260

Antiviral Activity of the Clinical Stage TLR7 Agonist Prodrug PRTX007 in a Murine Respiratory Syncytial Virus Animal Model

James R. Appleman[1], Stephen E. Webber[1], Haniah Abdullah[2], Pia Thommes[2]

[1]Primmune Therapeutics Inc., Carlsbad, CA, USA, [2]Evotec (UK), Cheshire, United Kingdom

***Presenting Author:** Dr. James Appleman, PhD*

Background

It is well known that respiratory syncytial virus (RSV) is a major pathogen in young children; however, there is rising recognition that RSV poses a risk to older adults.[1,2] Each year ~177,000 older adults are hospitalized in the US due to RSV infection.[1] Among older adults (≥50 years) hospitalized with RSV in the US, the mortality rate may be 6% to 8%.[2] Current treatment approaches for RSV are focused on infants and are prophylactic in nature.[3] There are no approved vaccines for RSV in children or older adults.[3]

In addition to the disease burden that results from cell death caused by viral infection in respiratory viral diseases, a significant burden also results from an exacerbated inflammatory response that extends beyond the duration of the viral infection itself. Therefore, two key parts for control of infection include acute control of viral replication and generation of a response that provides protection against future infections. Our treatment objective is to control viral replication early and avoid inflammatory pathogenesis. Success will be evaluated in RSV murine models. Activators of innate immunity have not been evaluated in this setting due to concerns about exacerbating inflammatory pathogenesis. In murine models, RSV infections are self-limiting due to an increase and accumulation of plasmacytoid dendritic cells (pDCs) in lung tissue[4]; furthermore, RSV infection induces activation of pDCs and secretion of interferon (IFN)α, limiting viral replication.[5] These data support a model where pDC activation and IFN induction play an important role in the treatment of human RSV. Toll-like receptor (TLR)7 agonists known to induce IFN through pDCS, however, would need to avoid the exaggerated hyperimmune pathologies associated with concurrent induction of the NF-kB pathway products such as TNFα, IL-1β, and IL-6.

PRTX007 is an oral prodrug of PRX034, a novel TLR7 agonist that uniquely elicits IFN-mediated immune induction from pDCs without inducing inflammation via the NF-κB pathway (**Figure 1**).[6] Accordingly, pharmacologic response to PRTX007 in healthy humans for the first time supports the use of a TLR7 agonist in treatment of respiratory viral infections (**see Poster #143, at this Congress**).[6]

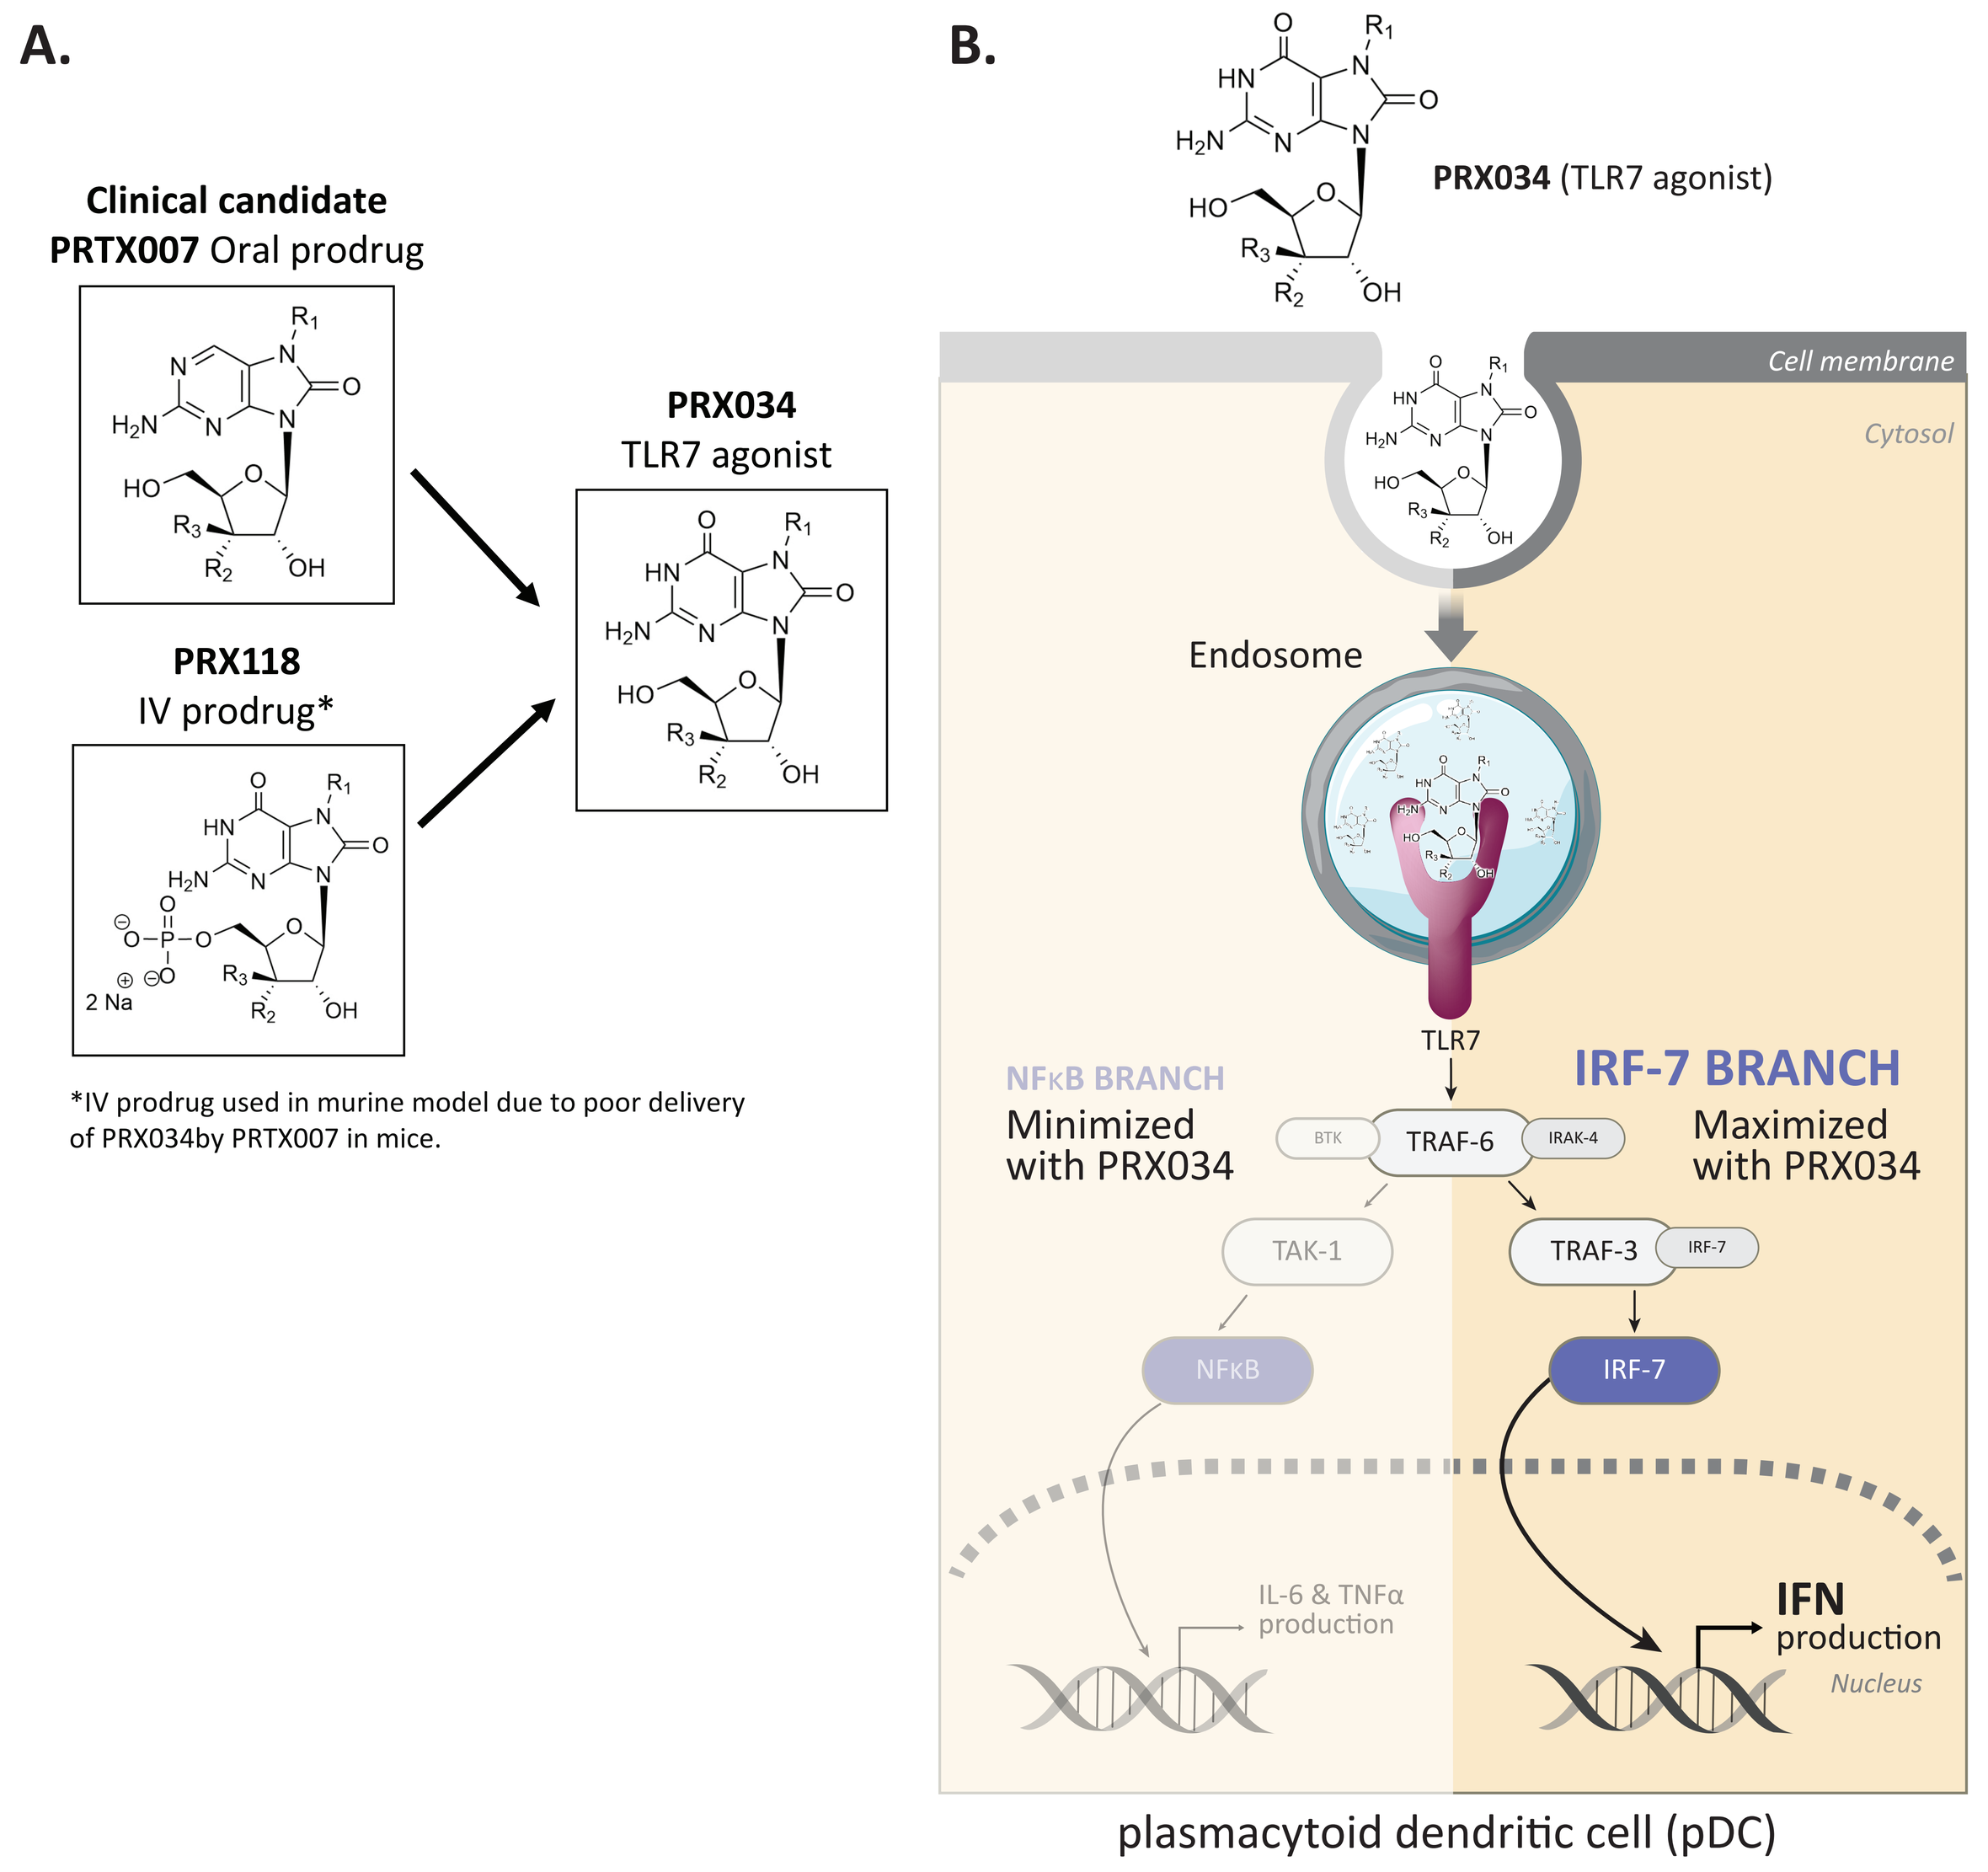
A.
Clinical candidate
PRTX007 Oral prodrug
PRX118
IV prodrug*
PRX034
TLR7 agonist
*IV prodrug used in murine model due to poor delivery of PRX034by PRTX007 in mice.
B.
PRX034 (TLR7 agonist)
Cell membrane
Cytosol
Endosome
TLR7
NFκB BRANCH
Minimized with PRX034
IRF-7 BRANCH
Maximized with PRX034
BTK
TRAF-6
IRAK-4
TAK-1
TRAF-3
IRF-7
NFκB
IRF-7
IL-6 & TNFα production
IFN production
Nucleus
plasmacytoid dendritic cell (pDC)

BTK=Bruton's tyrosine kinase; IL-6=interleukin 6; IFN=interferon; IRAK-4=interleukin-1 receptor-associated kinase 4; IRF-7=interferon regulatory factor 7; IV=intravenous; NF-κB=nuclear factor-κB; TAK1=TGF-β-activated kinase 1; TNFα= tumor necrosis factor alpha; TRAF-6=tumor necrosis factor receptor (TNFR)-associated factor 6.

Figure 1. PRX034 induction of innate immunity without production of proinflammatory factors.
A. Structures of PRTX007, PRX118, and PRX034. **B.** PRX034 binds to the host TLR7 receptor, triggering an intracellular signaling cascade within pDCs. PRX034 preferentially activates the IRF-7 branch, leading to robust IFN production with minimal NF-κB pathway activation.

Methods

- ***In vitro* viral challenge with conditioned media (CM)**. PRTX007's antiviral activity was assessed in a standard cellular model of viral infection using CM prepared by incubating human peripheral blood mononuclear cells (hPBMCs) with PRX034 and harvesting the supernatant. hPBMCs were isolated using standard procedure. Antiviral activity of CM, derived from PRX034-treated hPBMCs against RSV strain A2, was tested in relation to time of addition relative to infection. Cells were pre-treated with CM at 18, 8, 4 and 2 hours prior to infection or CM was added simultaneously with the virus (0 hour). Type I IFN (IFNα2a) was tested in parallel as a control treatment
- ***In vivo* murine RSV viral challenge**. For RSV murine model experiments, BALB/c mice received PRX118 (a very soluble form of the clinical candidate) administered by intravenous (IV) bolus (**Figure 2**). The time course evaluating antiviral activity builds upon conditions where antiviral activity was observed in the assessment of mechanistically related therapeutics (**Figure 2**, Top panel). Therefore, treatment was initiated 12 hours pre-infection for these experiments (**Figure 2**, Bottom panel)

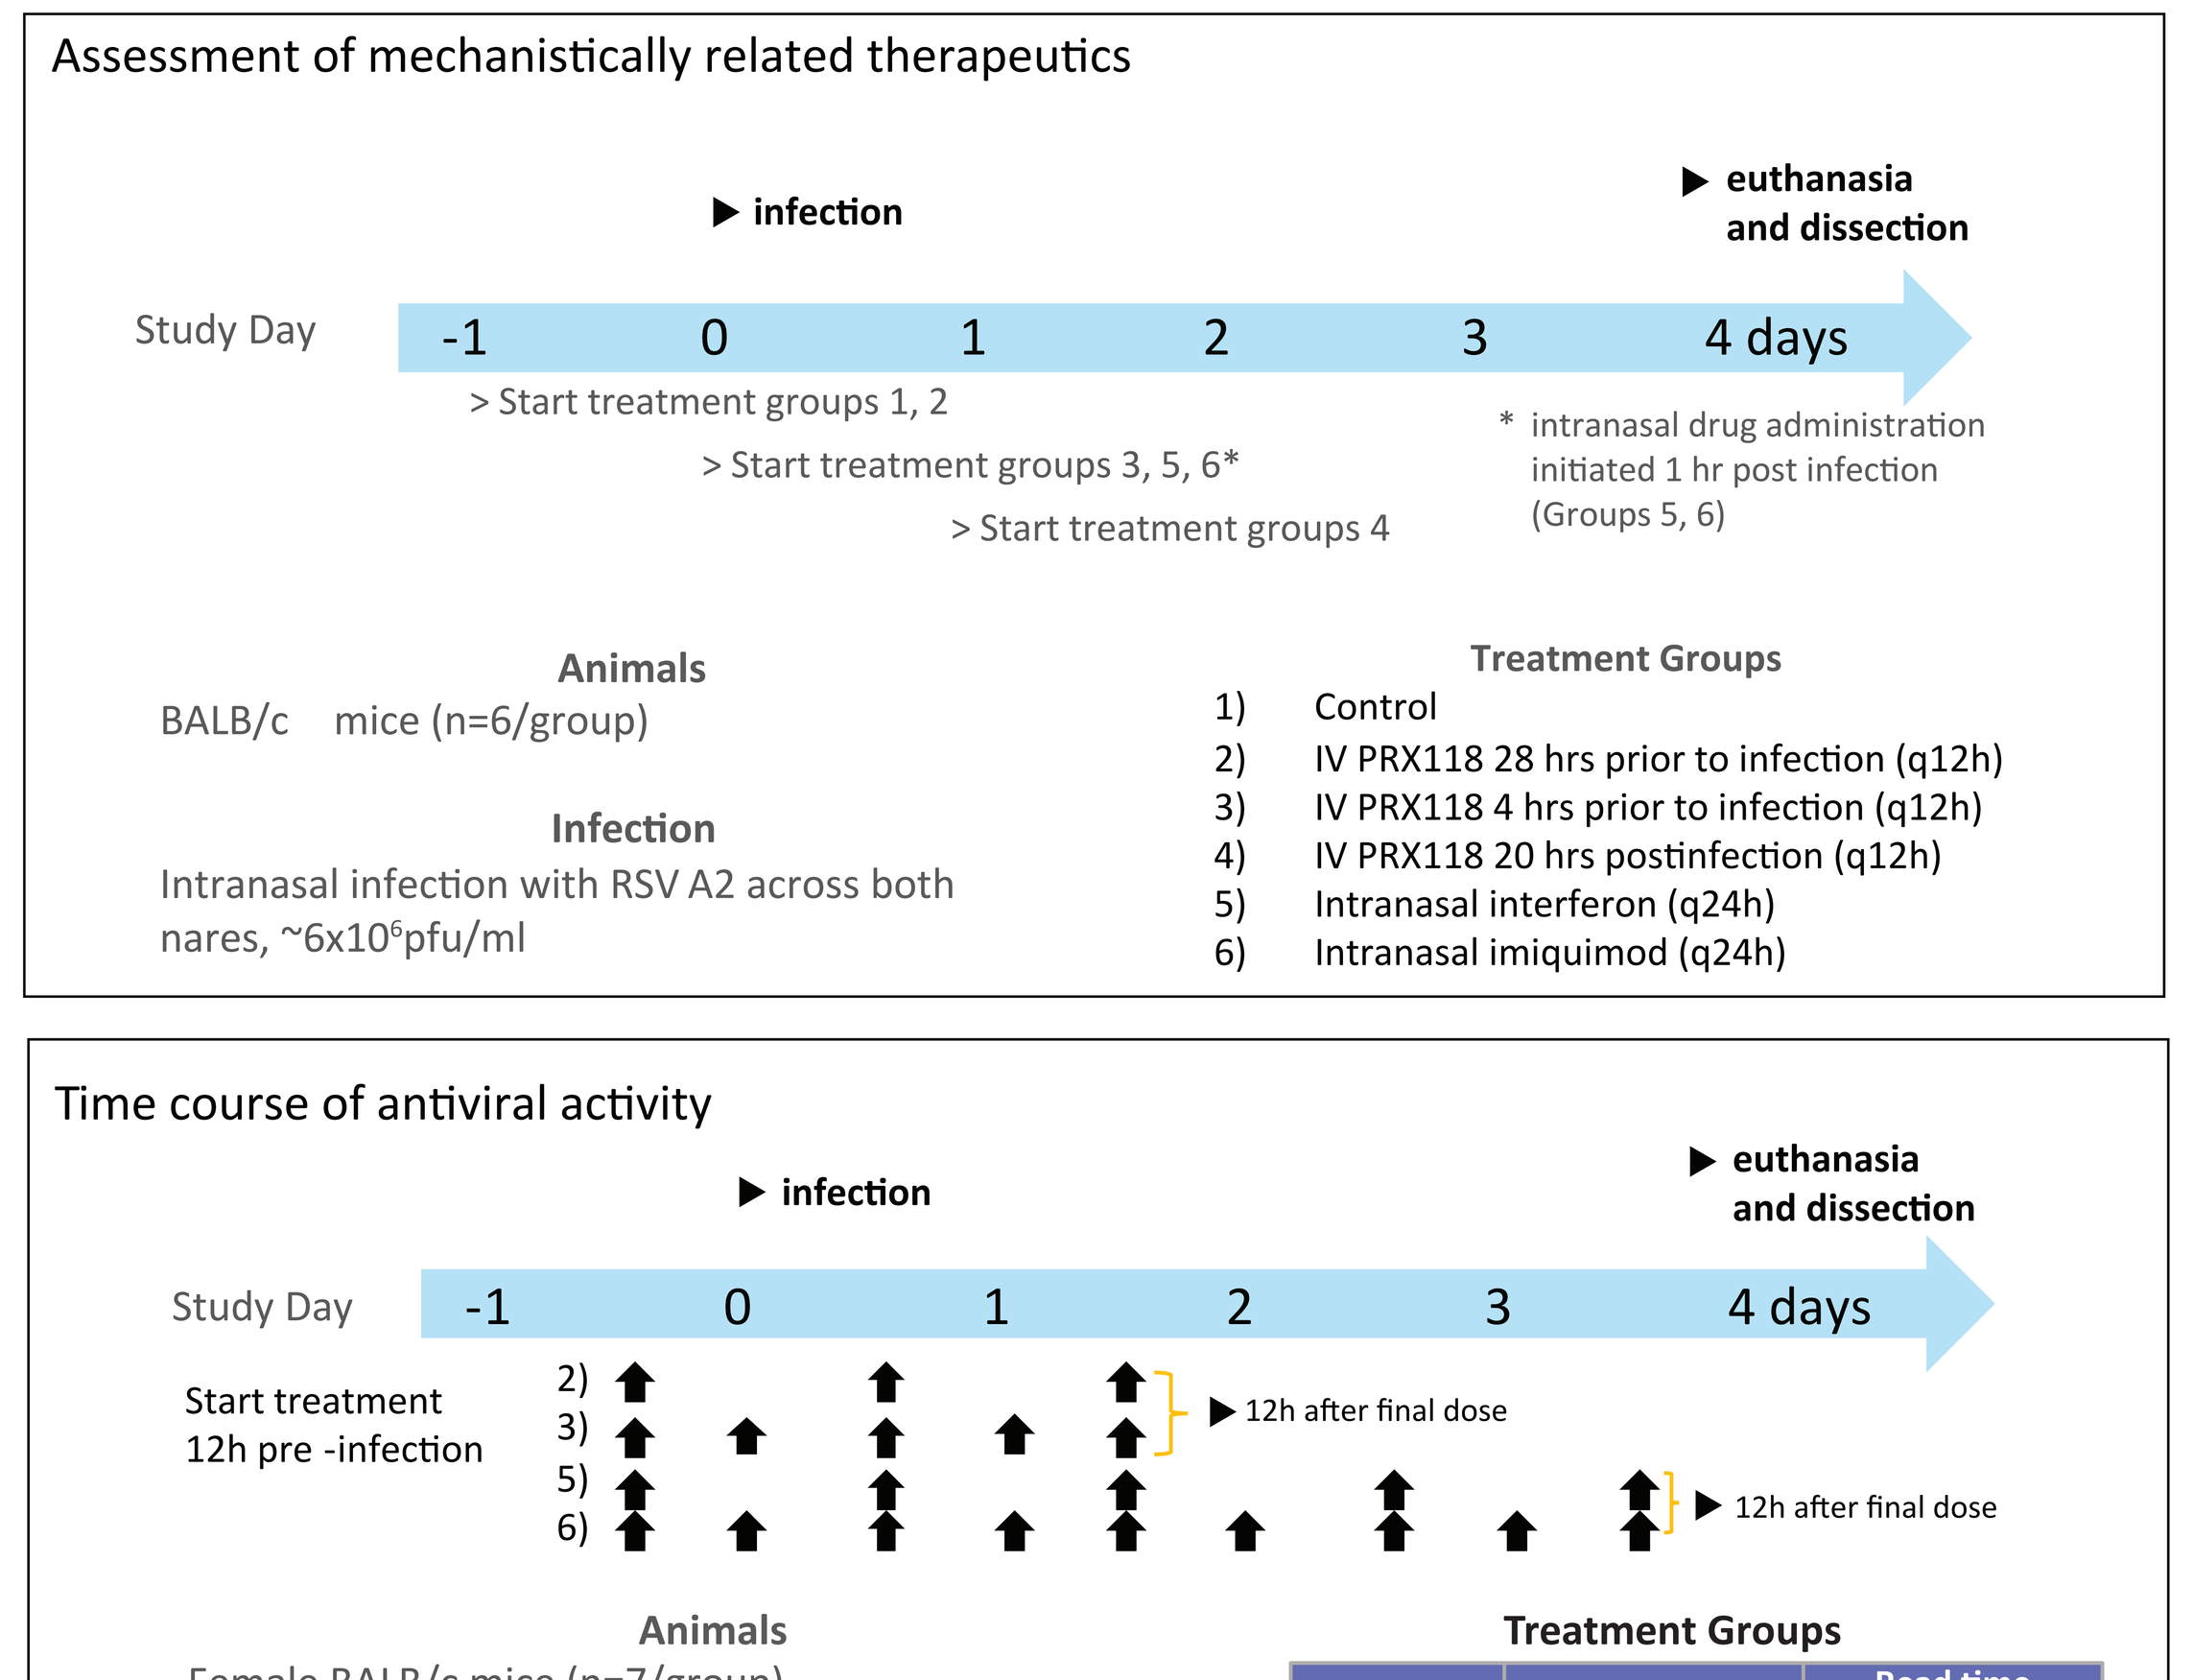
Assessment of mechanistically related therapeutics
▶ infection
▶ euthanasia and dissection
Study Day
-1
0
1
2
3
4 days
> Start treatment groups 1, 2
> Start treatment groups 3, 5, 6*
> Start treatment groups 4
* intranasal drug administration initiated 1 hr post infection (Groups 5, 6)
Animals
BALB/c mice (n=6/group)
Infection
Intranasal infection with RSV A2 across both nares, ~6x10^6pfu/ml
Treatment Groups
1) Control
2) IV PRX118 28 hrs prior to infection (q12h)
3) IV PRX118 4 hrs prior to infection (q12h)
4) IV PRX118 20 hrs postinfection (q12h)
5) Intranasal interferon (q24h)
6) Intranasal imiquimod (q24h)
Time course of antiviral activity
▶ infection
▶ euthanasia and dissection
Study Day
-1
0
1
2
3
4 days
Start treatment 12h pre -infection
2)
3)
5)
6)
▶ 12h after final dose
▶ 12h after final dose
Animals
Female BALB/c mice (n=7/group)
Test article
PRX118 (PRX034 prodrug) administered at 60 mg/kg by IV bolus, started 12 hours pre-infection groups
Infection
Intranasal infection with 50 µL RSV A2 across both nares, ~2x10^6 pfu/ml
Treatment Groups

Group	Treatment	Read time postinfection
1	Vehicle (control)	48h
2	PRX118 q24h	48h
3	PRX118 q12h	48h
4	Vehicle (control)	96h
5	PRX118 q24h	96h
6	PRX118 q12h	96h

h=hours; IV=intravenous; RSV A2=human respiratory syncytial virus strain A2; pfu=plaque-forming unit, q=every.

Figure 2. Study design for assessment of antiviral activity in mouse models for RSV infection.

Results

Antiviral Activity Demonstrated Against Non-Coronavirus Respiratory Viruses in vitro

	EC_{50} as IFNα2a Concentration in Assay (pg/mL)		
	CM IFNα2a	Exogenous IFNα2a	Exog. IFNα2a/CM
Influenza H1N1	19	54	2.9
RSV A2a	121	1950	16
Rhinovirus-14	57.3	1125	19.6

CM=conditioned media; Exog.=exogenous; EC_{50}= half maximal effective concentration; IFNα2= human interferon alpha-2; RSV A2= human respiratory syncytial virus strain A2.
[a]This is a prophylaxis.

Table. PRX034 *in vitro* viral challenge studies: Inhibition of viral replication following incubation with PRX034- CM with hPBMCs

- PRX034-CM inhibits RNA viruses *in vitro* (**Table**), including RSV, at a greater potency than what would be expected by treatment with IFNα2a alone (see ratio of potencies expressed as EC_{50} for exogenously added IFNα2a and CM based on its IFNα2a content; Exog. IFNα2a/CM in **Table**)
 - Most but not all activity is mediated through pDC-derived IFNs (IFNα blocking experiments, data not shown)

Pre-treatment With PRX034-CM Improves Antiviral Response

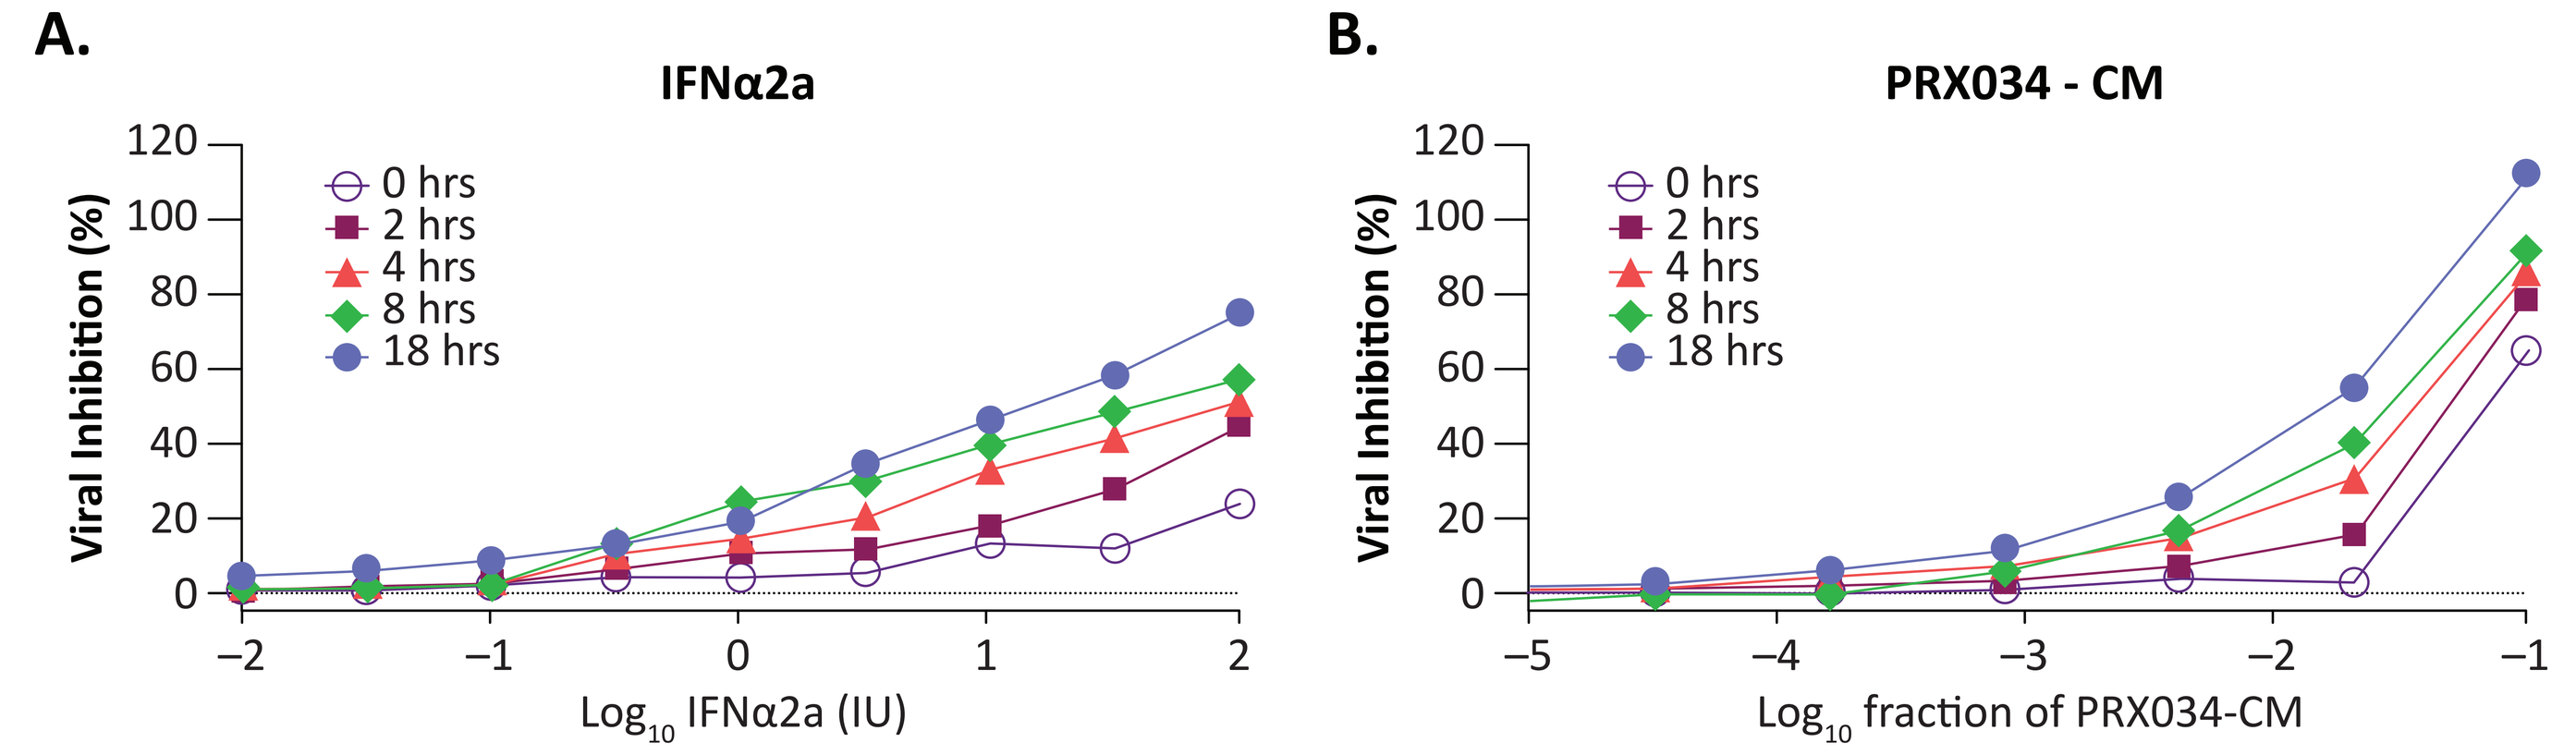
A.
IFNα2a
Viral Inhibition (%)
0 hrs
2 hrs
4 hrs
8 hrs
18 hrs
Log10 IFNα2a (IU)
B.
PRX034 - CM
Viral Inhibition (%)
0 hrs
2 hrs
4 hrs
8 hrs
18 hrs
Log10 fraction of PRX034-CM

CM=conditioned media; hr=hour; IFNα2a=interferon alpha 2a; IU=international units.

Figure 3. Antiviral activity of pre- and standard treatment conditions.
Cells were pre-treated with PRX034-CM at 18, 8, 4 and 2 hours prior to infection or CM was added simultaneously with the virus (0 hour, standard treatment). Type I IFN (IFNα2a) was tested in parallel as a control treatment. Following 72 hours postinfection, the cells' metabolic activity was measured by Viral ToxGlo™. Viral inhibition was determined relative to cell and virus controls. Each symbol represents an average of two intra-assay replicate data. **A.** IFNα2a control. **B.** Antiviral activity of CM in pre- and standard treatment conditions. PRX034-CM was diluted at 1:10, 1:50, 1:250 and 1:1250.

- Pre-treatment increases antiviral activity in a time-dependent manner for both the IFNα2a control (as expected; **Figure 3A**) and for PRX034-CM (**Figure 3B**)
- PRX034 alone has no effect on viral protection in the absence of immune cells (data not shown)
 - There is no direct antiviral activity intrinsic to PRX034
 - Since HEp-2 cells do not contain TLR7, no endogenous IFN is produced

Treatment With PRX034-CM Affects Cytopathic Effect Caused by RSV as Well as Viral Progeny Production

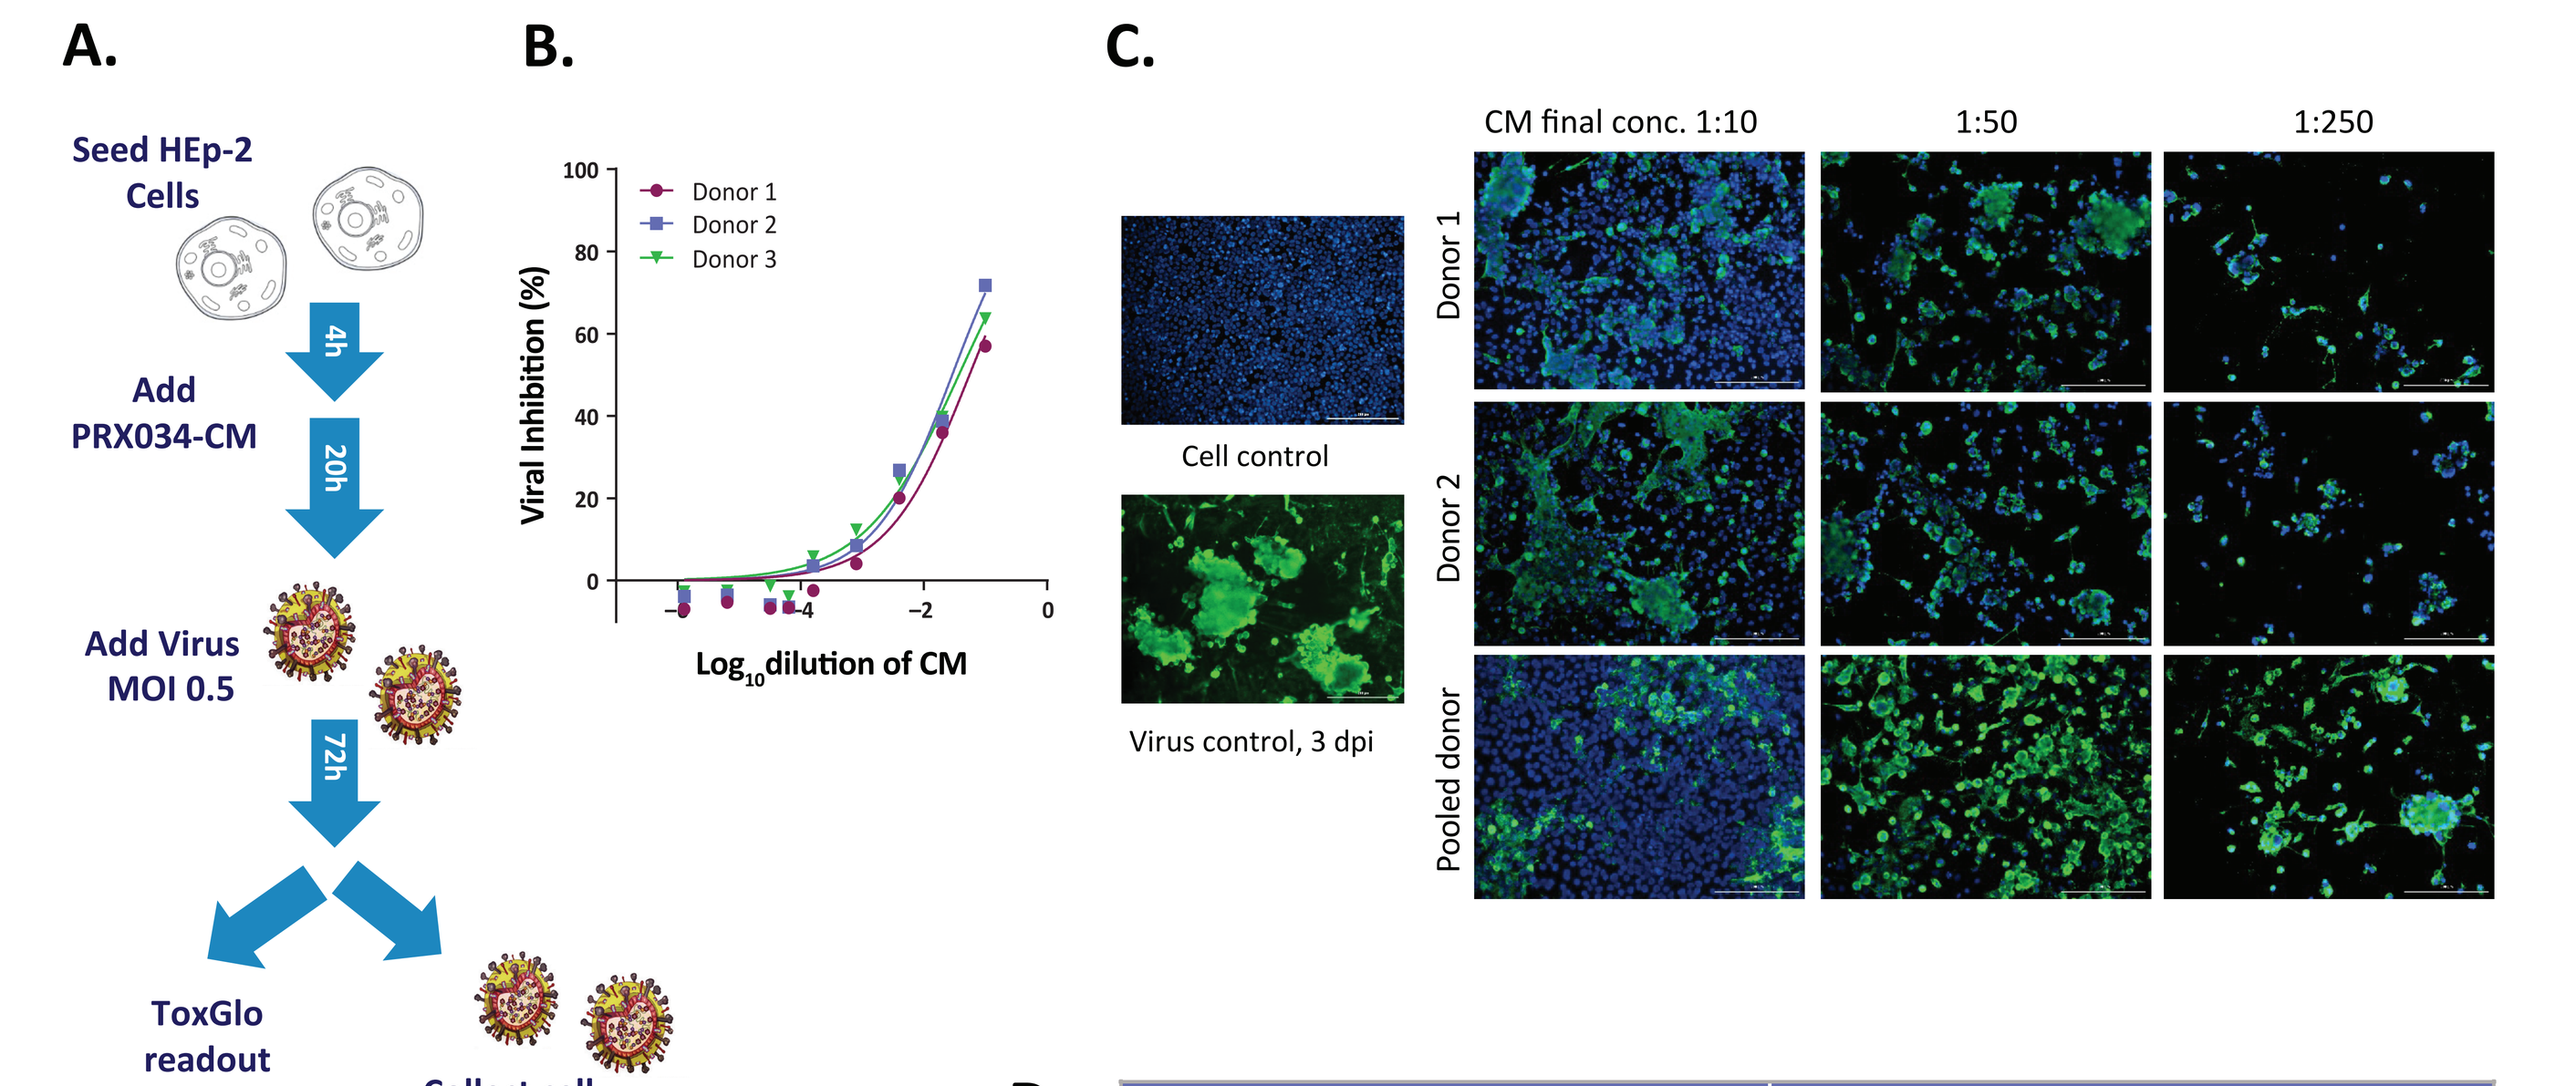
A.
Seed HEp-2 Cells
4h
Add PRX034-CM
20h
Add Virus MOI 0.5
72h
ToxGlo readout
Collect cell medium
Add to fresh HEp2-cells to perform plaque assay
B.
Donor 1
Donor 2
Donor 3
Viral Inhibition (%)
Log10dilution of CM
C.
CM final conc. 1:10
1:50
1:250
Cell control
Virus control, 3 dpi
Donor 1
Donor 2
Pooled donor
D.

Source of PRX034-Treated CM	EC_{50}(fold-dilution PRX034-CM)	
	Parent*	Progeny[†]
Donor 1 CM	18.2	1263
Donor 2 CM	34.6	259
Pooled Donor CM	27.0	6120

*ToxGlo readout; [†]Plaque assay

CM=conditioned media; conc.=concentration; dpi=days postinfection; EC_{50}=half maximal concentration; HEp-2=human epithelial; hpi=hours post-infection; MOI=multiplicity of infection; RSV=respiratory syncytial virus; RSV-F=respiratory syncytial virus-fusion protein.

Figure 4. Pre-treated CM inhibited RSV infection at 72 hours postinfection (hpi).
A. Experimental design. HEp-2 cells were infected with RSV following pre-treatment with CM derived from PRX034-treated hPBMCs at 18 hours relative to RSV infection in three 96-well plates. **B.** Parental viral inhibition. At 72 hpi the cells' metabolic activity was measured by Viral ToxGlo in two of the plates and viral inhibition was determined relative to cell and virus controls. **C.** Plaque assay of progeny. Concurrently the cell medium was collected from the third plate and transferred to fresh HEp-2 and incubated for further 72 hours for plaque assay. Viral protein was stained by rabbit anti-human RSV-F monoclonal antibody and detected with anti-rabbit IgG, Alexa Fluor 488 (green labeled). Cell nuclei were stained by DAPI (blue). Magnification x10. Size bars=200 microns **D.** Calculated EC_{50} derived from ToxGlo and plaque assay presented in the table. Under these conditions, maximal observed inhibition in the ToxGlo assay is ~70% for the parental progeny and is a limitation of the amount of CM that can be added.

- RSV viral progeny is reduced after PRX034-CM treatment
- The plaque assay result (direct measurement of virus titer) confirms the antiviral effect of the PRX034-CM observed by Viral ToxGlo (indirect measurement based on cell's metabolic activity)
- It also suggests that that the plaque assay method is significantly more sensitive than the ToxGlo assay to assess the effect of CM on RSV replication (**Figure 4**)

In vitro *Assay Demonstrates Synergistic Benefit of Combining PRTX007 with Direct-Acting Antiviral Against RSV*

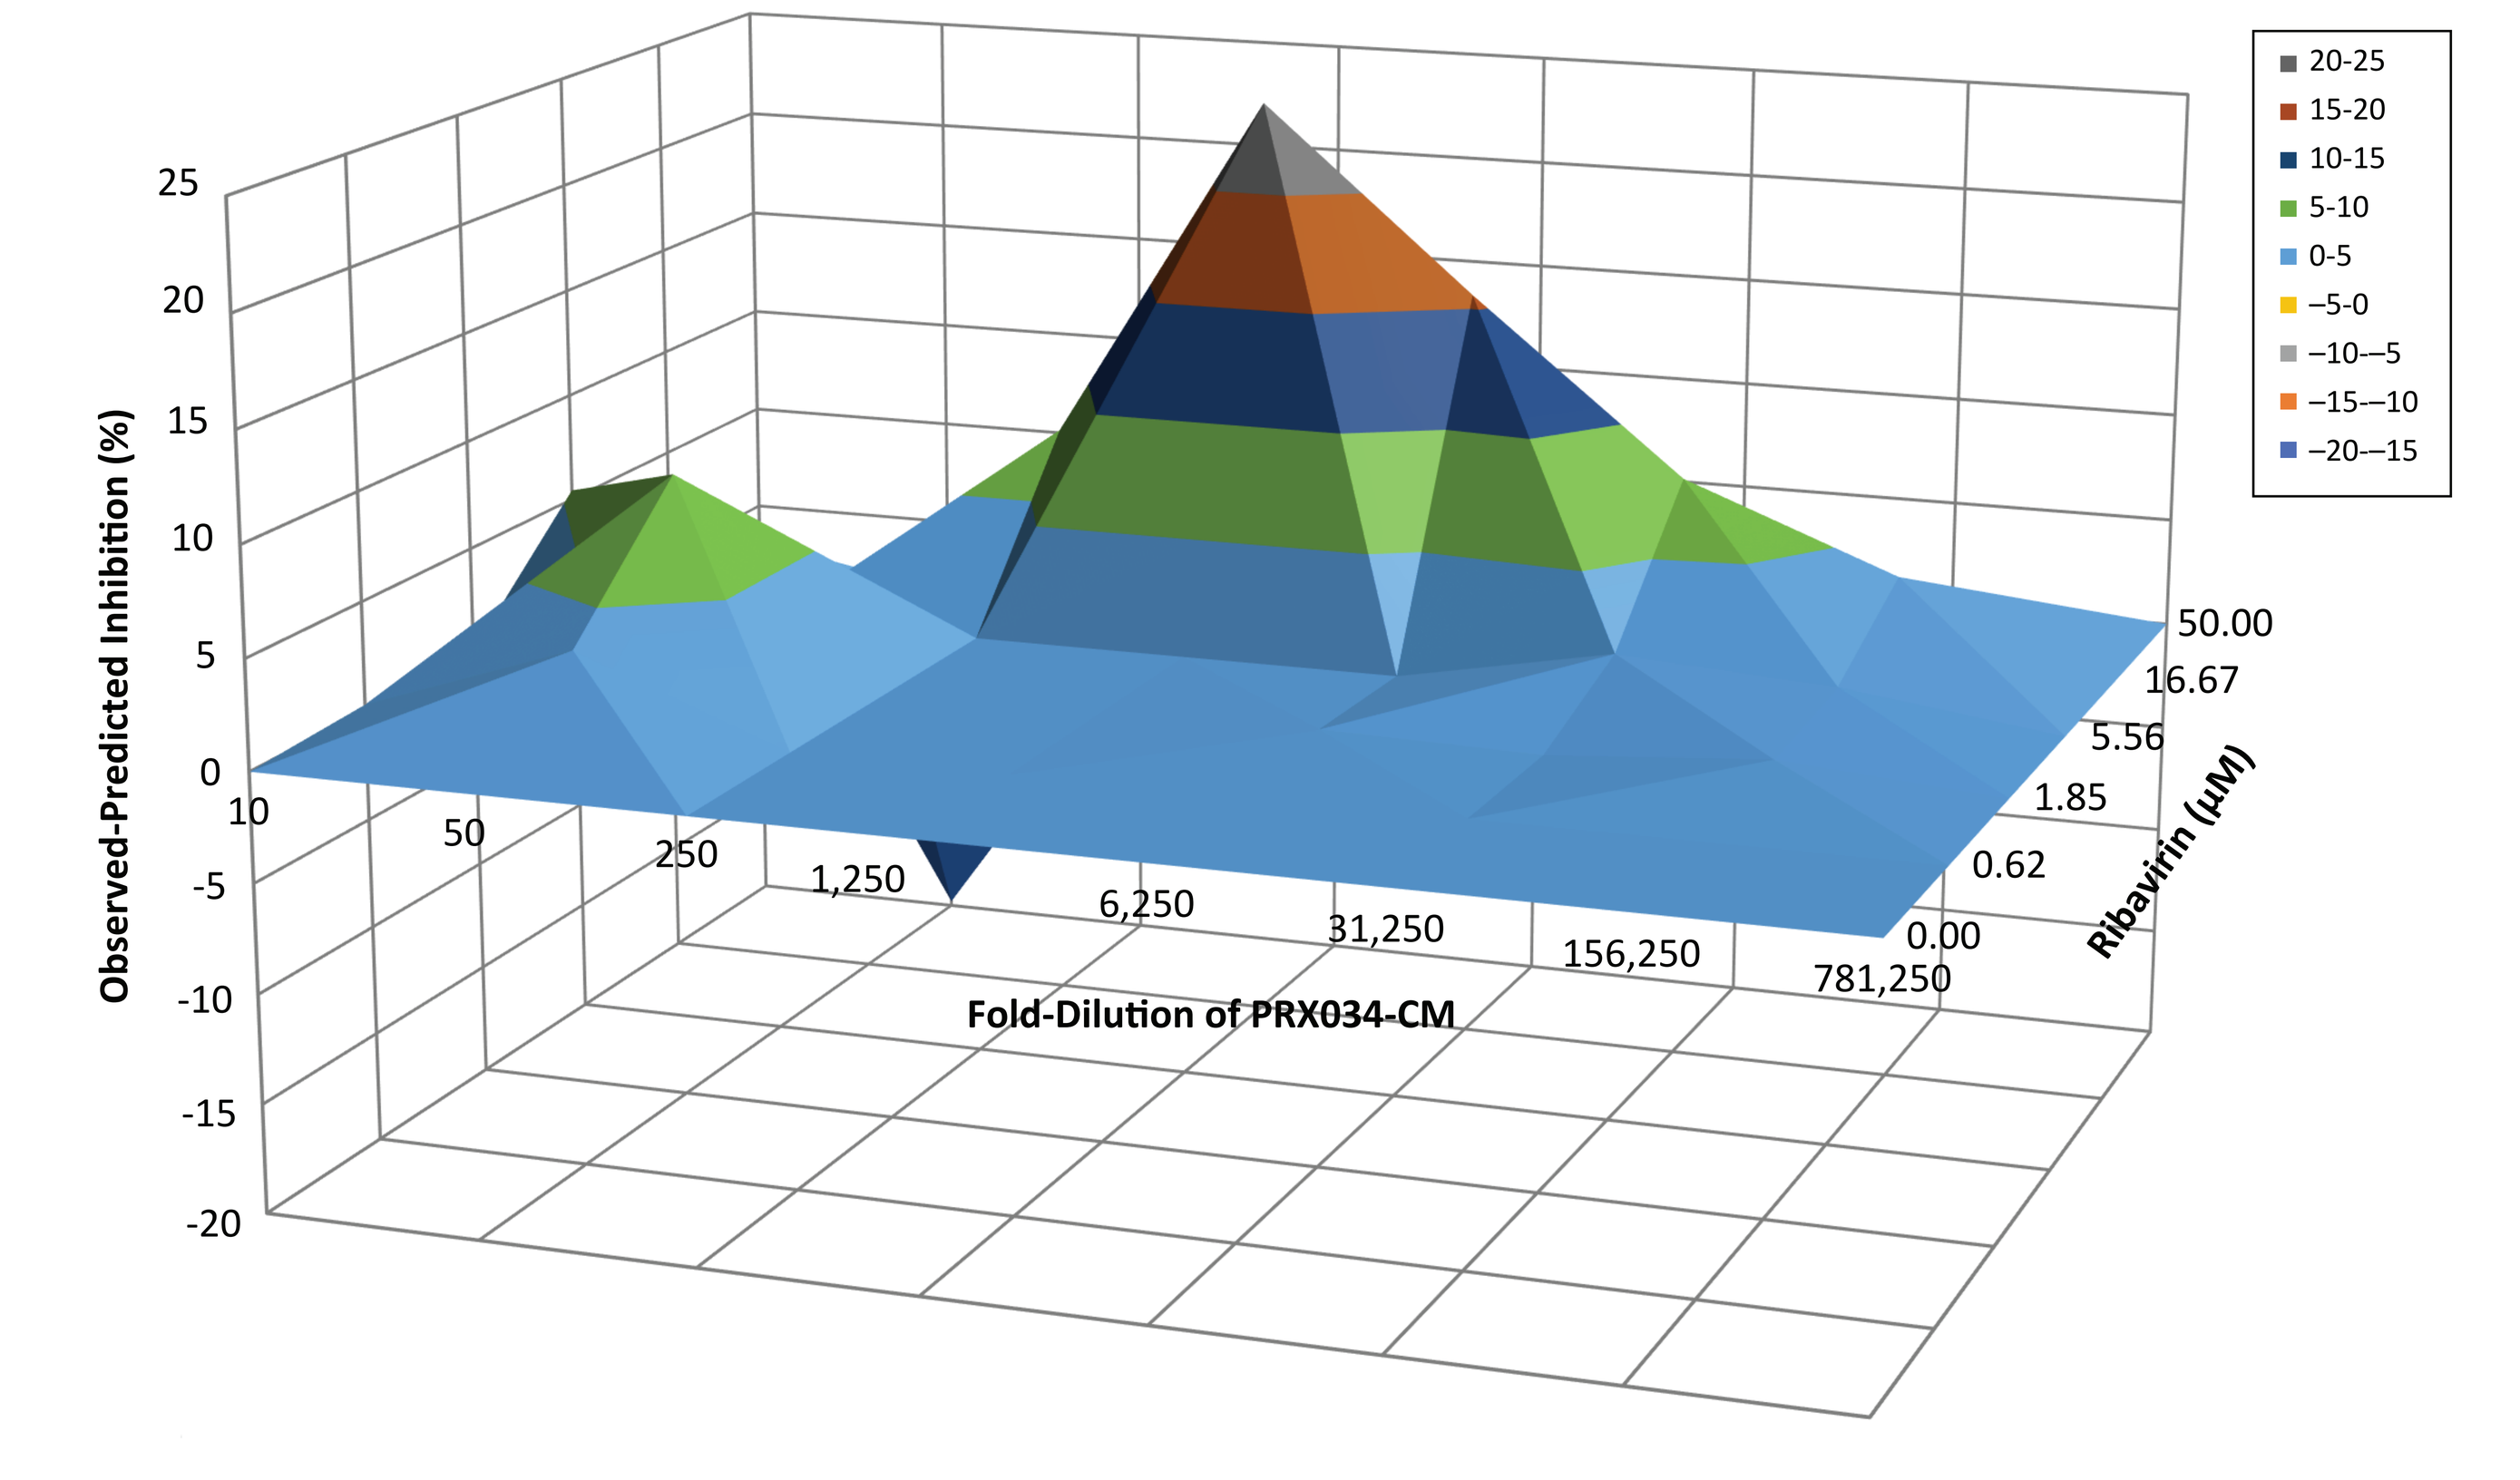
Observed-Predicted Inhibition (%)
Fold-Dilution of PRX034-CM
Ribavirin (µM)
20-25
15-20
10-15
5-10
0-5
−5-0
−10–−5
−15–−10
−20–−15

CM=conditioned media; CPE=cytopathic effect; HEp-2=human epithelial; hpi=hours postinfection; PBMC=peripheral blood mononuclear cell; RSV=respiratory syncytial virus.

Figure 5. Combination of PRX034-CM and ribavirin inhibits CPE caused by RSV infection.
HEp-2 cells were pre-treated with CM derived from PRX034-treated hPBMCs for 18 hours prior to addition of ribavirin followed by RSV infection. After 72 hpi the cells' metabolic activity was measured by Viral ToxGlo™. The raw luminescence data were analyzed using MacSynergy II. The plot displays the difference between experimentally measured inhibition of the combination and predicted inhibition based upon the dose responses of the two agents alone. Log volume below 0 is considered as antagonism, between 0 to 25% is additive, and greater than 25% is synergistic.

- CM plus ribavirin suggests interactions to be additive, approaching frank synergy where inhibition by either agent alone is modest
- The log volume >25% (27.32%) indicates the interaction is a statistically significant, greater than expected interaction, and thus synergistic

Confirmation of Antiviral Activity in an Animal Model (Mouse Model of RSV Infection)

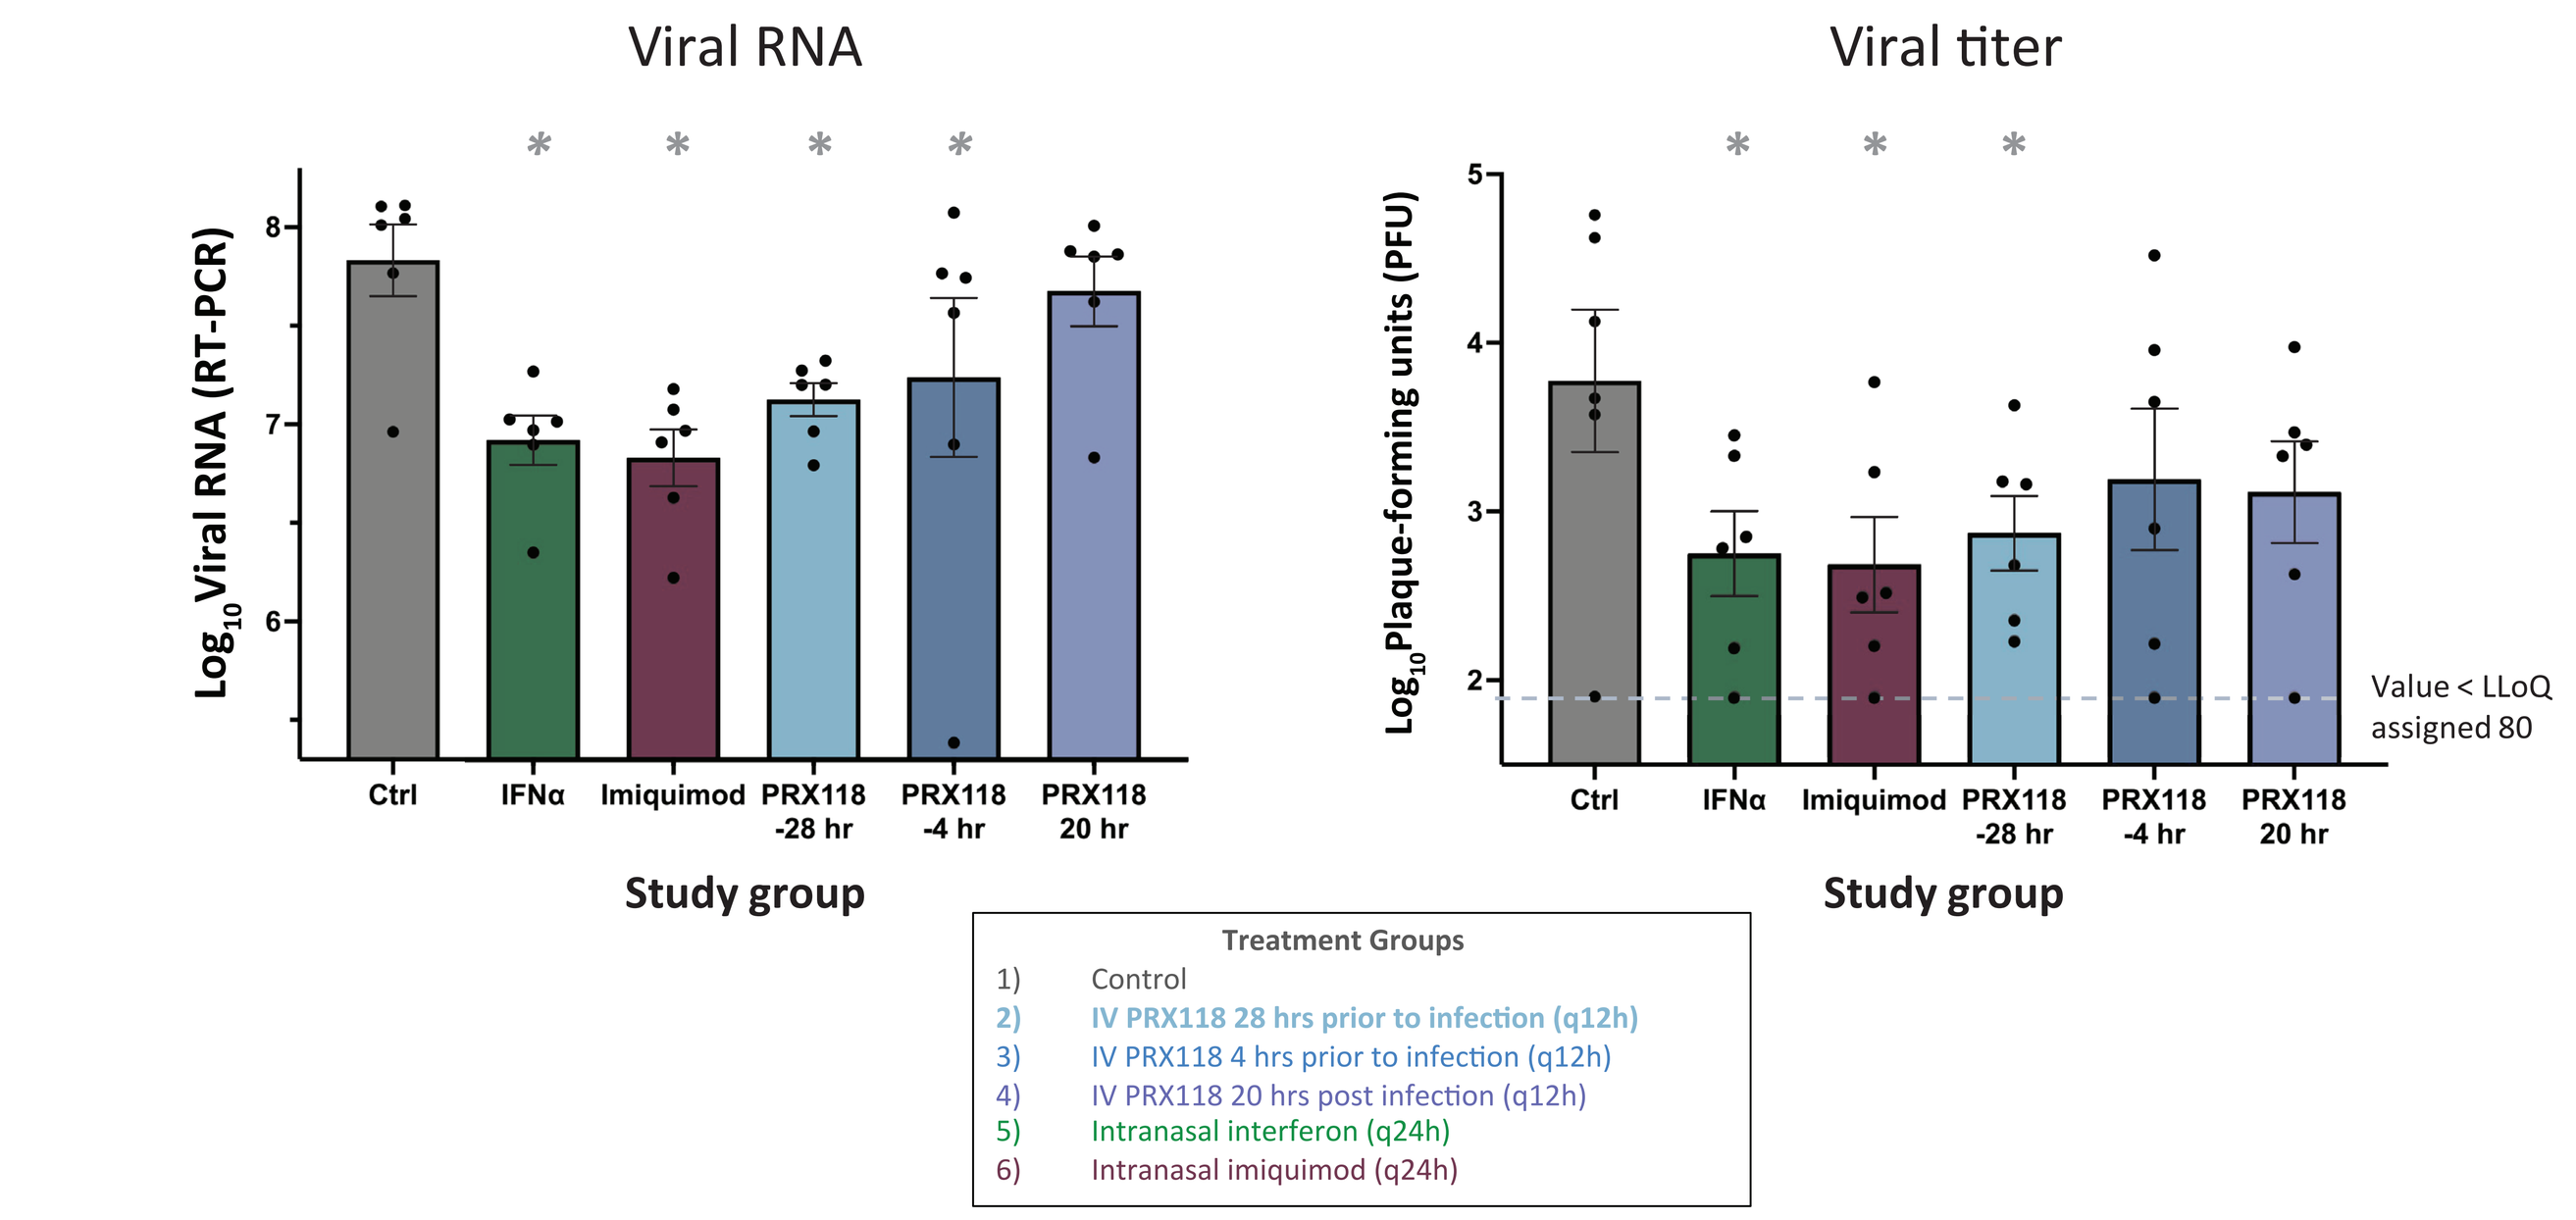
Viral RNA
Log10 Viral RNA (RT-PCR)
Ctrl
IFNα
Imiquimod
PRX118 -28 hr
PRX118 -4 hr
PRX118 20 hr
Study group
Viral titer
Log10Plaque-forming units (PFU)
Value < LLoQ assigned 80
Study group
Treatment Groups
1) Control
2) IV PRX118 28 hrs prior to infection (q12h)
3) IV PRX118 4 hrs prior to infection (q12h)
4) IV PRX118 20 hrs post infection (q12h)
5) Intranasal interferon (q24h)
6) Intranasal imiquimod (q24h)

Ctrl=control; GM=geometric mean; h/hr=hour(s); IFNα=interferon alpha; IV=intravenous; LLoQ=lower limit of quantitation; q=every; RSV=respiratory syncytial virus; RT-PCR=reverse transcription polymerase chain reaction.

Figure 6. Antiviral as systemic agent is comparable to mechanistically related therapeutics administered directly to site of infection.
PRX034 was delivered by IV administration of the prodrug PRX118 because PRTX007 conversion to PRX034 is inefficient in rodents. Error bars indicate errors of GM, which are graphed as bars moving from left to right. Single asterisk (*) indicates P≤.05 by Dunnett's test. Dashed line indicates LLoQ, values <LLoQ were assigned 80.

- In the RSV murine model, PRX034's antiviral activity was comparable with that achieved with intranasal IFNα or intranasal imiquimod, as assessed by viral RNA and viral titer from the lungs of infected animals

A.

Viral titer (plaque-forming units from lung)

Group	Treatment	Read time postinf	Geometric mean (GM)	Log_{10} GM	Log_{10} SD	Log_{10} decrease
1	Vehicle (control)	48h	1.40×10^3	3.15	0.55	
2	PRX118 q24h	48h	6.06×10^1	1.78	1.70	**1.36**
3	PRX118 q12h	48h	6.29×10^0	0.8	1.36	**2.35**
4	Vehicle (control)	96h	2.00×10^4	4.30	0.76	
5	PRX118 q24h	96h	2.54×10^2	2.41	1.71	**1.90**
6	PRX118 q12h	96h	9.76×10^2	2.99	1.42	**1.31**

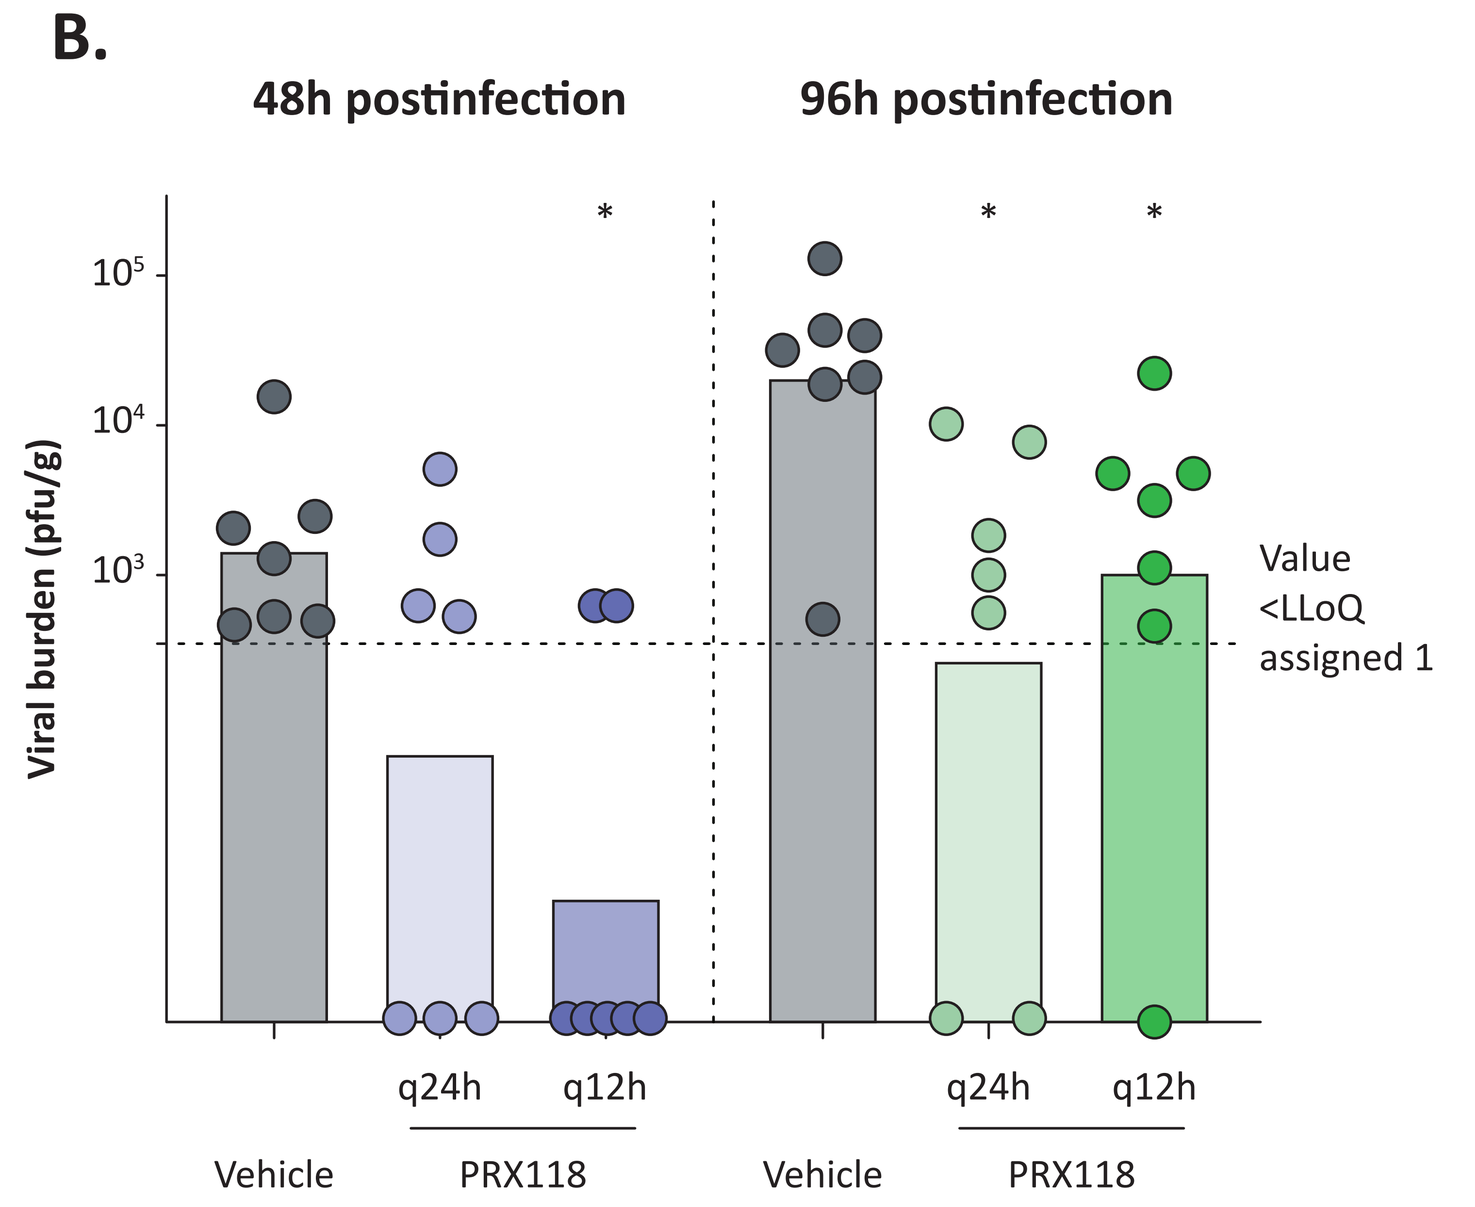
B.
48h postinfection
96h postinfection
Viral burden (pfu/g)
Value <LLoQ assigned 1
Vehicle
q24h
q12h
PRX118
Vehicle
q24h
q12h
PRX118

GM=geometric mean; h=hour(s); hpi=hours postinfection; IV=intravenous; LLoQ= lower limit of quantitation; q=every; RSV=respiratory syncytial virus; SD=standard deviation.

Figure 7. Time course of antiviral activity of PRX034 in mouse model of RSV infection.
Treatment was initiated 12 hours prior to infection with either vehicle (control), or PRX118 at q24h and q12h (n = 7 mice/group). Viral titers (plaque-forming units [PFU] from lung) were collected at 48 and 96 hpi. PRX034 was delivered by IV administration of the prodrug PRX118 because PRTX007 conversion to PRX034 is inefficient in rodents. **A.** Table of viral titer results, including geometric mean and standard deviation. **B.** Individual viral titers from RSV-infected mice shown as colored dots. The GMs for each treatment are shown as bars moving from left to right and correspond to treatment groups in table. Asterisks indicate statistical significance with a *P*=.05 by Kruskal-Wallis test. Dashed line indicates LLoQ, values <LLoQ were assigned a 1.

- This is a time course of antiviral activity mouse model of RSV infection
 - Significant reduction in amount of infectious virus in lung (measured as PFUs) demonstrated at 48 and 96 hpi (**Figure 7**)
 - Most relevant study endpoint indicating antiviral response
 - Decrease in viral RNA also observed, albeit of lower magnitude
- No evidence of adverse effects was observed with treatment
 - Slight increase in circulating levels of TNFα observed with q12h but not q24h dosing (no increase in IL-6 with either treatment schedule)

Conclusions

- Demonstrated antiviral activity in cellular studies
 - Activity was greater than predicted based on IFNα2a content alone
 - Pre-treatment increases potency of the PRX034-CM antiviral response
 - Viral progeny retain sensitivity
- Demonstrated antiviral activity in animal studies
 - No evidence of adverse effects after treatment in animal model
 - Viral load reduction with systemic PRX034 was comparable to intranasal IFNα and imiquimod in animal model
- Support for combining PRXT007 with other therapeutic agents in treatment of RSV shown from *in vitro* studies
- Findings support clinical investigation of PRTX007 for the treatment of RSV and other acute respiratory viruses

References

1. Centers for Disease Control and Prevention. August 2017. Accessed February 15, 2022. https://www.cdc.gov/rsv/factsheet-older-adults.pdf/; 2. Colosia AD, et al. *PLoS ONE*. 2017;12(8):e0182321; 3. Behzadi MA, et al. *Front Microbiol*. 2019;10:1327. 4. Smit JJ, et al. *J Exp Med*. 2006;203(5):1153-1159; 5. Wang H, et al. *J Immunol*. 2006;177(9):6263-62670; 6. Appleman J, et al. **Poster #143**. Presented at: 35th International Conference on Antiviral Research; March 21-25, 2022; Seattle, Washington.

Disclosures & Acknowledgments

This study was funded by Primmune Therapeutics.
Medical writing services were provided by SCIENT Healthcare Communications and funded by Primmune Therapeutics, Inc., (Carlsbad, CA).

Presented at the 35th International Conference on Antiviral Research | March 21–25, 2022 | Seattle, Washington, USA