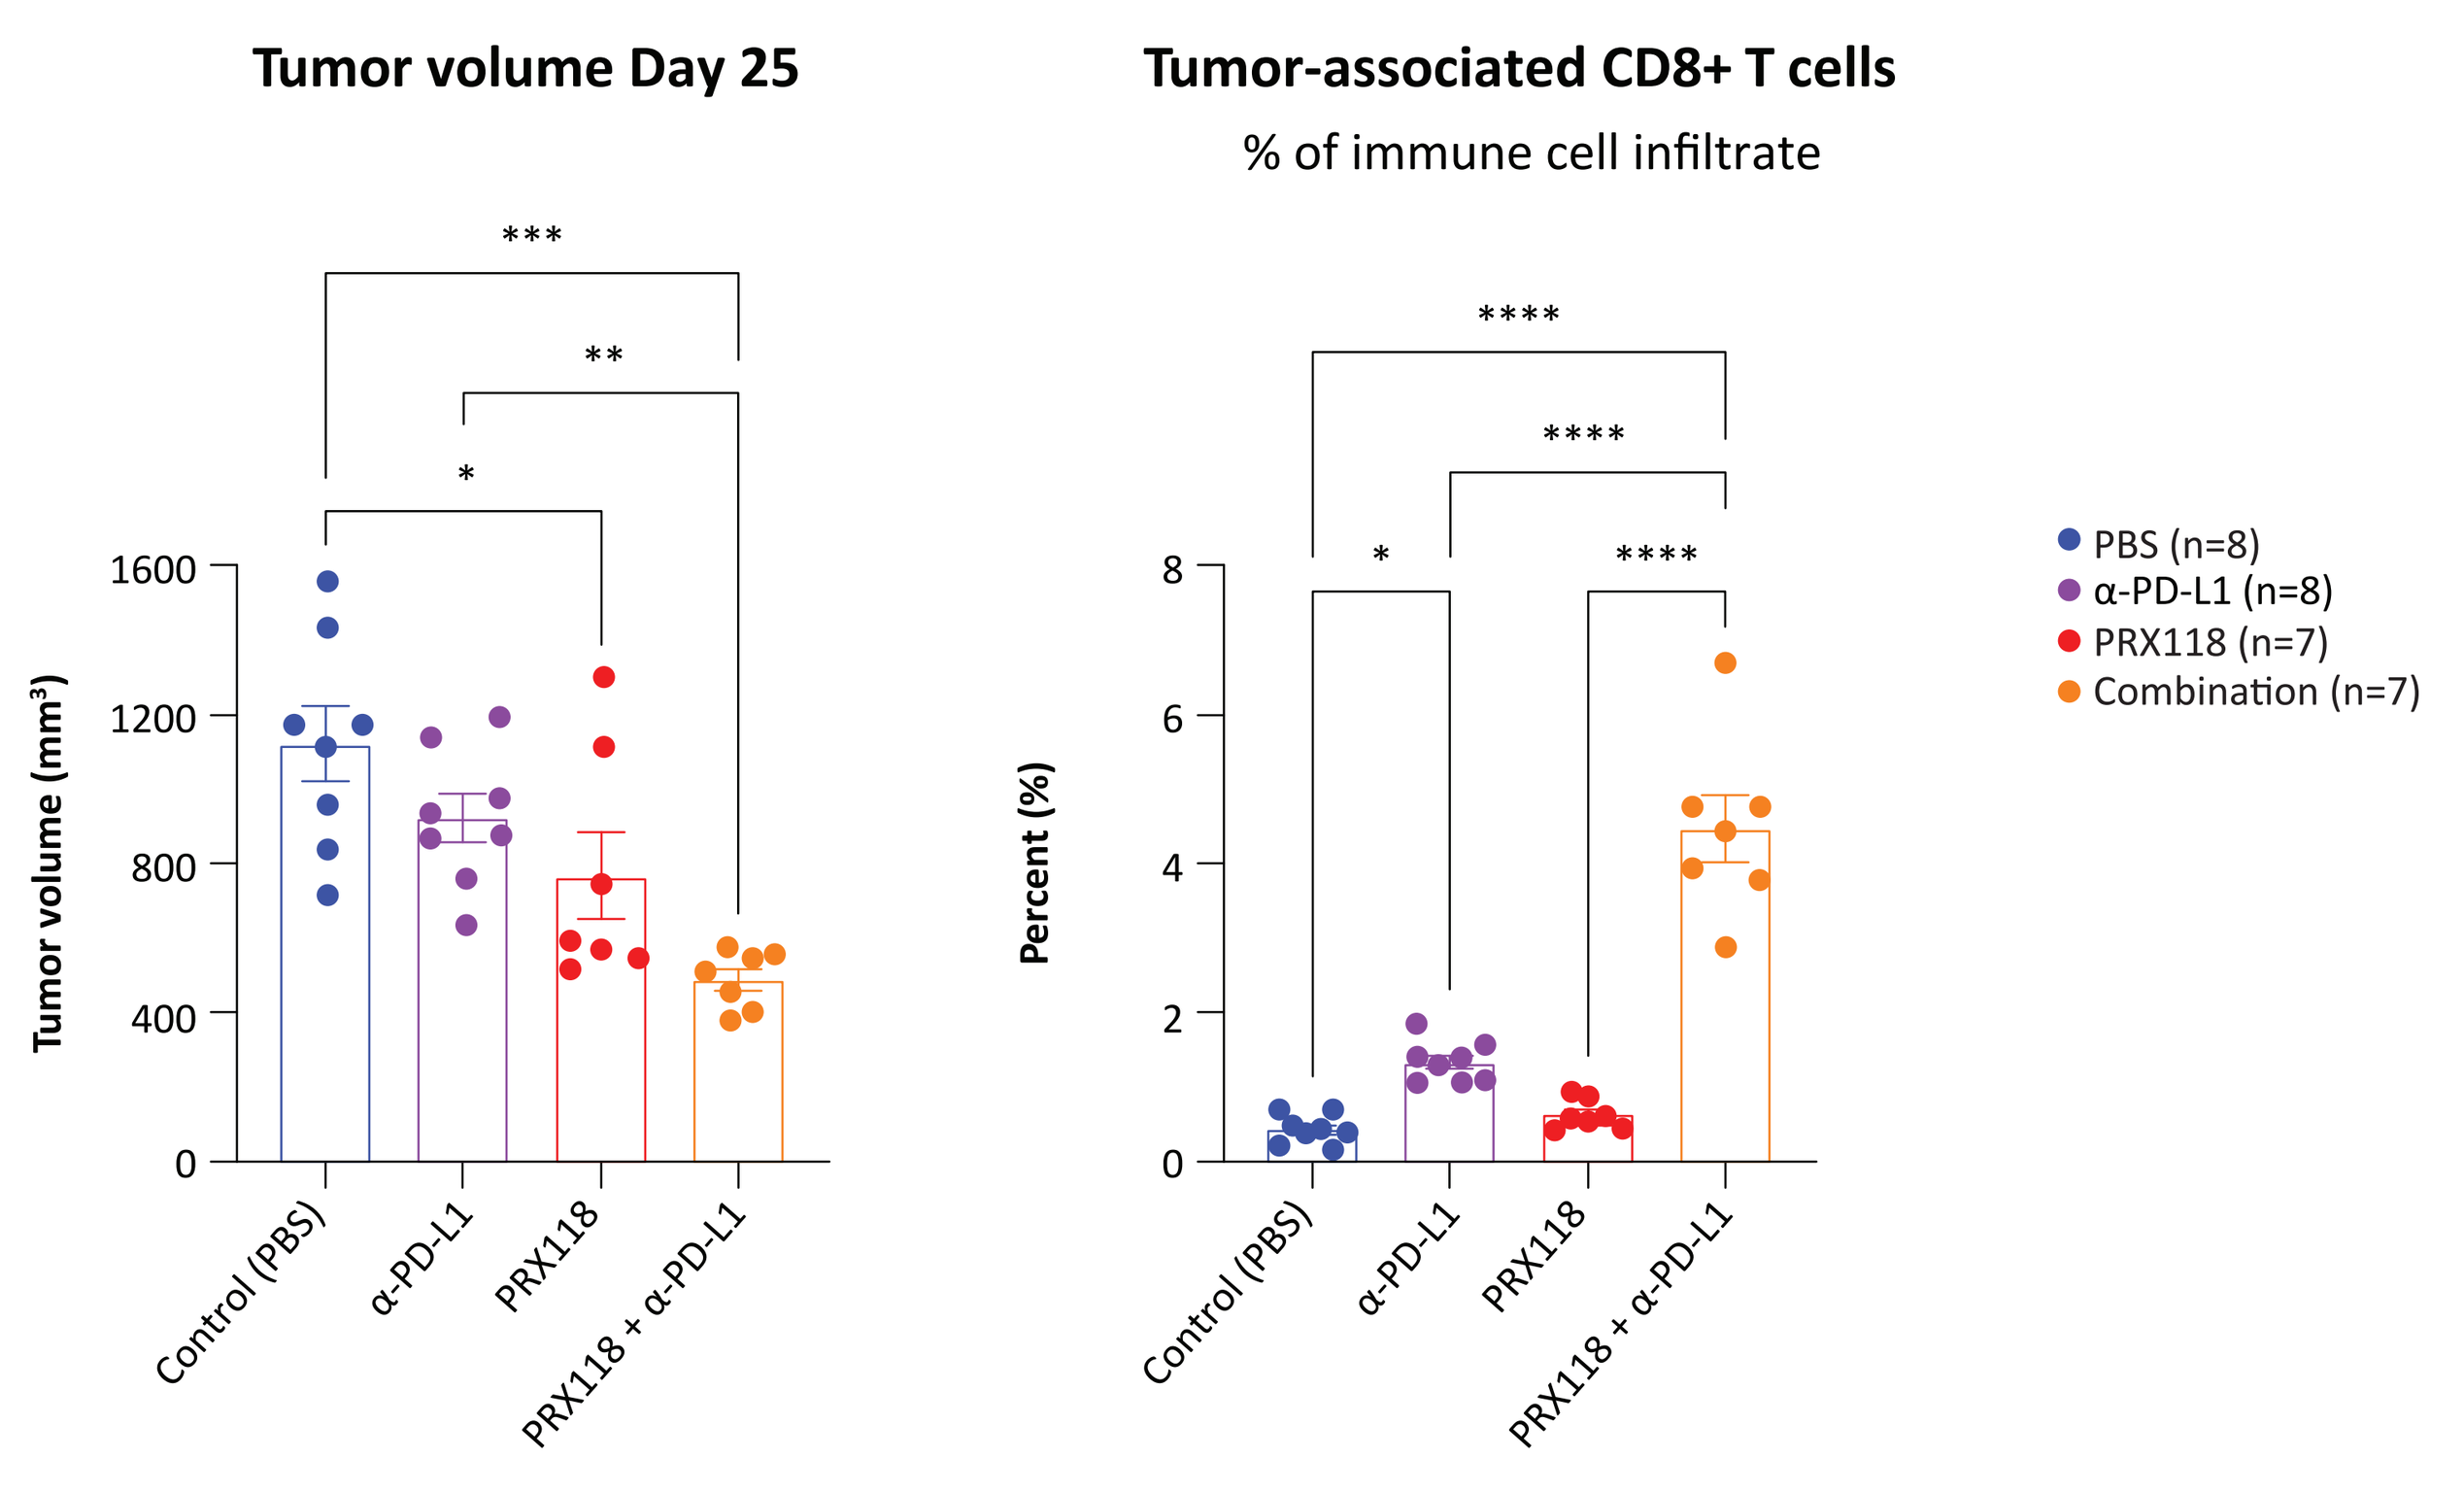
Tumor volume Day 25
Tumor volume (mm³)
Control (PBS)
α-PD-L1
PRX118
PRX118 + α-PD-L1
Tumor-associated CD8+ T cells
% of immune cell infiltrate
Percent (%)
PBS (n=8)
α-PD-L1 (n=8)
PRX118 (n=7)
Combination (n=7)

*P ≤ 0.05; **P ≤ 0.01; ***P ≤ 0.001; ****P ≤ 0.0001

- Significant antitumor activity of TLR7 agonist monotherapy was observed (exceeding that of PD-L1 inhibitor alone) (**Figure 3, left panel**)
 - Increased antitumor activity was obtained when combining these 2 mechanisms
- The percent of tumor growth inhibition observed at Day 25 compared to placebo was 18% for α-PD-L1, 31% for PRX118, and 56% for combination therapy (PRX118 + α-PD-L1)
- Orthogonal antitumor mechanisms were induced (**Figure 3, right panel**)
- An increase in CD8+ T-cell tumor infiltration was observed with combination therapy compared with monotherapy
 - Systemic activation of CD8+ T cells resulted in increased CD8+ T-cell infiltration in the presence of the CPI, resulting in increased killing of tumor cells
- PRX034 and closely related compounds have also demonstrated activity in CT26 colon carcinoma and B16F10 melanoma rodent tumor models (data not shown)

Overview of Pharmacodynamic (PD) Markers and Relative Sensitivity to PRTX007 Dose in the Phase 1a Clinical Study

Figure 4. PD markers and their sensitivity to optimized TLR7 agonists

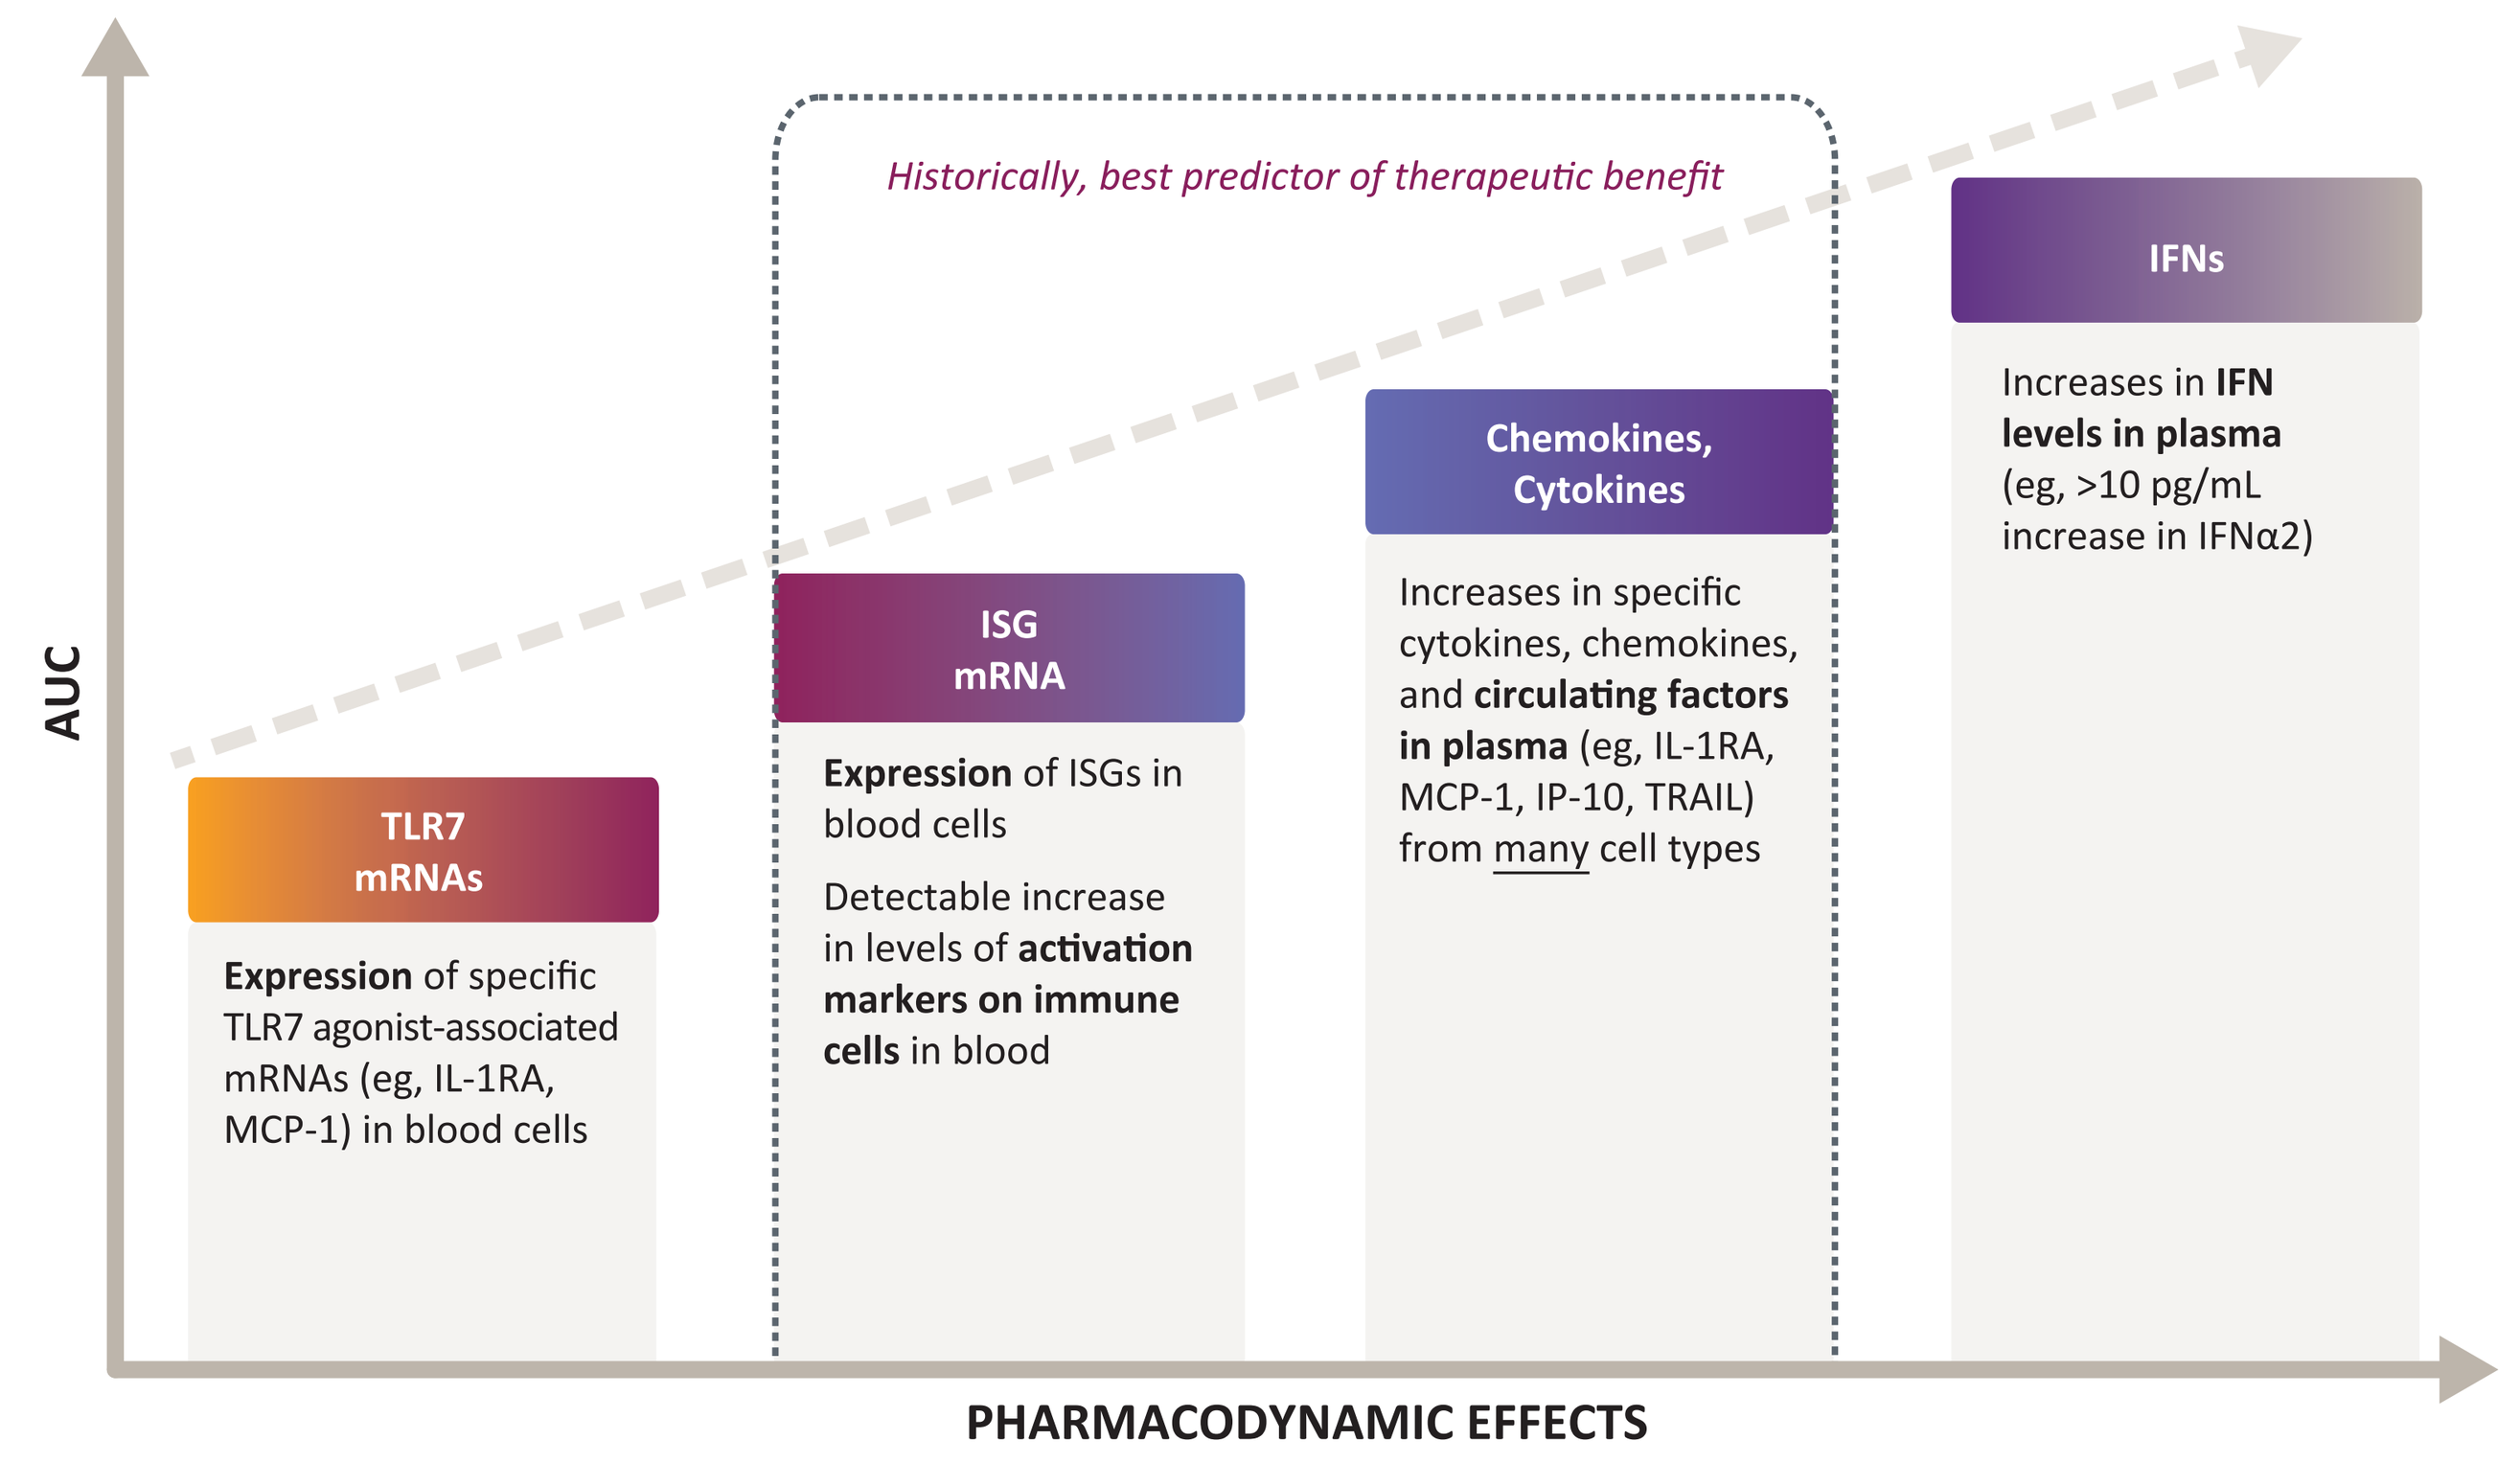
Historically, best predictor of therapeutic benefit
AUC
TLR7 mRNAs
Expression of specific TLR7 agonist-associated mRNAs (eg, IL-1RA, MCP-1) in blood cells
ISG mRNA
Expression of ISGs in blood cells
Detectable increase in levels of activation markers on immune cells in blood
Chemokines, Cytokines
Increases in specific cytokines, chemokines, and circulating factors in plasma (eg, IL-1RA, MCP-1, IP-10, TRAIL) from many cell types
IFNs
Increases in IFN levels in plasma (eg, >10 pg/mL increase in IFNα2)
PHARMACODYNAMIC EFFECTS

AUC=area under the plasma drug concentration-time curve; ISG=interferon-stimulated gene.

Methods

- This was a first-in-human, phase 1, single-center, prospective, randomized, double-blind, placebo-controlled study of 9 single-ascending dose (SAD) cohorts and 4 multiple-ascending dose (MAD) cohorts of PRTX007 administered every other day (QOD) orally to adult healthy volunteers (HVs) in Sydney, Australia[2]
 - The 500-mg MAD cohort terminated early (after 5th dose) because of COVID-19
- Primary objectives were to assess clinical safety and tolerability of PRTX007 in HVs
- Secondary objectives were to (1) assess the pharmacokinetic (PK) characteristics of both PRTX007 and PRX034, (2) assess the PD responses of PRX034 over single and multiple doses in normal HVs, and (3) compare the PK profiles of free-base and salt forms of PRTX007
- Rationale for conducting a phase 1 study in HVs included: (1) identification of drug-specific AEs and PD markers, which is not possible to do in combination therapy in oncology patients, (2) increased clinical sampling, allowing investigators to build a comprehensive PD profile, and (3) identification of the specific active dosing range for use in cancer studies, which will help significantly reduce time, costs, and patient burden in future studies

Figure 5. Study design for phase 1 trial in healthy volunteers (double-blind, placebo-controlled)

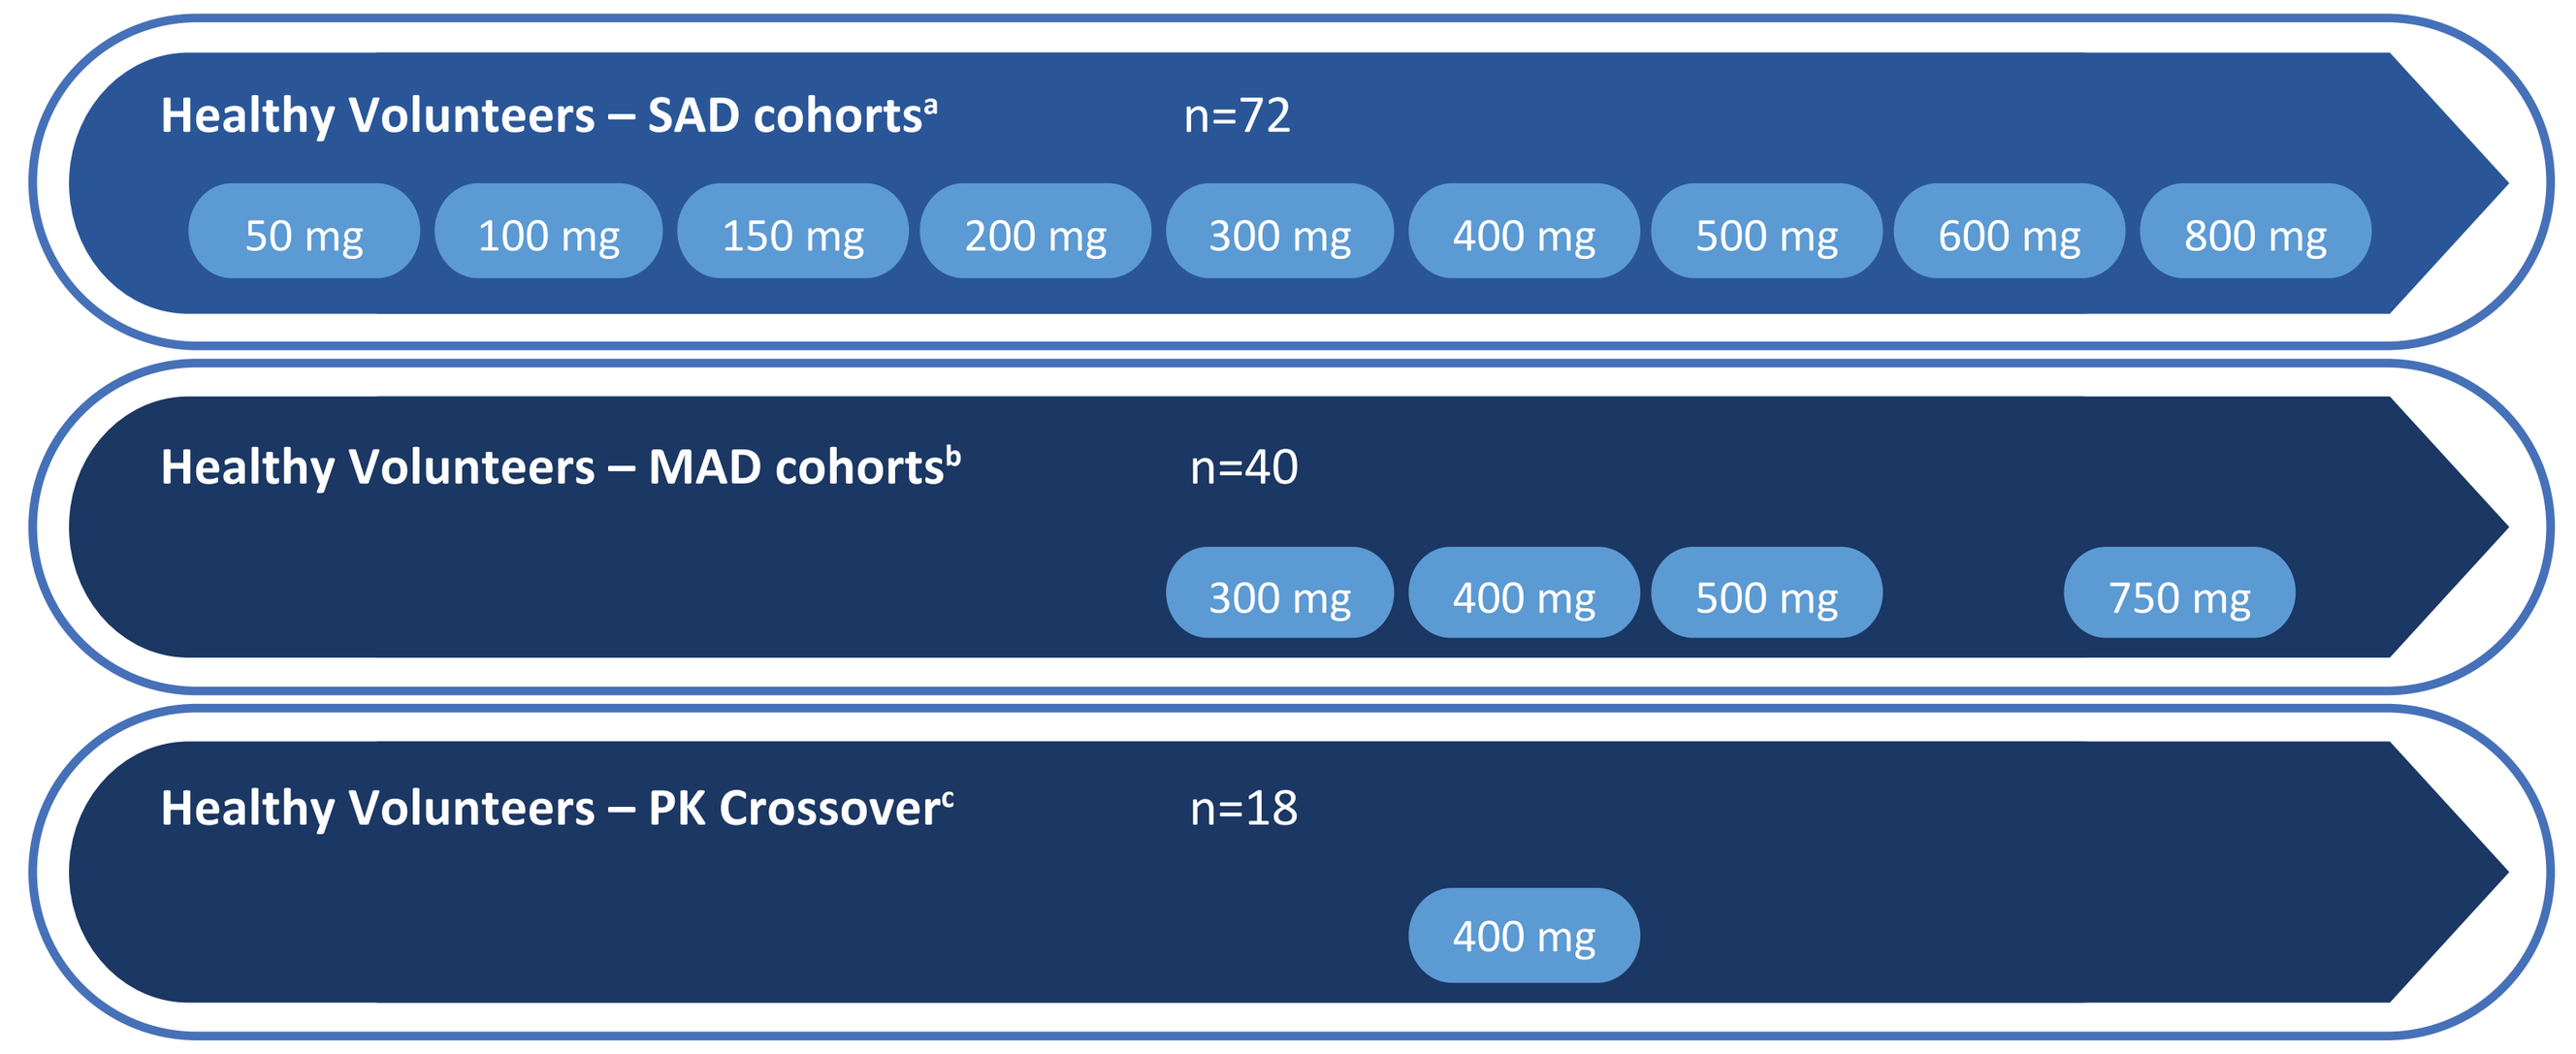
Healthy Volunteers – SAD cohorts[a] n=72
50 mg
100 mg
150 mg
200 mg
300 mg
400 mg
500 mg
600 mg
800 mg
Healthy Volunteers – MAD cohorts[b] n=40
300 mg
400 mg
500 mg
750 mg
Healthy Volunteers – PK Crossover[c] n=18
400 mg

[a]Each SAD cohort contained 6 treated and 2 placebo HVs/group; food effect study at 100 mg.
[b]Each MAD cohort contained 8 treated and 2 placebo HVs/group; administered QOD over 13 days (7 doses).
[c]Comparison of free-base and salt forms of PRTX007; single dose, crossover design.

Results

Clinical Pooled Safety Summary

AE profile for the 104 of 130 HVs receiving study drug.
The total number of AEs recorded in treated HVs was 110, with 42 attributed to PRTX007.

- There were no Grade 3 or higher AEs
- There were no clinically meaningful changes observed in creatinine, BUN, or uric acid
- There was no dose modification or discontinuation due to treatment-related AEs

Table 1. Most common AEs coded as treatment-related compared with placebo

AEs (X from N=130)	Placebo (n=26)	PRTX007 (n=104)	PRTX007 Percent (X/104=%) Grade Response: Grade 1	Grade 2	Grade 3
Nervous System Disorders					
Headache (40)[a]	4	17[a]	12%	5%	0
Hepatobiliary Disorders					
Elevated ALT (5)[b]	0	5[b]	5%[c]	0	0
General Disorders					
Fever (transient) (4)	1	3[d]	3%	0	0

ALT=alanine transaminase
[a]19 additional headaches in the PRTX007-treated group were coded as unrelated to drug. There was no dose dependence.
[b]No significant changes in aspartate transferase, bilirubin, or alkaline phosphatase were observed.
[c]4 of 5 HVs exhibited levels between 1.5x and 2x the upper limits of normal (ULN); 1 HV was at ~2.8x the ULN.
[d]Transient fever; resolved within 24-36 hours and did not recur upon subsequent dosing.

Individual HV Clinical Data for the 750-mg MAD Cohort

Well-behaved PK of PRX034 Following Oral Administration of 750 mg

As presented previously,[2] the PKs of PRTX007 and PRX034 were well behaved with exposure increasing proportionally to PRTX007 dose without accumulation upon repeated dosing (mean PRX034 AUC (hr*ng/ml) on D1: 13,692; D13: 13,933 for 750-mg dose cohort)

PD Responses in 750-mg Cohort

Induction of mRNAs for IFN-Stimulated Genes (ISGs) and Other TLR7-Associated Genes

Figure 6. mRNA transcript induction heat maps for 750 mg in the multi-dose cohort maintained a coordinated response

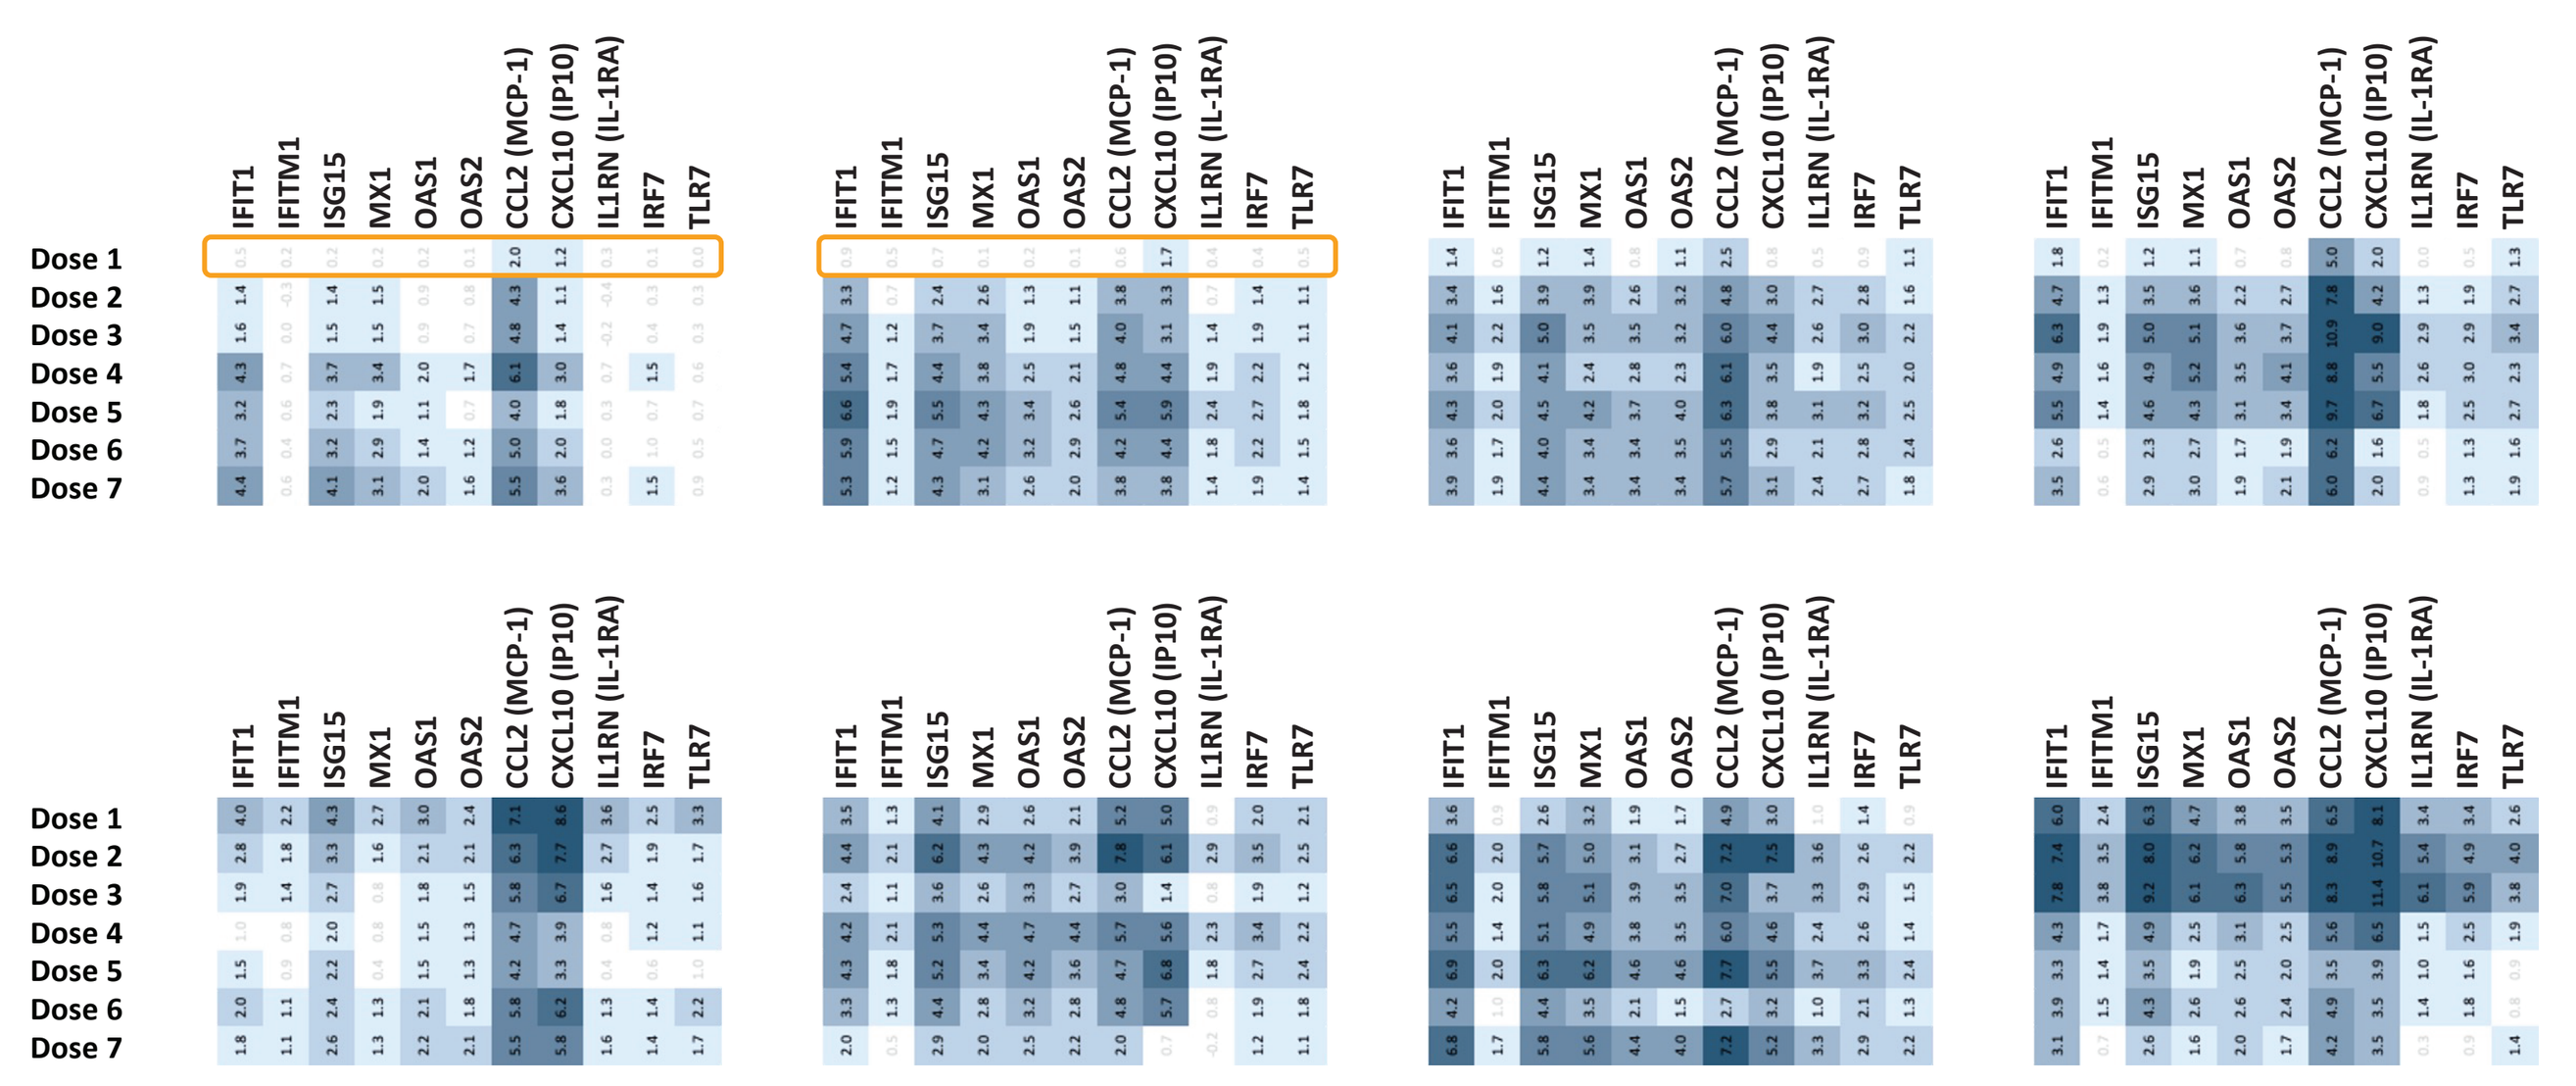
IFIT1
IFITM1
ISG15
MX1
OAS1
OAS2
CCL2 (MCP-1)
CXCL10 (IP10)
IL1RN (IL-1RA)
IRF7
TLR7
Dose 1
Dose 2
Dose 3
Dose 4
Dose 5
Dose 6
Dose 7

ISGs

IFIT1	Interferon-induced protein with tetratricopeptide repeats 1
IFITM	Interferon-induced transmembrane protein 1
ISG15	ISG15 ubiquitin-like modifier
MX1	MX dynamin-like GTPase 1
OAS1	2'-5'-oligoadenylate synthetase 1
OAS2	2'-5'-oligoadenylate synthetase 2

Mechanistic (other)

CCL2 (MCP-1)	C-C motif chemokine ligand 2; MCP-1
CXCL10 (IP10)	C-X-C motif chemokine ligand 10; IP-10
IL1RN (IL-1RA)	Interleukin 1 receptor antagonist; IL1RA
IRF7	Interferon regulatory factor 7
TLR7	Toll-like receptor 7

Color Scale

Min	Max	Color
0.0	1.0	
1.0	2.0	
2.0	3.0	
3.0	4.0	
4.0	5.0	
5.0	6.0	
6.0	7.0	
7.0	10.5	

Values are $\text{Log}_2\ R_{max,0\text{-}24hr}$ (Log_2 maximal fold-change from pretreatment baseline during 24-hr period following administration of PRTX007).
All HVs receiving PRTX007 in the 200- to 800-mg SAD cohorts, and the 300- to 750-mg MAD cohorts (n=68) are shown. Color intensity is as shown in scale.
ISG=interferon-stimulated gene.

- Coordinated expression of ISGs and genes known to be related to TLR7 agonism were observed in immune cells in blood in all HVs
 - The well-controlled expression seen with a single dose was maintained during repeated doses
- Two HVs that were poorly responsive to the first dose (first dose response highlighted in orange) established a robust response upon repeated dosing
 - The increased responsiveness in HVs 1 and 2 was not due to increased drug exposure, but rather due to increased sensitivity to drug
- There was no change in expression of NF-κB–regulated gene products

Circulating Markers in Plasma

PRTX007 Immune Cytokine Profile Demonstrates Compatibility for Combination Treatment With CPIs

Figure 7. Individual time course of cytokine profiles for all subjects in the 750-mg PRTX007 cohort (n=8)[a,b]

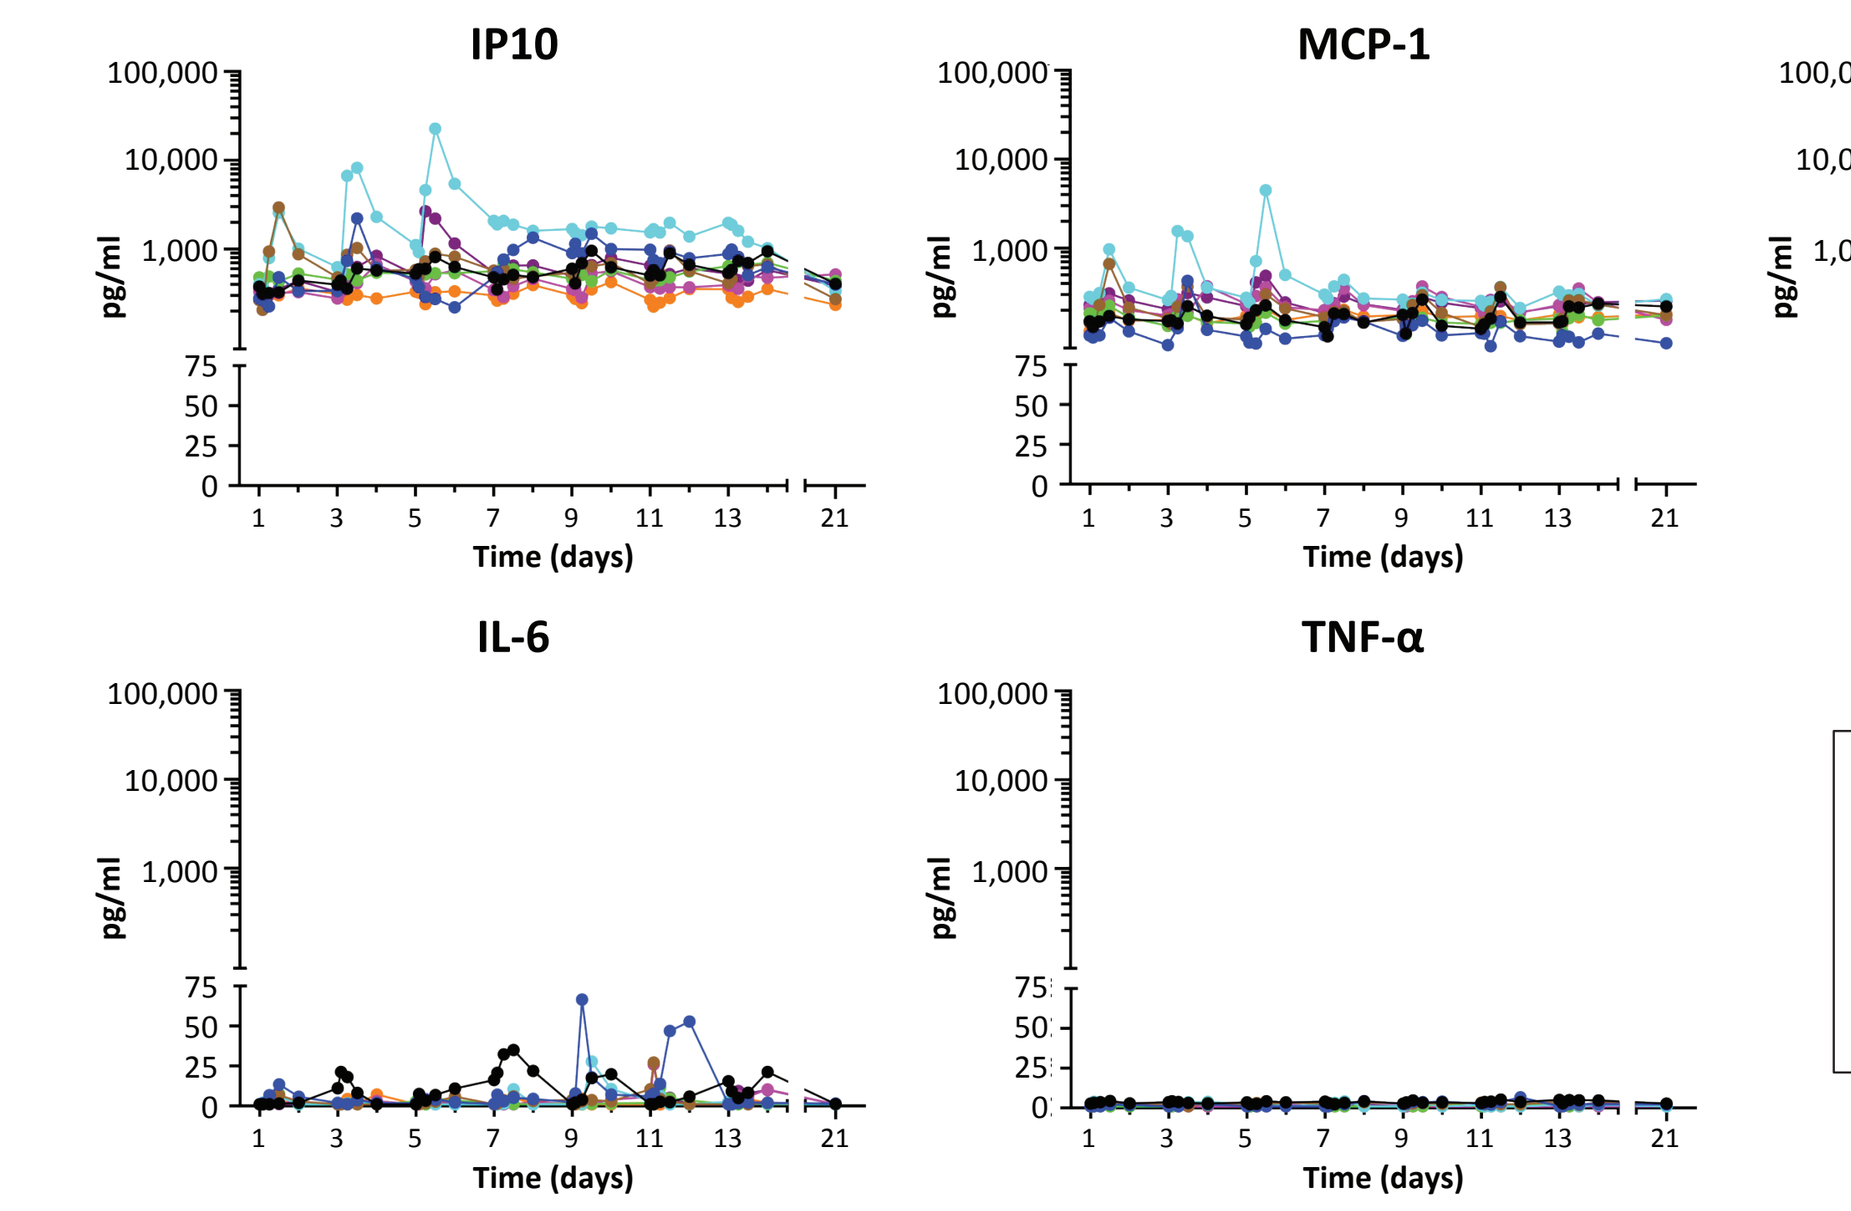
IP10
MCP-1
IL-1RA
IL-6
TNF-α
pg/ml
Time (days)

- The lack of proinflammatory factor production at active doses of PRTX007 make administration of this novel TLR7 agonist compatible with CPIs

[a]PRTX007 was administered for 2 weeks to 8 HVs (n=8) at 7 doses at 750 mg/dose QOD. Each colored line represents an individual HV.
[b]Day 1 was the predose data point and serves as a baseline. Day 21 was 8 days post last dose and shows cytokine levels return to baseline.

- A coordinated downstream immune response is suggested by the induction of selected circulating markers by the oral PRTX007 TLR7 agonist prodrug
 - Increases in IP10, monocyte chemoattractant protein 1 (MCP-1), and IL-1RA in plasma were observed
 - IL-6 and TNF-α remained essentially unchanged from pretreatment levels
 - No substantive increase in circulating Type I IFN or IL-1β were measured; these markers were less than lower limit of quantitation

Activation and Proliferation Markers Demonstrate Systemic CD8+ T Cell and NK Cell Engagement in 750-mg PRTX007 Cohort

Figure 8. Activation and proliferation in CD8+ T Cells and NK Cells in 750-mg PRTX007 cohort (n=8)

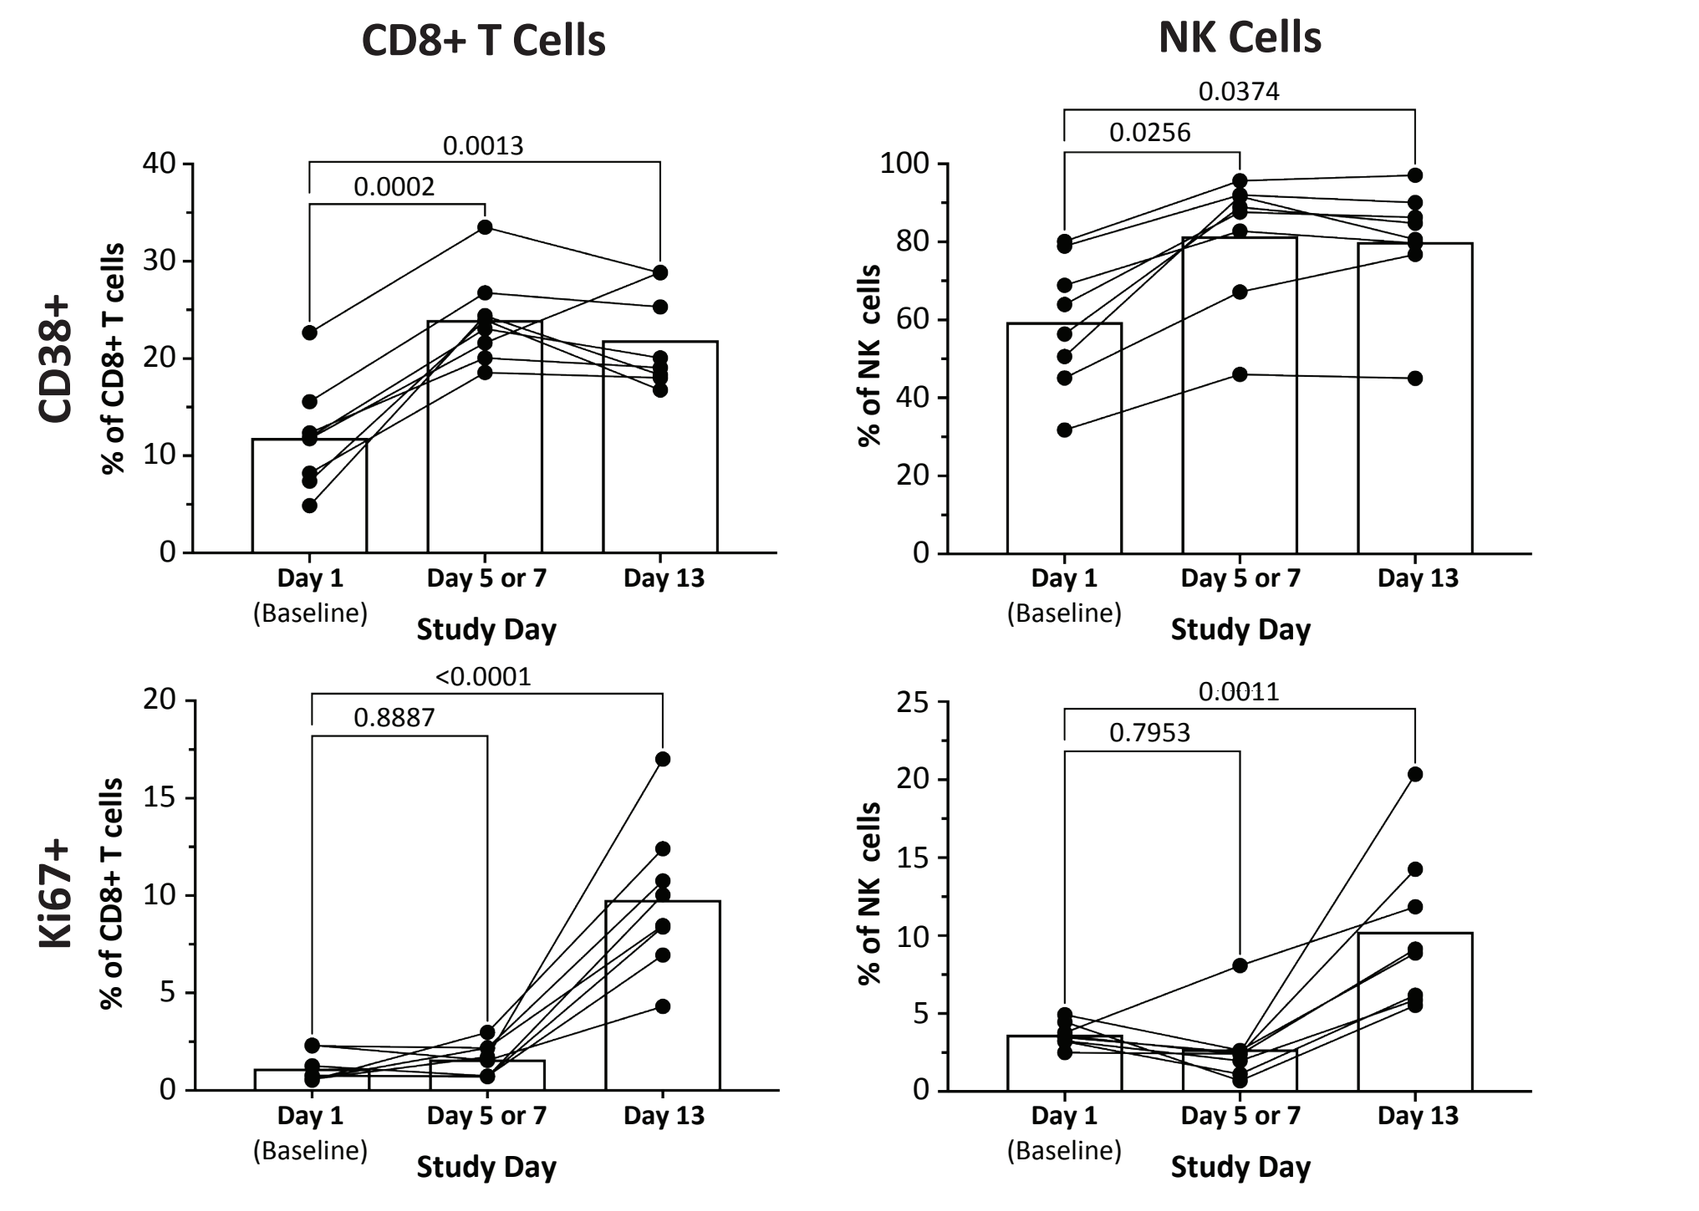
CD8+ T Cells
NK Cells
CD38+
% of CD8+ T cells
% of NK cells
Ki67+
0.0013
0.0002
0.0374
0.0256
<0.0001
0.8887
0.0011
0.7953
Day 1 (Baseline)
Day 5 or 7
Day 13
Study Day

- The proportion of CD8+ T cells and NK cells, with significant expression of the activation marker CD38 and proliferation marker Ki67, increased markedly from pretreatment to end of dosing in all HVs. During this period:
 - CD38+ CD8+ T cells increased from 11.8% to 21.9% and CD38+ NK cells increased from 59.4% to 80.0%
 - Ki67+ expressing CD8+ T cells increased from 1.1% to 9.8% and Ki67+ expressing NK cells increased from 3.6% to 10.3%
- In contrast, minimal change in activation and proliferation markers was observed for CD4+ T cells and B cells (data not shown; data on file). See also **Figure 1**

Conclusions & Discussion

- PRTX007 demonstrated a favorable safety profile in all HVs receiving drug in phase 1
- In the 750-mg MAD cohort, activation of the innate and adaptive immune response, including important effector cell populations, were observed without systemic increases in proinflammatory factors
 - Induction of ISGs without significant increases in circulating IFNs was observed
 - No increase in expression or circulating levels of proinflammatory cytokines (eg, TNF-α) was noted
 - CD8+ NK and T-cell activation (CD38+ markers) increased markedly from pretreatment to end of dosing in all HVs
- Both the clinical characteristics and unique pattern of immune induction by PRTX007 support its use in combination with CPIs
 - Local antitumor activity of intratumorally administered pDC-activating agents in combination with systemic CPIs is well recognized, but overall clinical benefit has been limited
 - Use of PRTX007 in combination with CPIs in the murine tumor model expressing the HPV E7 oncogene revealed robust antitumor activity. Increased CD8+ T-cell infiltration of tumors occurred only in the presence of the CPI
 - We believe the proposed combination will increase therapeutic benefit in cancer immunotherapy
- These data justify further exploration of PRTX007 as a potential cancer therapeutic; a future study will include a combination with a CPI in solid tumors at active doses identified in this HV study

References

1. Appleman JA, et al. Abstract 582: Selection of a novel toll-like receptor 7 (TLR7) agonist PRX034 for immunotherapy of cancer. *Cancer Res*. 2020;80(16S):582. **2.** Lemech C, et al. Abstract CT189: PRTX007, an optimized TLR7 agonist for systemic immunotherapy of cancers: Interim analysis of phase I study in healthy volunteers. *Cancer Res*. 2022;82(suppl 12):CT189-CT189. **3.** Paolini F, et al. Immunotherapy in new pre-clinical models of HPV-associated oral cancers. *Hum Vaccin Immunother*. 2013;9(3)534-543. **4.** Kim SS, et al. B cells improve overall survival in HPV-associated squamous cell carcinomas and are activated by radiation and PD-1 blockade. *Clin Cancer Res*. 2020;26(13):3345-3359.

Disclosures & Acknowledgements

This study was funded by Primmune Therapeutics.
We would like to acknowledge Sharabi lab members, Department of Radiation Medicine, University of California at San Diego.
Medical writing services were provided by SCIENT Healthcare Communications and funded by Primmune Therapeutics, Inc., (Carlsbad, CA).

Presented at AACR Annual Meeting 2023 | April 14-19, 2023 |Orlando, Florida, USA

PRTX007, an Optimized TLR7 Agonist for Systemic Immunotherapy of Cancers: Interim Analysis of Phase I Study in Healthy Volunteers

Charlotte R. Lemech[1], Christopher Argent[1], Curtis L. Scribner[2], Richard Daniels[3], James R. Appleman[3]

[1]Scientia Clinical Research, LTD, Randwick, NSW, Australia; [2]Curtis L. Scribner, Oakland, CA, USA; [3]Primmune Therapeutics Inc., Carlsbad, CA, USA

8165

Background

- The TLR7 (toll-like receptor 7) pathway to stimulate innate and adaptive immunity for treatment of cancer and multiple viral diseases, has been well characterized
- Systemically administered TLR7 agonists present a variety of challenges as clinical drug candidates to provide therapeutic benefit
- Primmune Therapeutics has designed and developed an orally administered, systemically distributed TLR7 agonist that elicits a robust, well-tolerated immune response involving activation of plasmacytoid dendritic cells (pDCs) throughout the body[1]; it is expected to also activate pDCs resident in the tumor microenvironment in cancer patients
- PRTX007 (oral prodrug) and the corresponding well-tolerated TLR7 agonist, PRX034, achieve acceptable potency with dose- dependent control of immune induction without driving a pro-inflammatory response

Activation of Plasmacytoid Dendritic Cells (pDCs) by TLR7 Agonists Elicits Effective Anti-tumor Response

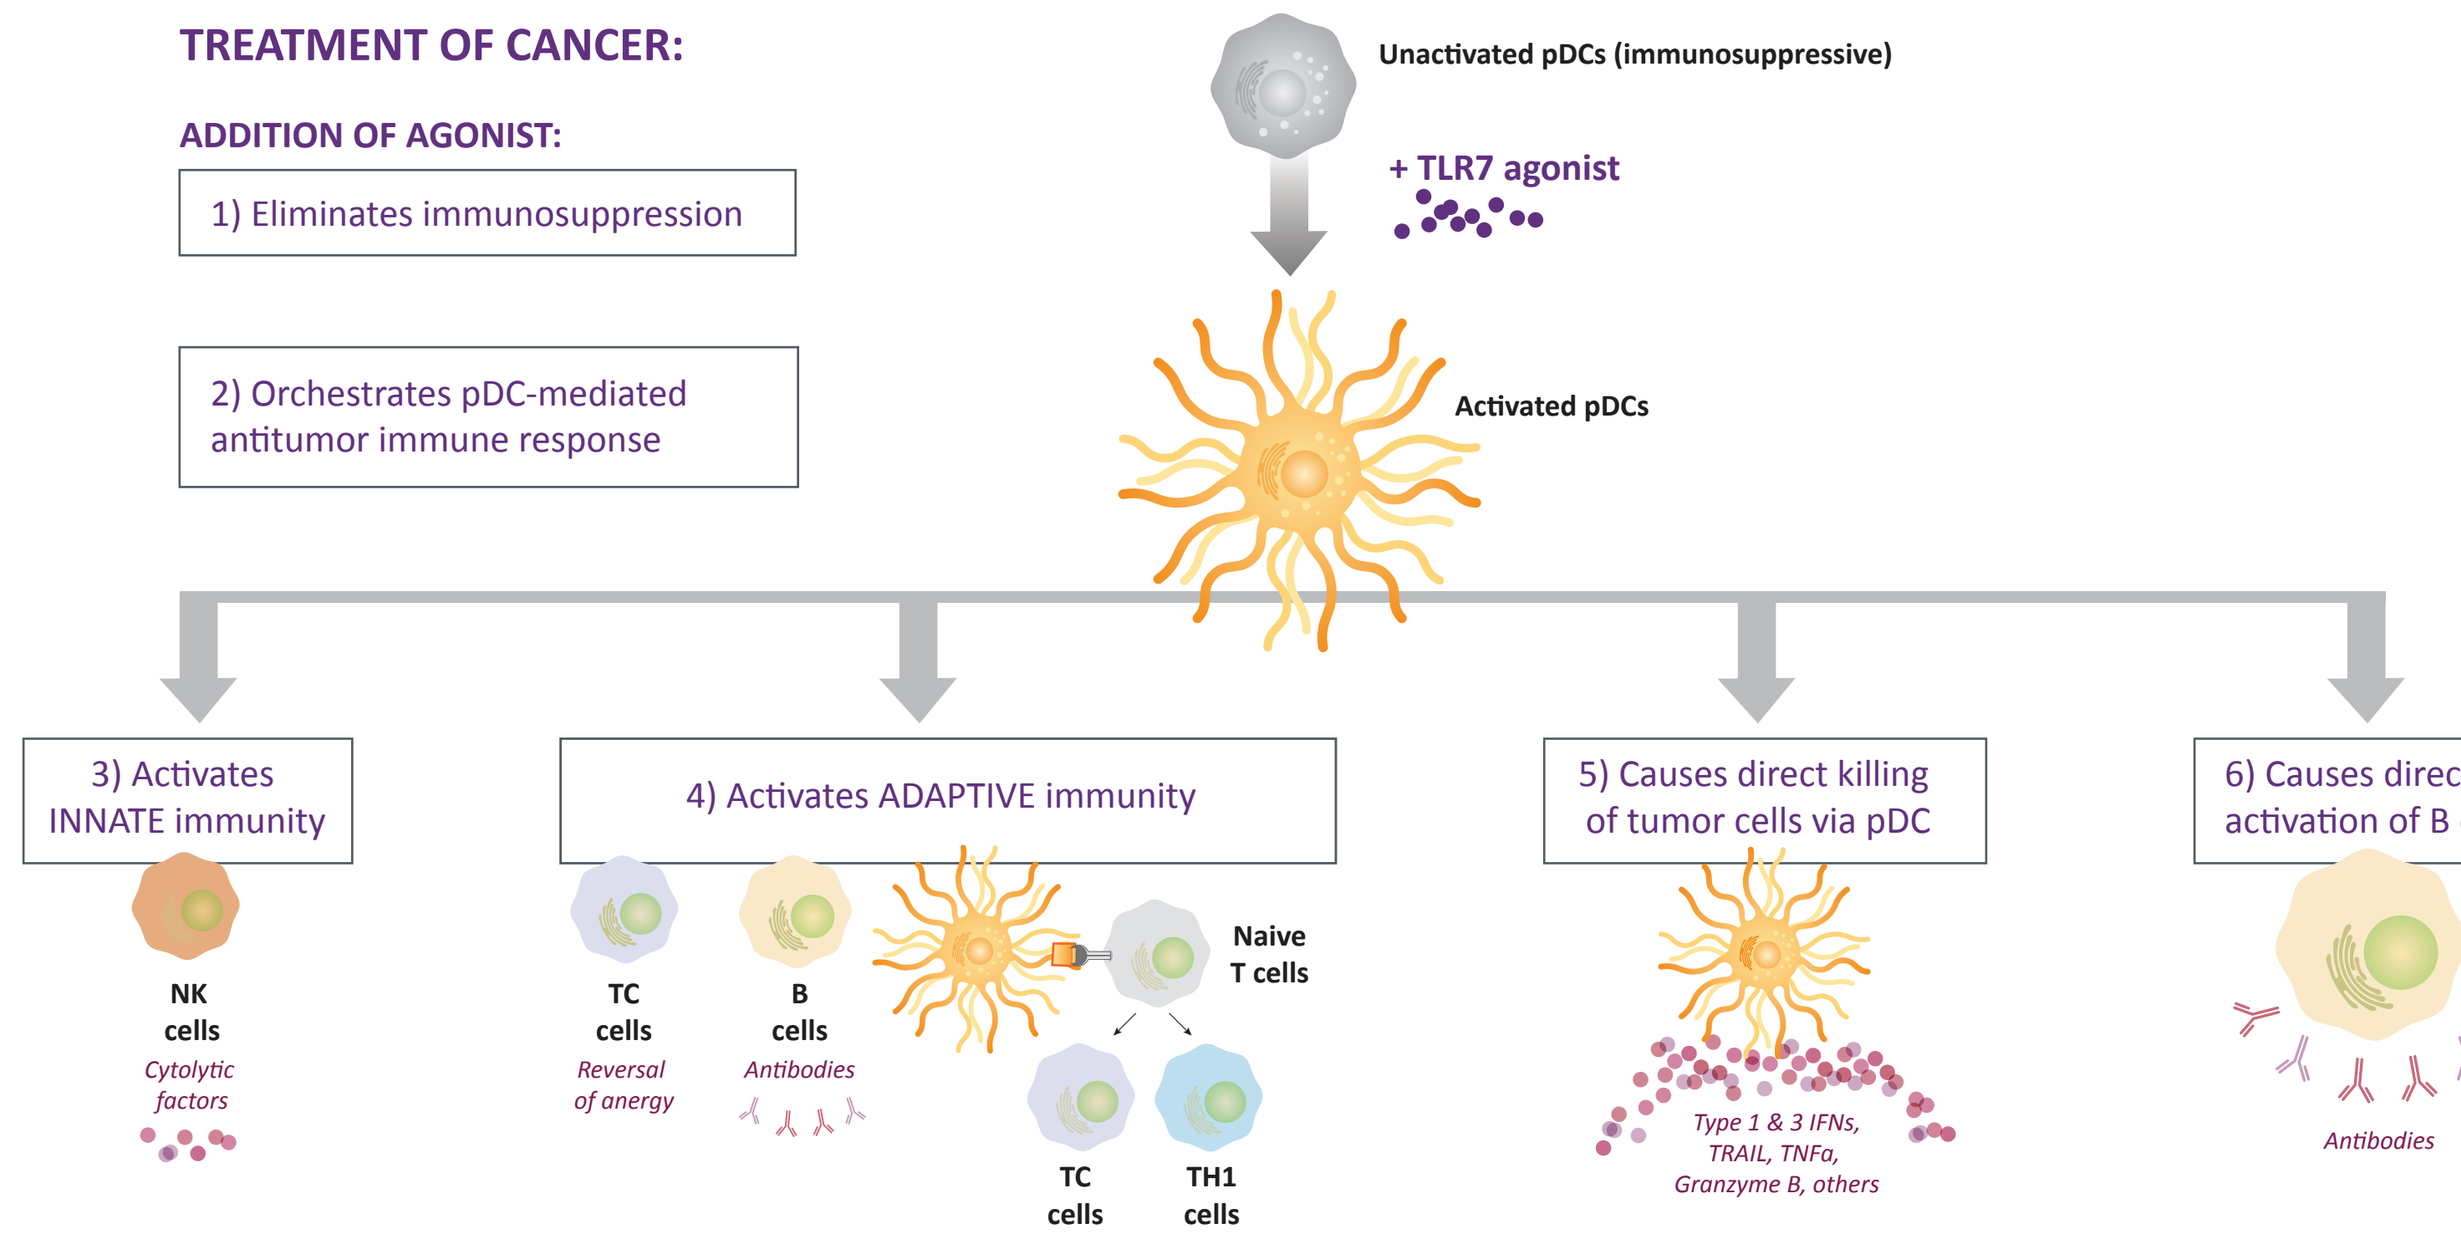
TREATMENT OF CANCER:
ADDITION OF AGONIST:
1) Eliminates immunosuppression
2) Orchestrates pDC-mediated antitumor immune response
Unactivated pDCs (immunosuppressive)
+ TLR7 agonist
Activated pDCs
3) Activates INNATE immunity
NK cells
Cytolytic factors
4) Activates ADAPTIVE immunity
TC cells
Reversal of anergy
B cells
Antibodies
Naive T cells
TC cells
TH1 cells
5) Causes direct killing of tumor cells via pDC
Type 1 & 3 IFNs, TRAIL, TNFα, Granzyme B, others
6) Causes direct activation of B cells
Antibodies

IFN=interferon; NK=natural killer; TC=cytotoxic T cell; TH1= Type 1 T helper cells; TLR7=toll-like receptor 7; TNFα=tumor necrosis factor alpha; TRAIL=tumor necrosis factor–related apoptosis-inducing ligand.

PRX034 is Highly Preferential for pDC-mediated Interferon Induction While Minimizing Inflammatory Cytokine Production When Compared to Traditional TLR7 Agonists

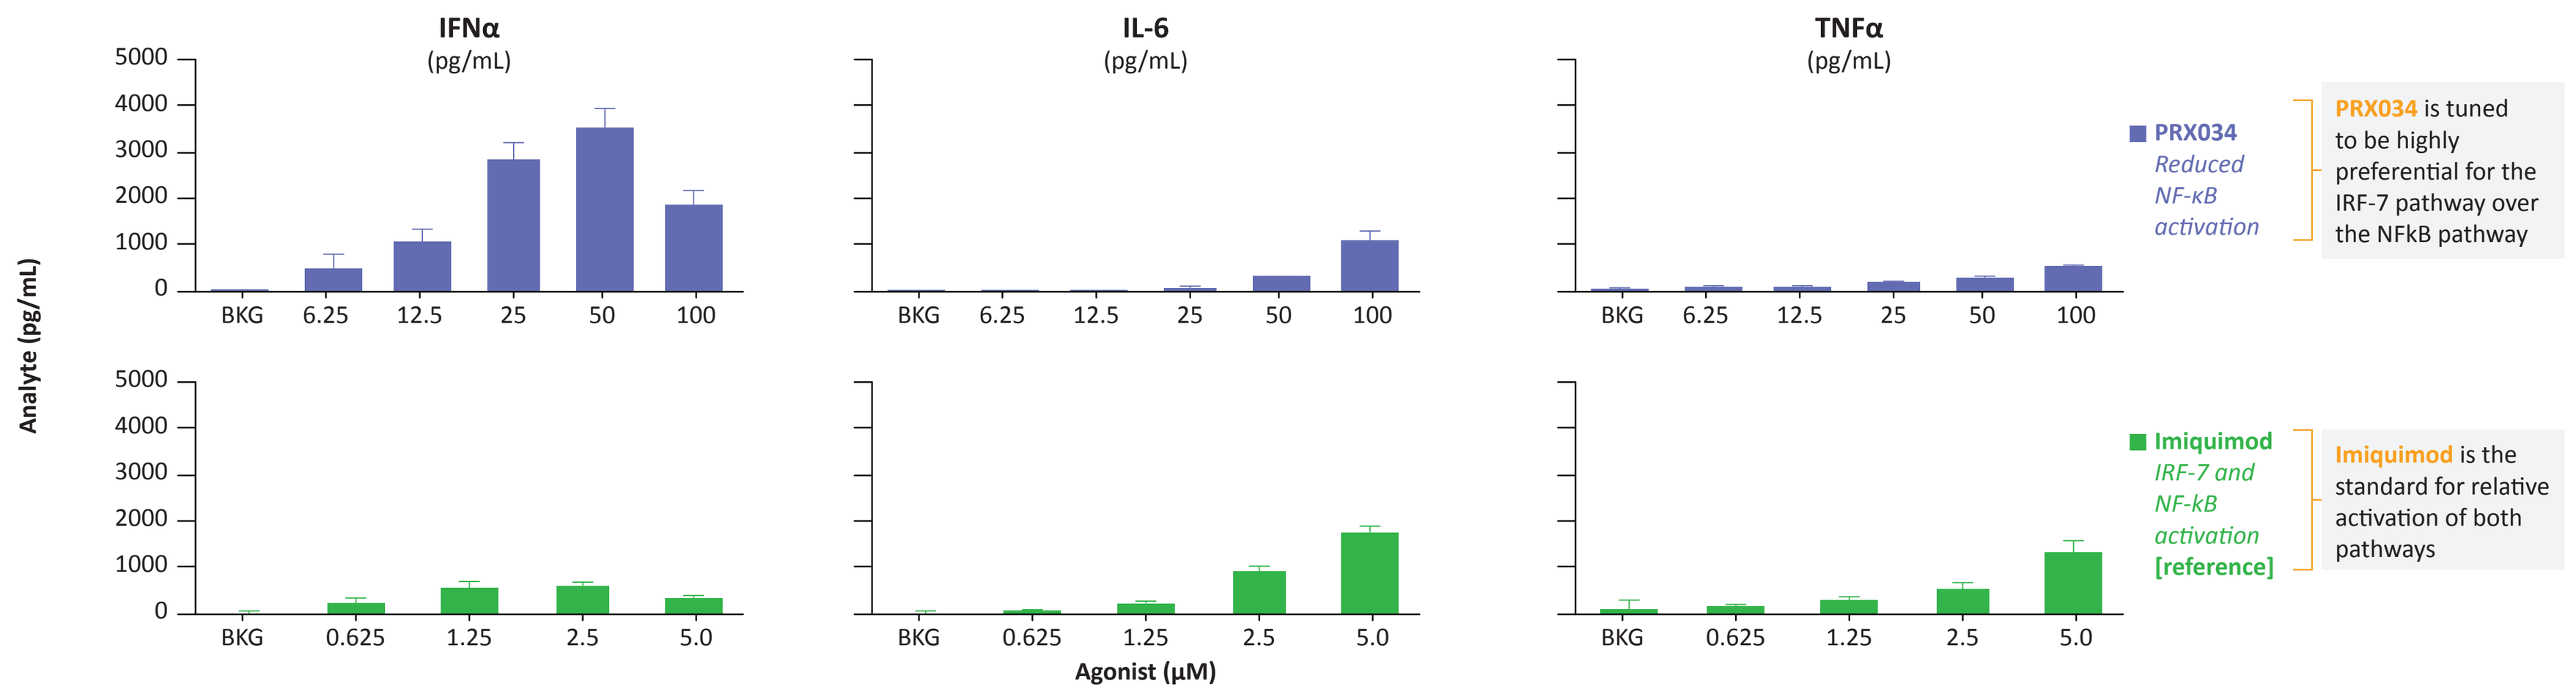
IFNα (pg/mL)
IL-6 (pg/mL)
TNFα (pg/mL)
Analyte (pg/mL)
Agonist (μM)
PRX034
Reduced NF-κB activation
PRX034 is tuned to be highly preferential for the IRF-7 pathway over the NFkB pathway
Imiquimod
IRF-7 and NF-kB activation [reference]
Imiquimod is the standard for relative activation of both pathways

TLR7-specific agonists elicit biased cellular pharmacology. Human peripheral blood mononuclear cells (hPBMCs) cytokine release profiles of PRX034 (blue bars) and imiquimod (green bars) are shown for 49 donors. hPBMC incubation with agonist was for 24 hours. Error bars are standard error of the mean.

Overview of Pharmacodynamic Markers and Relative Sensitivity to PRTX007 Dose in the Phase 1a Clinical Study

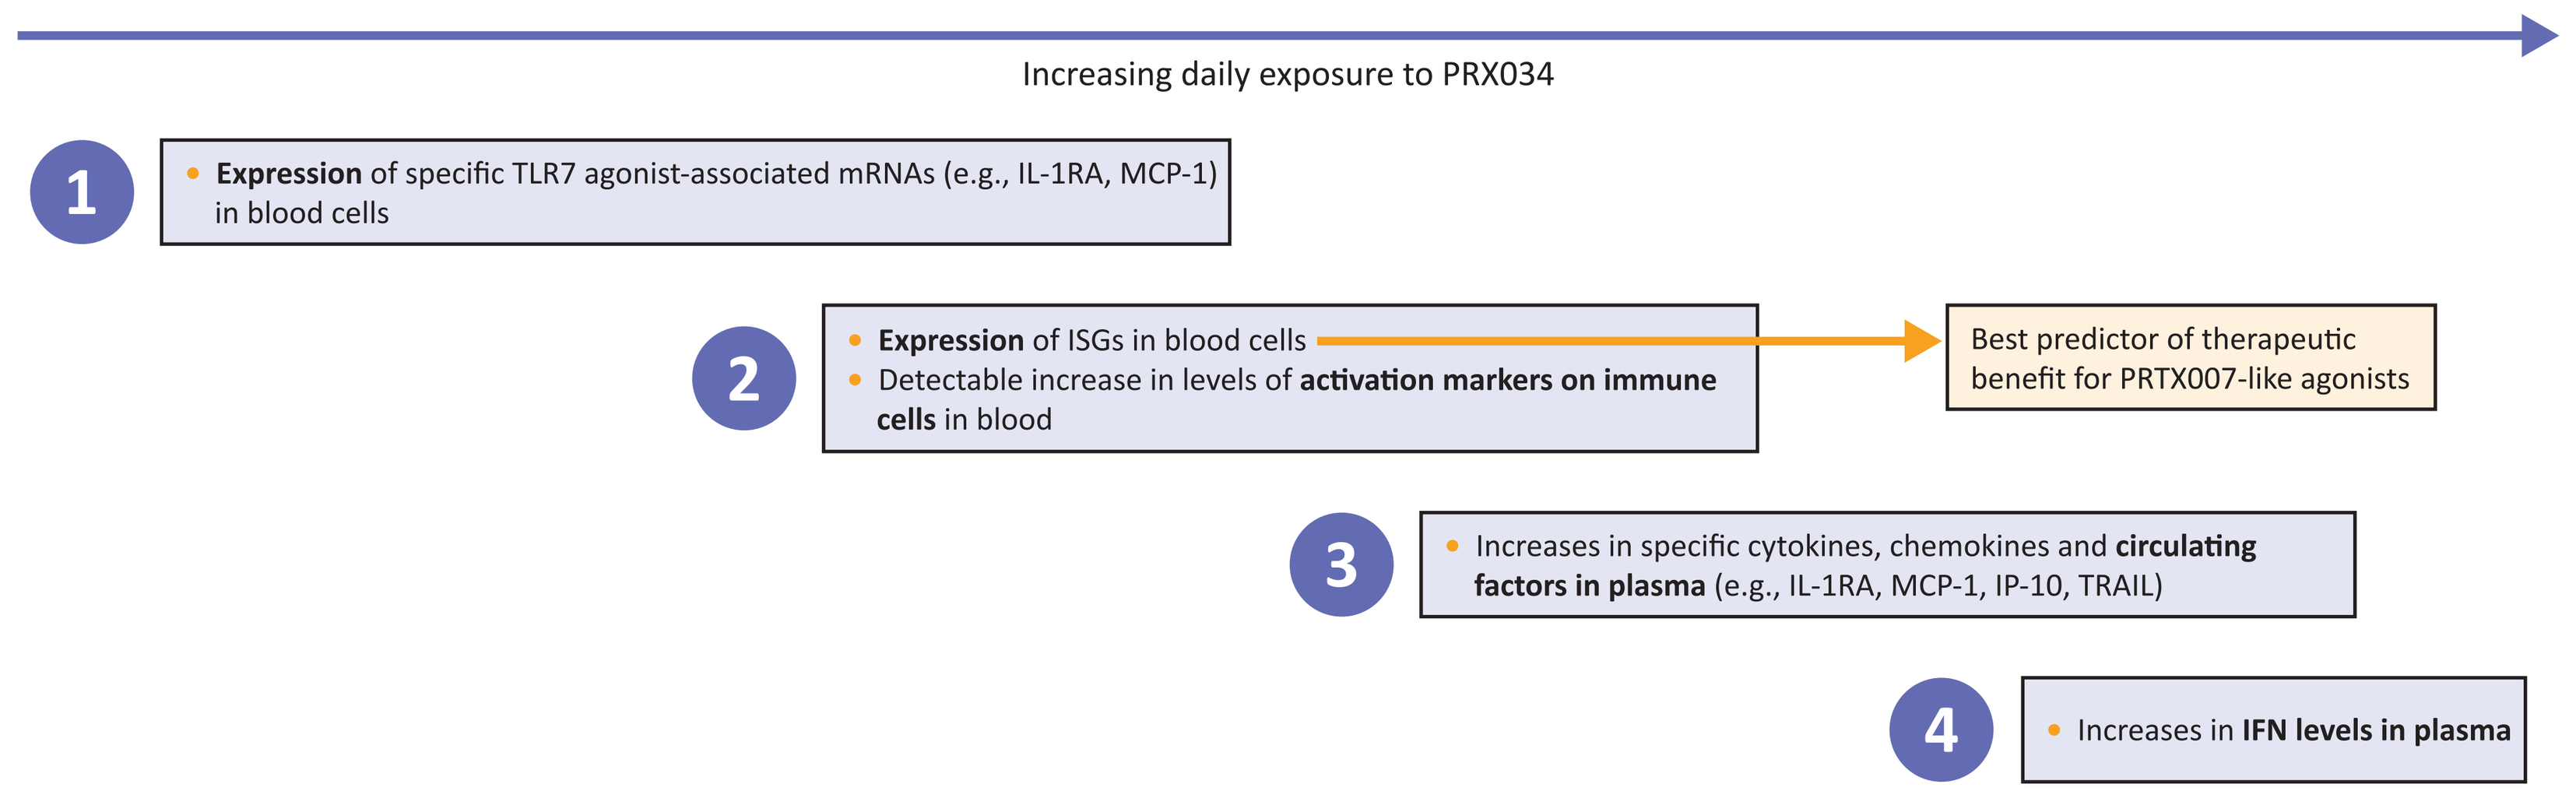
Increasing daily exposure to PRX034
1
Expression of specific TLR7 agonist-associated mRNAs (e.g., IL-1RA, MCP-1) in blood cells
2
Expression of ISGs in blood cells
Detectable increase in levels of activation markers on immune cells in blood
Best predictor of therapeutic benefit for PRTX007-like agonists
3
Increases in specific cytokines, chemokines and circulating factors in plasma (e.g., IL-1RA, MCP-1, IP-10, TRAIL)
4
Increases in IFN levels in plasma

ISG=interferon-stimulated gene

Methods

- This is a first-in-human, phase 1, single-center, prospective, randomized, double-blind, placebo-controlled study of 9 single-ascending dose (SAD) cohorts and 4 multiple-ascending dose (MAD) cohorts of PRTX007 administered orally to adult healthy volunteers (HVs) that is ongoing in Sydney, Australia
 - 500 mg MAD cohort truncated after 5th dose because of COVID
- Primary objective is to assess clinical safety and tolerability of PRTX007 in HVs
- Secondary objectives are to (1) assess the pharmacokinetic (PK) characteristics of both PRTX007 and PRX034, and (2) assess the pharmacodynamic (PD) responses of PRX034 over single and multiple doses in normal HVs

Study Design for Phase 1 SAD and MAD Trial in Healthy Volunteers (Double-blind, Placebo-controlled)

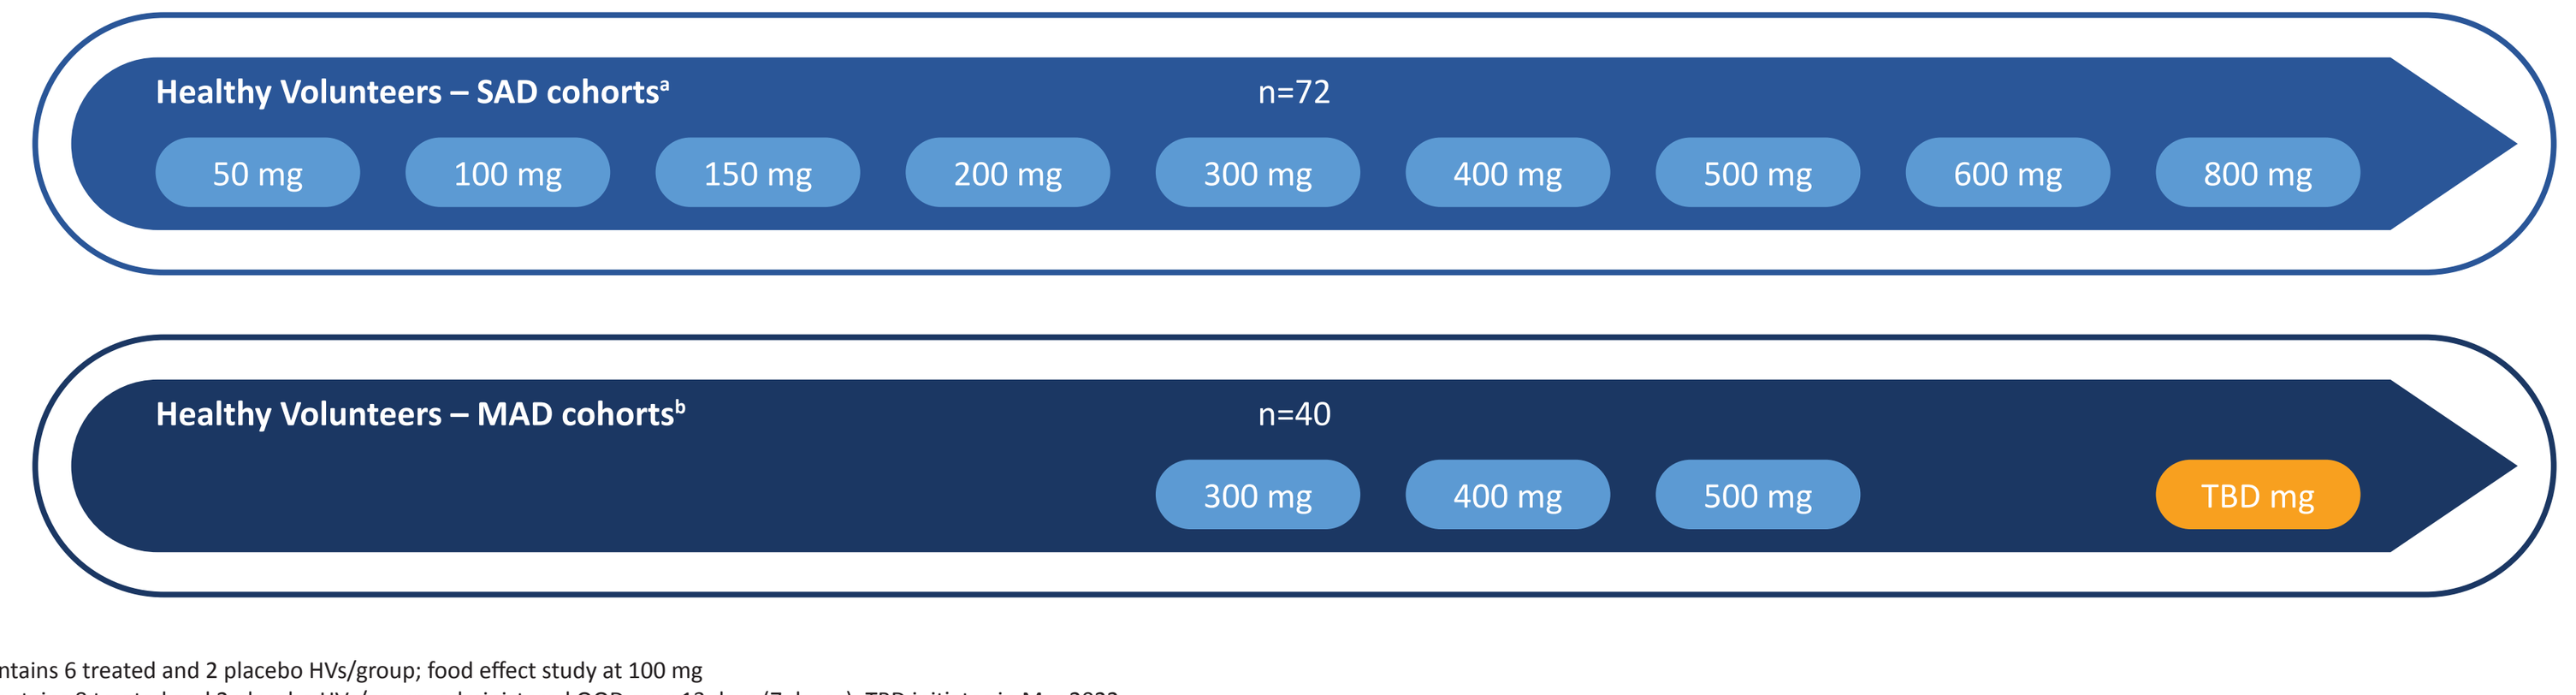
Healthy Volunteers – SAD cohorts[a]
n=72
50 mg
100 mg
150 mg
200 mg
300 mg
400 mg
500 mg
600 mg
800 mg
Healthy Volunteers – MAD cohorts[b]
n=40
300 mg
400 mg
500 mg
TBD mg

[a]: Each SAD cohort contains 6 treated and 2 placebo HVs/group; food effect study at 100 mg
[b]: Each MAD cohort contains 8 treated and 2 placebo HVs/group; administered QOD over 13 days (7 doses); TBD initiates in Mar 2022

Results

Favorable Safety Profile and Tolerability

Safety and adverse event (AE) data for 102 subjects across nine SAD and three MAD cohorts

- Most AEs are incidental and not dose related. No moderate or severe AEs; no serious AEs (SAEs)
- Most common drug-related AE is headache, which is seen in 11.8% of subjects
 - Mild (9.8%, n=10), Moderate (2.0%, n=2)
 - Transient in nature and resolved either without intervention or with use of OTC meds
 - These occur in both treated and placebo groups (n=10 vs n=20) with no dose dependence
- Elevated ALT noted in two subjects at both the 300 mg MAD and 400 mg MAD dose cohorts
 - Not seen in the 500 mg MAD cohort
 - No associated changes in AST, bilirubin or alkaline phosphatase
 - No stopping or dose modifications required, and ALT levels resolved to within normal range post-dosing
- Three mild to moderate fevers recorded at the 500 mg MAD dose

Well-behaved PK of PRX034 Following Oral Administration of PRTX007

Human pharmacokinetics (SAD cohorts 50-800 mg; MAD cohorts 300-500 mg PRTX007)

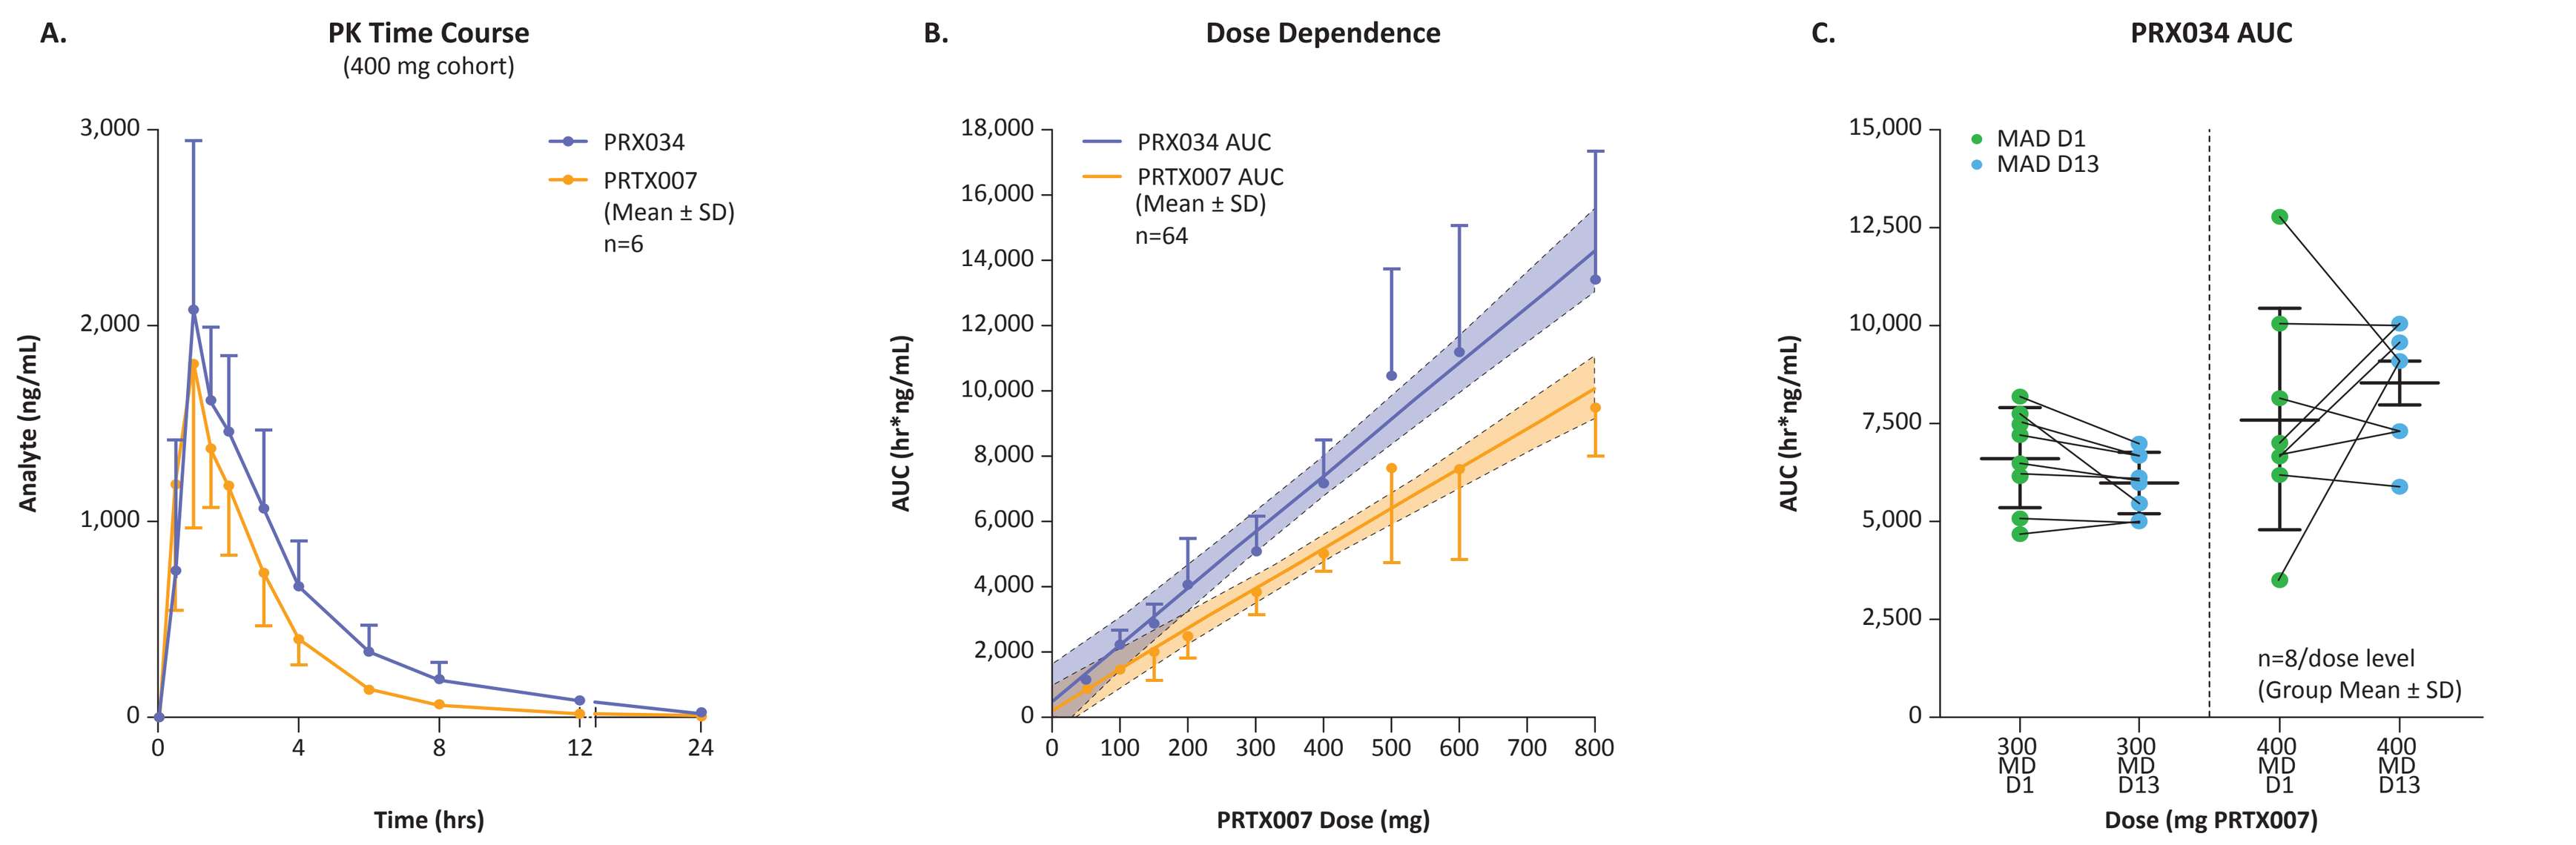
A.
PK Time Course
(400 mg cohort)
PRX034
PRTX007
(Mean ± SD)
n=6
Analyte (ng/mL)
Time (hrs)
B.
Dose Dependence
PRX034 AUC
PRTX007 AUC
(Mean ± SD)
n=64
AUC (hr*ng/mL)
PRTX007 Dose (mg)
C.
PRX034 AUC
MAD D1
MAD D13
n=8/dose level
(Group Mean ± SD)
Dose (mg PRTX007)

- Rapid absorption and conversion of prodrug PRTX007 to agonist PRX034 following oral administration
- Targeted short duration of pulsatile exposure to PRX034
 - Duration of systemic exposure to PRX034 at pharmacologically active levels is consistent with activation of innate immune response without counter-regulation

- Dose-proportional increase in exposure to prodrug and active agonist
- Agonist/Prodrug AUC~ 1.7 for all subjects

- Exposure unchanged between 1st (D1) and 7th (D13) doses

Induction of IFN-Stimulated Gene Products (ISG) and Other TLR7-Associated Cytokines Without NF-κB-Mediated Inflammatory Cytokines (IL-1β, IL-6, TNFα)

Heat map of selected transcripts demonstrates coordinated response following first dose

(data through 800 mg SAD cohort and through 500 mg MAD cohort)

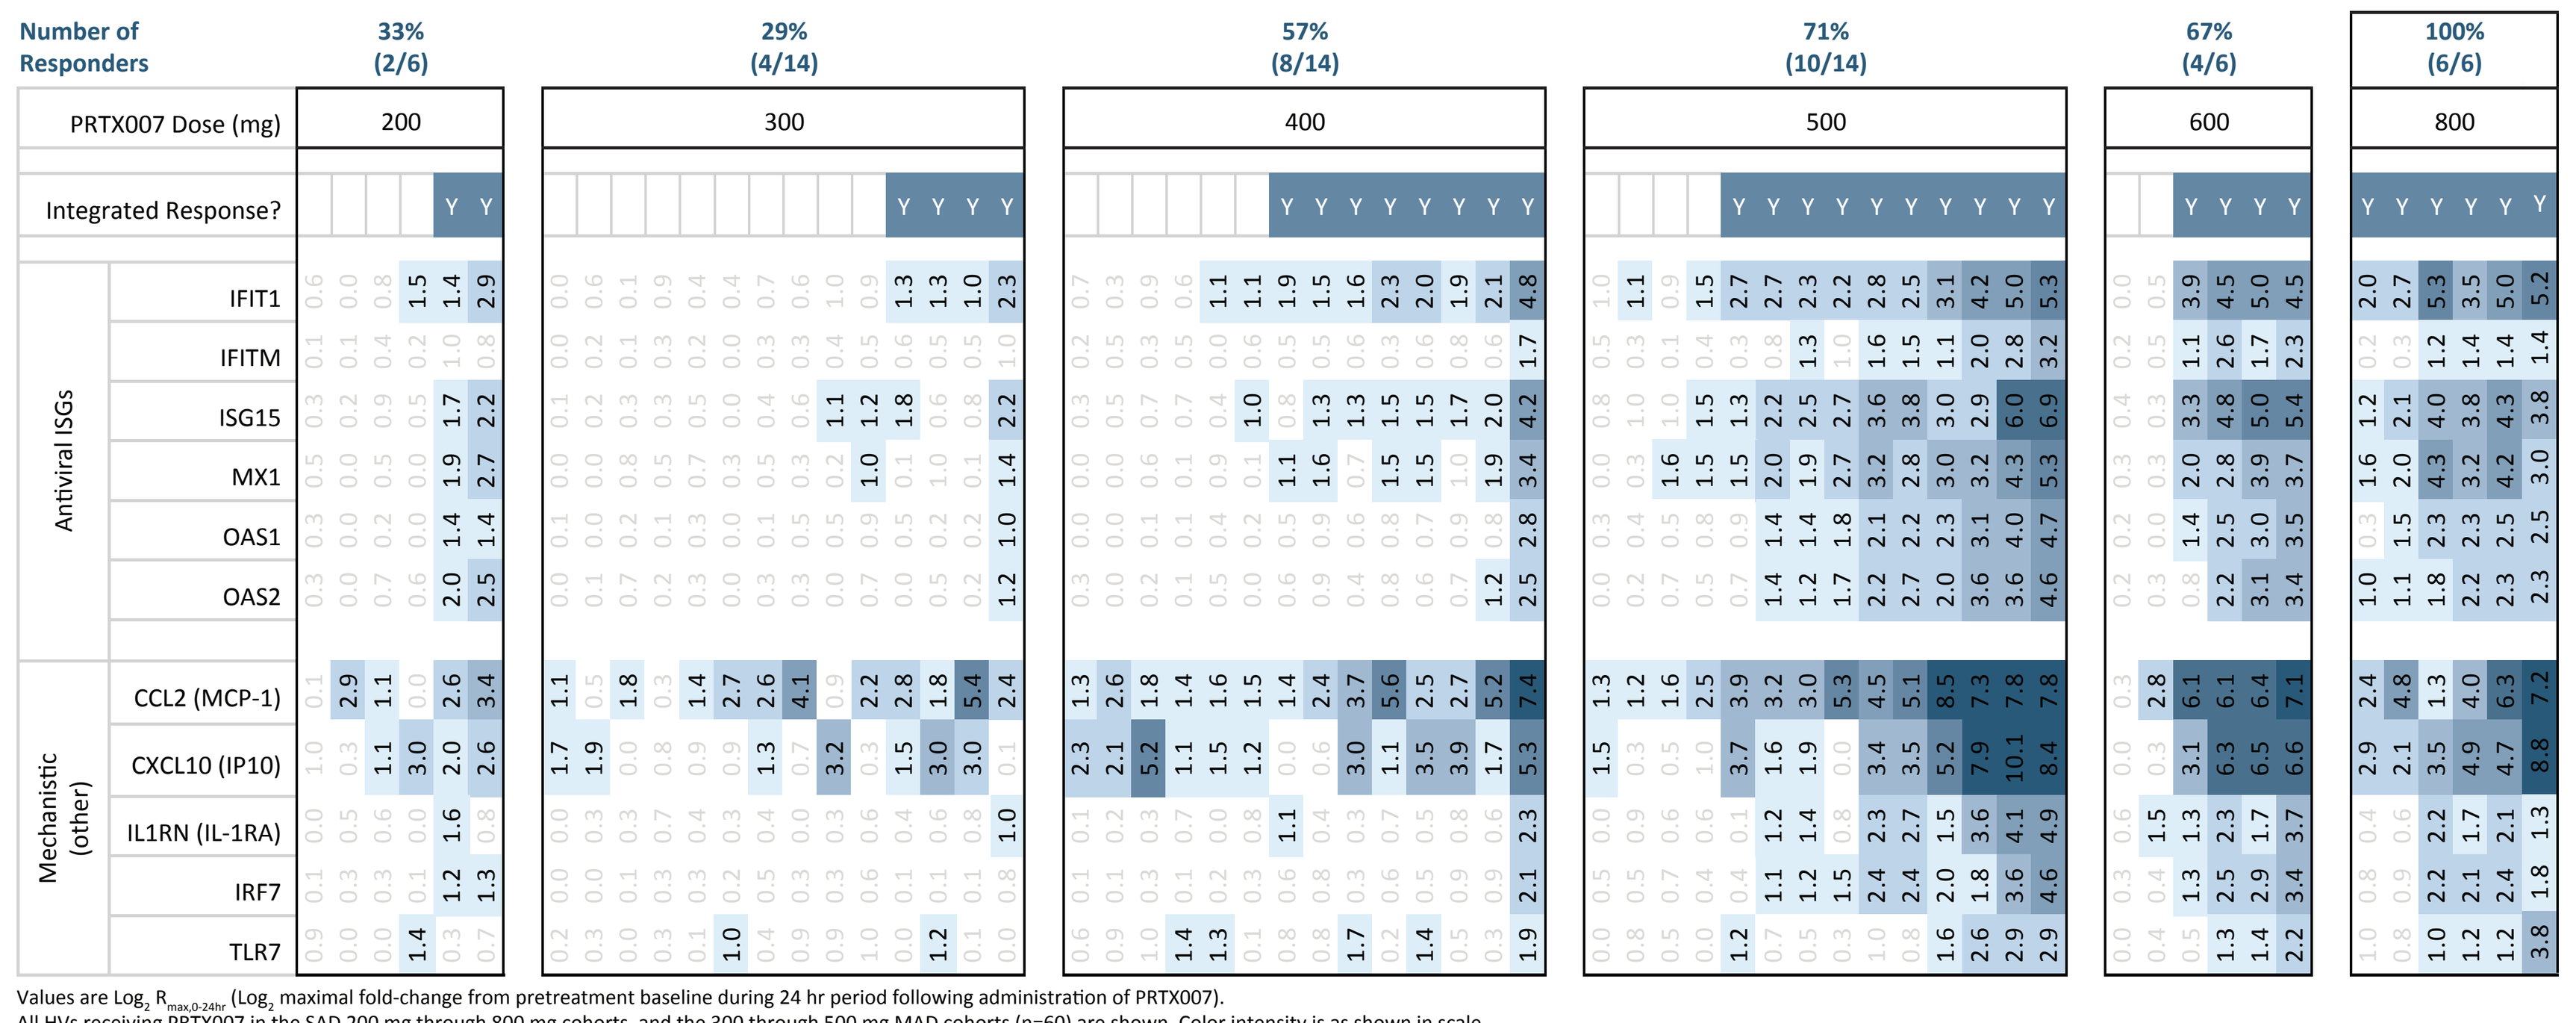
Number of Responders
33% (2/6)
29% (4/14)
57% (8/14)
71% (10/14)
67% (4/6)
100% (6/6)
PRTX007 Dose (mg)
200
300
400
500
600
800
Integrated Response?
Log2 Fold-Change
Antiviral ISGs
IFIT1
IFITM
ISG15
MX1
OAS1
OAS2
Mechanistic (other)
CCL2 (MCP-1)
CXCL10 (IP10)
IL1RN (IL-1RA)
IRF7
TLR7

Values are Log_2 $R_{max,0\text{-}24hr}$ (Log_2 maximal fold-change from pretreatment baseline during 24 hr period following administration of PRTX007).
All HVs receiving PRTX007 in the SAD 200 mg through 800 mg cohorts, and the 300 through 500 mg MAD cohorts (n=60) are shown. Color intensity is as shown in scale.
ISG=interferon-stimulated gene product.

Antiviral ISGs

IFIT1	interferon-induced protein with tetratricopeptide repeats 1
IFITM	interferon-induced transmembrane protein 1
ISG15	ISG15 ubiquitin-like modifier
MX1	MX dynamin like GTPase 1
OAS1	2'-5'-oligoadenylate synthetase 1
OAS2	2'-5'-oligoadenylate synthetase 2

Mechanistic (other)

CCL2 (MCP-1)	C-C motif chemokine ligand 2; MCP-1
CXCL10 (IP10)	C-X-C motif chemokine ligand 10; IP-10
IL1RN (IL-1RA)	interleukin 1 receptor antagonist; IL-1RA
IRF7	interferon regulatory factor 7
TLR7	toll-like receptor 7

Color Scale

Min	Max	Color
0.0	1.0	
1.0	2.0	
2.0	3.0	
3.0	4.0	
4.0	5.0	
5.0	6.0	
6.0	7.0	
7.0	10.5	

- Pharmacodynamic response increases with dose as measured by:
 - Proportion of HVs responding to first- or single-dose administration
 - Magnitude of induction within responders
- The integrated response rate at 800 mg in the SAD cohort is 100%
- These ISG transcript increases are observed in the absence of a corresponding increase in circulating interferons (data not shown)

Similar findings on a gene-by-gene basis (IP10 expression as example)

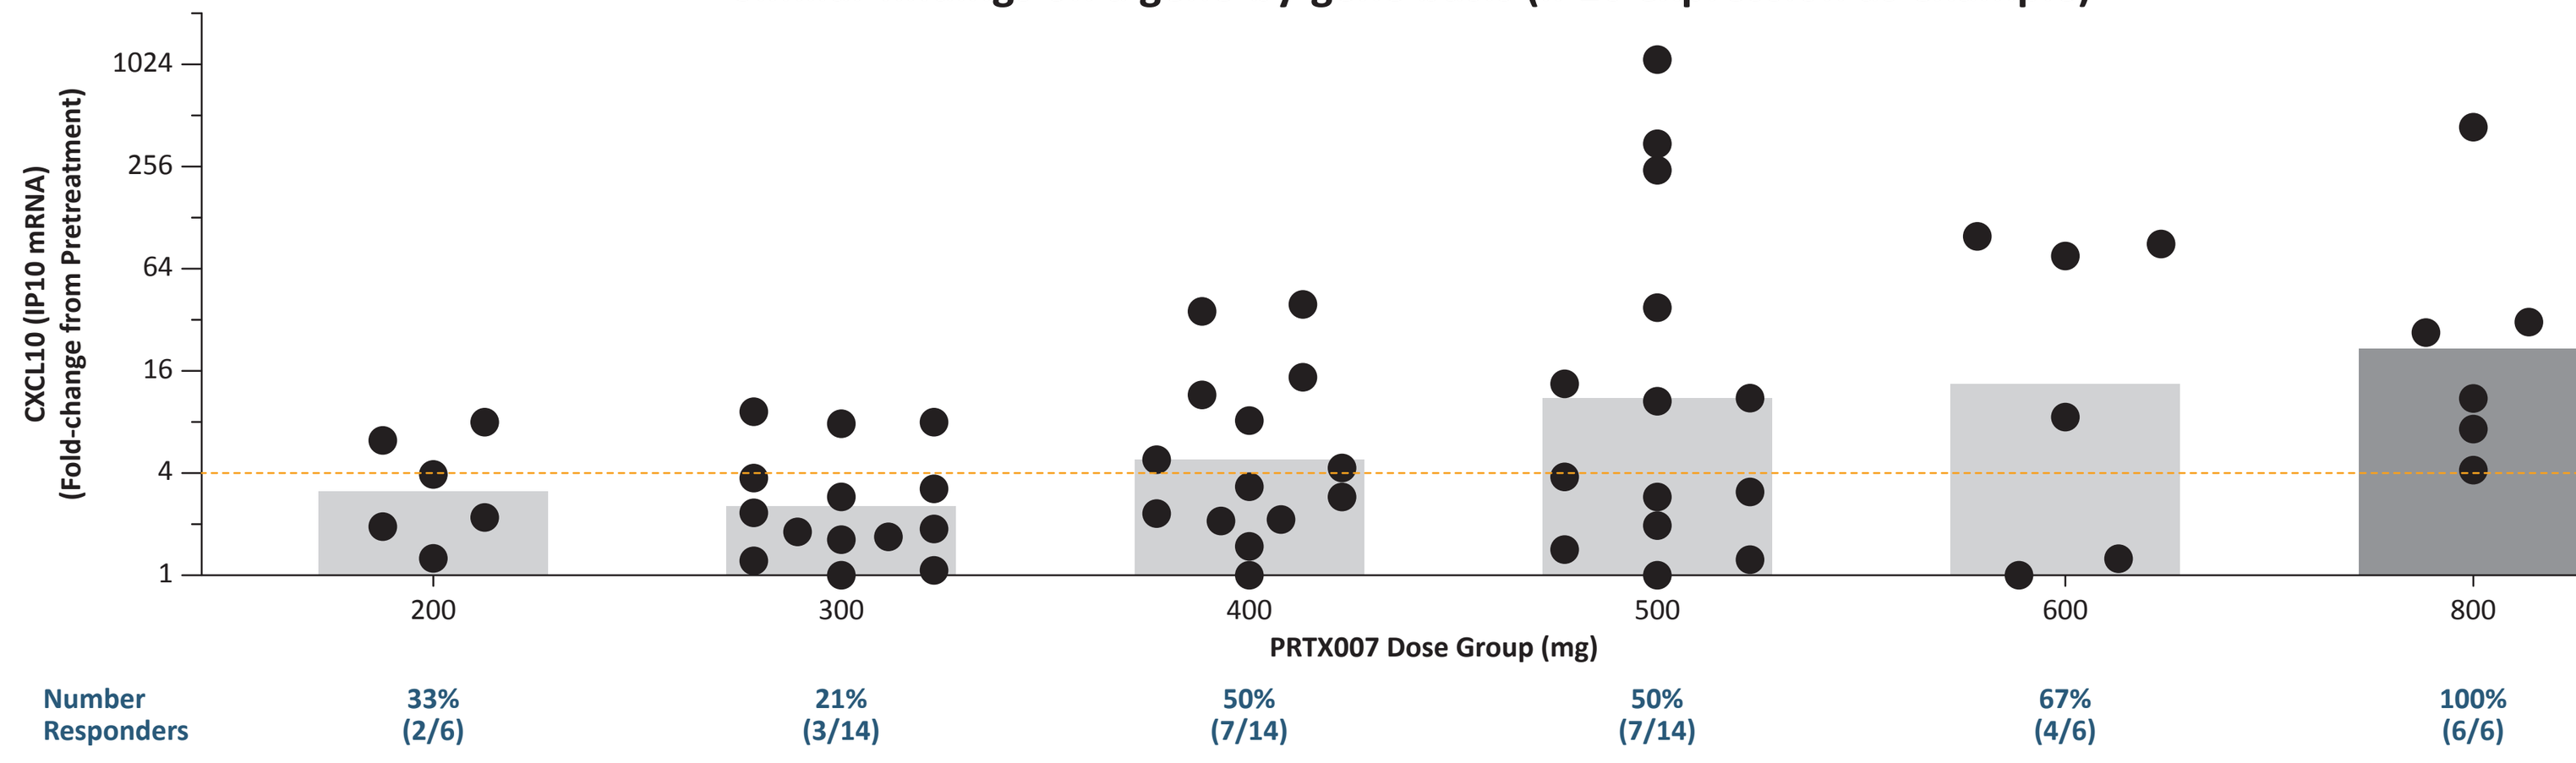
CXCL10 (IP10 mRNA) (Fold-change from Pretreatment)
PRTX007 Dose Group (mg)
Number Responders
33% (2/6)
21% (3/14)
50% (7/14)
50% (7/14)
67% (4/6)
100% (6/6)

*Gold dashed line represents threshold of response for this gene

IP-10 (CXCL10): Breadth of immune induction without induction of NF-κB pathway products

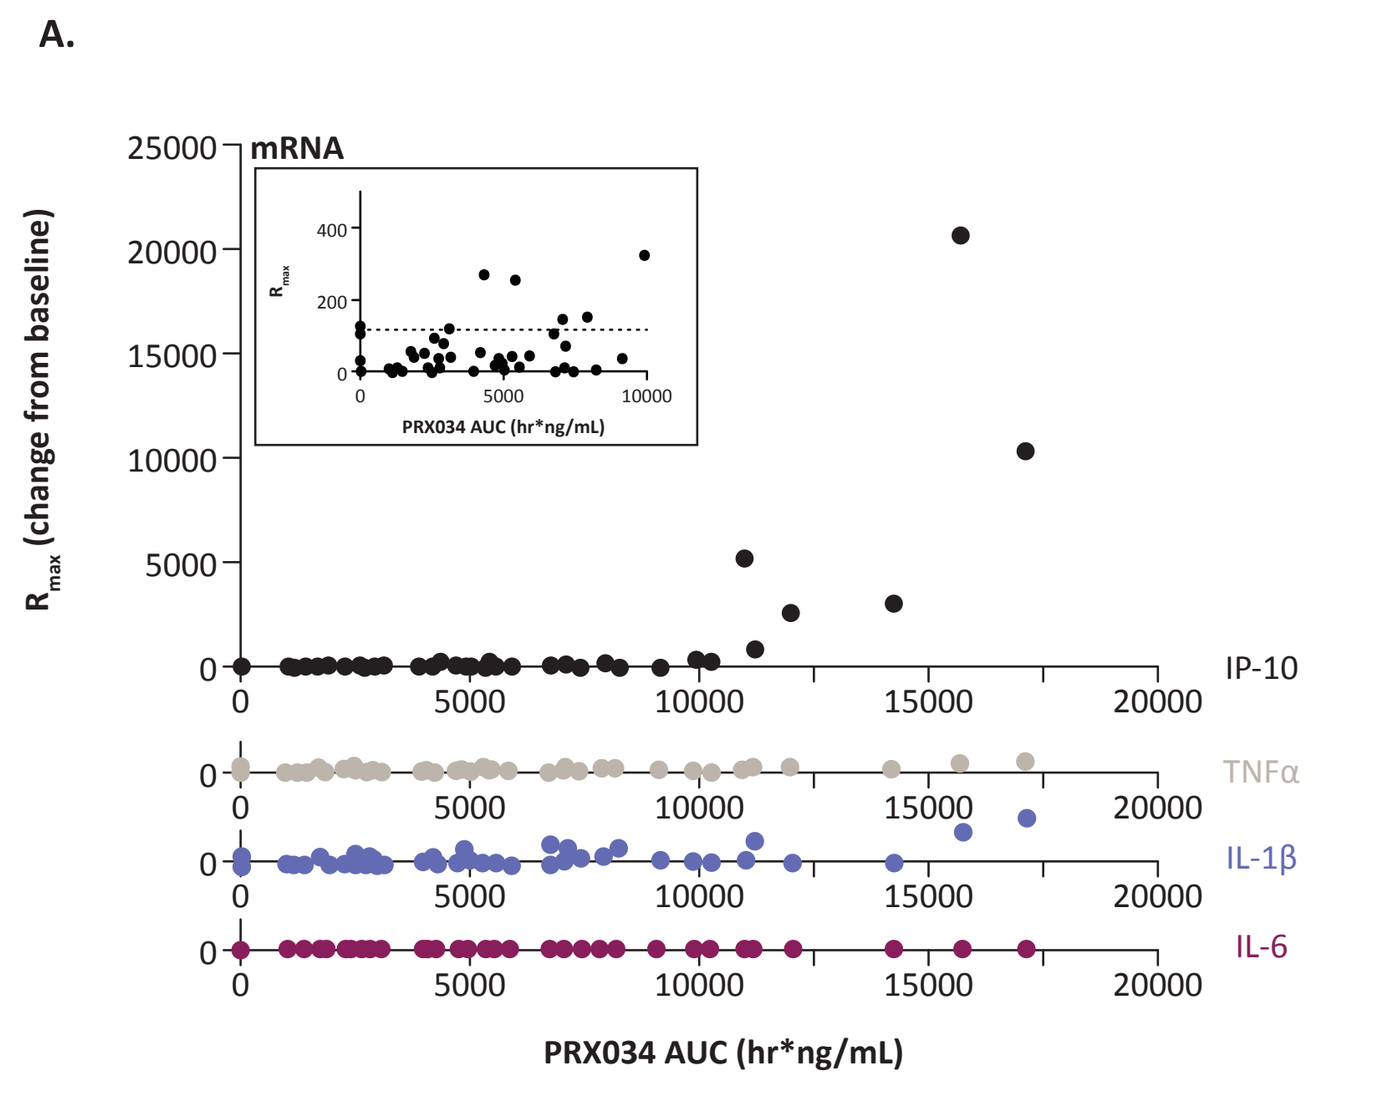
A.
mRNA
R_{max} (change from baseline)
IP-10
TNFα
IL-1β
IL-6
PRX034 AUC (hr*ng/mL)

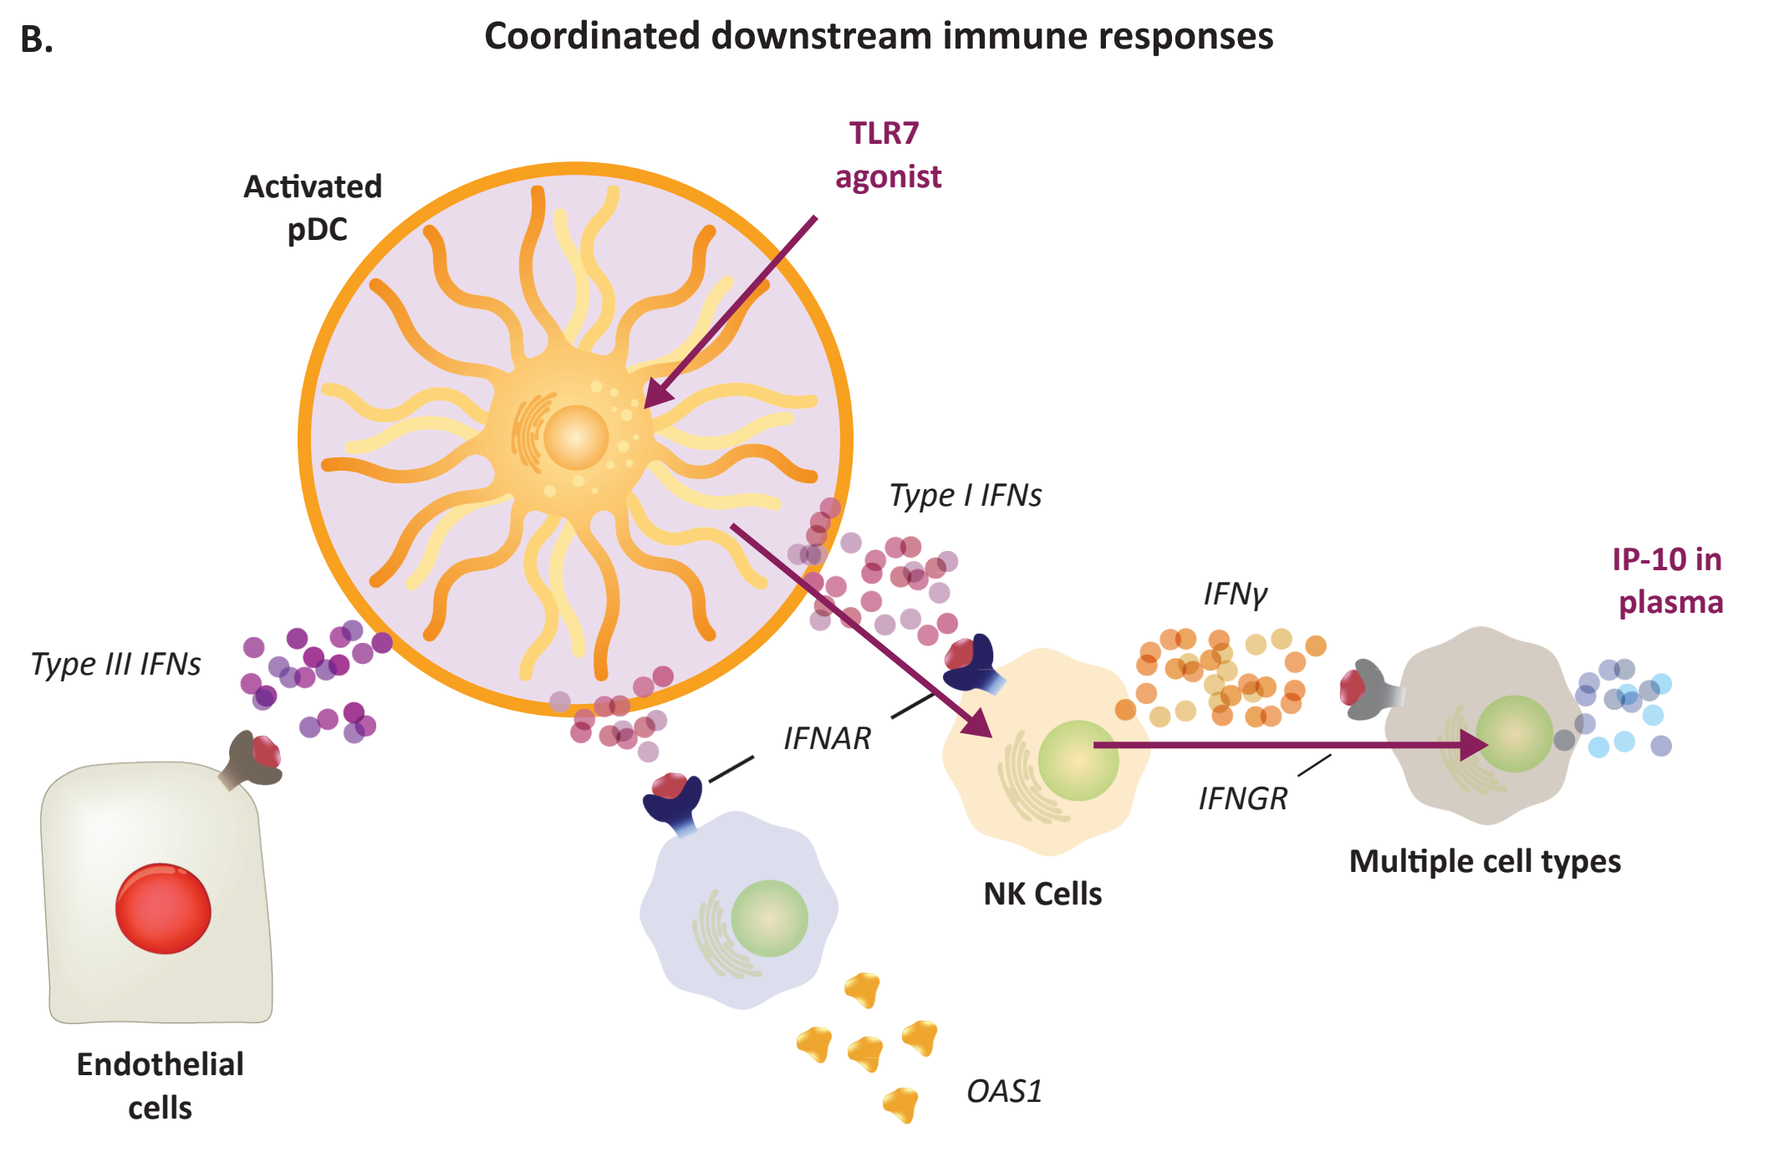
B.
Coordinated downstream immune responses
Activated pDC
TLR7 agonist
Type I IFNs
Type III IFNs
IFNAR
IFNγ
IFNGR
IP-10 in plasma
NK Cells
Multiple cell types
Endothelial cells
OAS1

- Increased IP-10 in plasma indicates action through extended cellular network
- Not accompanied by increased inflammatory factors

AUC=area under the plasma drug concentration-time curve; IFNAR=IFN alpha receptor; IFNGR=IFN gamma receptor; IP-10=interferon gamma-induced protein 10; OAS1=2'-5'-oligoadenylate synthetase 1; Rmax=maximum response.
A. mRNA expression of markers after a SAD of PRTX007 at 50 to 600 mg (n = 64; includes placebo-treated HVs). The inset shows that some statistically significant responses (values above dashed line) of lower magnitudes also occur at lower exposures to PRX034. The threshold for significance is set from the placebo group (n = 16) as the geometric mean (GM) of IP10 mRNA + 2*SD of the GM; values above this line represent true responders. **B.** Coordinated downstream immune response.

- Selected circulating markers induced by PRX034 exposure in 50 to 600 mg SAD in HVs
 - IP-10, IL-1RA, monocyte chemoattractant protein 1 (MCP1), and tumor necrosis factor–related apoptosis-inducing ligand (TRAIL) are expressed at high levels in plasma (data not shown for IL-1RA, MCP1, or TRAIL)
 - IP-10 protein levels and mRNA levels increase in response to drug exposure (**A**)
- Increased IP-10 in plasma indicates activation of multiple cell types downstream from activated pDCs (see cell types in, **B**)
- Inflammatory factor production (IL-6, TNFα, IL-1β) is not observed even in the face of this profound immune stimulation (**B**)

When PRTX007 is Administered Repeatedly on a QOD (Every Other Day) Dosing Schedule, the Magnitude of ISG Induction Increases Until a Steady State is Reached

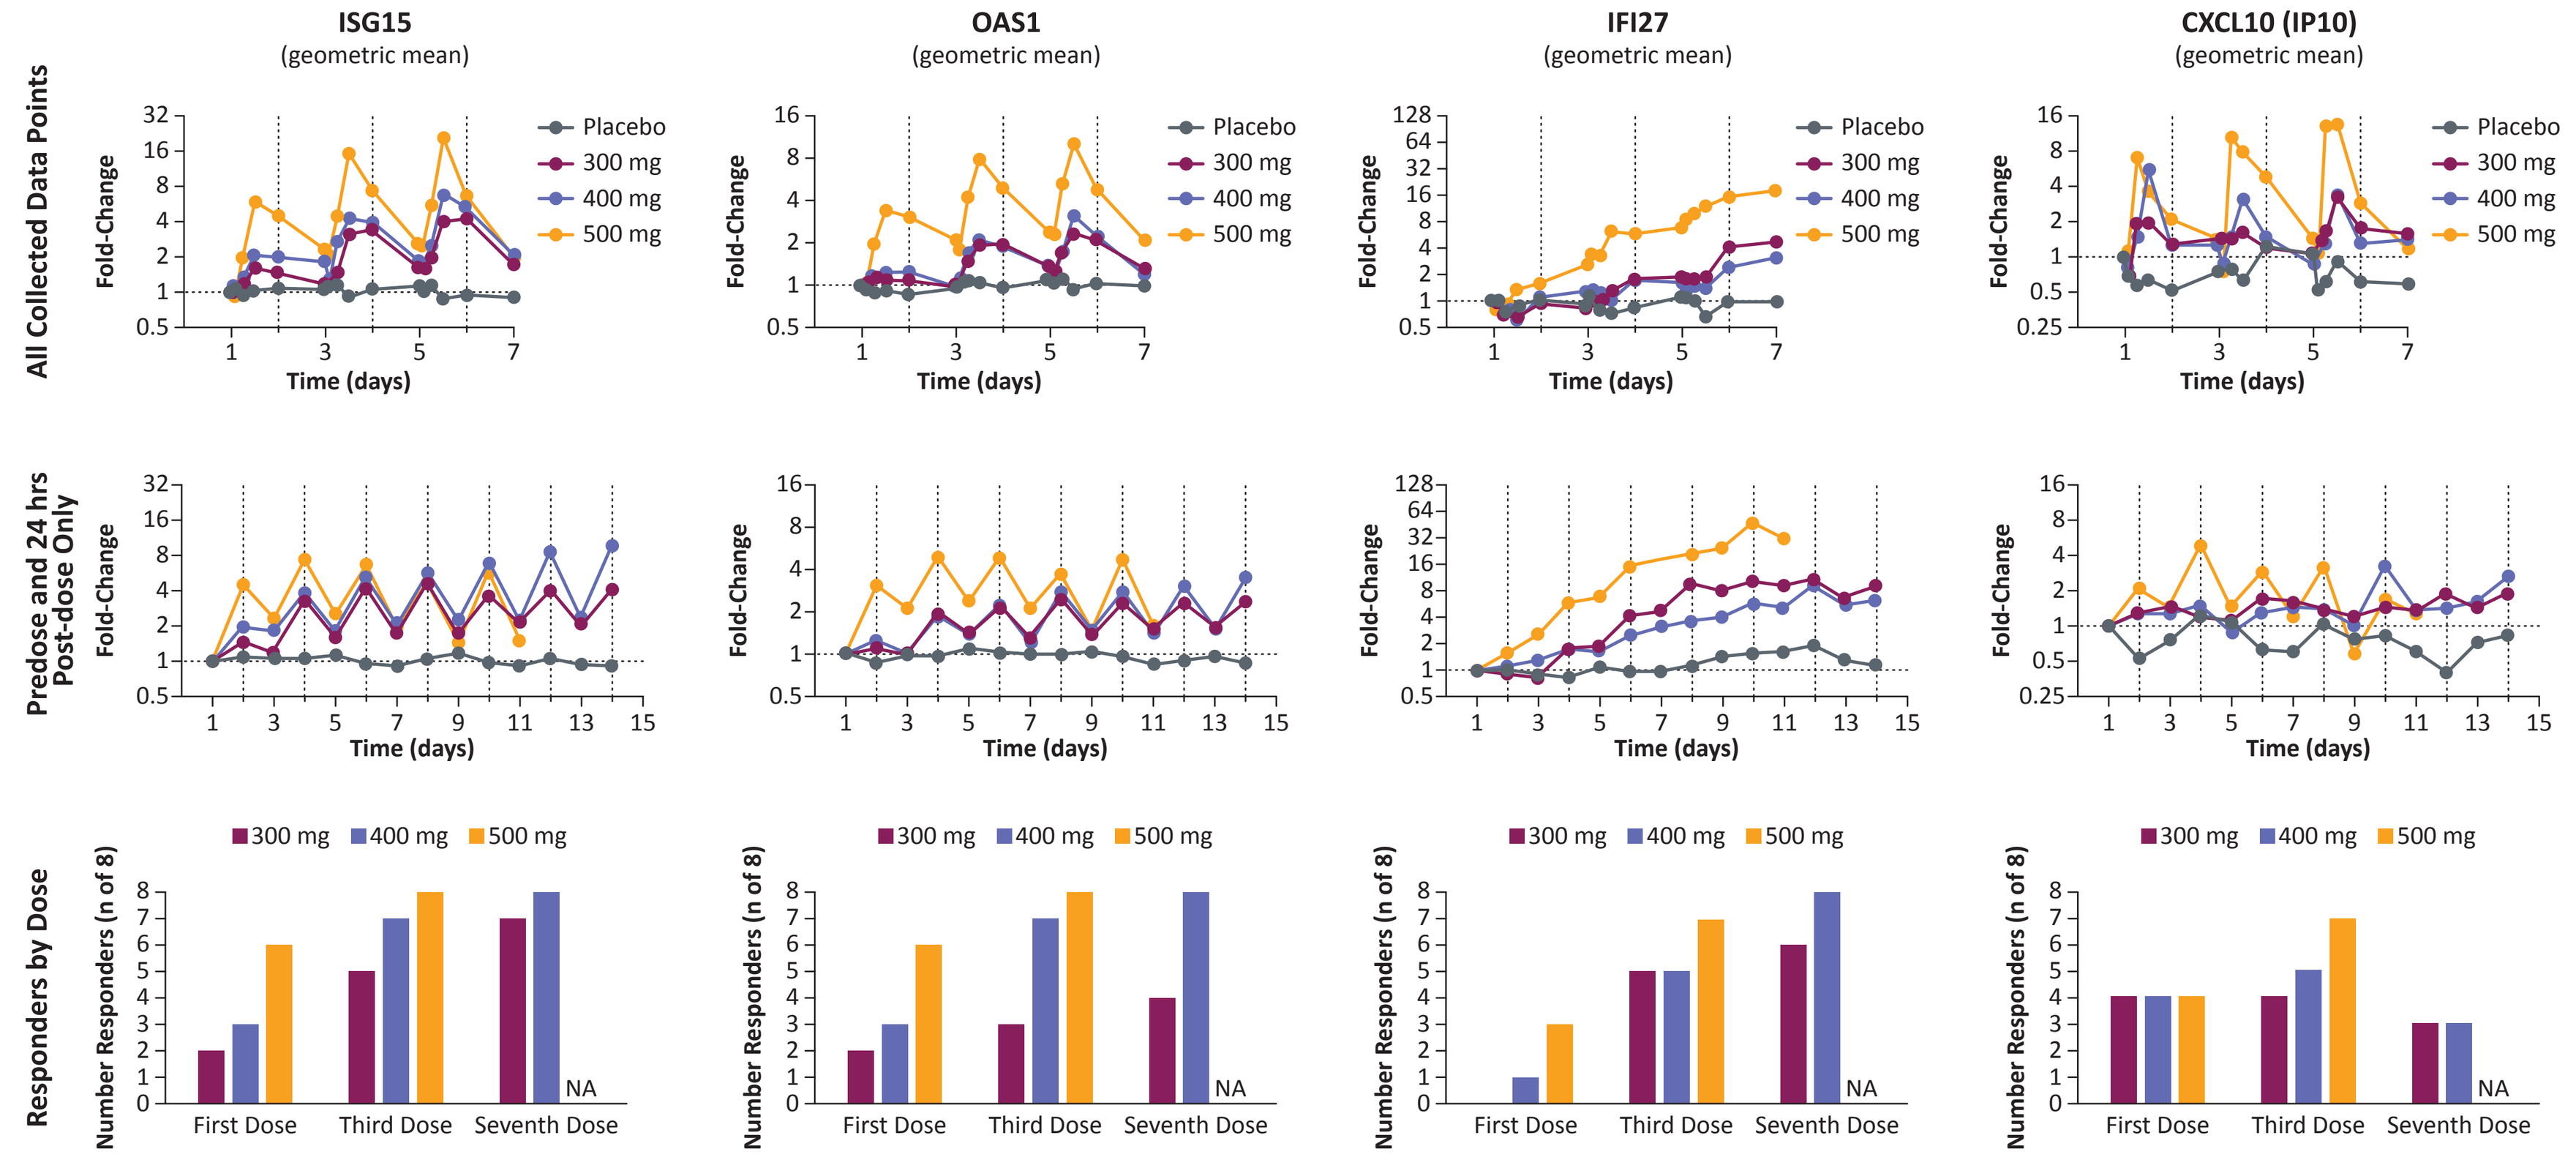
ISG15 (geometric mean)
OAS1 (geometric mean)
IFI27 (geometric mean)
CXCL10 (IP10) (geometric mean)
All Collected Data Points
Predose and 24 hrs Post-dose Only
Responders by Dose
Fold-Change
Time (days)
Placebo
300 mg
400 mg
500 mg
Number Responders (n of 8)
First Dose
Third Dose
Seventh Dose
NA

AUC = area under the plasma drug concentration-time curve.

Population pharmacodynamic responses to repeated doses at 300, 400 and 500 mg/dose are shown. Detailed daily kinetics are available following administration of the first three doses (Day 1-6, top row). Sparse kinetics are available on subsequent days (sparse kinetics depicted for D1-14 for 300 and 400 mg dose groups and D1-10 for 500 mg dose group, middle row).

- No induction is observed for classic inflammatory factors IL-6, TNFα and IL-1β (data not shown)
- In general, the magnitude of response increases with each dose until a steady-state is achieved (typically by dose 3-4, see middle row)
 - For ISG15, OAS1 and CXCL10 (IP-10), expression peaks at 12-24 hours followed by a decline until the next dose of PRTX007 is administered (top row)
 - For IFI27 (interferon alpha inducible protein 27), accumulation is observed throughout the entire time course until steady state is achieved (top and middle rows)
- For the classical ISGs (ISG15, OAS1, IFI27), the number of responders (HVs achieving a defined threshold for increase in mRNA level versus baseline) increases with repeated dosing until steady state is achieved (bottom row)
 - For CXCL10 (IP-10) which is not directly linked to type I interferons, this pattern is less obvious (bottom row)

Conclusions & Discussion

- At the interim analysis, PRTX007 demonstrated a favorable safety profile when administered orally to all 9 SAD and 3 MAD cohorts tested
 - No SAEs; lack of AEs historically associated with TLR7 agonists
- Stable systemic immune induction without evidence of counter-regulation is achieved upon QOD dosing
 - Induction of ISGs without significant increases in circulating IFNs
 - No increase in expression or circulating levels of proinflammatory cytokines (eg, TNFα, IL-6, IL-1β)
- Both the clinical characteristics and unique pattern of immune induction by PRTX007 support its use in combination with immune checkpoint inhibitors (ICPIs)
 - Local antitumor activity of intratumorally administered pDC-activating agents in combination with systemic ICPIs is well recognized but overall clinical benefit has been limited
 - We believe the proposed combinations will increase therapeutic benefit in cancer immunotherapy by maintaining sustained, systemic immune pressure involving both innate and adaptive immunity

Reference

1. Appleman JA, et al. Abstract 582: Selection of a novel toll-like receptor 7 (TLR7) agonist PRX034 for immunotherapy of cancer. *Cancer Res*. 2020; 80 (16S): 582.

Disclosures & Acknowledgements

This study was funded by Primmune Therapeutics, Inc.
Medical writing services were provided by SCIENT Healthcare Communications and funded by Primmune Therapeutics, Inc., (Carlsbad, CA).

Presented at AACR Annual Meeting 2022 | April 8–13, 2022 | New Orleans, LA