582

Contact: jappleman
@primmunerx.com

Selection of a novel toll-like receptor 7 (TLR7) agonist PRX034 for immunotherapy of cancer

James R. Appleman
Stephen E. Webber

primmune Therapeutics

ABSTRACT

At Primmune, we believe that maximal therapeutic benefit in cancer immunotherapy will be achieved by maintaining sustained, systemic immune pressure involving both innate and adaptive immunity. We have therefore chosen PRX034✣, an orally administered TLR7 small molecule agonist, for clinical development. This candidate will ultimately be used in combination with immune checkpoint inhibitors (ICPIs) for the treatment of cancer. This TLR7 agonist preferentially activates plasmacytoid dendritic cells, the master coordinator of innate and adaptive immunity. Further, PRX034 and related analogs preferentially induce cellular production of Type I interferons while minimizing production of proinflammatory cytokines through the NFκβ pathway in vitro and in vivo.

We have previously reported values for a series of unusual properties that must be matched for any drug candidate successfully targeting TLR7 and intended for chronic and systemic administration (1). During lead optimization, each compound was evaluated in the following assays in order to assess whether requirements had been met: cellular assays (TLR reporter cells, human and monkey peripheral blood mononuclear cells), pharmacokinetic assays (IV administration of active agonist and PO administration of corresponding prodrug in cynomolgus monkeys), and pharmacodynamic response in cynomolgus monkeys. The declaration of PRX034 as a clinical candidate was ultimately dependent upon successfully meeting pre-selected targets for compound potency and reproducibility of induced immune response in a repeat dose study in cynomolgus monkeys. Specifically, PRX034 was administered orally as a prodrug on a QOD schedule for two weeks at two dose levels selected in a previous dose range-finding study. TLR7 agonist pharmacokinetics and the kinetics and dynamics of immune induction following each administered dose were constant within targeted levels throughout the study. Consequently, PRX034 has progressed into IND-enabling studies.

Based upon our previous experience with TLR7 agonists meeting our profile, we expect safety in pivotal repeat-dose toxicology studies to be sufficient to enable PRX034 to be evaluated in healthy volunteers (HVs). This clinical study will include SAD cohorts followed by two MAD cohorts, the latter including two dosing schedules. Feasibility and utility of this HV study require both appropriate preclinical safety findings and relevant pharmacodynamic markers that can be readily measured in volunteers. This study will ensure that subsequent cancer patients receive only pharmacologically active doses and that evaluation in combination with ICPIs is facilitated.

(1) Appleman J, Webber, S. Discovery of a series of novel toll-like receptor 7 agonists for systemic immunotherapy of cancer [abstract]. In: Proceedings of the Annual Meeting of the American Association for Cancer Research 2019. Mar 29 - Apr 3; Atlanta, GA. Abstract # 3262.

✣Note: The actual clinical candidate is PRTX007, an orally administered prodrug of the TLR7 agonist PRX034.

BACKGROUND

Our Program Originally Focused Upon Treatment of Cancer

Activation of Plasmacytoid Dendritic Cells (pDCs) by TLR7 Agonists Elicits Effective Anti-tumor Responses

TLR7 agonists engage a variety of antitumor responses, most of which are initiated by activation of plasmacytoid dendritic cells.

Inactivated pDCs in the tumor microenvironment are pro-tumorogenic, whereas activated pDCs elicit a plethora of antitumoral activities

pDC Activation Induces pDC Cell-surface Expression of:

Receptor	Bind To	Key Role
CD40	CD40L	Increases IFN secretion when responsive cell is engaged
CD80	CD28 & CTLA4	Involved in B- and T-cell activation, proliferation and differentiation
CD86	CD28	Involved in T-cell activation

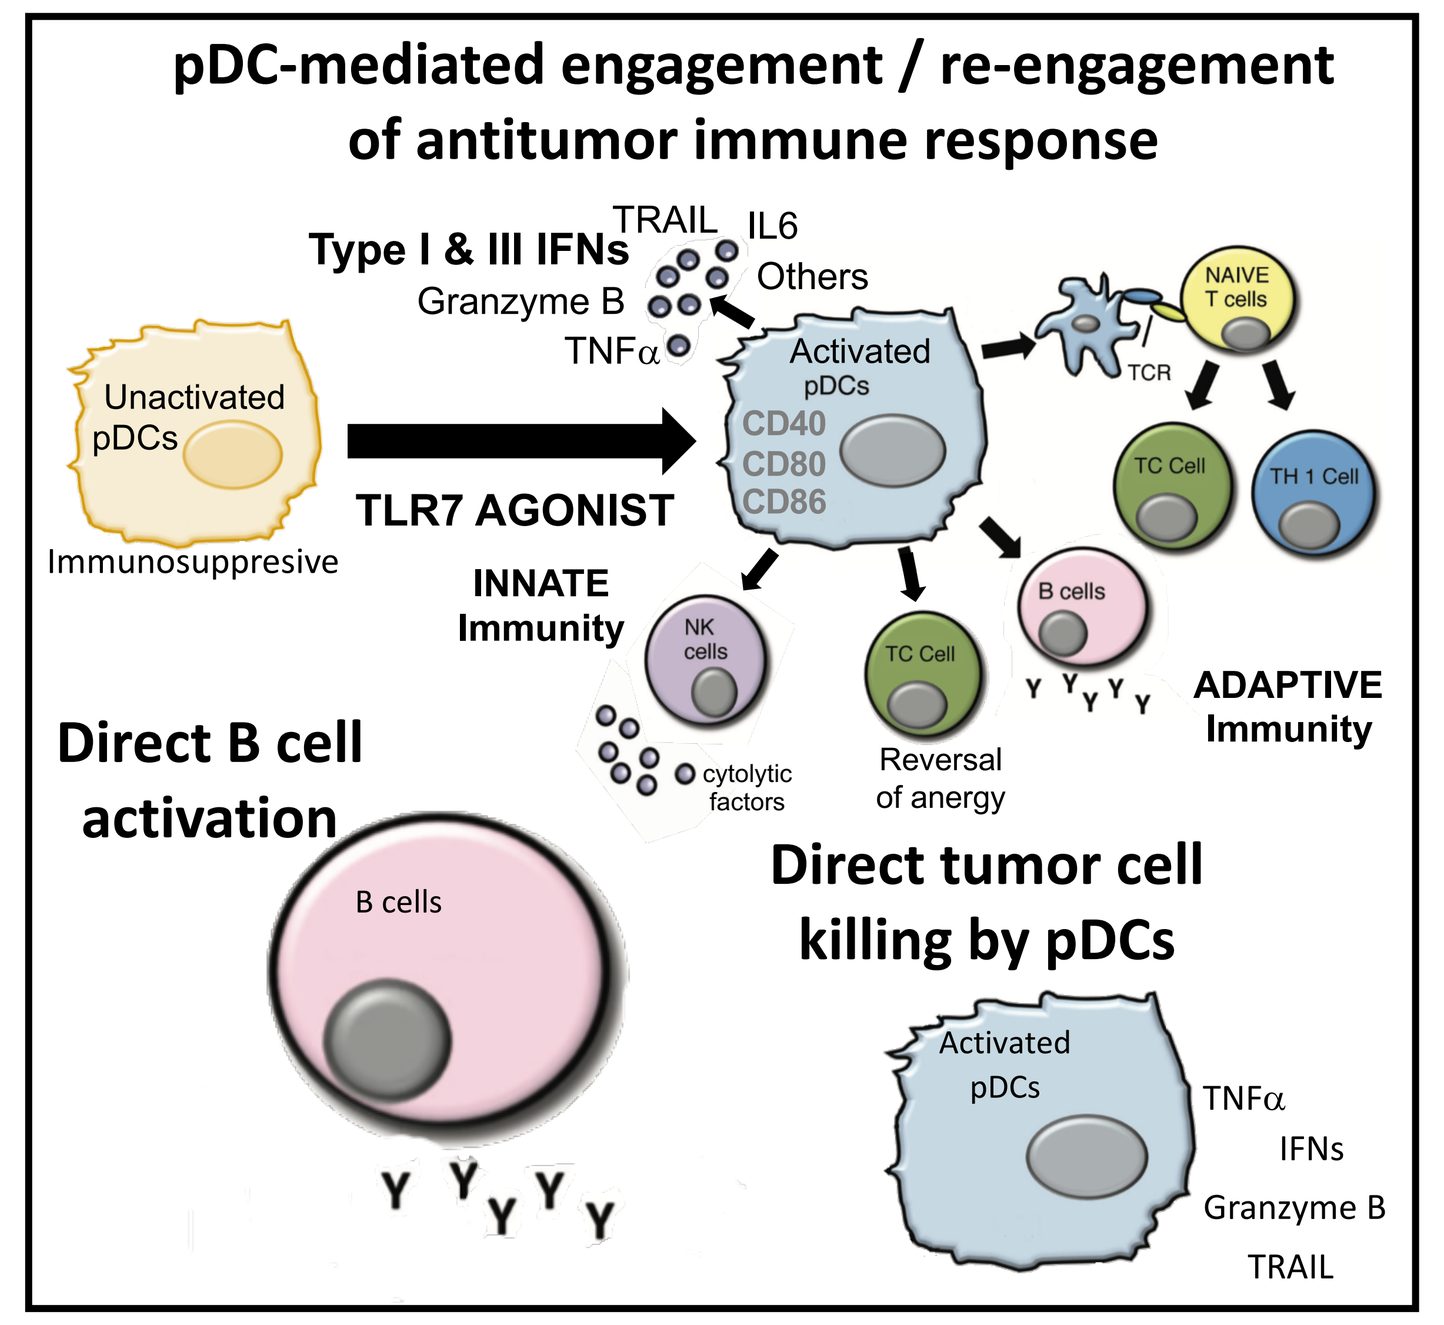
pDC-mediated engagement / re-engagement of antitumor immune response
Type I & III IFNs
TRAIL
IL6
Granzyme B
Others
TNFα
Unactivated pDCs
Immunosuppresive
TLR7 AGONIST
Activated pDCs
CD40
CD80
CD86
TCR
NAIVE T cells
TC Cell
TH 1 Cell
INNATE Immunity
NK cells
cytolytic factors
TC Cell
Reversal of anergy
B cells
ADAPTIVE Immunity
Direct B cell activation
B cells
Direct tumor cell killing by pDCs
Activated pDCs
TNFα
IFNs
Granzyme B
TRAIL

Because of the COVID-19 Pandemic, We Have Turned to SARS-CoV-2 Therapy

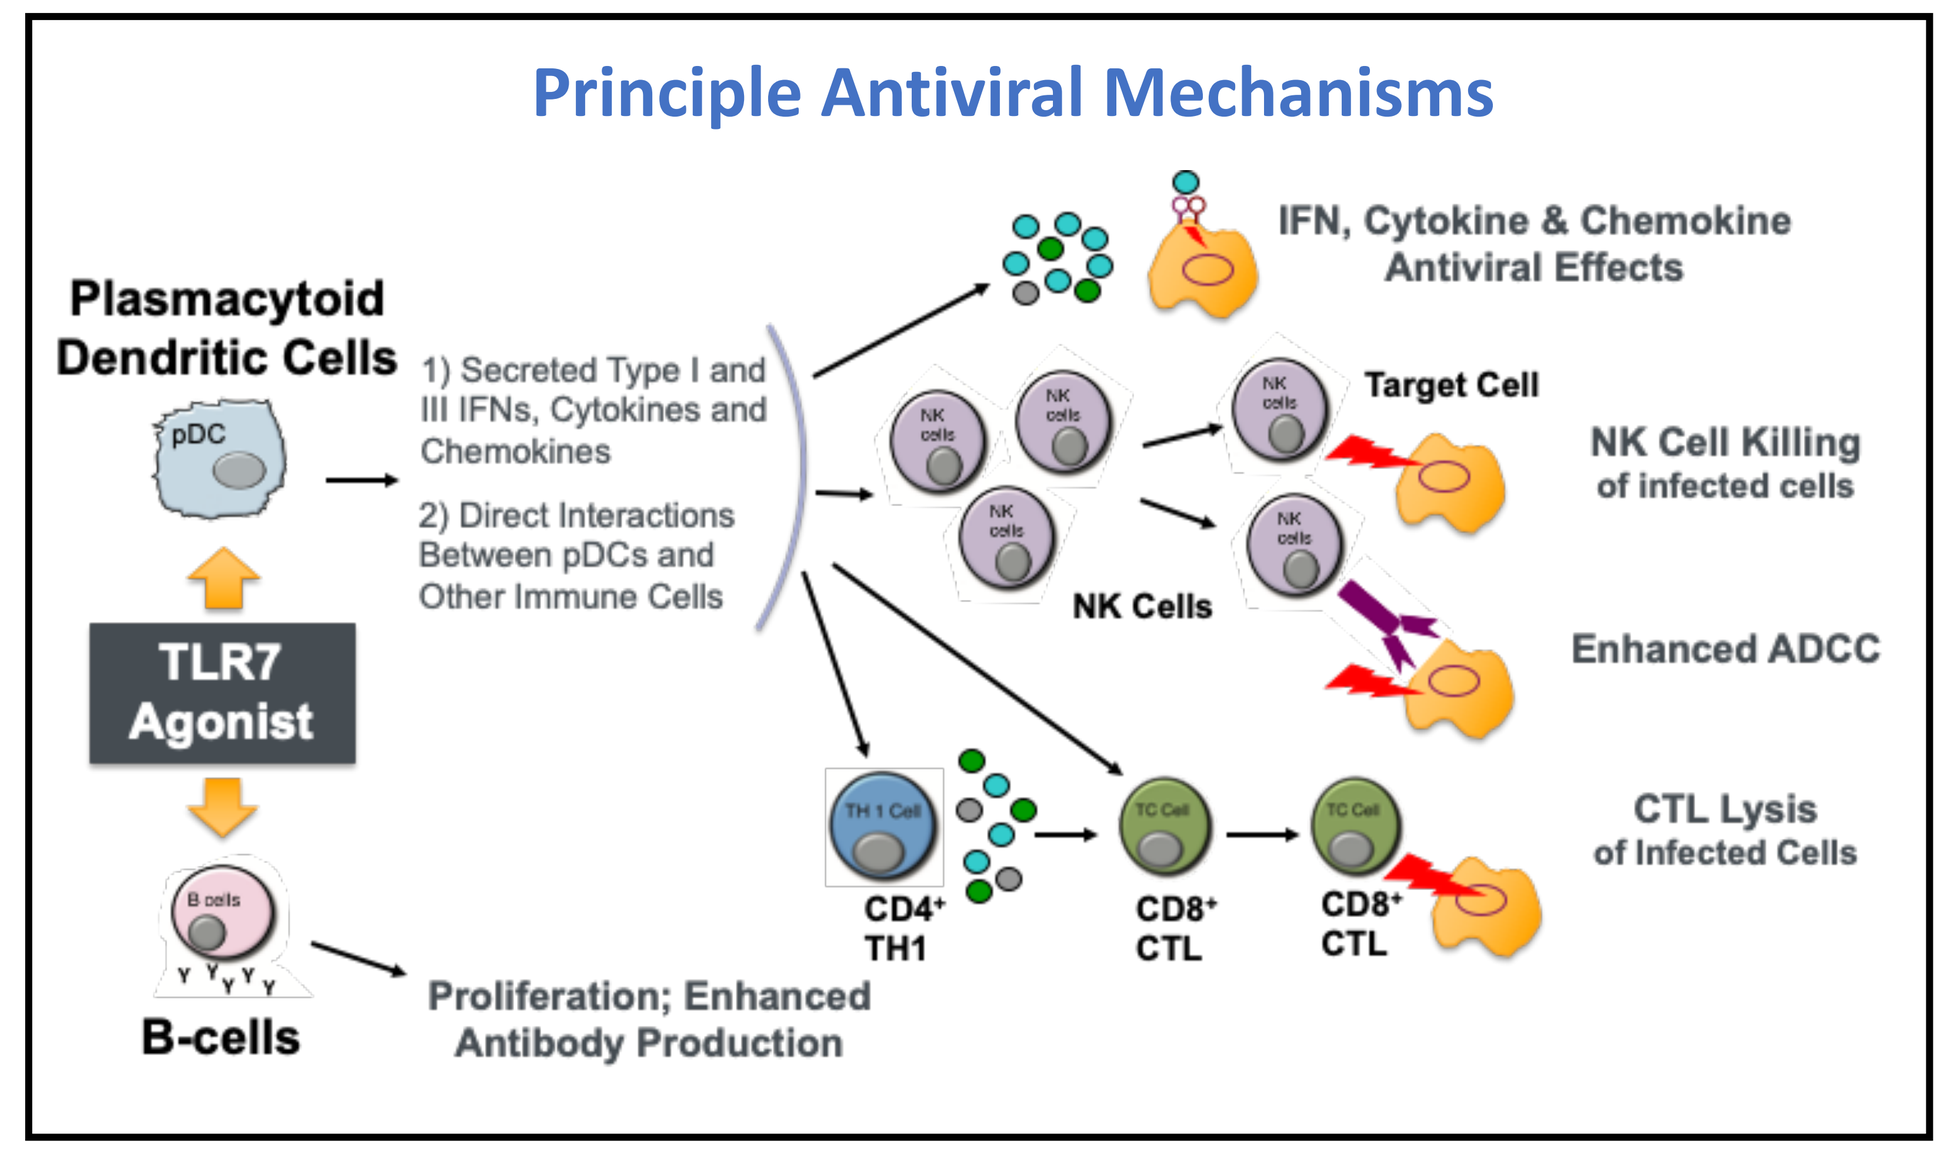
Principle Antiviral Mechanisms
Plasmacytoid Dendritic Cells
pDC
TLR7 Agonist
B-cells
1) Secreted Type I and III IFNs, Cytokines and Chemokines
2) Direct Interactions Between pDCs and Other Immune Cells
IFN, Cytokine & Chemokine Antiviral Effects
Target Cell
NK Cell Killing of infected cells
NK Cells
Enhanced ADCC
CTL Lysis of Infected Cells
CD4+ TH1
CD8+ CTL
CD8+ CTL
Proliferation; Enhanced Antibody Production

- Activation of general antiviral cellular responses primed by pDCs (plasmacytoid dendritic cells) responding to TLR7 agonists.
- NK (natural killer) cell activation and enhanced antibody-mediated killing of pathogen infected cells via antibody-dependent cellular cytotoxicity (ADCC) with therapeutic mAbs as well as endogenous antibodies.
- CTL (cytotoxic T-cell) activation.
- Increase in proliferation and activation of B-cells

- ❖ SARS-CoV-2 evades normal endogenous mechanisms that prevent or limit viral infection & replication
- ❖ SARS-CoV-2s retain sensitivity to these mechanisms when triggered by administration of a TLR7 agonist
- ❖ Primmune's clinical candidate uniquely reduces rather than exacerbates the risk of CRS, a major source of morbidity and mortality with COVID-19

Scope of Data Used in Selection of PRX034 & PRTX007

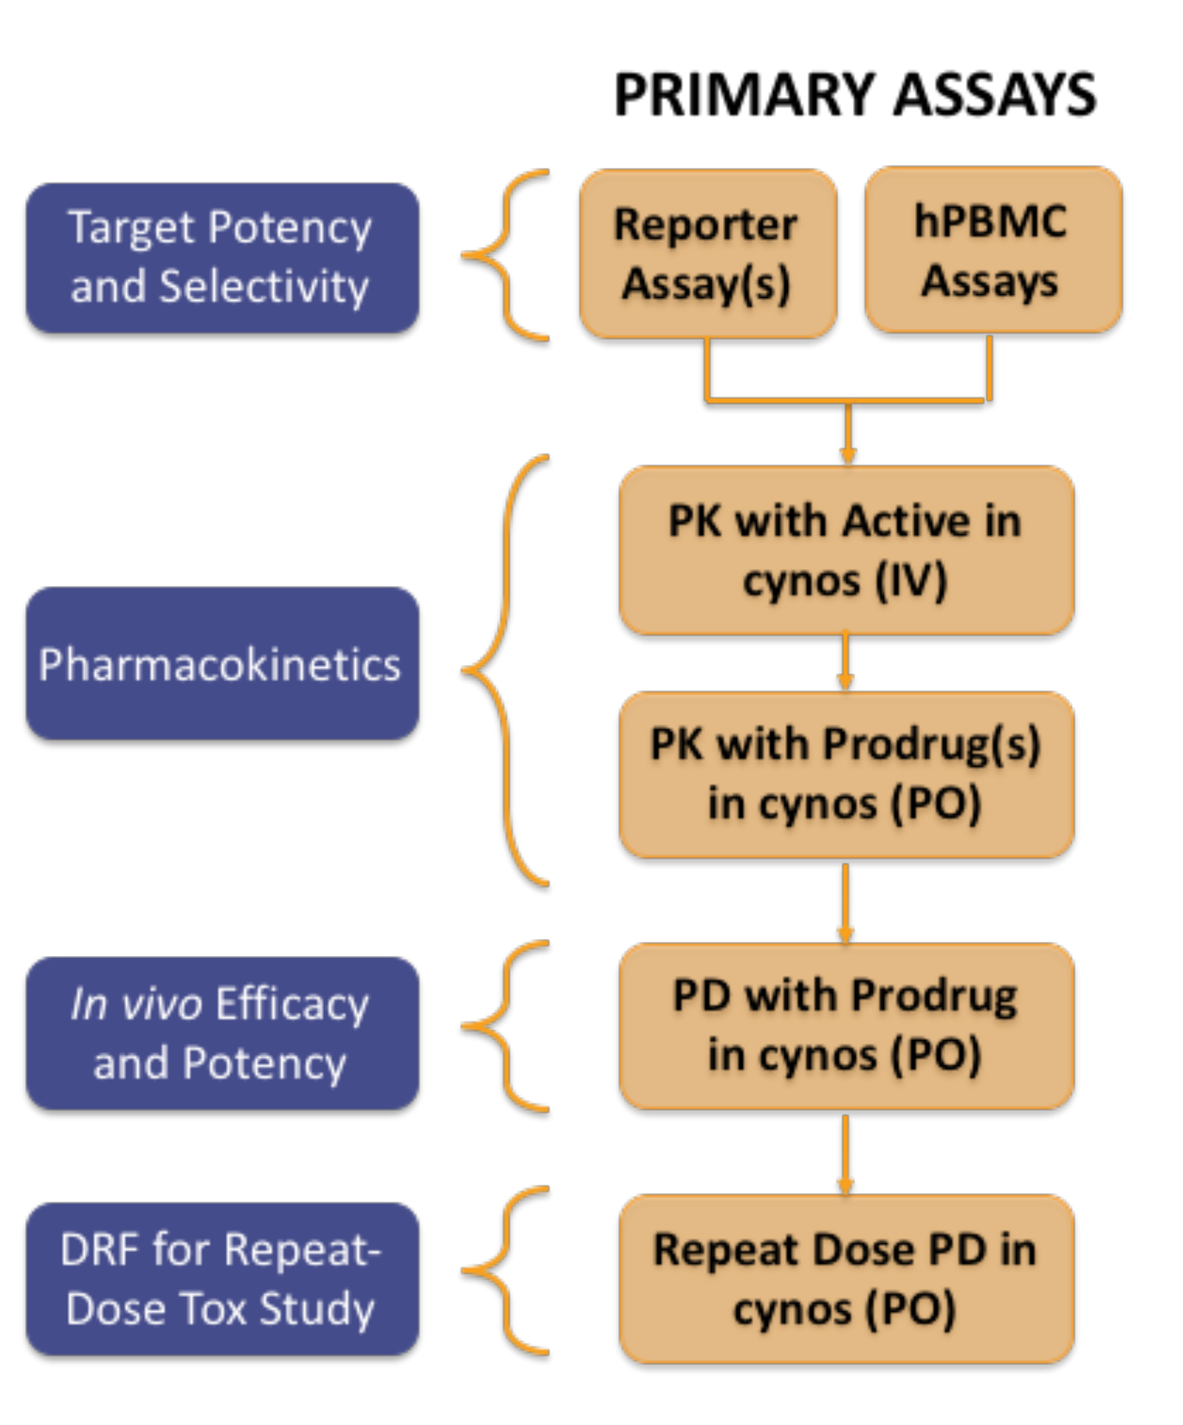
PRIMARY ASSAYS
Target Potency and Selectivity
Reporter Assay(s)
hPBMC Assays
Pharmacokinetics
PK with Active in cynos (IV)
PK with Prodrug(s) in cynos (PO)
In vivo Efficacy and Potency
PD with Prodrug in cynos (PO)
DRF for Repeat-Dose Tox Study
Repeat Dose PD in cynos (PO)
Secondary and Optional Assays
Direct Binding by ITC
Monkey PBMC
Physicochemical Properties
CYP Substrate or Inhibitor Eval
MDCKII Prodrug Permeability
In Vitro Prodrug Conversion
Cytotox Assay
Safety47 Panel
PK/PD in Mice

* Benchmarks for all assays were set based on experience with ANA773

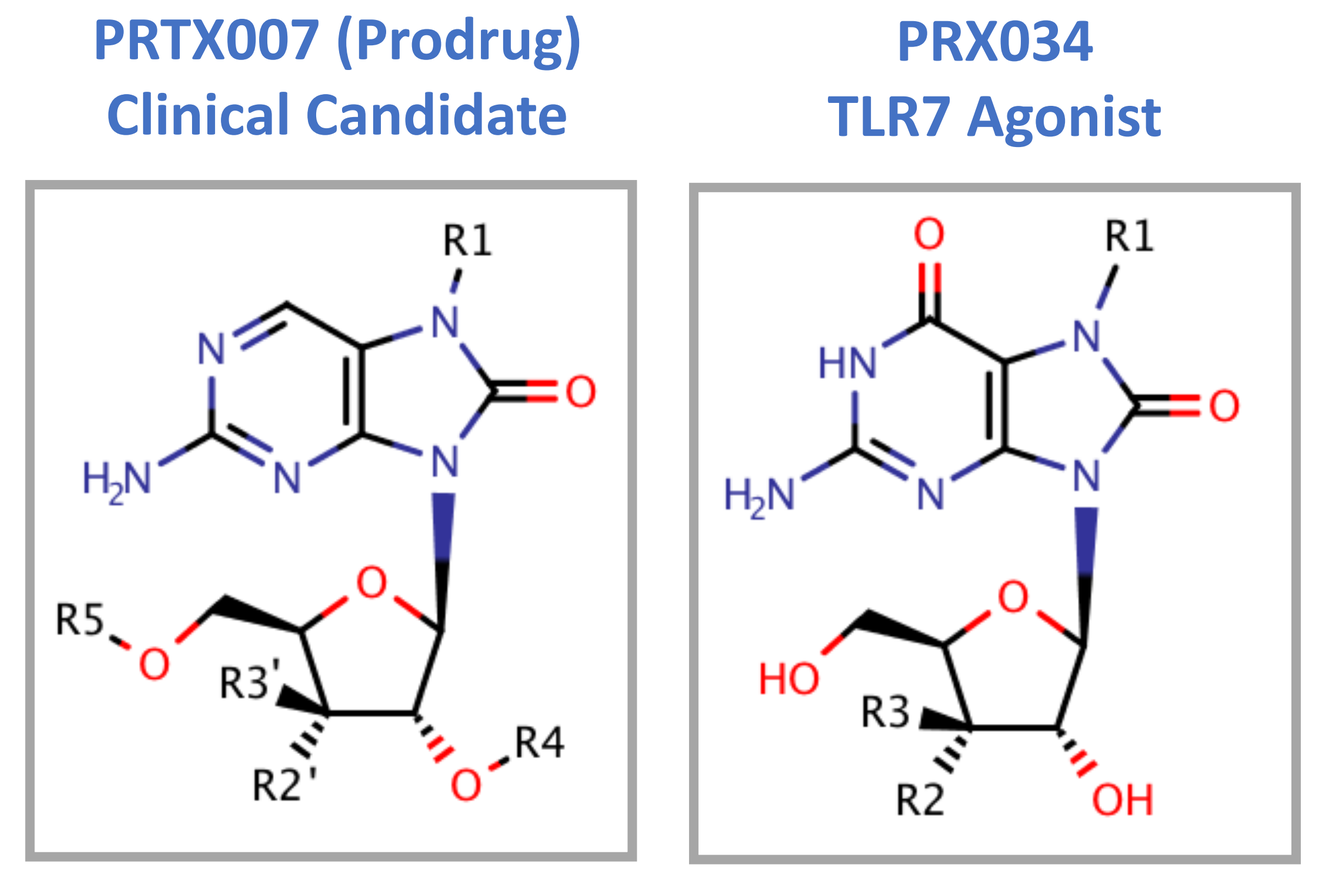
PRTX007 (Prodrug) Clinical Candidate
PRX034 TLR7 Agonist

(specific structures not disclosed)

PRX034 is administered as **PRTX007**, an inactive prodrug, to avoid excessive activation of GI-associated immune cells upon oral dosing

Biophysical Measures of Agonist Interaction with TLR7

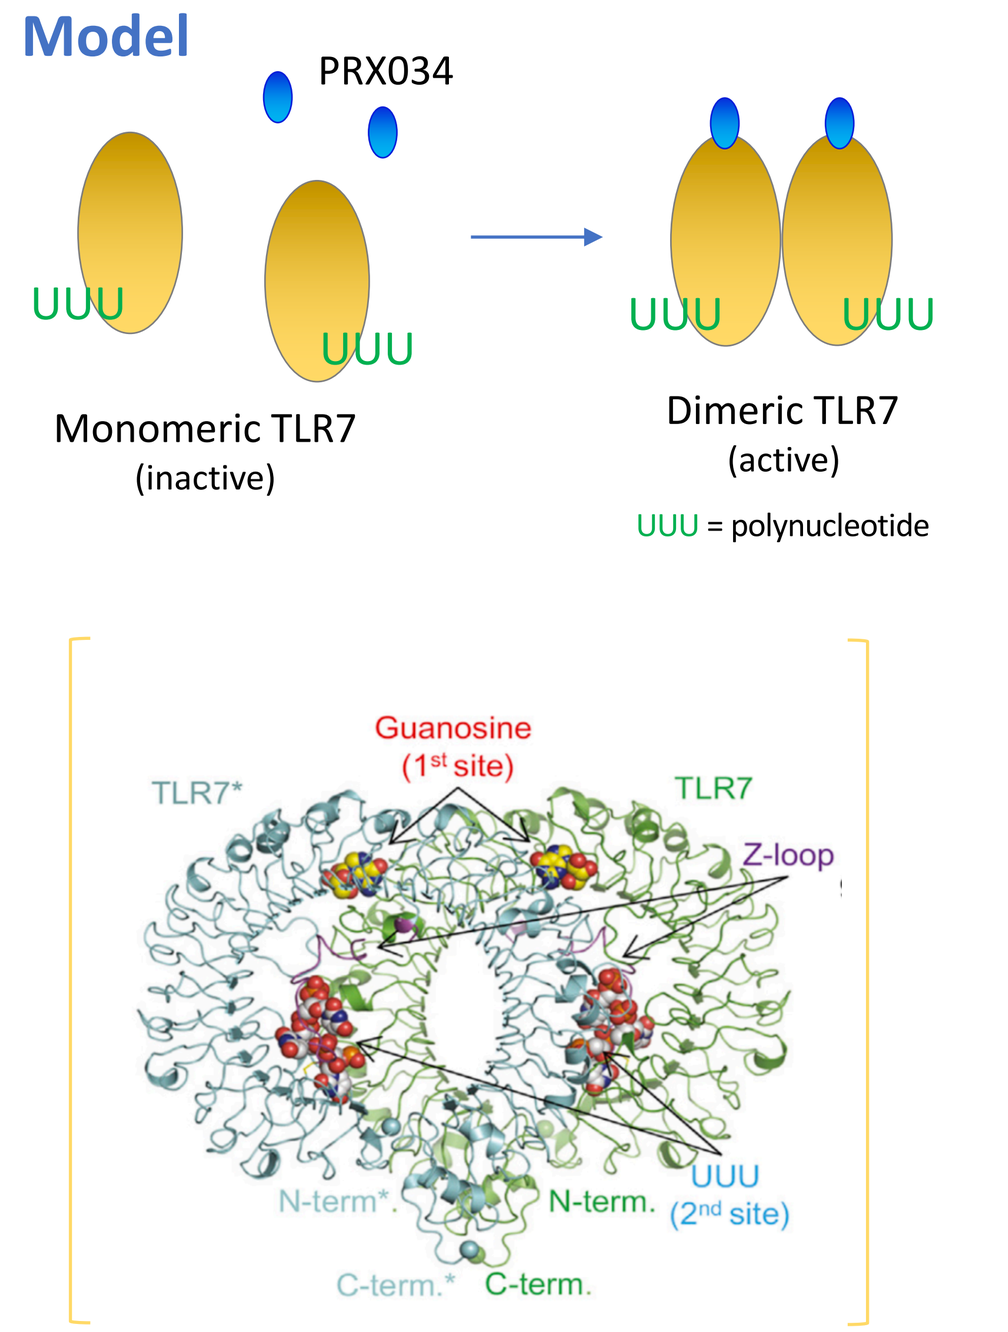
Model
PRX034
Monomeric TLR7 (inactive)
Dimeric TLR7 (active)
UUU = polynucleotide
Guanosine (1st site)
TLR7*
TLR7
Z-loop
N-term*
N-term.
UUU (2nd site)
C-term.*
C-term.

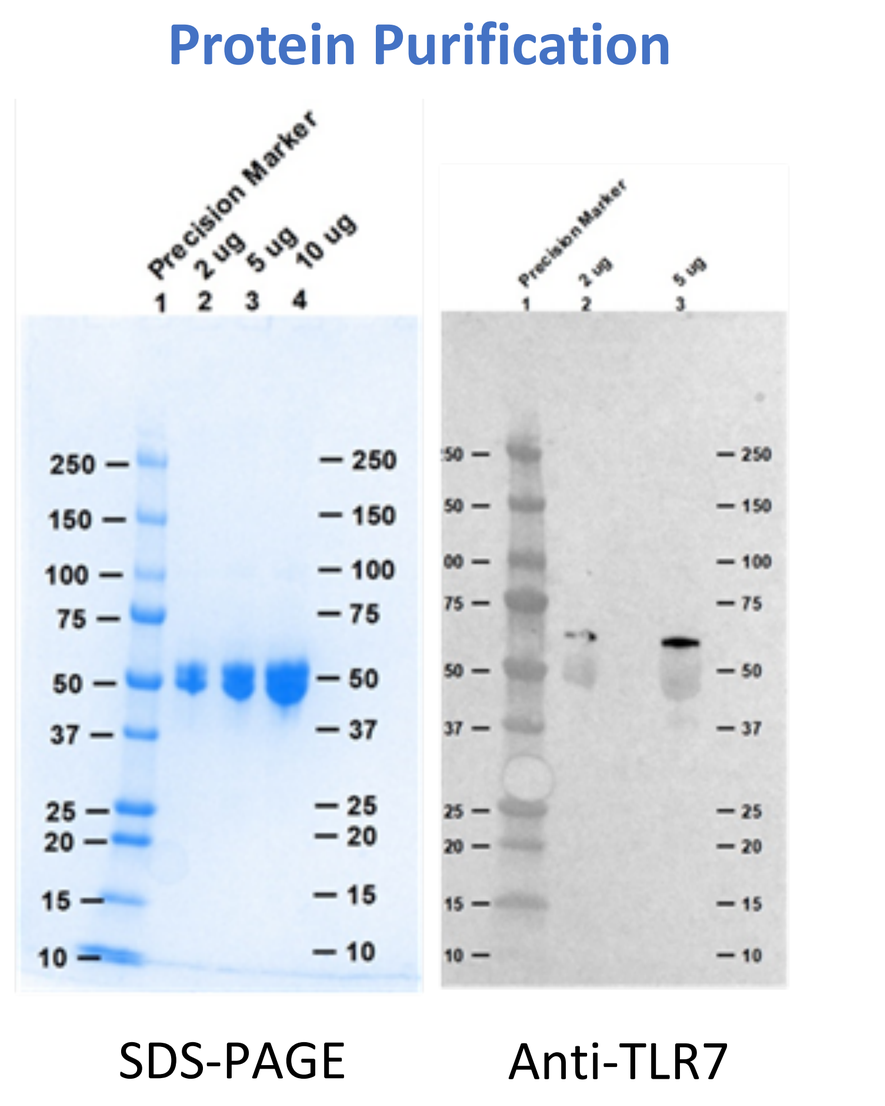
Protein Purification
Precision Marker
2 ug
5 ug
10 ug
SDS-PAGE
Anti-TLR7

Expression: 15 L S2 stable cells
Purification steps: FLAG-Affinity/Thrombin cleavage/SEC
Purity: > 98 %
Buffer: 10mM Tris -HCl pH 8.0, 150 mM NaCl

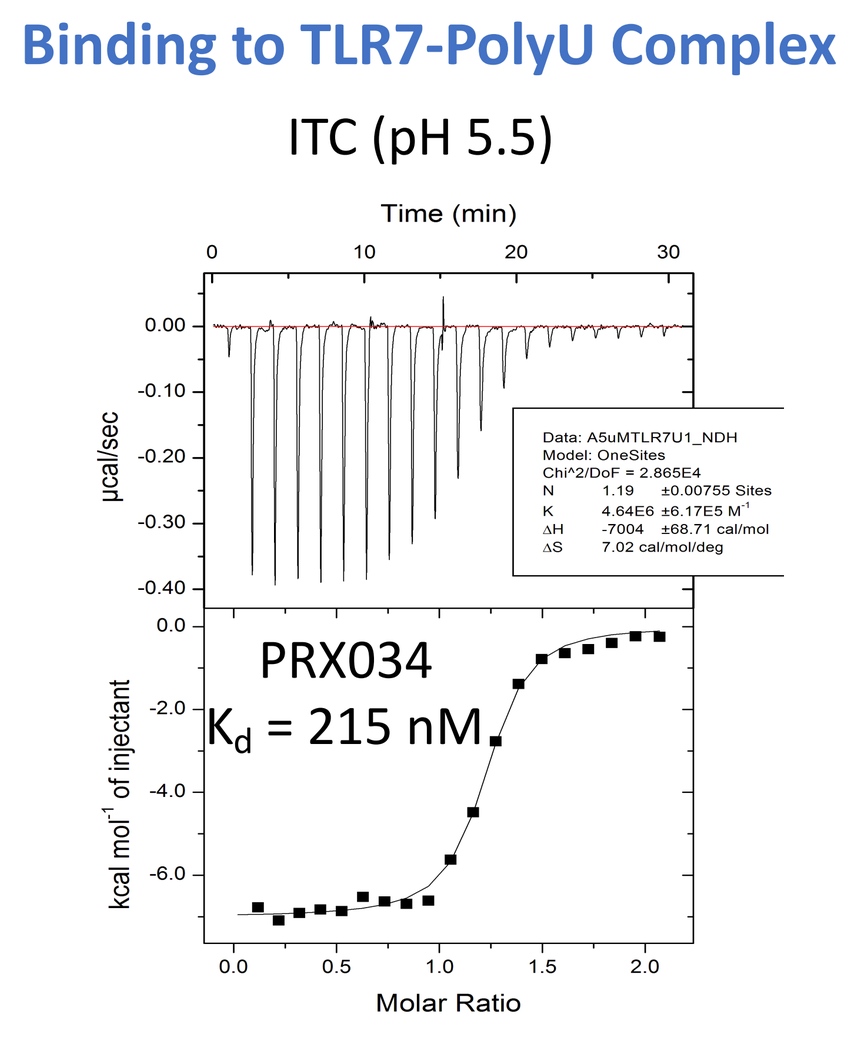
Binding to TLR7-PolyU Complex
ITC (pH 5.5)
Time (min)
μcal/sec
kcal mol-1 of injectant
Molar Ratio
PRX034
Kd = 215 nM

Binding of PRX034 also induces TLR7 dimerization as demonstrated by analytical SEC (not shown)

PRX034 Binds at Same Small Molecule Site as Guanosines

- As with other guanosine analogs, formation of active TLR7 dimer requires occupancy of both small molecule binding site and polynucleotide binding site
- Features identified in this and other co-crystal structures have contributed to the design of compounds in our next-generation series of agonists

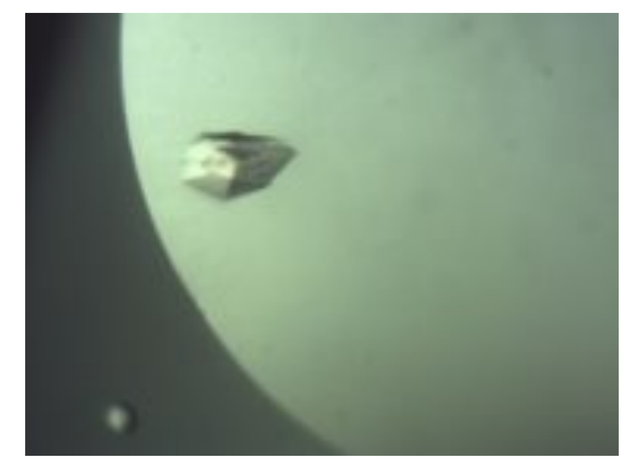

TLR7-polyU-PRX034 Co-crystal

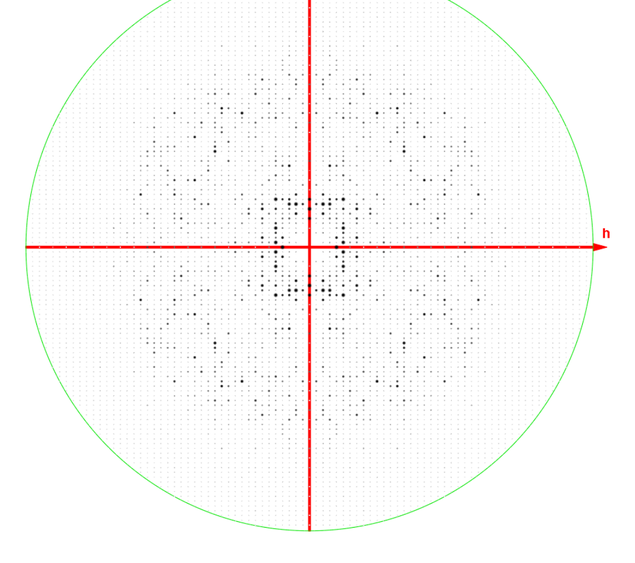

Diffraction Pattern

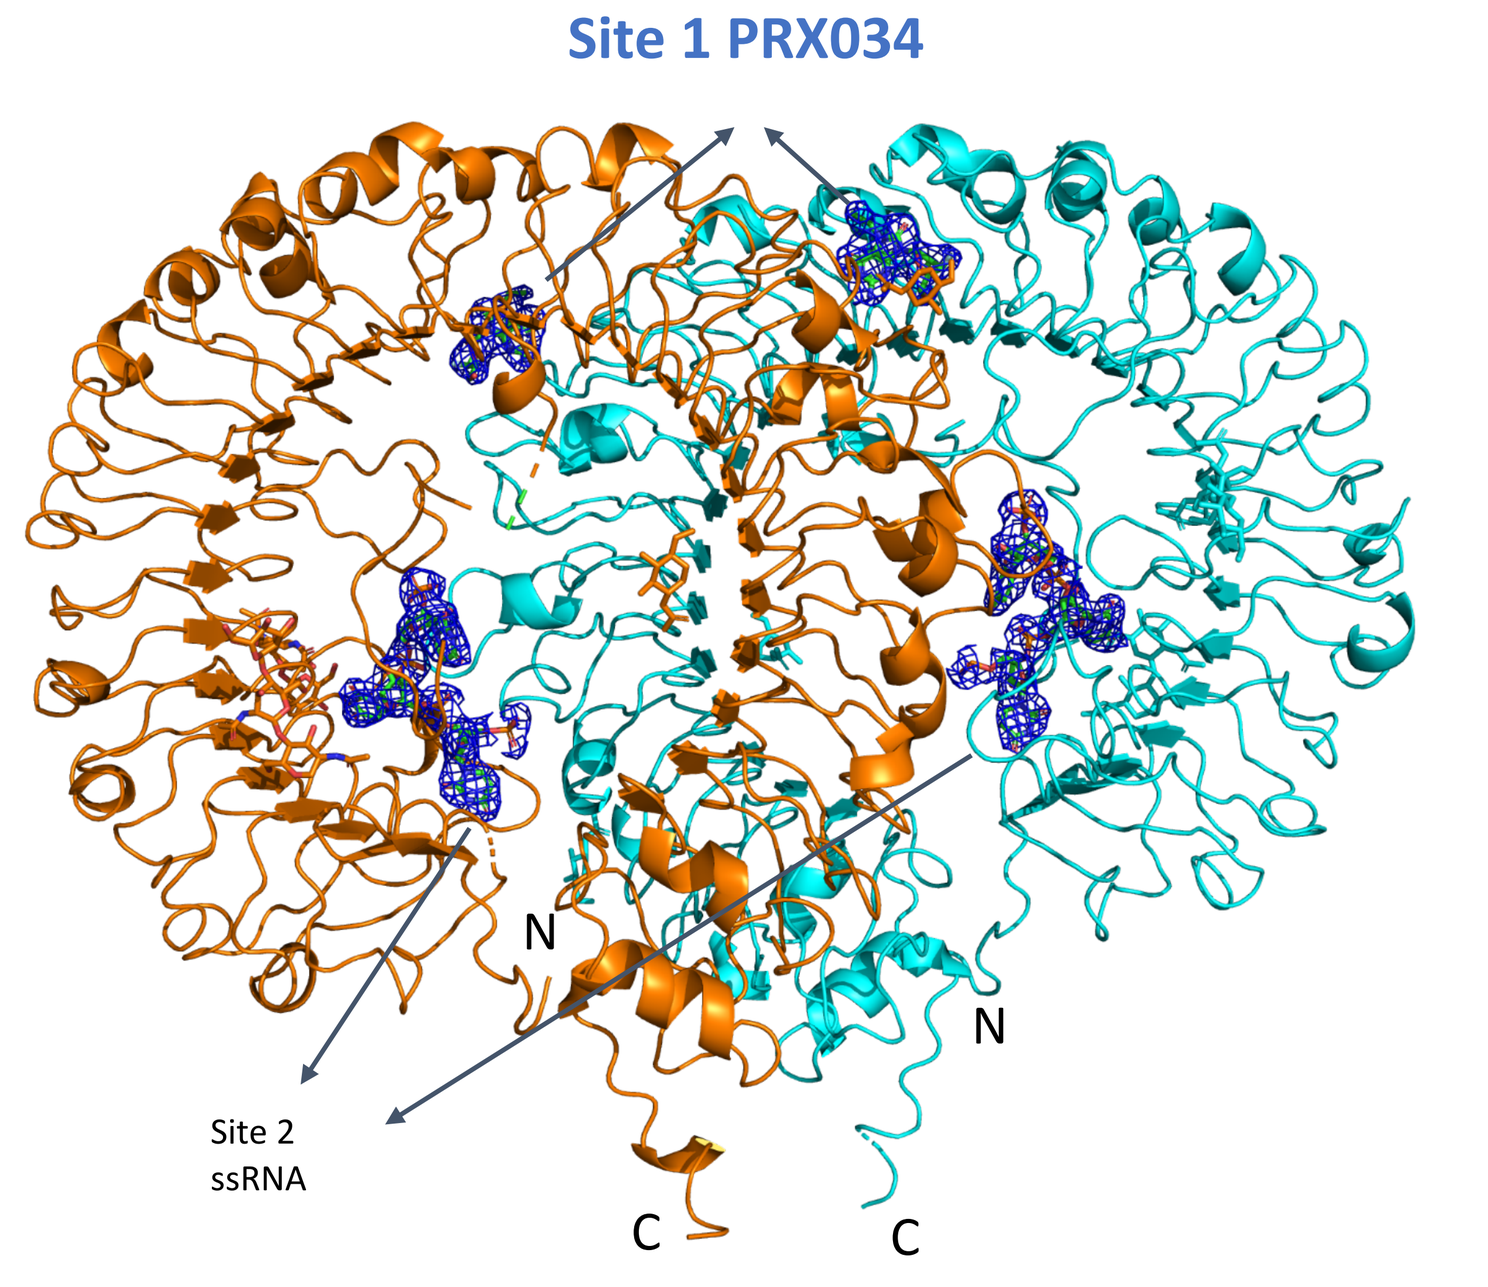
Site 1 PRX034
Site 2 ssRNA
N
N
C
C

TLR7-polyU-PRX034 Structure

PRX034 Activity in Human PBMCs

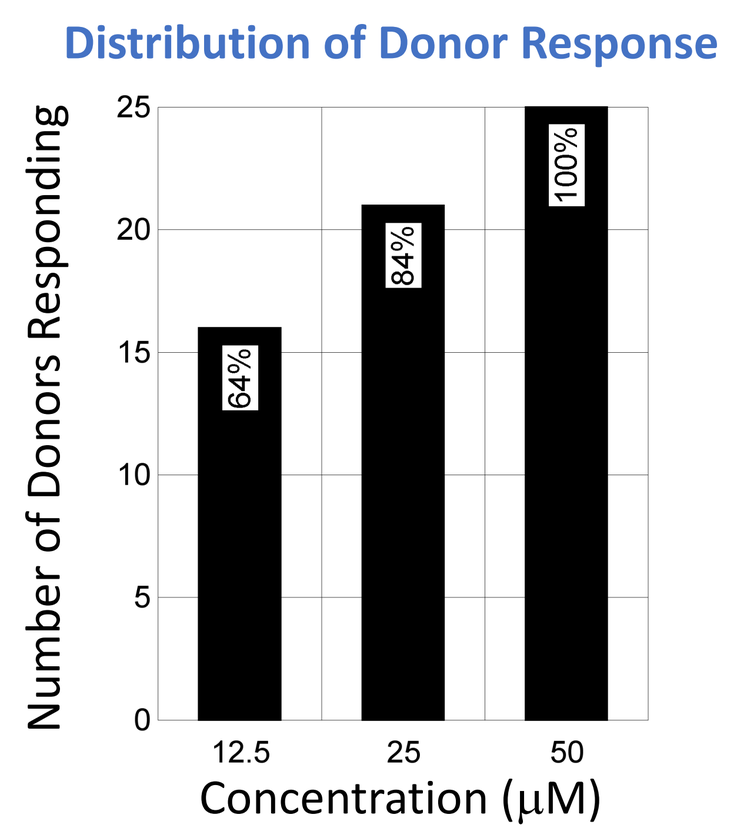
Distribution of Donor Response
Number of Donors Responding
64%
84%
100%
12.5
25
50
Concentration (μM)

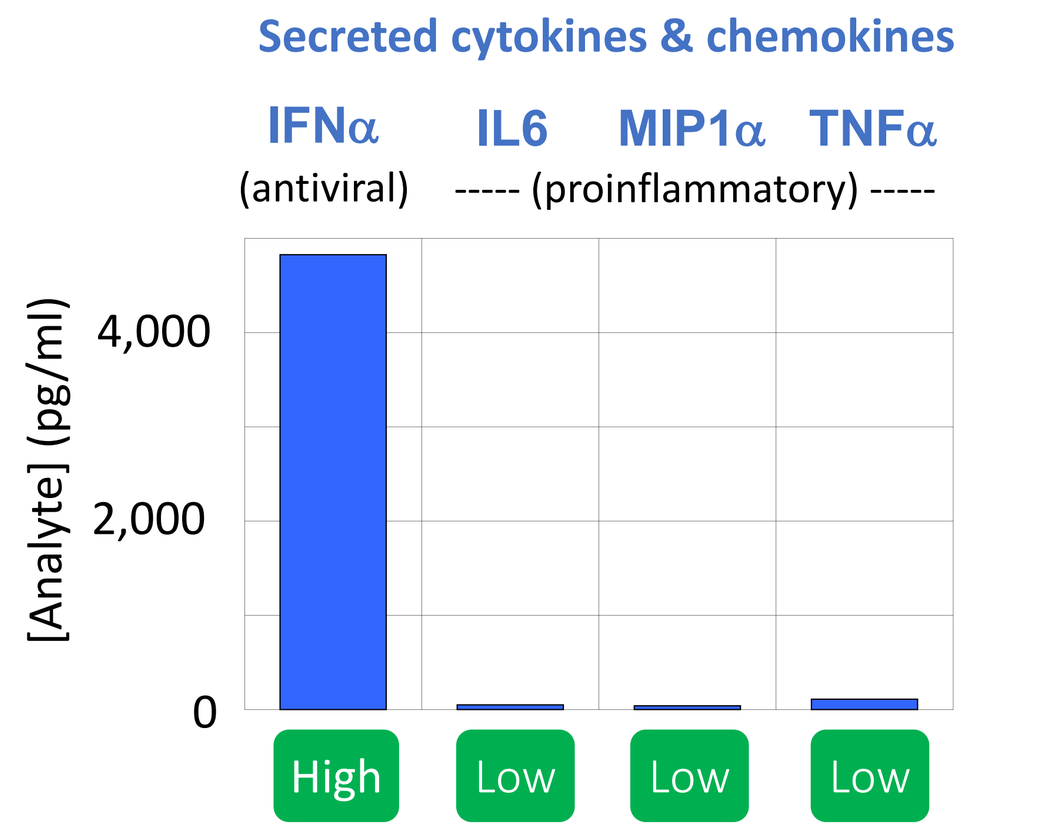
Secreted cytokines & chemokines
IFNα (antiviral)
IL6
MIP1α
TNFα
----- (proinflammatory) -----
[Analyte] (pg/ml)
High
Low
Low
Low

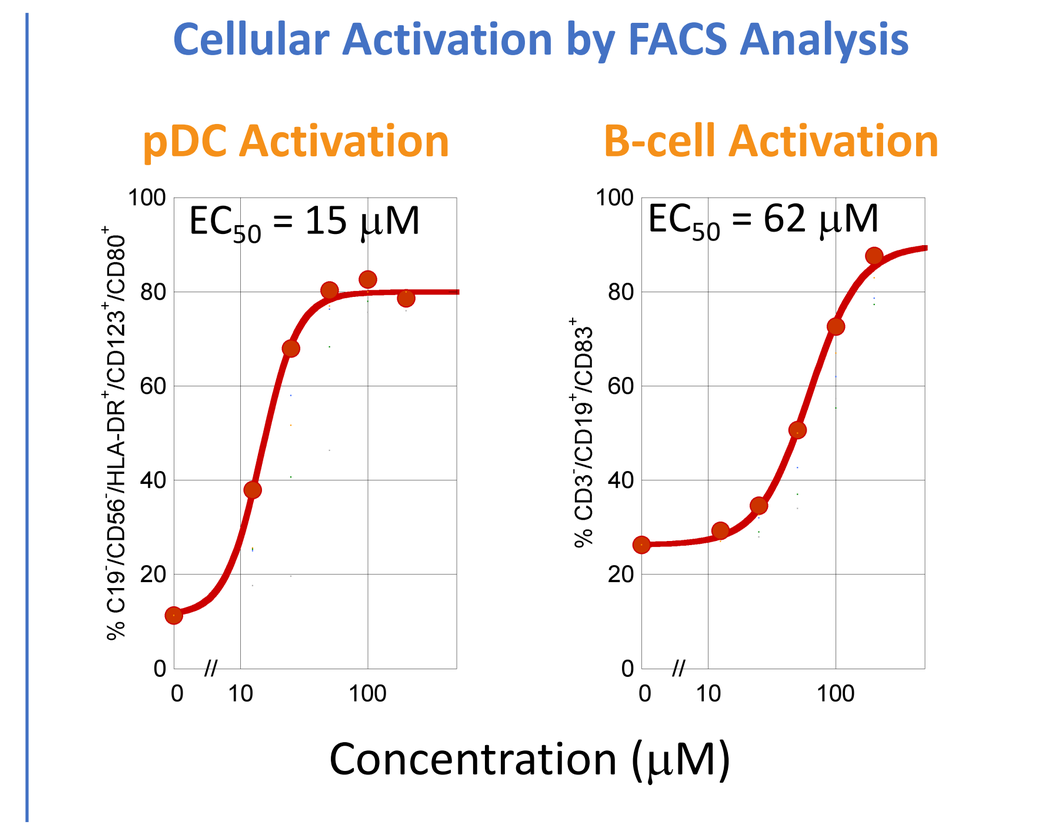
Cellular Activation by FACS Analysis
pDC Activation
EC50 = 15 μM
B-cell Activation
EC50 = 62 μM
Concentration (μM)

Weighted MEC (μM)		Ratio
Human	*Monkey*	
18	112	6.3

MEC = minimal effective concentration of PRX034 eliciting IFN in cell culture

- Activation of pDCs and secretion of significant IFNα occurs in the same concentration range; B-cell activation occurs at a somewhat higher concentration
- The profile of secreted cytokines and chemokines is notable for the magnitude of IFNa production and the relative absence of proinflammatory cytokines and chemokines
- PRX034 is 6.3-fold more potent (MEC 6.3-fold lower) in human PBMCs than in PBMCs from cynomolgus monkeys

PK of PRX034 after PRTX007 Oral Dosing in NHPs

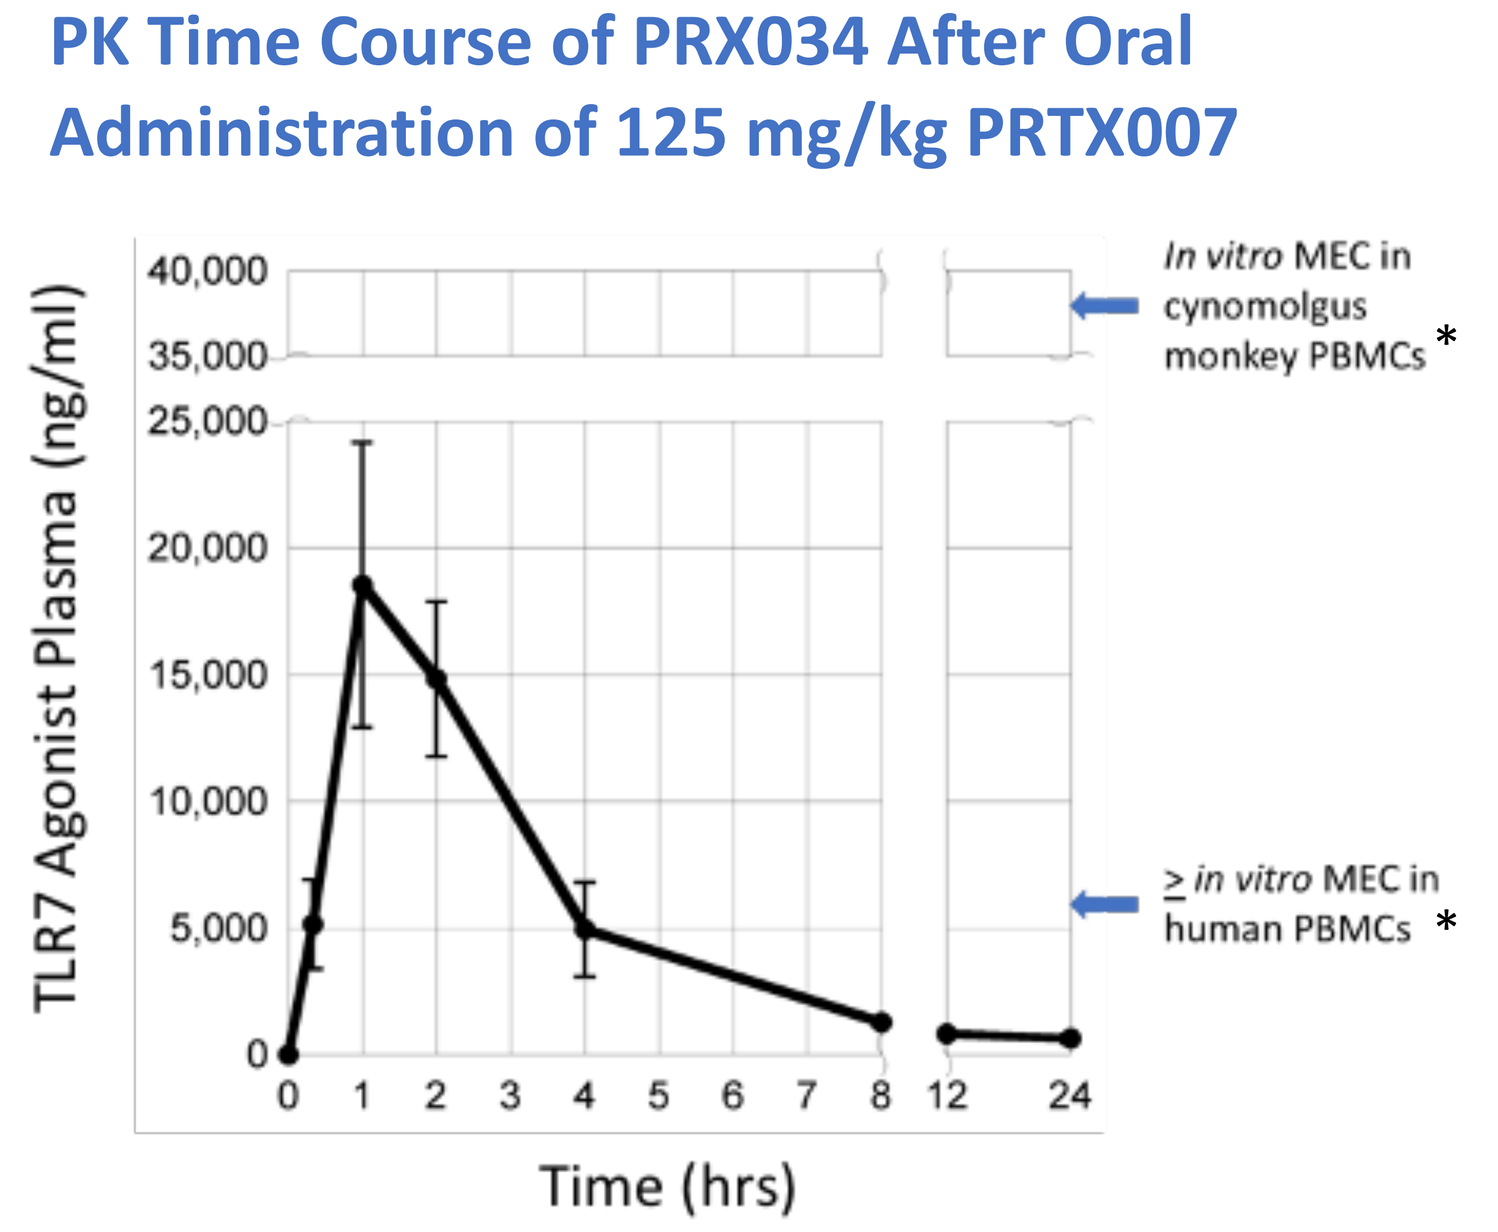
PK Time Course of PRX034 After Oral Administration of 125 mg/kg PRTX007
TLR7 Agonist Plasma (ng/ml)
Time (hrs)
In vitro MEC in cynomolgus monkey PBMCs *
≥ in vitro MEC in human PBMCs *

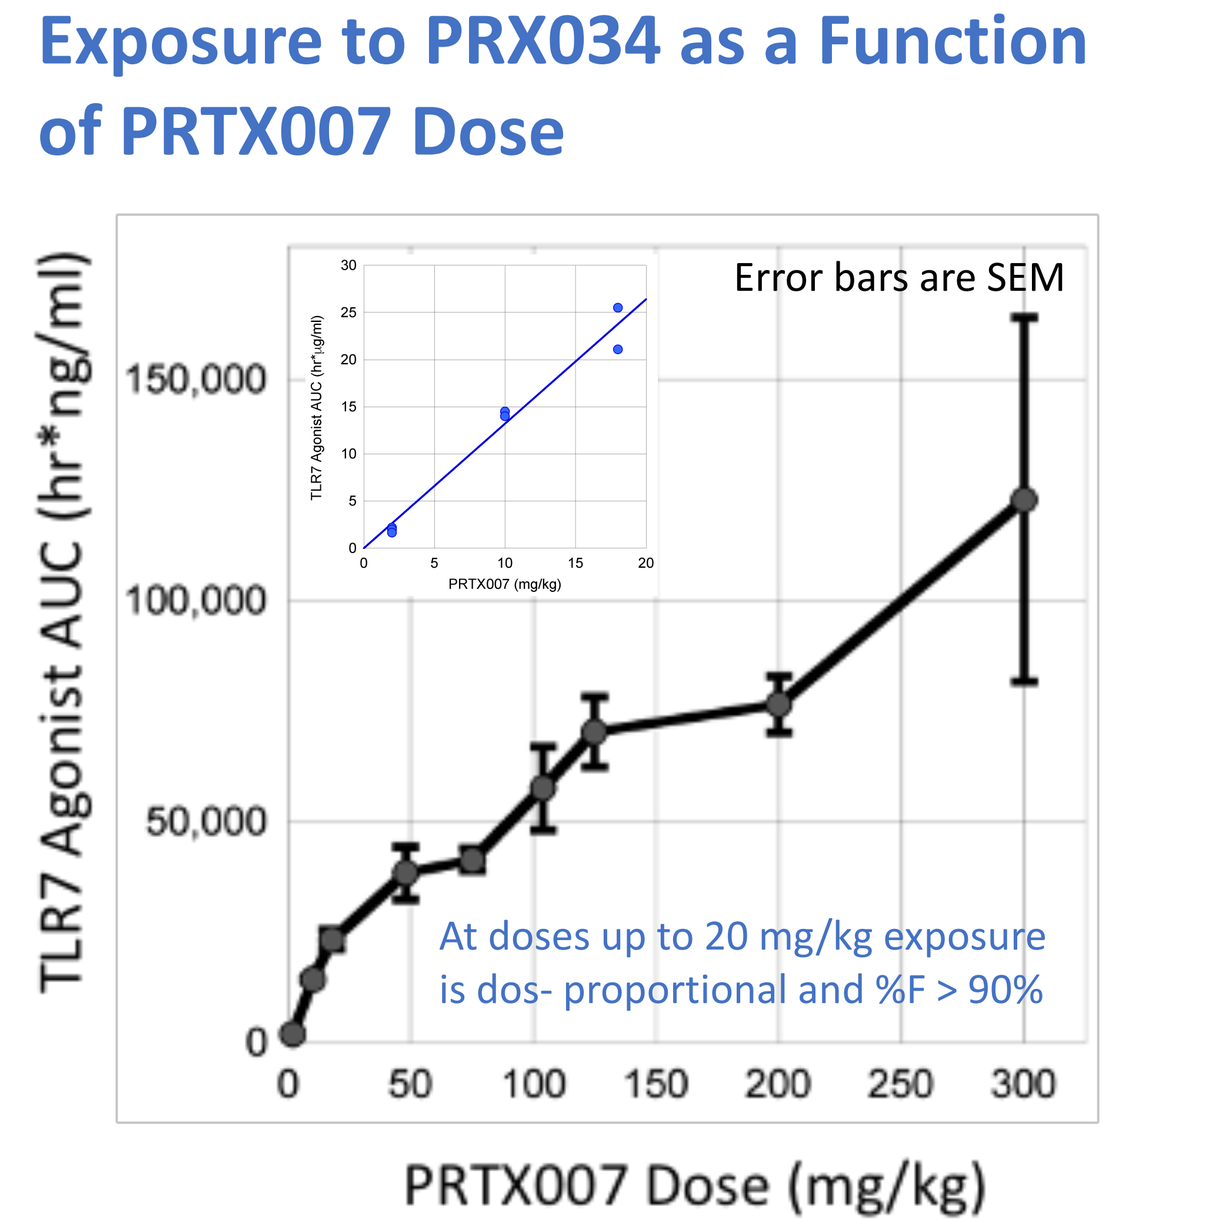
Exposure to PRX034 as a Function of PRTX007 Dose
Error bars are SEM
TLR7 Agonist AUC (hr*ng/ml)
At doses up to 20 mg/kg exposure is dos- proportional and %F > 90%
PRTX007 Dose (mg/kg)

- After oral administration, PRTX007 is rapidly absorbed and converted to the active agonist PRX034 which distributes systemically at pharmacologically active levels
- As with many other TLR7 agonists, pDC activation *in vivo* occurs at lower concentrations than required for activiation in culture. For example, note that peak plasma levels of PRX034 achieved with 125 mg/kg of PRTX007, a highly pharmacologically active dose, are significantly less than the MEC in monkey PBMCs *in vitro*

Multidose PK/PD Response in Cynomolgus Monkeys

Primates are the only relevant species for evaluation

Summary

- Therapeutic and supra-therapeutic levels well-tolerated; no adverse effects in any dose group
- Well-behaved PK across all dose groups
- Dose-dependent robust immune induction:
 - Type I IFN production and typical TLR7 mediated responses
 - Minimal engagement of NFκβ proinflammatory pathway
 - Consistent immune induction on QOD schedule, characteristic increased response on second dose administered 24 hrs after first dose on QD schedule
 - Variability in response consistent with that reported for the clinical-stage investigational TLR7 agonist prodrug ANA773
- Dose dependence consistent with expected PO daily human dose of 200 – 400 mg*

Basic Study Design

Dosing Schedule: 6 x QOD (days 1–11), 3 x QD (days 11–13)

Day	1	2	3	4	5	6	7	8	9	10	11	12	13	14
Dose	x		x		x		x		x		x	x	x	
PK (hr)														
0	x										x			
0.33	x										x			
1	x										x			
2	x		x		x		x		x		x	x	x	
4	x										x			
8	x										x			
12	x										x			
24	x										x			
PD (hr)														
0	x		x		x		x		x		x			
4	x		x		x		x		x		x	x	x	
8	x		x		x		x		x		x	x	x	
24	x		x		x		x		x		x	x	x	x

PD Response: Repeated Dosing at 125 mg/kg PRTX007

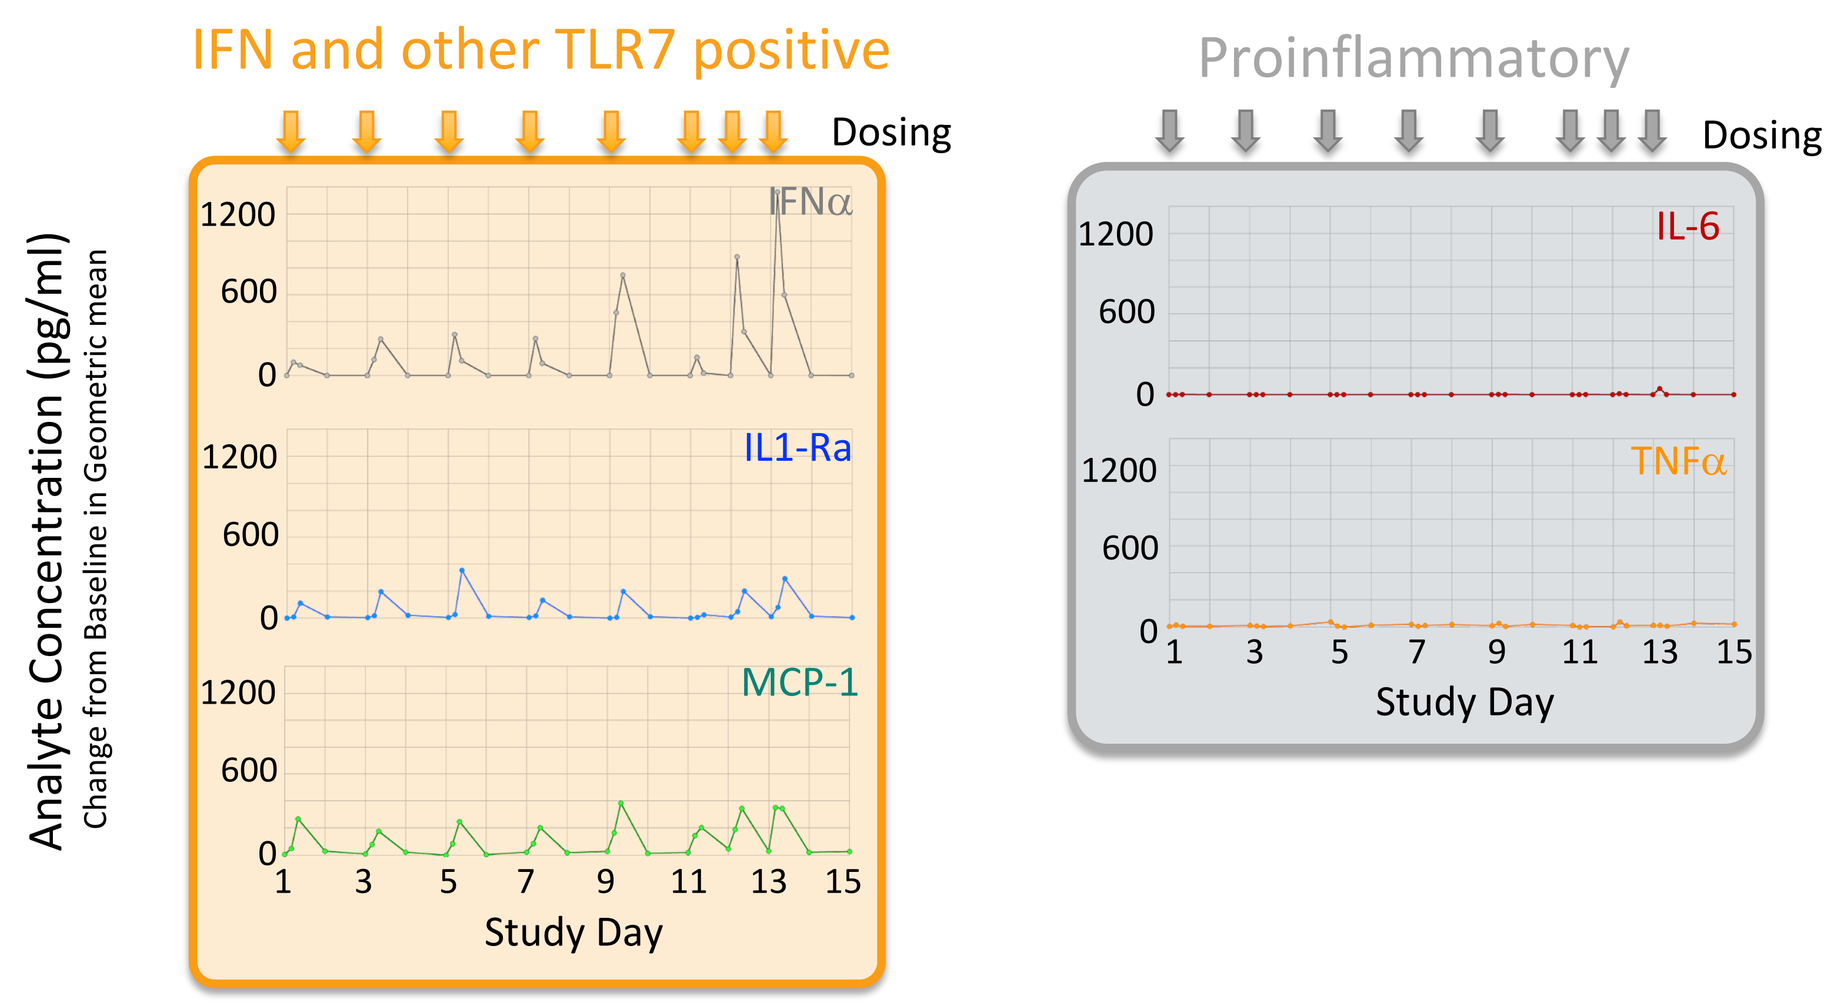
IFN and other TLR7 positive
Dosing
IFNα
IL1-Ra
MCP-1
Proinflammatory
IL-6
TNFα
Analyte Concentration (pg/ml)
Change from Baseline in Geometric mean
Study Day

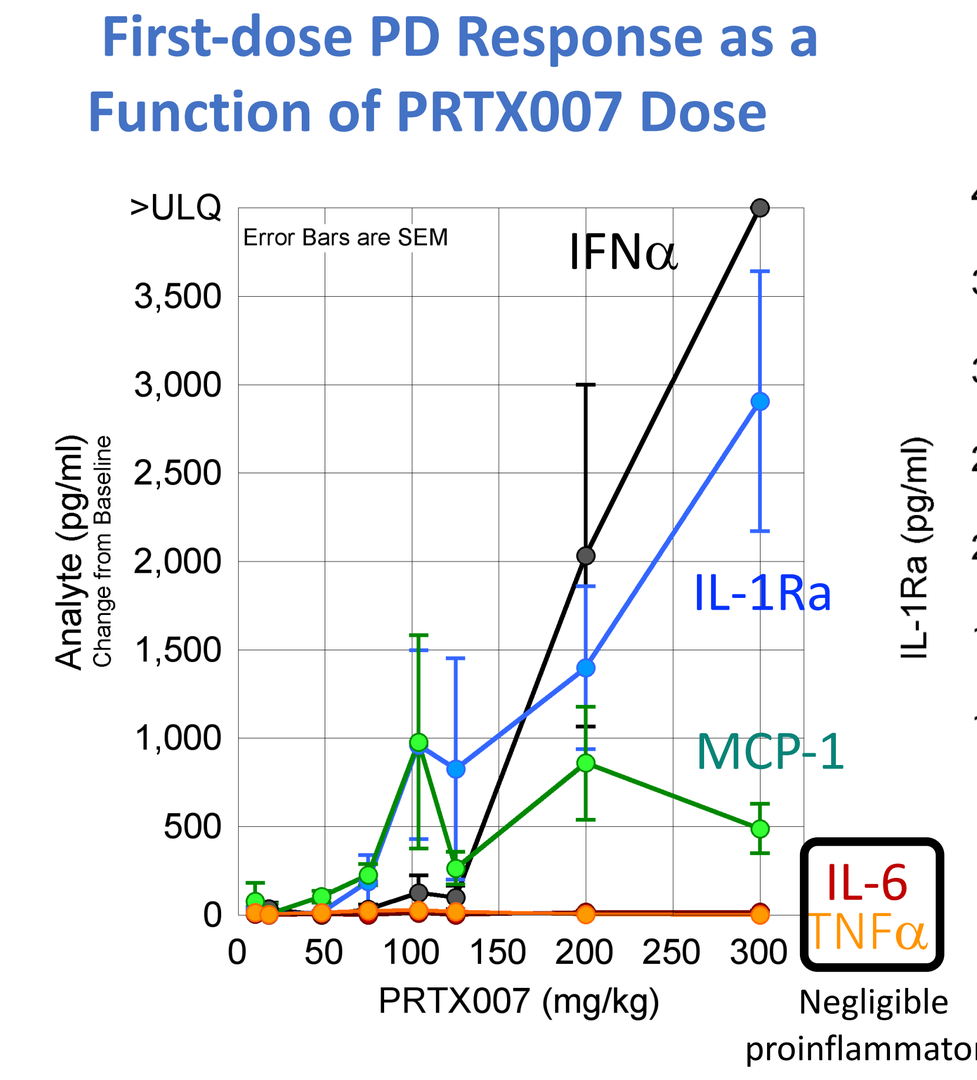
First-dose PD Response as a Function of PRTX007 Dose
Error Bars are SEM
IFNα
IL-1Ra
MCP-1
IL-6
TNFα
Analyte (pg/ml)
PRTX007 (mg/kg)
Negligible proinflammatory

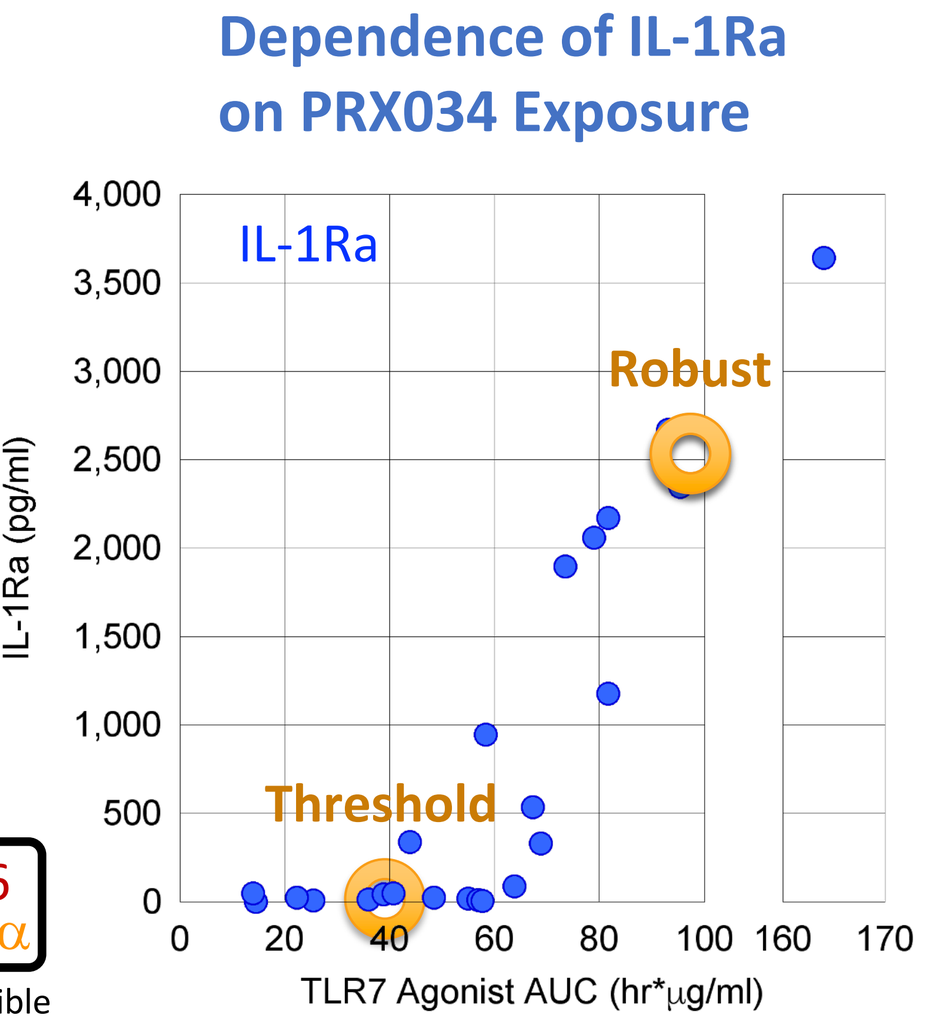
Dependence of IL-1Ra on PRX034 Exposure
IL-1Ra
Robust
Threshold
IL-1Ra (pg/ml)
TLR7 Agonist AUC (hr*μg/ml)

- The preferred pharmacologic response elicited by PRTX007 *in vitro* (lack of proinflammatory cytokines) is also observed in these multi-dose studies *in vivo*
 - For example, dose levels of 200 and 300 mg/kg produce extremely robust but focused immune induction with circulating IFN levels in some animals well in excess of 4,000 pg/ml.
 - In contrast, the circulating levels of proinflammatory factors like TNFa and IL-6 are at or less than the limits of detection
- An antiviral state is created by pDC activation at doses where circulating IFN is not elevated
 - This corresponds to doses where IL-1Ra and MCP-1 are elevated but IFNα increase is not observed
 - This is attributable to paracrine transfer of intererons and other factors directly from pDCs to target cells
- The pharmacodynamic response upon repeated dosing is reproducible
- Induction of IL-1Ra, a potent antagonist of IL-1β, may alleviate COVID19-associated inflammation
- Based upon difference in intrinsic potency across species and with allometric scaling, we expect human dose to be 100 – 250 mg

How Does PRTX007 Avoid Inflammation?

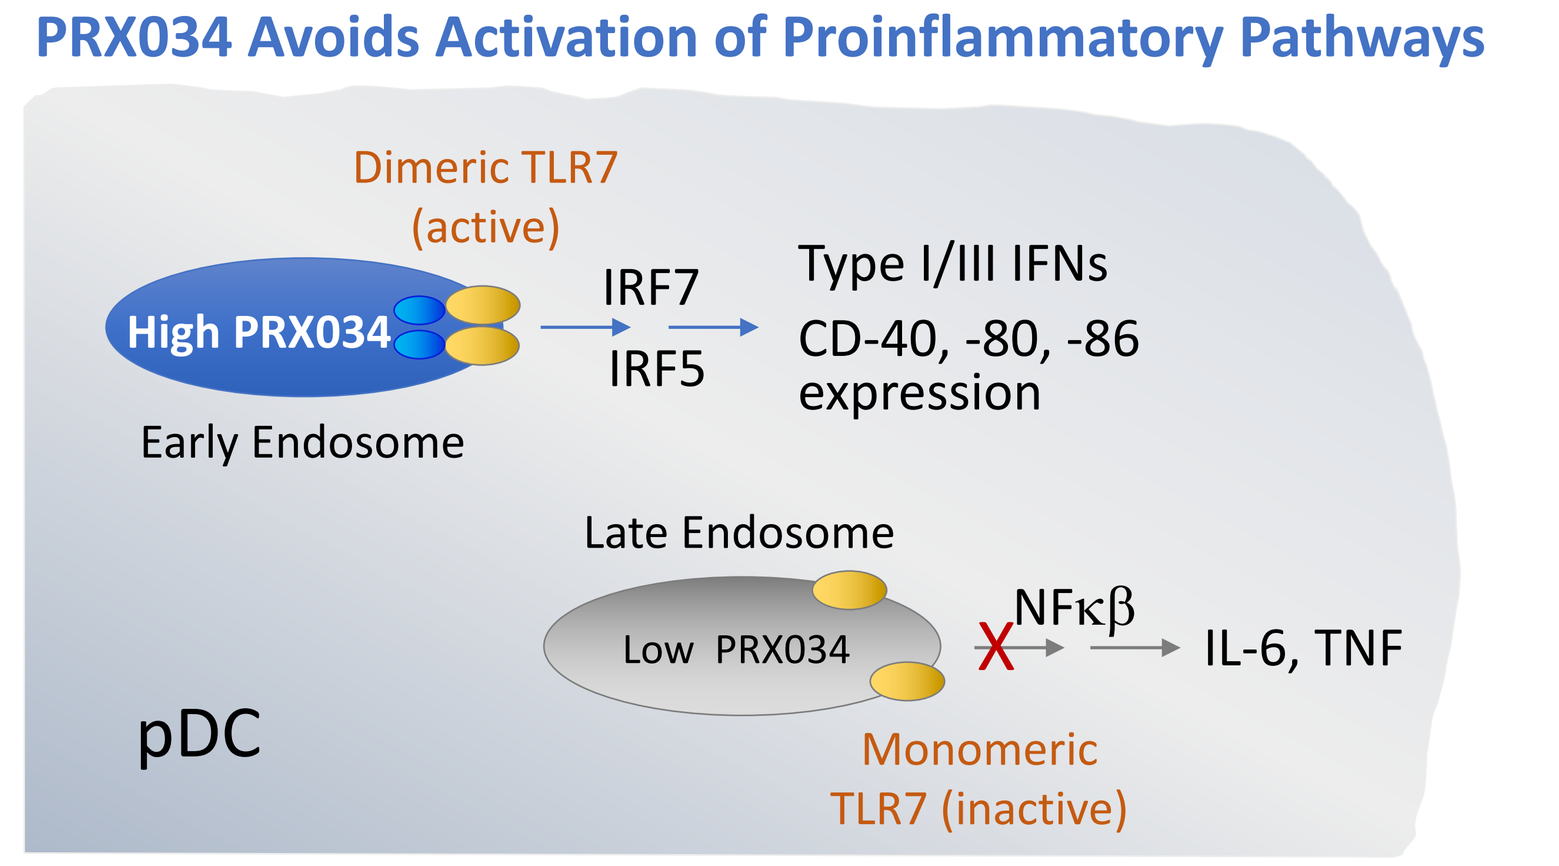
PRX034 Avoids Activation of Proinflammatory Pathways
Dimeric TLR7 (active)
High PRX034
IRF7
IRF5
Type I/III IFNs
CD-40, -80, -86 expression
Early Endosome
Late Endosome
Low PRX034
NFκβ
IL-6, TNF
pDC
Monomeric TLR7 (inactive)

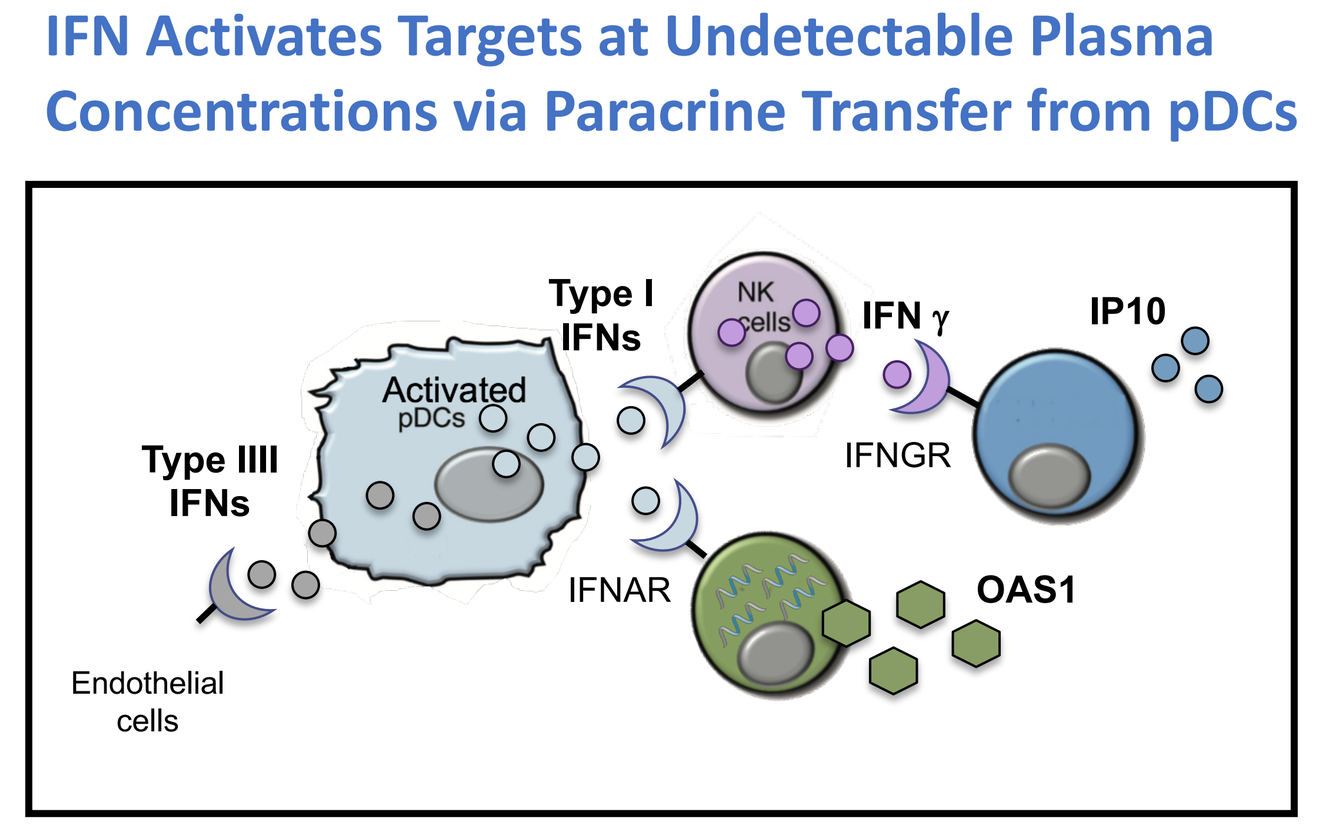
IFN Activates Targets at Undetectable Plasma Concentrations via Paracrine Transfer from pDCs
Type I IFNs
NK cells
IFN γ
IP10
Activated pDCs
IFNGR
Type IIII IFNs
IFNAR
OAS1
Endothelial cells

STATUS

- ❖ We are currently conducting IND-enabling studies with PRTX007
- ❖ Initial evaluation in humans will be SAD and MAD studies in healthy volunteers expected to complete in early 2021; this will confirm relevant dose levels and characteristics of induced immune state
- ❖ The current development plan enables treatment of both COVID-19 patients and patients with cancer

ACKNOWLEDGEMENTS

EVOTEC

In Vitro Pharmacology: Filippo Andreetta, Maria Elisa Perico, Elisa Bianchini

Protein Sciences: Rahul Yadav, Velupillai Srikannathasan, Karthik Rajasekar

DMPK: Marisa Dal Lago, Stefano Fontana, Serenella Zambon, Jerome Menegotto

Alliance Management: Sadaf Kasraie

Computational Chemistry: Iuni Margaret Laura Trist

GVK BIO

Synthetic/Medicinal Chemistry: Hari Krishna Chava, Satyanarayana Reddy Avuluri, Santhosh Konna, Raju Guduri, M. Shambabu Joseph

Team, many thanks to all of you - you did the heavy lifting! Jim & Steve