6019

Discovery of a Series of Novel Toll-Like Receptor 7 Agonists for Systemic Immunotherapy of Cancer

James R. Appleman
Stephen E. Webber

primmune Therapeutics

ABSTRACT

While ICPIs (immune checkpoint inhibitors) have fundamentally changed the practice of cancer therapy for tumors arising from many different tissues, ways to increase both response rate and durability are critically needed. On theoretical grounds, combining stimulators of innate immunity with activators of adaptive immunity should lead to better outcomes. Consistent with this hypothesis, encouraging results have been obtained with intratumoral injections of TLR9 agonists in combination with ICPIs in treatment-naïve patients and in patients that have failed previous ICPI therapy. However, there are substantial limitations inherent in any drug that must be administered intratumorally, particularly where repeated administration is preferred. We therefore have invented a novel series of TLR7 agonists that engage the same elements of the immune system as TLR9 agonists while having the advantages of oral delivery and safe systemic engagement of the immune system. The ability to successfully achieve such a profile while maintaining a reasonable therapeutic window was previously demonstrate by Anadys Pharmaceuticals with ANA773. This drug candidate was evaluated in healthy volunteers, chronic HCV and HBV patients and treatment-experienced solid tumor patients. Based upon our analysis of desirable pharmacologic features of ANA773 and challenges faced by molecules like 3M-852A, where no therapeutic window could be identified, we have derived a critical set of unusual target properties that are evaluated in our testing cascade:

- Intrinsic potency for TLR7 activation in a defined range that maintains ability to vary degree of engagement with concentration
- Specificity for TLR7 and, in particular, a lack of activity at TLR8
- Defined target profile of cytokine and chemokine induction in human peripheral blood mononuclear cells
- Efficient delivery in primates to systemic circulation by prodrugs that intrinsically lack TLR7 agonist activity themselves
- PK volume of distribution ~ 1 L/kg
- Relatively short PK $t_{1/2}$ with substantially more long-lived pharmacodynamic response

We hypothesize that a molecule with the above criteria can be dosed QOD continuously over a 24-month period to appropriately engage innate immunity that is well-tolerated by the patient and increases response rate and durability.

From our original starting point – a relatively weak TLR7 agonist with no oral bioavailability – we have invented a novel series of molecules meeting our target profile. We are currently engaged in evaluation of three leading candidates in advanced models with the objective of selecting one to proceed into IND-enabling studies.

BACKGROUND

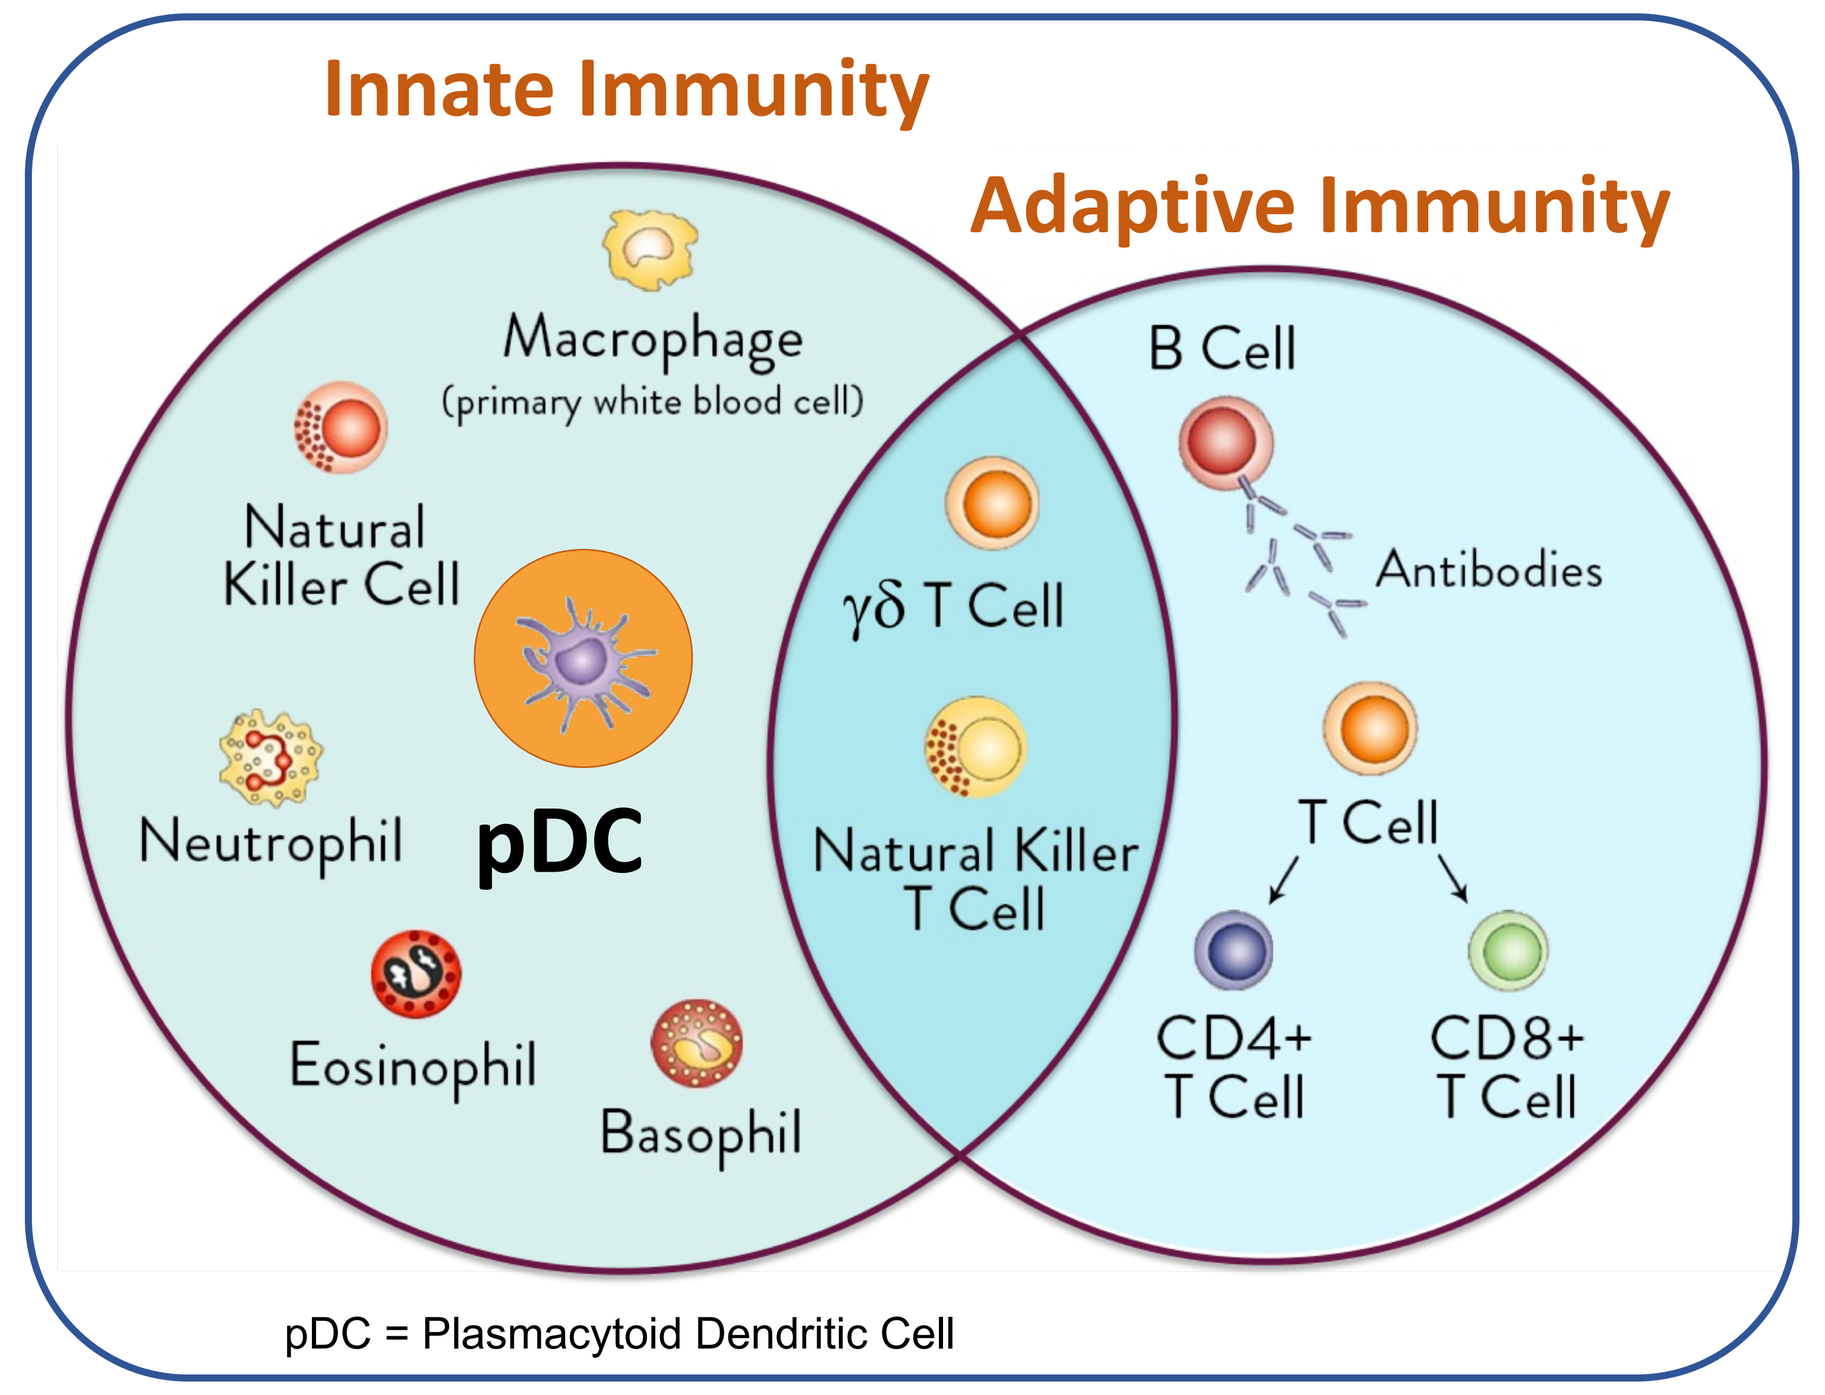
Innate Immunity
Adaptive Immunity
Macrophage
(primary white blood cell)
Natural Killer Cell
Neutrophil
pDC
Eosinophil
Basophil
γδ T Cell
Natural Killer T Cell
B Cell
Antibodies
T Cell
CD4+ T Cell
CD8+ T Cell
pDC = Plasmacytoid Dendritic Cell

Activation of both **Innate** and **Adaptive** immunity is expected to improve treatment outcome.

We are targeting activation of plasmacytoid dendritic cells (**pDCs**), which orchestrate the interplay between the two arms of the immune system.

TLR7 agonists engage a variety of antitumor responses, most of which are initiated by activation of plasmacytoid dendritic cells.

Inactivated pDCs in the tumor microenvironment are pro-tumorogenic, whereas activated pDCs elicit a plethora of antitumoral activities

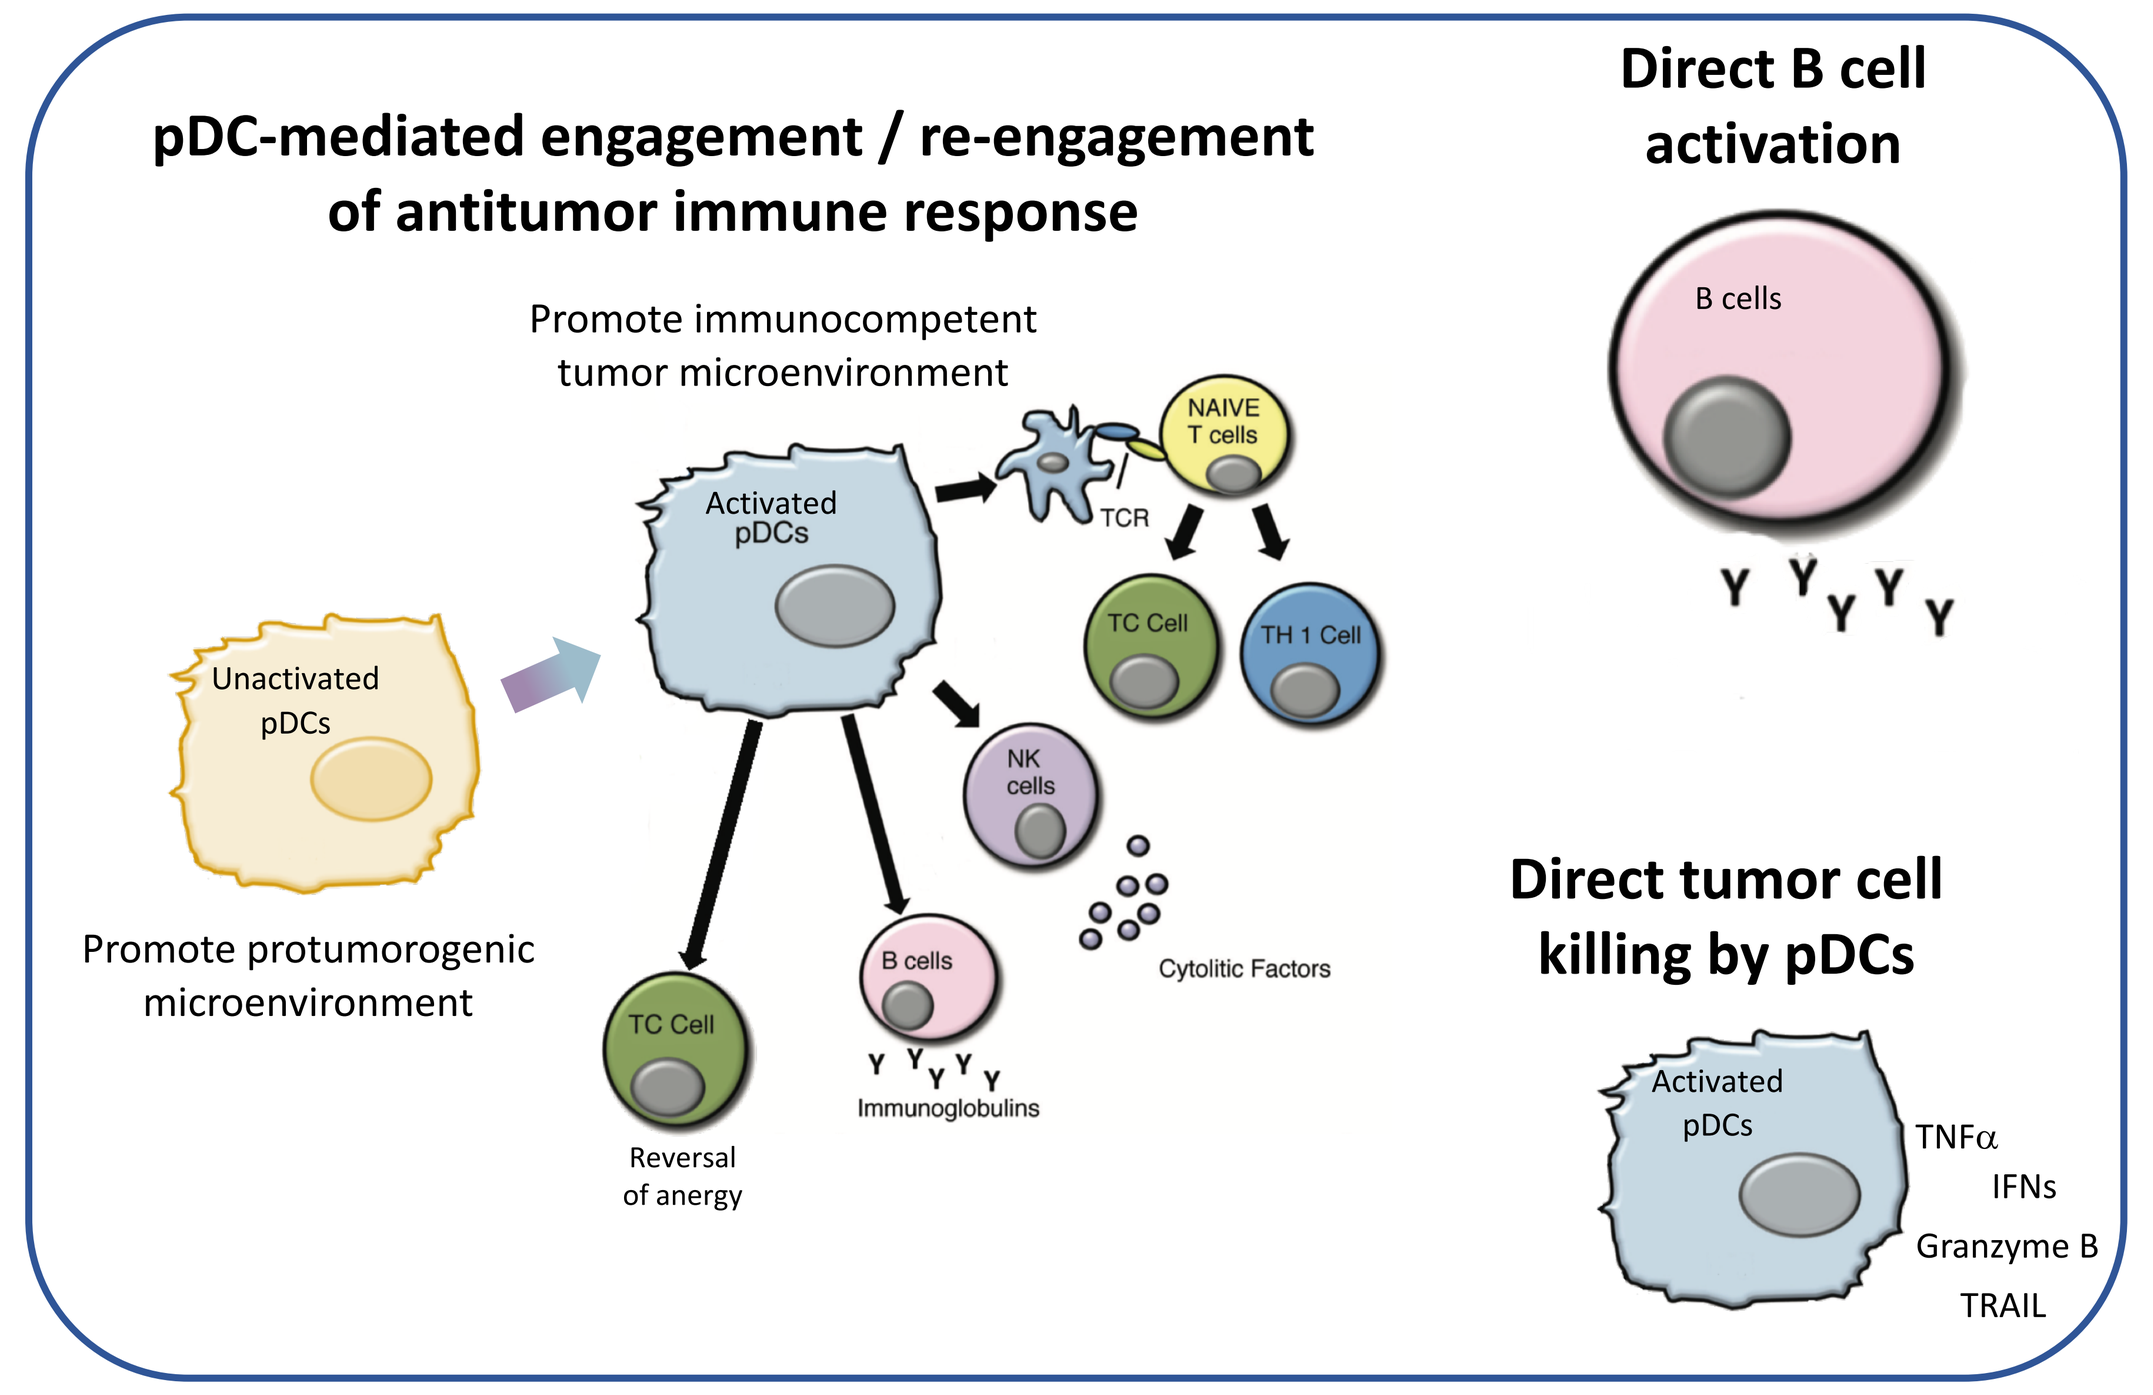
pDC-mediated engagement / re-engagement of antitumor immune response
Promote immunocompetent tumor microenvironment
Activated pDCs
NAIVE T cells
TCR
TC Cell
TH 1 Cell
NK cells
Cytolitic Factors
B cells
Immunoglobulins
TC Cell
Reversal of anergy
Unactivated pDCs
Promote protumorogenic microenvironment
Direct B cell activation
B cells
Direct tumor cell killing by pDCs
Activated pDCs
TNFα
IFNs
Granzyme B
TRAIL

DISCOVERY THESIS

- ANA773 from Anadys was an interesting tool compound in establishing clinical POC
- The single feature targeted for improvement in Primmune's program is intrinsic cellular potency to address following clinical limitations:
 - Ability to investigate full range of **immune induction** limited by the amount of drug required per dose
 - **Plateau in C_{max}** at the high end of the investigated dose range, blunting impact of increased applied dose

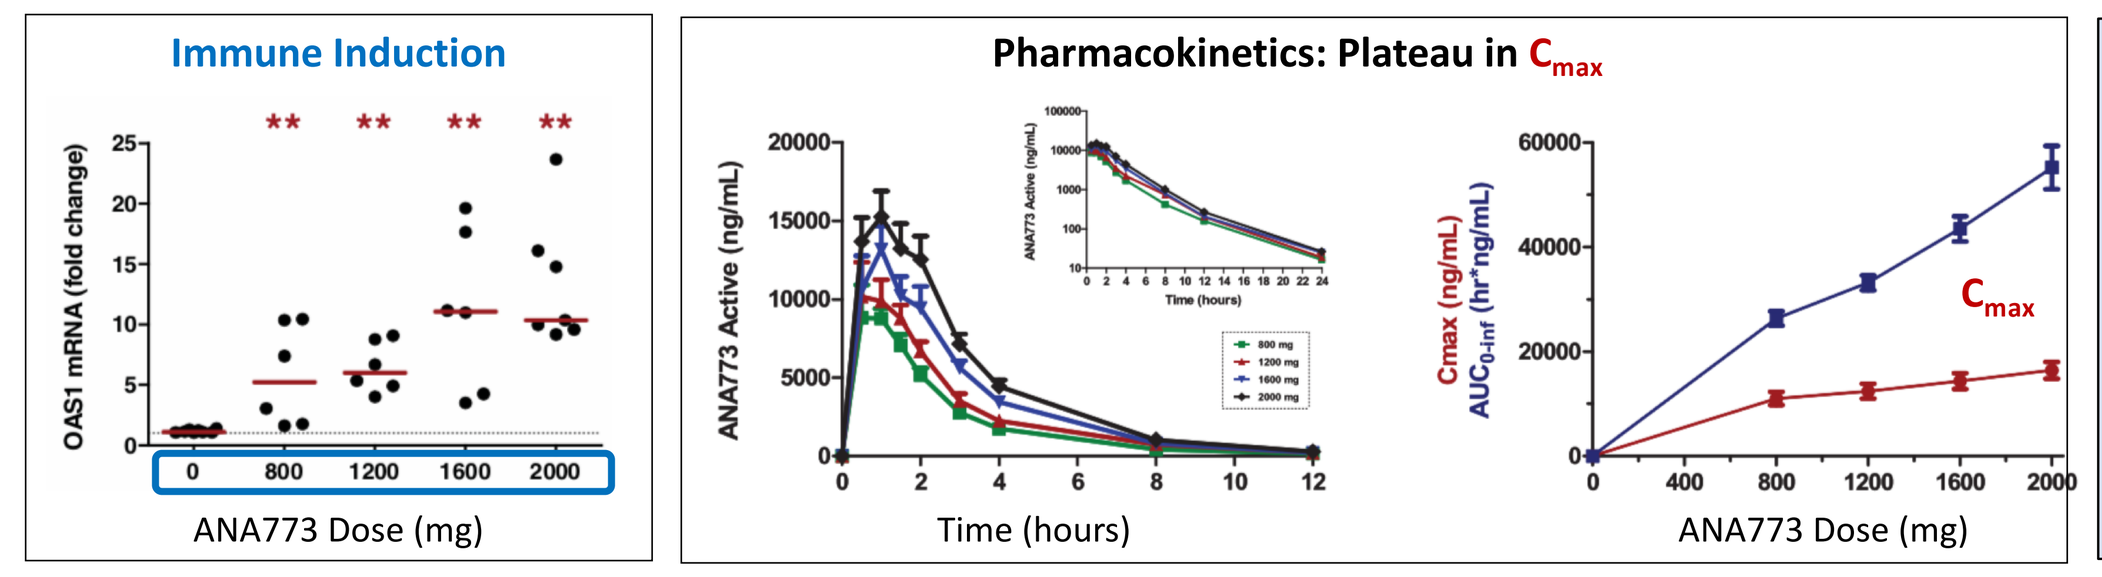
Immune Induction
OAS1 mRNA (fold change)
ANA773 Dose (mg)
Pharmacokinetics: Plateau in Cmax
ANA773 Active (ng/mL)
Time (hours)
Cmax (ng/mL)
AUC0-inf (hr*ng/mL)
Cmax
ANA773 Dose (mg)

Reference: ANA773, an Oral Inducer of Endogenous Interferons that Acts Via TLR7, Reduced Serum Viral Load in Patients Chronically Infected with HCV. Janssen et al. Hepatology (2009) 50(4): 1022A Abstract 1560 Poster presentation at AASLD 2009

- Other features (e.g. target selectivity, cytokine/chemokine profile elicited by agonist, PK and efficient oral delivery in cynomolgus monkeys) of ANA773 and its parent TLR7 agonist, ANA122, to be matched by Primmune's compounds.

TESTING CASCADE

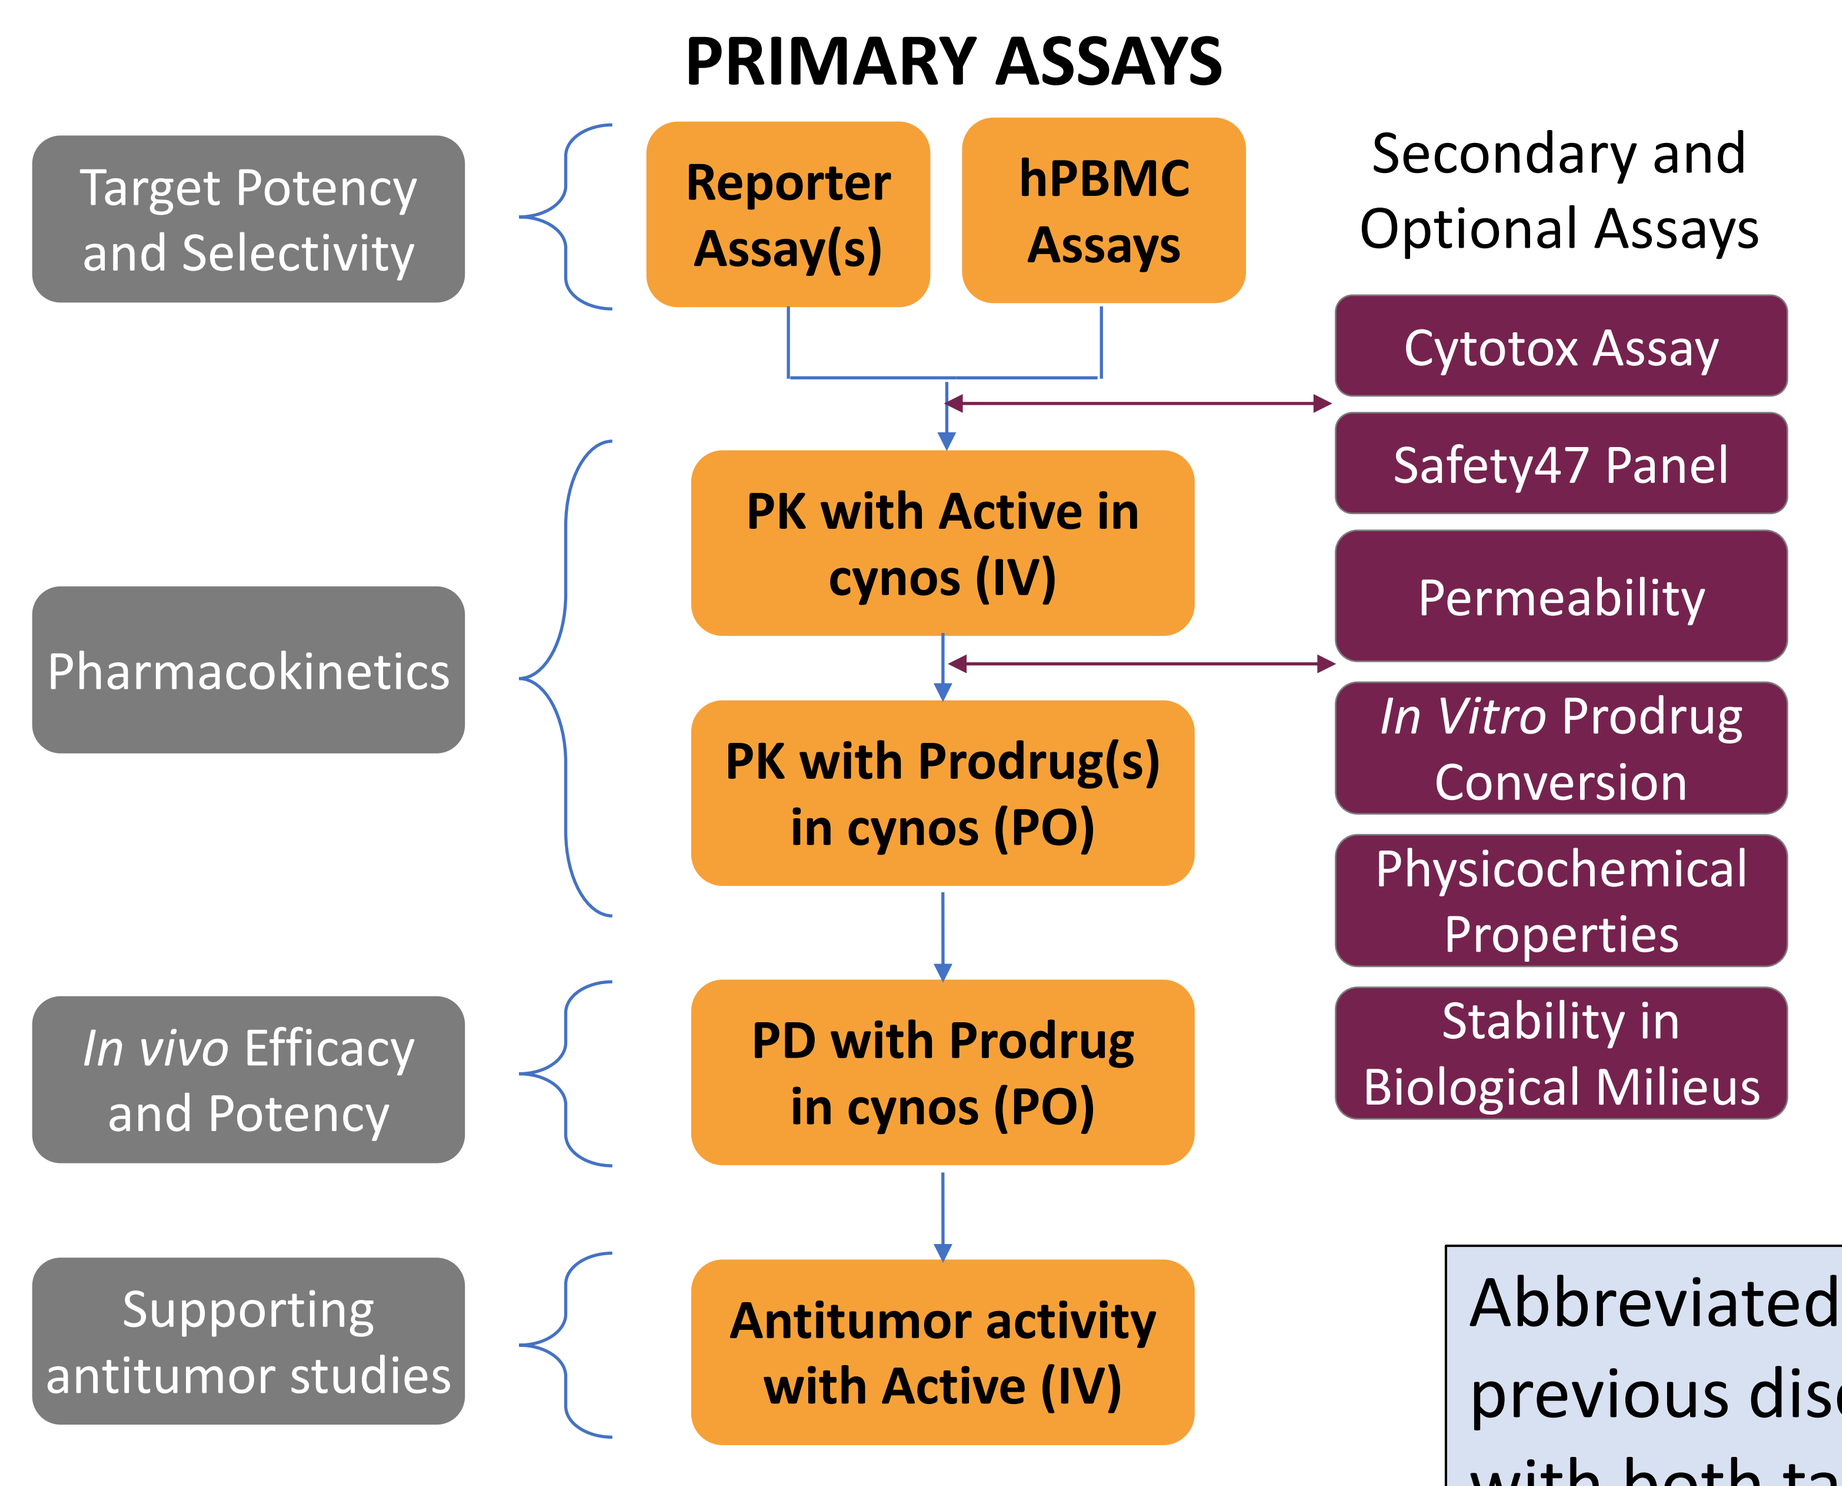
PRIMARY ASSAYS
Target Potency and Selectivity
Reporter Assay(s)
hPBMC Assays
Pharmacokinetics
PK with Active in cynos (IV)
PK with Prodrug(s) in cynos (PO)
In vivo Efficacy and Potency
PD with Prodrug in cynos (PO)
Supporting antitumor studies
Antitumor activity with Active (IV)
Secondary and Optional Assays
Cytotox Assay
Safety47 Panel
Permeability
In Vitro Prodrug Conversion
Physicochemical Properties
Stability in Biological Milieus

- Demonstration that compounds are not intrinsically cytotoxic
- General safety predictor
- Evaluation of possible limitations in oral delivery by prodrugs

Abbreviated testing cascade enabled by previous discovery and clinical experience with both target and compound class

MEDICINAL CHEMISTRY EVOLUTION

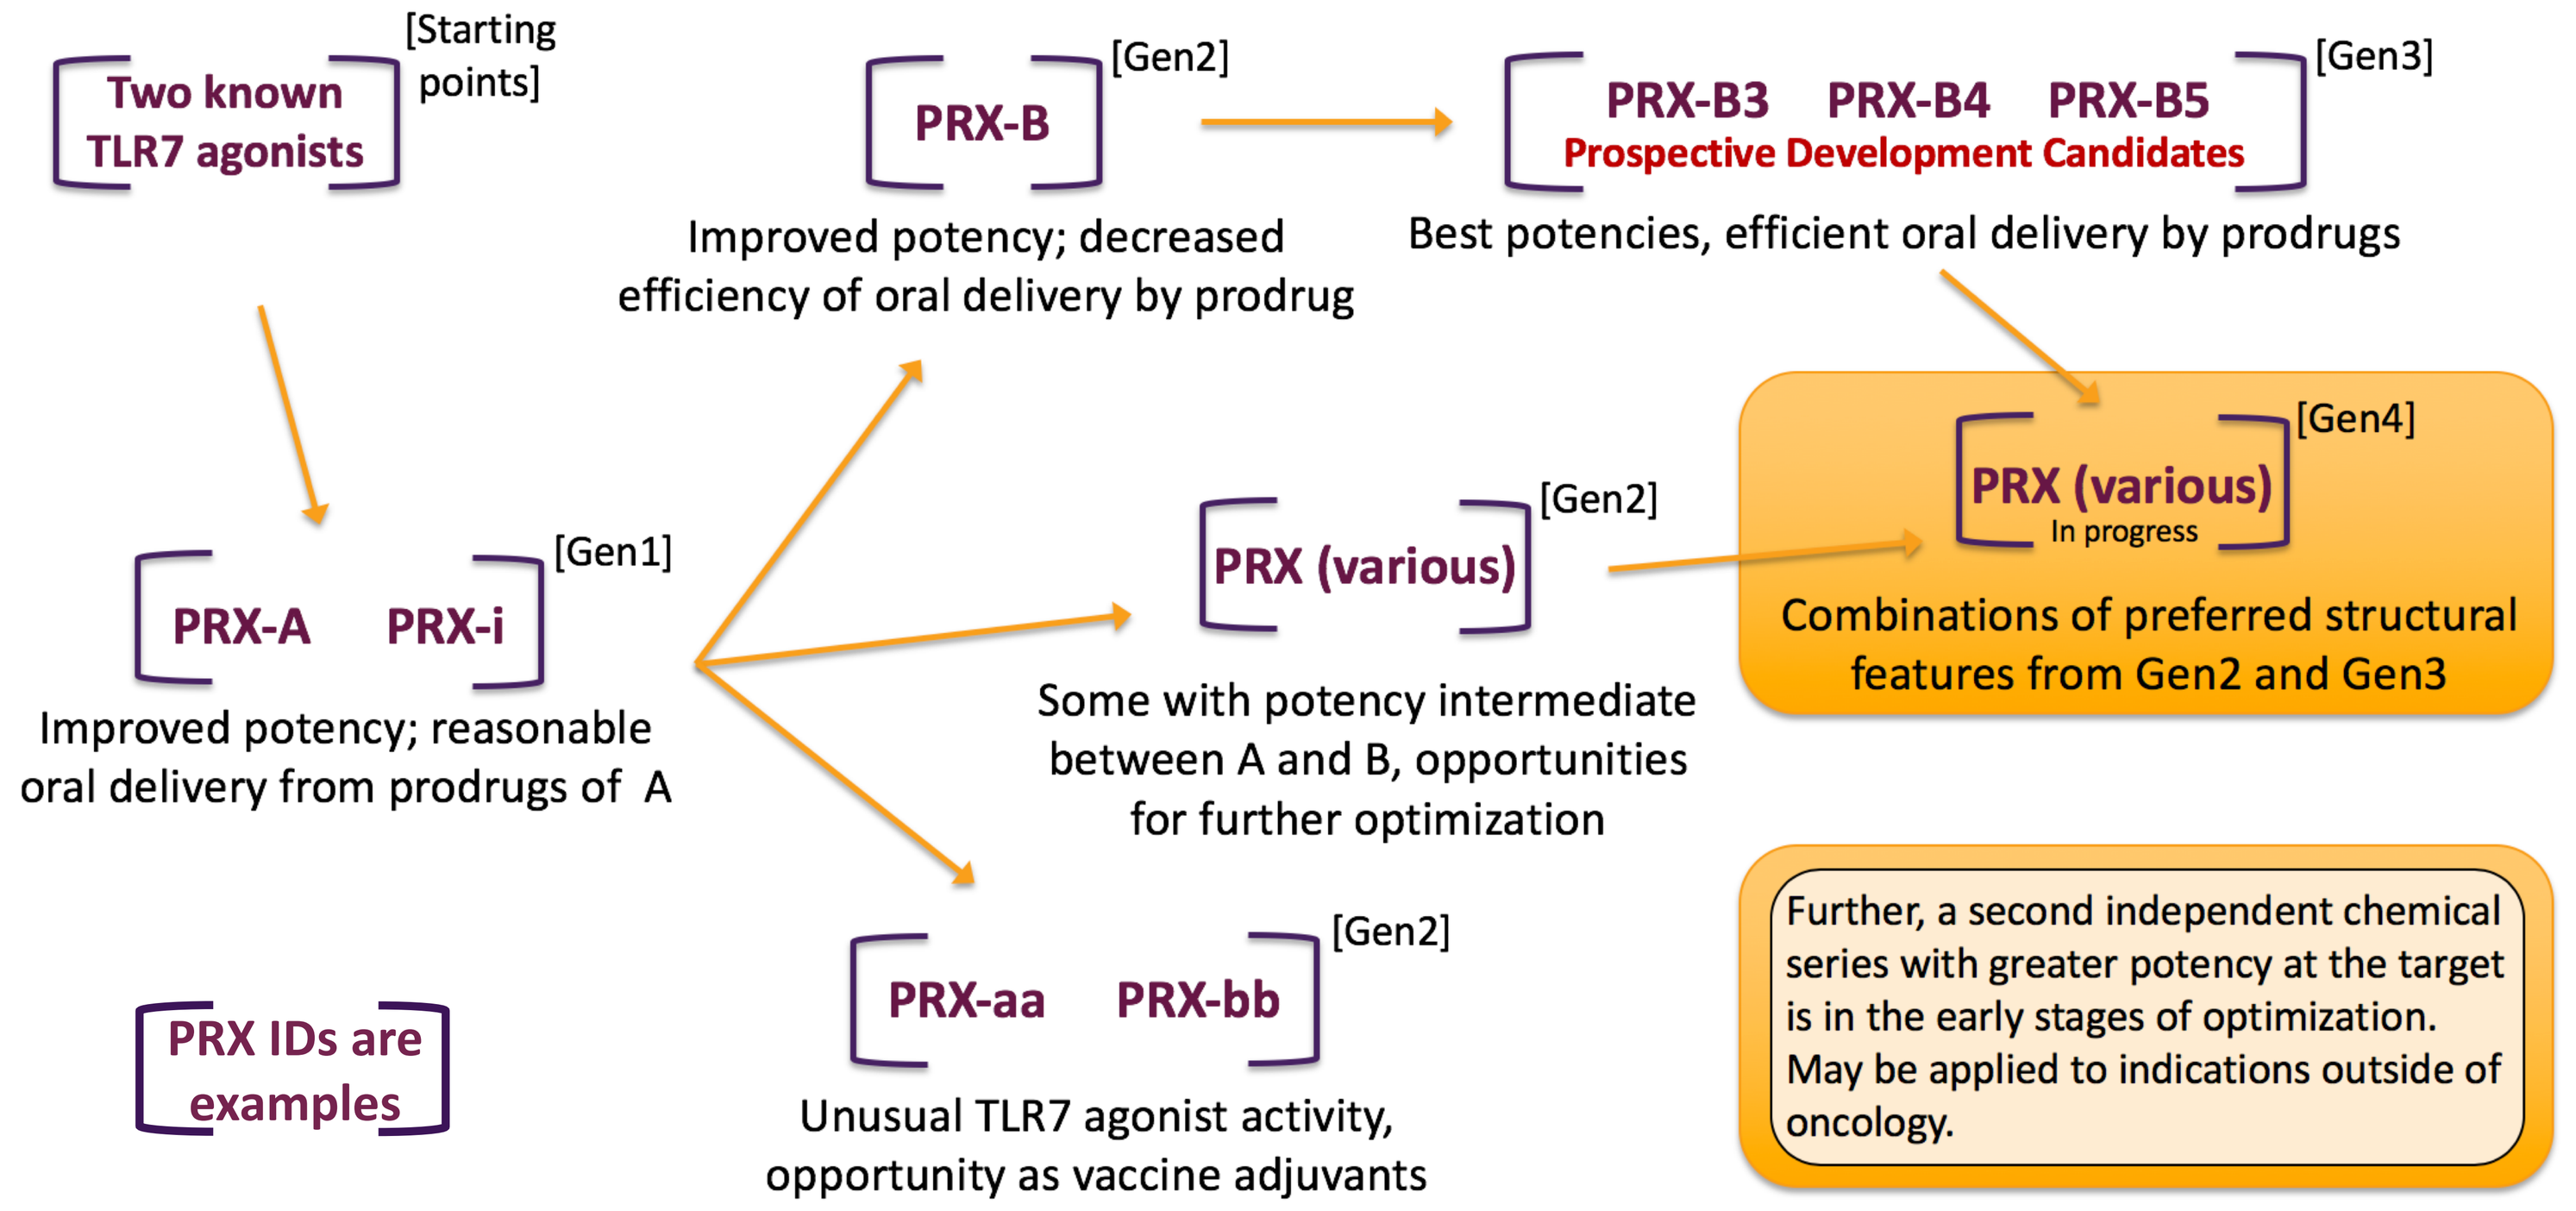
Two known TLR7 agonists
[Starting points]
PRX-A PRX-i
[Gen1]
Improved potency; reasonable oral delivery from prodrugs of A
PRX-B
[Gen2]
Improved potency; decreased efficiency of oral delivery by prodrug
PRX-B3 PRX-B4 PRX-B5
Prospective Development Candidates
[Gen3]
Best potencies, efficient oral delivery by prodrugs
PRX (various)
[Gen2]
Some with potency intermediate between A and B, opportunities for further optimization
PRX (various)
In progress
[Gen4]
Combinations of preferred structural features from Gen2 and Gen3
PRX-aa PRX-bb
[Gen2]
Unusual TLR7 agonist activity, opportunity as vaccine adjuvants
PRX IDs are examples
Further, a second independent chemical series with greater potency at the target is in the early stages of optimization. May be applied to indications outside of oncology.

hPBMC TESTING (selected compounds)

➤ Target potency achieved in 3rd generation TLR7 agonists

ID	Weighted MEC* (arbitrary units)	Count of Donors with MEC: 1	2	4	8	≥ 8	Percent of Donors with MEC: 1	2	4	8	≥ 8
ANA122	12.7				1	2				33%	67%
ANA134	10.1			1		2			33%		67%
A	5.3			8	3	1			67%	25%	8%
B	2.7		10	8				56%	44%		
B2	5.0			2	1				67%	33%	
B3	2.5	2	3	5			20%	30%	50%		
B4	1.9	4	3	3			40%	30%	30%		
B5	2.6		6	4				60%	40%		
C	8.0			1	1	1			33%	33%	33%
D	5.3			4		1			80%		20%
f	6.3			1	2				33%	67%	
g	3.0		3	4				43%	57%		
h	8.0			2	2	2			33%	33%	33%
i	6.3			1	2				33%	67%	
o	5.0			2	1				67%	33%	
q	6.3			1	2				33%	67%	
Starting point	> 16										

Note

* MEC: minimal effective concentration

Note: PRX-B4 induces a greater amount of IFNα production than PRX-B3 at their respective MECs

➤ Target profile for chronic systemic use achieved

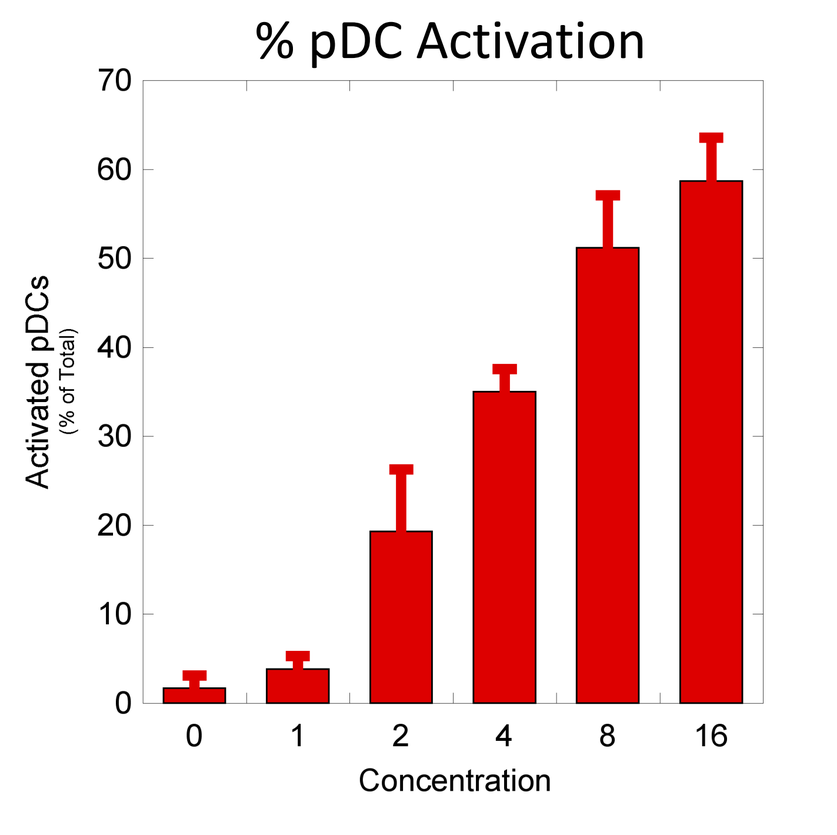
% pDC Activation
Activated pDCs (% of Total)
Concentration

Representative data shown for PRX-B

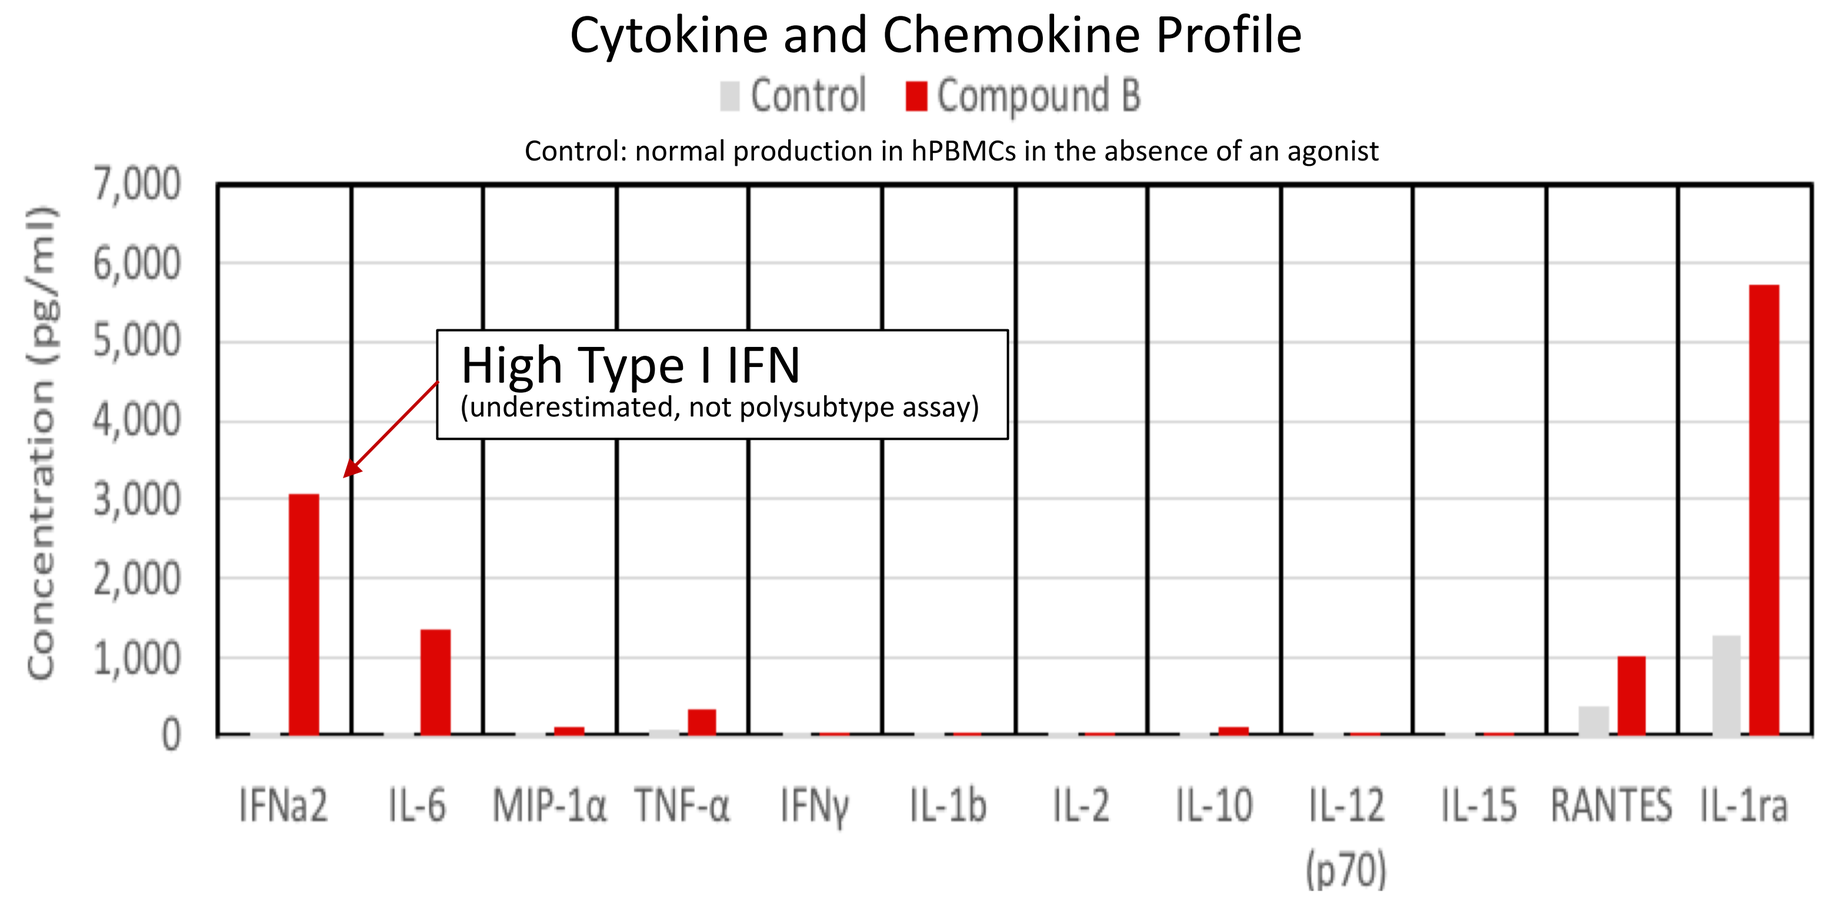
Cytokine and Chemokine Profile
Control
Compound B
Control: normal production in hPBMCs in the absence of an agonist
Concentration (pg/ml)
High Type I IFN
(underestimated, not polysubtype assay)
IFNa2
IL-6
MIP-1α
TNF-α
IFNγ
IL-1b
IL-2
IL-10
IL-12 (p70)
IL-15
RANTES
IL-1ra

PK IN CYNOMOLGUS MONKEYS

➤ Appropriate PK, highly efficient oral delivery of 3rd gen cmpds

- Efficient systemic delivery of active TLR7 agonists was achieved with Primmune's prodrug strategy

Compound	% F
PRX-B	14%
PRX-B3	88%
PRX-B4	99%
ANA122	98%

- PK properties measured in monkeys for Primmune compounds are expected to translate to humans

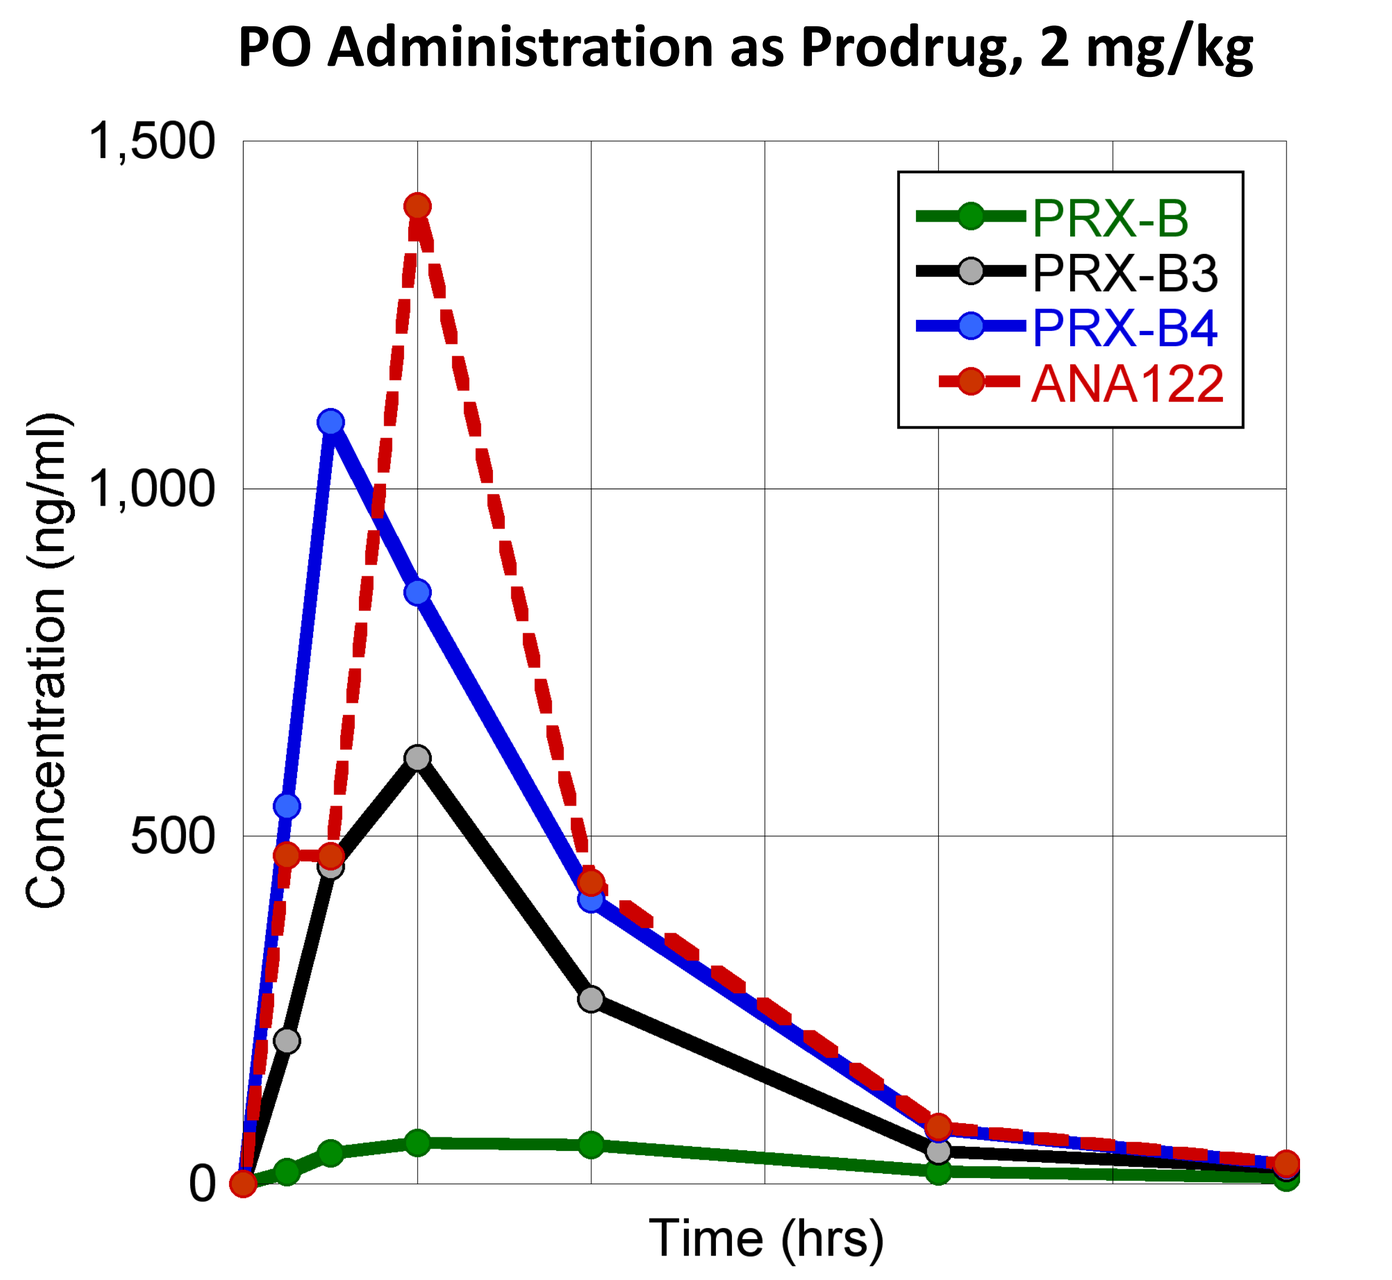
PO Administration as Prodrug, 2 mg/kg
PRX-B
PRX-B3
PRX-B4
ANA122
Concentration (ng/ml)
Time (hrs)

SYNGENEIC RODENT TUMOR MODEL

➤ Activity demonstrated as monotherapy and in combination with PD1 inhibitor in challenging B16F10 model

Dosing Schedule

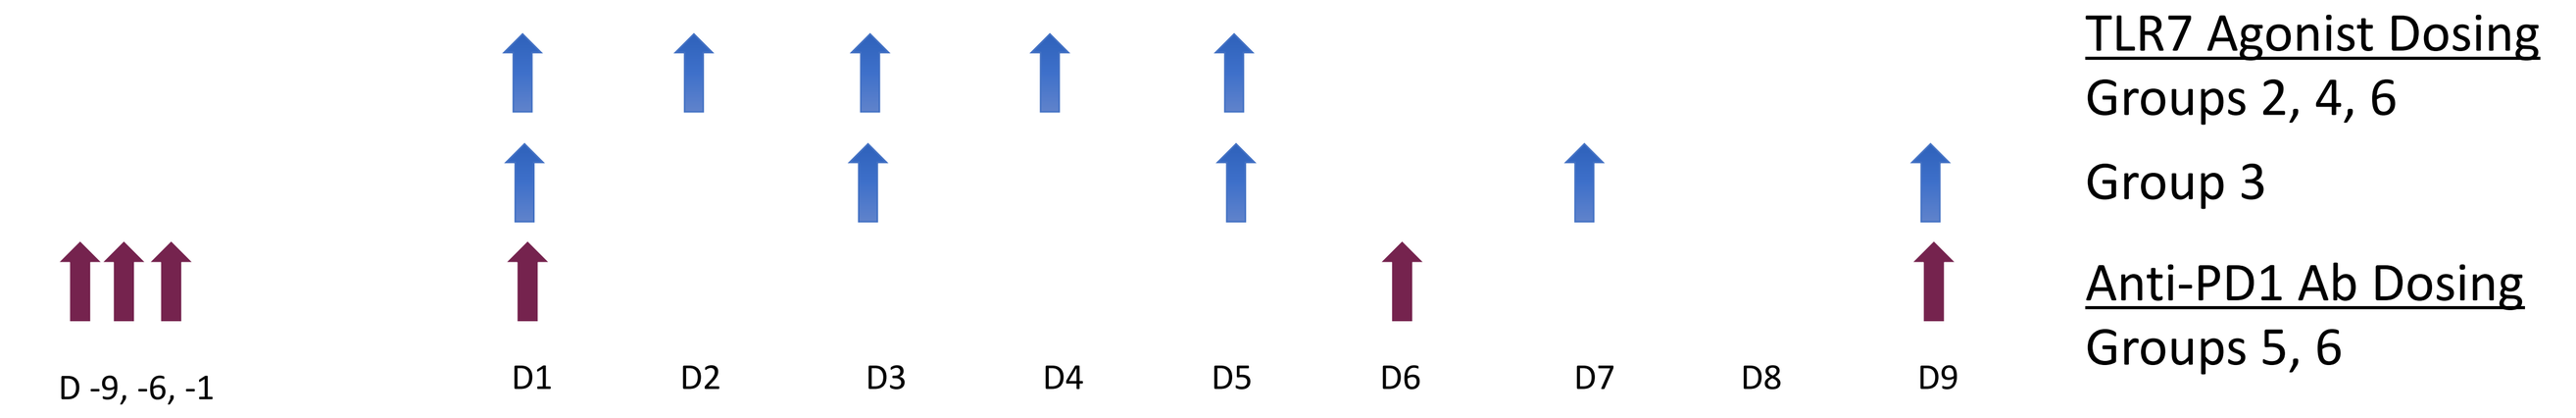
D -9, -6, -1
D1
D2
D3
D4
D5
D6
D7
D8
D9
TLR7 Agonist Dosing
Groups 2, 4, 6
Group 3
Anti-PD1 Ab Dosing
Groups 5, 6

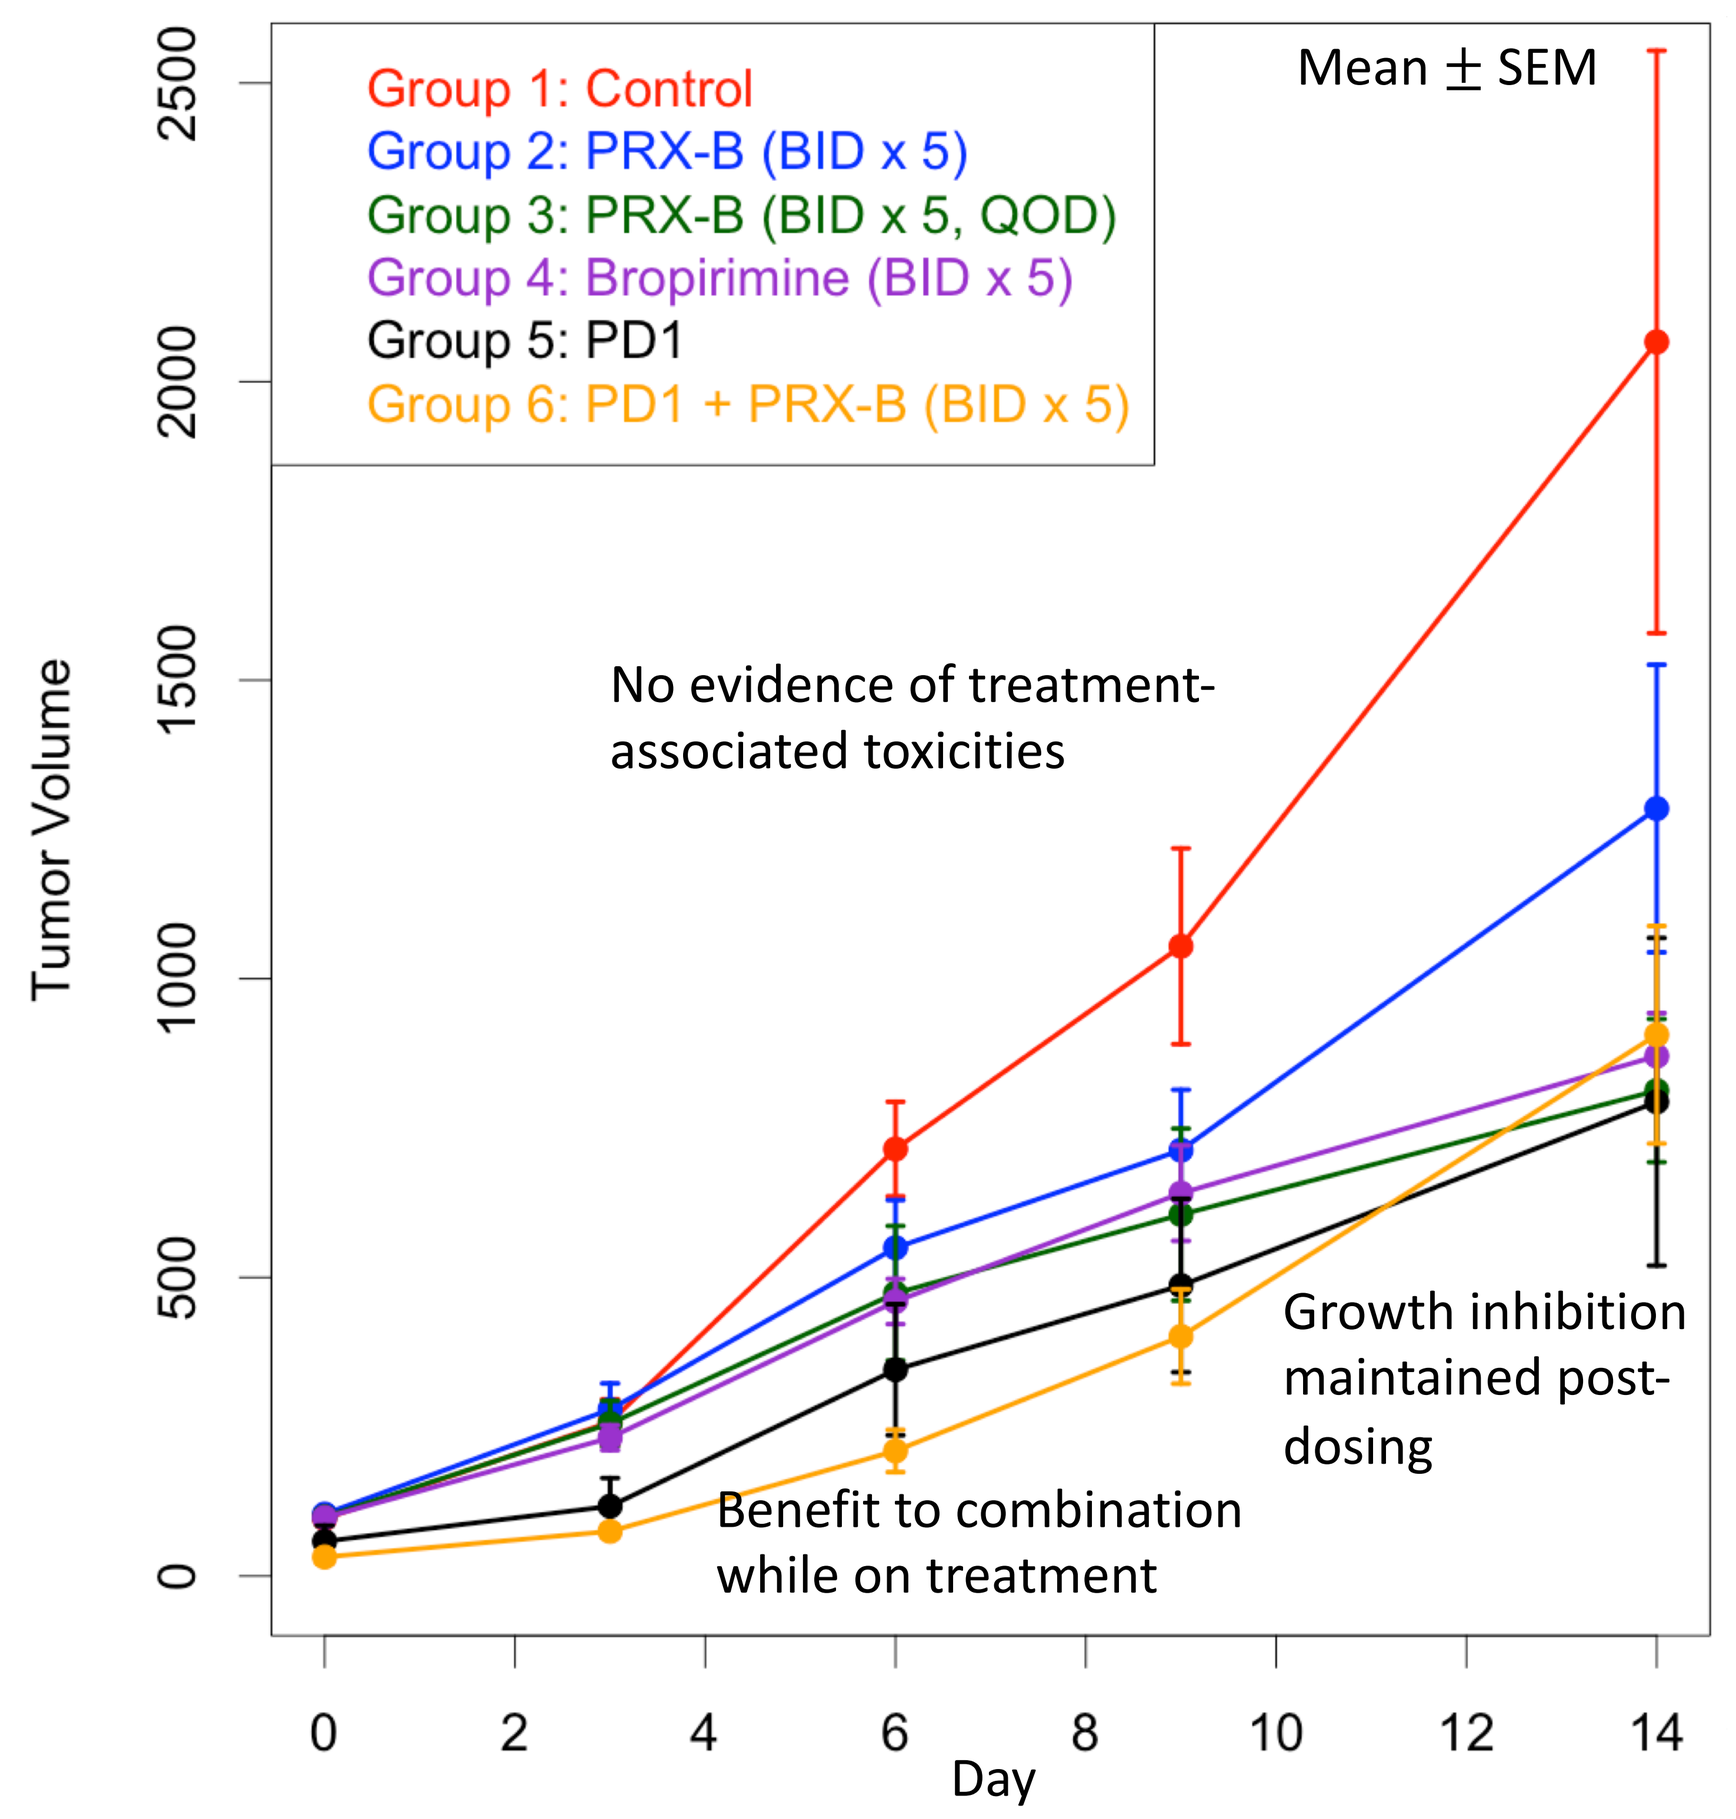
Group 1: Control
Group 2: PRX-B (BID x 5)
Group 3: PRX-B (BID x 5, QOD)
Group 4: Bropirimine (BID x 5)
Group 5: PD1
Group 6: PD1 + PRX-B (BID x 5)
Mean ± SEM
No evidence of treatment-associated toxicities
Growth inhibition maintained post-dosing
Benefit to combination while on treatment
Tumor Volume
Day

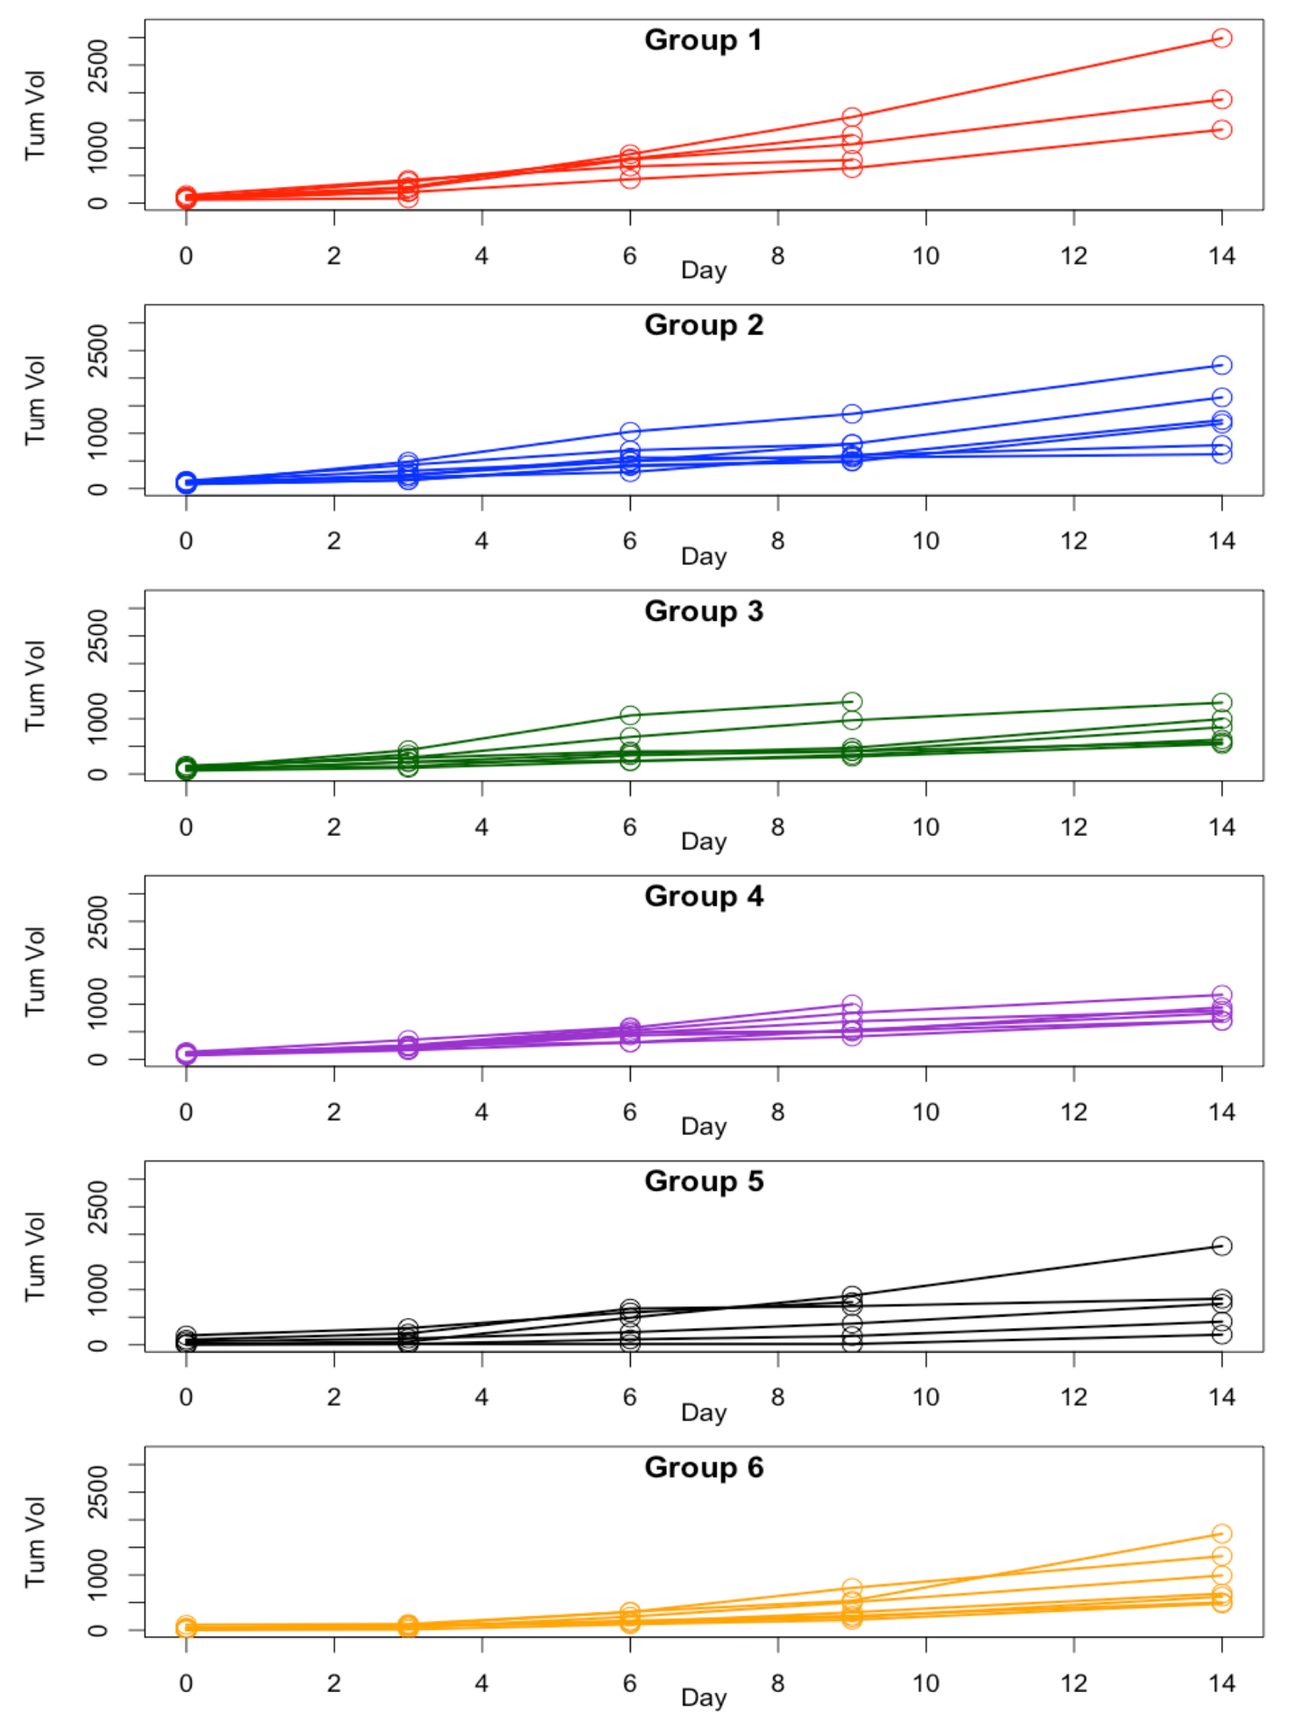
Group 1
Group 2
Group 3
Group 4
Group 5
Group 6
Tum Vol
Day

Methods. 50,000 B16F10 cells were injected subcutaneously into the flank of B57BL/6 mice. For groups 5 and 6, treatment with the murine anti-PD1 antibody RMP1-14 was started 3 days after implantation (D-9 above). Mice were randomized to groups 1-4 when tumors were ~ 100 mm^3 (D0 above, 12 days post implantation). Treatment of groups 2, 3, 4 and 6 with TLR7 agonists PRX-B and bropirimine was initiated on D1. PRX-B, bropirimine and RMP1-14 were administered at 40 mg/kg/dose IV, 100 mg/kg/dose PO and 10 mg/kg/dose IP, respectively.

STATUS

- Studies with the prodrug of **PRX-B**, a second-generation TLR7 agonist used as a benchmark in primate studies, identified features to be optimized in third generation compounds
- **PRX-B3**, **B4** and **B5**, all analogs of **B,** now meet our target profile
- Further characterization of **B3**, **B4** and their prodrugs, including testing in cynomolgus monkeys and rodent tumor models, is currently in progress

➤ We are on track to select a preferred candidate upon completion of these experiments

ACKNOWLEDGEMENTS

Much of the laboratory experimentation reported herein was conducted at Aptuit, an Evotec Company. We would like to acknowledge the critical contributions of core project team members Filippo Andreetta, Elisa Bianchini, Viola Bigatto, Marisa Dal Lago, Sadaf Kasraie and John Kamins in experimental design and interpretation, project management and alliance management.